Human Research Investigation on the Effects of Cigarette Smoking, E-Cigarette Vaping, and Smoking Cessation on Vascular Cell Biology

C Poussin[1], J Hoeng[1], S Ansari[1], E Guedj[1], L Felber Medlin[1], NV Ivanov[1], P Picavet[1], MC Peitsch[1]

TM Curran[2], LA Baglia[2], RA Berg[2], N Blumberg[2], SL Spinelli[2]

[1]*Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland (part of Philip Morris International group of companies)*

[2]*Department of Pathology and Laboratory Medicine University of Rochester School of Medicine and Dentistry, Rochester, NY.*

Abstract

Inhaled toxicants present in tobacco smoke accelerate and exacerbate inflammation and oxidative stress. Over time, these processes promote metabolic disorders, chronic pulmonary obstructive disease, and cardiovascular disease (CVD) development and progression (1). The goal of this clinical research study is to investigate the biological networks and signaling profiles that drive vascular dysregulation that are (i) perturbed by smoking, (ii) reversible with respect to smoking cessation and (iii) are largely unknown for long term e-cigarette usage. These results will impact the development of new approaches and testing of reduced-risk products to mitigate smoking-related health risks. Circulating blood cells and biofluids were obtained from smokers, former smokers and electronic cigarette users and compared with those obtained from never smokers (a total of 160 subjects). The investigations of smoke and e-vapor exposure onto peripheral human blood cells involve transcriptomics, lipidomics, and proteomics, as well as measures of inflammation (*e.g.,* cytokine production), oxidative stress (*e.g.,* DNA methylation, monoamine oxidase activity), and clinical endpoints to identify critical biomarkers and specific molecular signatures. Data are derived from five white blood cell subtypes (*e.g.* neutrophils, monocytes, CD4 and CD8 T cells, B cells), red blood cells, platelets, secreted vesicles, and biofluids (serum, plasma and urine). Our initial findings confirm that smokers exhibit increased levels of cardiovascular markers (*e.g.,* homocysteine, C-reactive protein), and an increase in platelet aggregation. These markers are reduced in former smokers who quit smoking for a period of at least two years. Molecular profiling investigations are ongoing with the aim of discovering predictive models that will be leveraged in future clinical research.

Material & Methods (in brief)

Human volunteers were recruited and consented in accordance with the Helsinki declaration under a protocol approved by the University of Rochester Institutional Review Board. Up to 100 mL of venous blood was drawn from the forearm of the consenting study subject via venipuncture and urine was collected. All information and samples were assigned a unique identifier for each study subject to protect the anonymity of the subject. Whole blood was used for complete blood cell count analysis, serum, clinical endpoints, DNA methylation and miRNA studies. Whole blood was also centrifuged to obtain blood component fractions, such as platelet rich plasma, platelet poor plasma, cell populations and extracellular vesicles. An overview of the project is provided in the schematic below.

Subject
- Demographics
- Questionnaire

→ **Whole blood**

Urine → Biomarkers of exposure

Clinical analytes
- Complete blood counts
- Exposure biomarkers
- Biomarkers of potential harm (health risk)

Platelet biology research
- **Platelet-rich plasma**
 - Platelet aggregation (ADP/ATP)
 - Extracellular vesicles (NanoSight)
 - CVD and inflammation markers (Luminex, immunoassays)
- **Platelet-monocyte complexes, PMC (FACS)**

Biobanked samples
- **Serum and plasma**
- **Whole blood in PaxGene tube**
- **Leukocyte sub-population isolation**
 - Monocyte CD14
 - Neutrophils CD66b
 - Lymphocytes B CD19
 - Lymphocytes T CD4 and CD8
- **RBCs and platelets**

Analysis

University of Rochester Medical Center (URMC) Clinical Laboratories

URMC Research Laboratories

Systems Toxicology Research (PMI)
- Transcriptomics. miRNA
- DNA methylome
- Proteomics
- Lipidomics
- Luminex /immunoassays
- Functional assays

Results

Subject Recruitment Criteria and Demographics

Table 1: Inclusion and exclusion criteria for the study.

Inclusion criteria				
Age	21 - 45 (extended to 55) years old			
Weight	At least 110 lbs			
Group	Never Smokers (NS)	Cigarette Smoker (S)	Former Smokers (FS)	E-cigarette User (EC)
	- Never smoked - No other product use	- Currently smoking ≥ 10 cigarettes/day for ≥ 3 years - No other product use	- Previously smoked ≥ 10 cigarettes/day for ≥ 3 years - Quit smoking for ≥ 2 years - No other product use	- Used E-cigs ≥ 6 months - Previously smoked ≥ 10 cigarettes/day for ≥ 3 years - Quit smoking for ≥ 6 months - No other product use
Targeted N	40	40	40	40
Exclusion criteria				
General	Pregnancy			
Drugs	- Aspirin, ibuprofen or a steroid medication within the last 10 days - Any medication in the last 30 days			
Health status	- Acute illness (e.g. viral infection) - Pathologies which interfere with the scope of the study (listed in the study protocol)			

Table 2: Demographics by study group.

Variables and statistics	S	FS	NS	EC
Recruitment, %	100	45	100	17
Age, years (mean [SD])	36.8 [7.4]	34.3 [6.6]	29.2 [6.7]	28.3 [5.1]
Sex, %				
- Female	52	67	70	14
- Male	48	33	30	86
BMI, kg/m^2 (mean [SD])	28.3 [5.4]	30.0 [5.2]	24.8 [3.7]	25.5 [5.7]
Waist circumference, cm (mean[SD])	89.2 [13.4]	93.6 [16.5]	78.8 [10.8]	87.8 [11.3]
Race, %				
- Asian	0	6	9	0
- Black or African American	27	6	7	14
- White	68	89	78	71
- Other	5	0	7	14

Table 3. Smoking history for S and FS.

Variables and statistics	S	FS
Tobacco smoking history		
- Pack years (mean [SD])	14.5 [9.8]	5.1 [2.8]
- Daily cigarette consumption (mean [SD])	15.6 [5.3]	13.3 [3.3]
Quitting, years (mean [SD])	NA	7 [6.9]

Biomarkers of Exposure

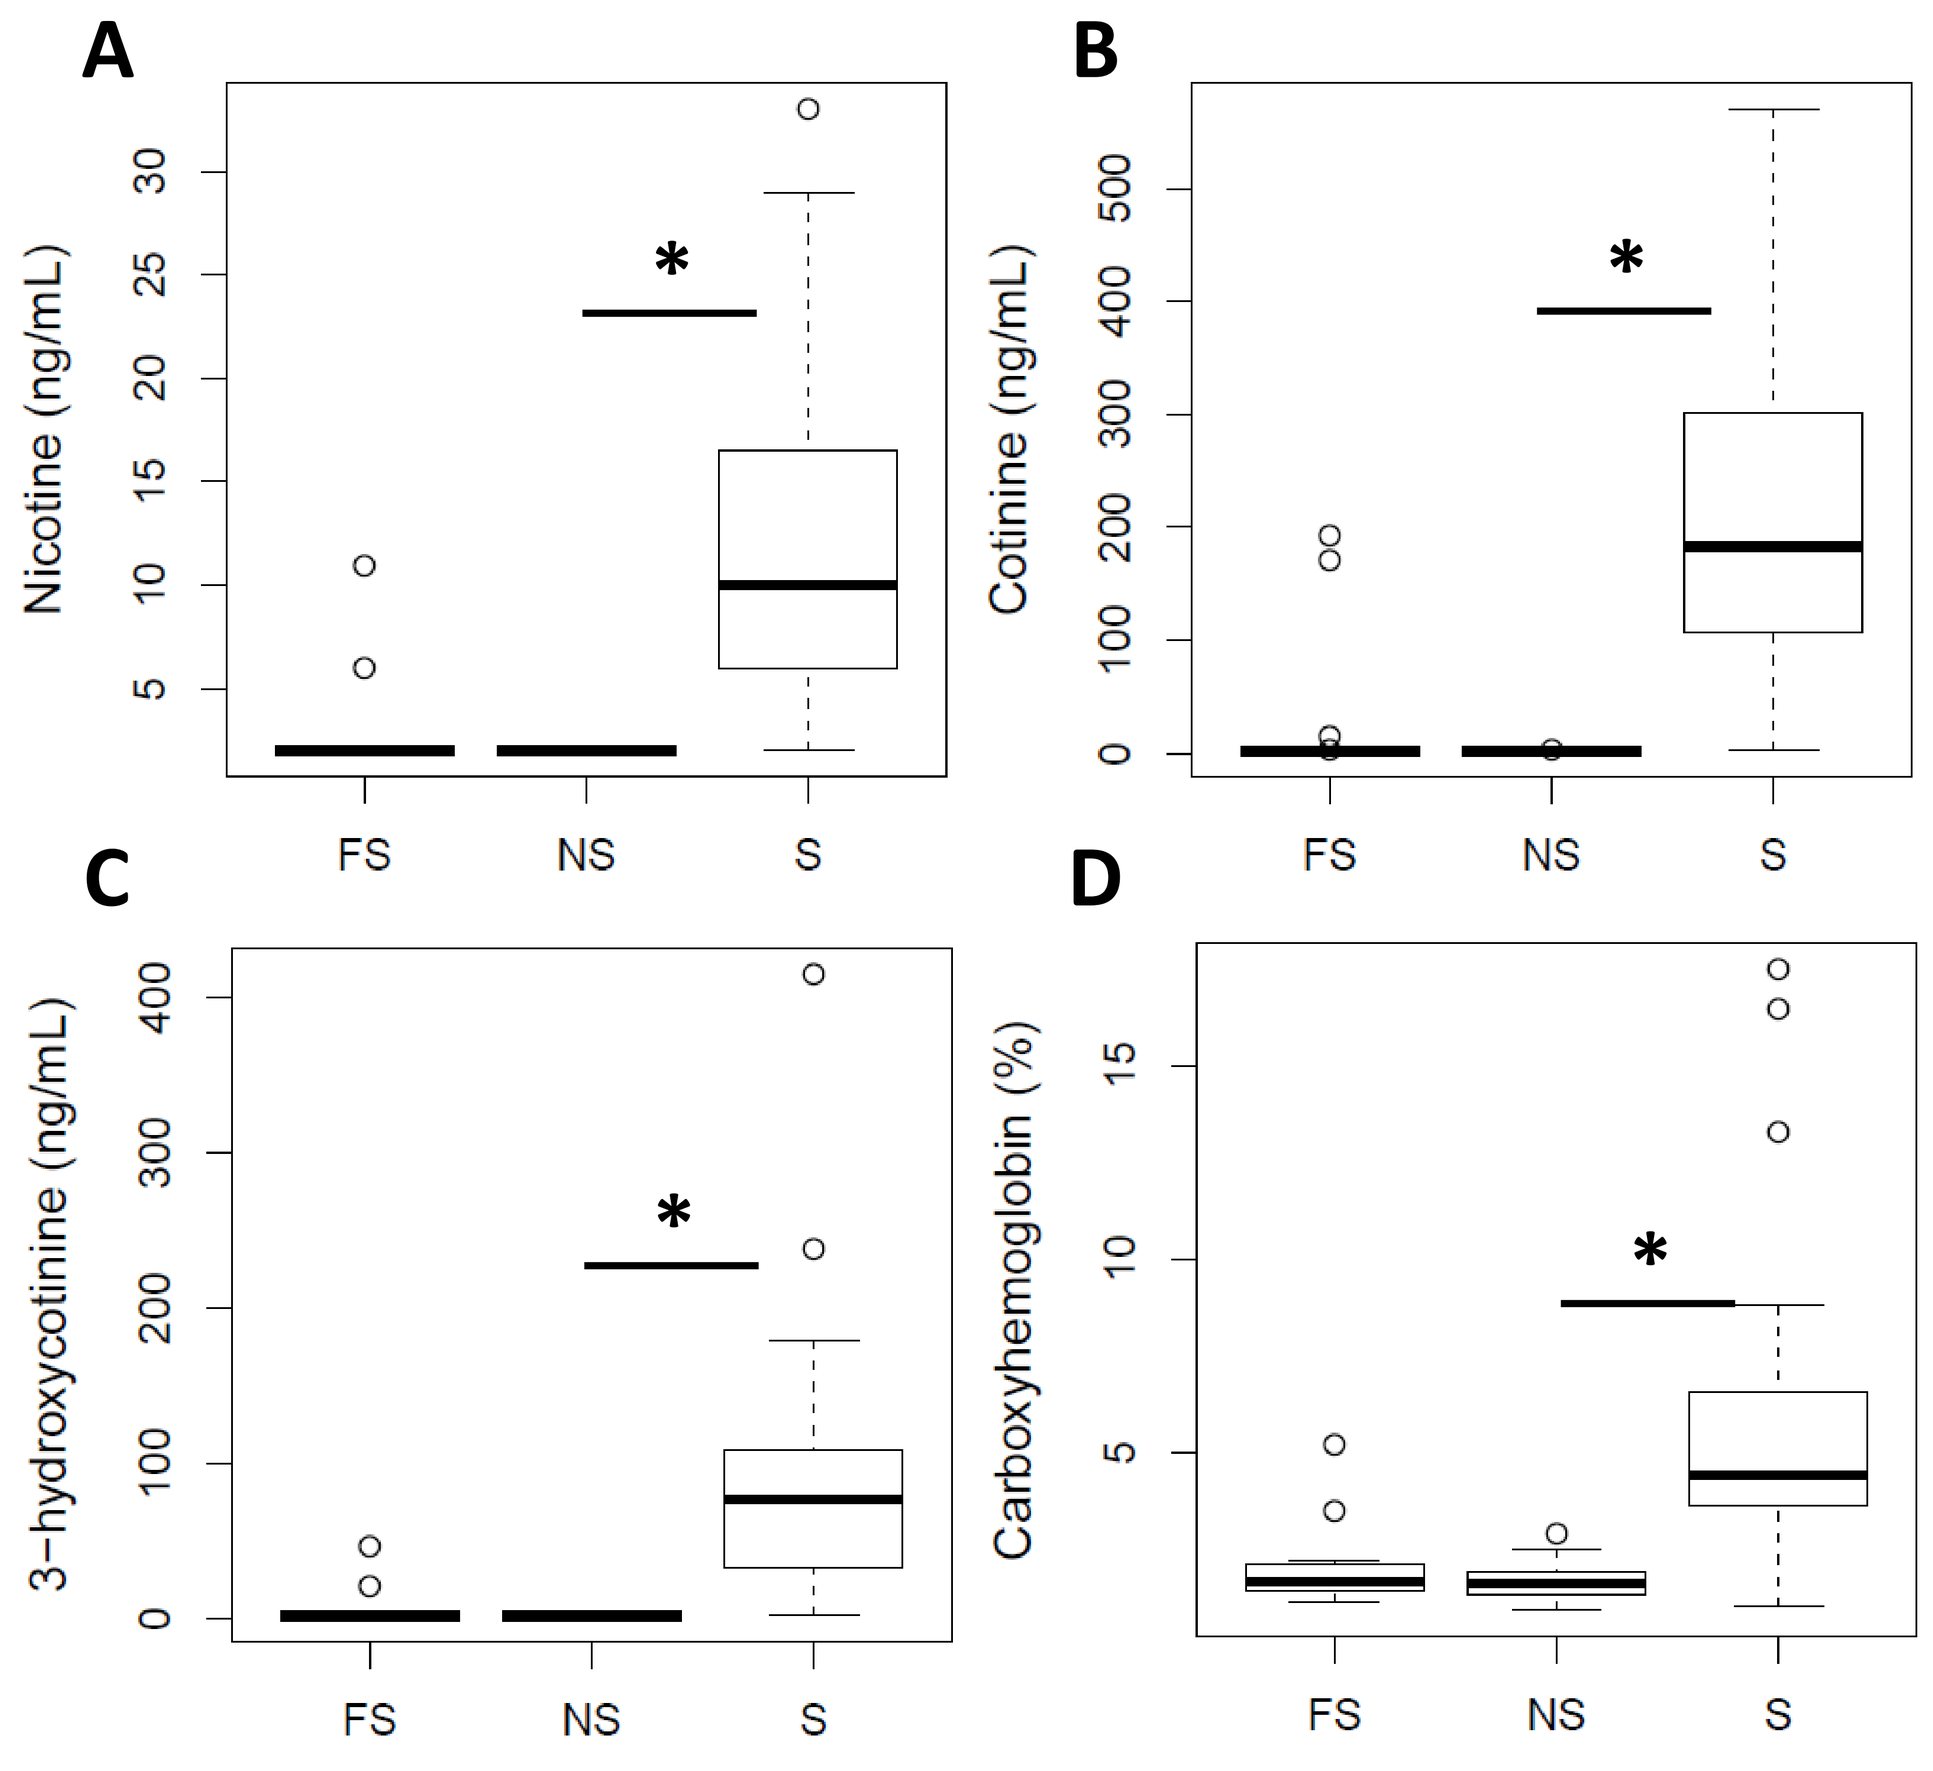
A
Nicotine (ng/mL)
B
Cotinine (ng/mL)
C
3−hydroxycotinine (ng/mL)
D
Carboxyhemoglobin (%)
FS
NS
S
*

Figure 1. The concentration of (A) nicotine and nicotine metabolites, such as (B) cotinine and 3-hydroxycotinine, were measured in serum/plasma of s, NS and FS by the URMC Clinical Laboratories. As expected, the concentrations of all analytes are significantly higher in S vs. NS or FS (unpaired t-test, two-tailed; *p≤0.05).

Biomarkers of Potential Harm

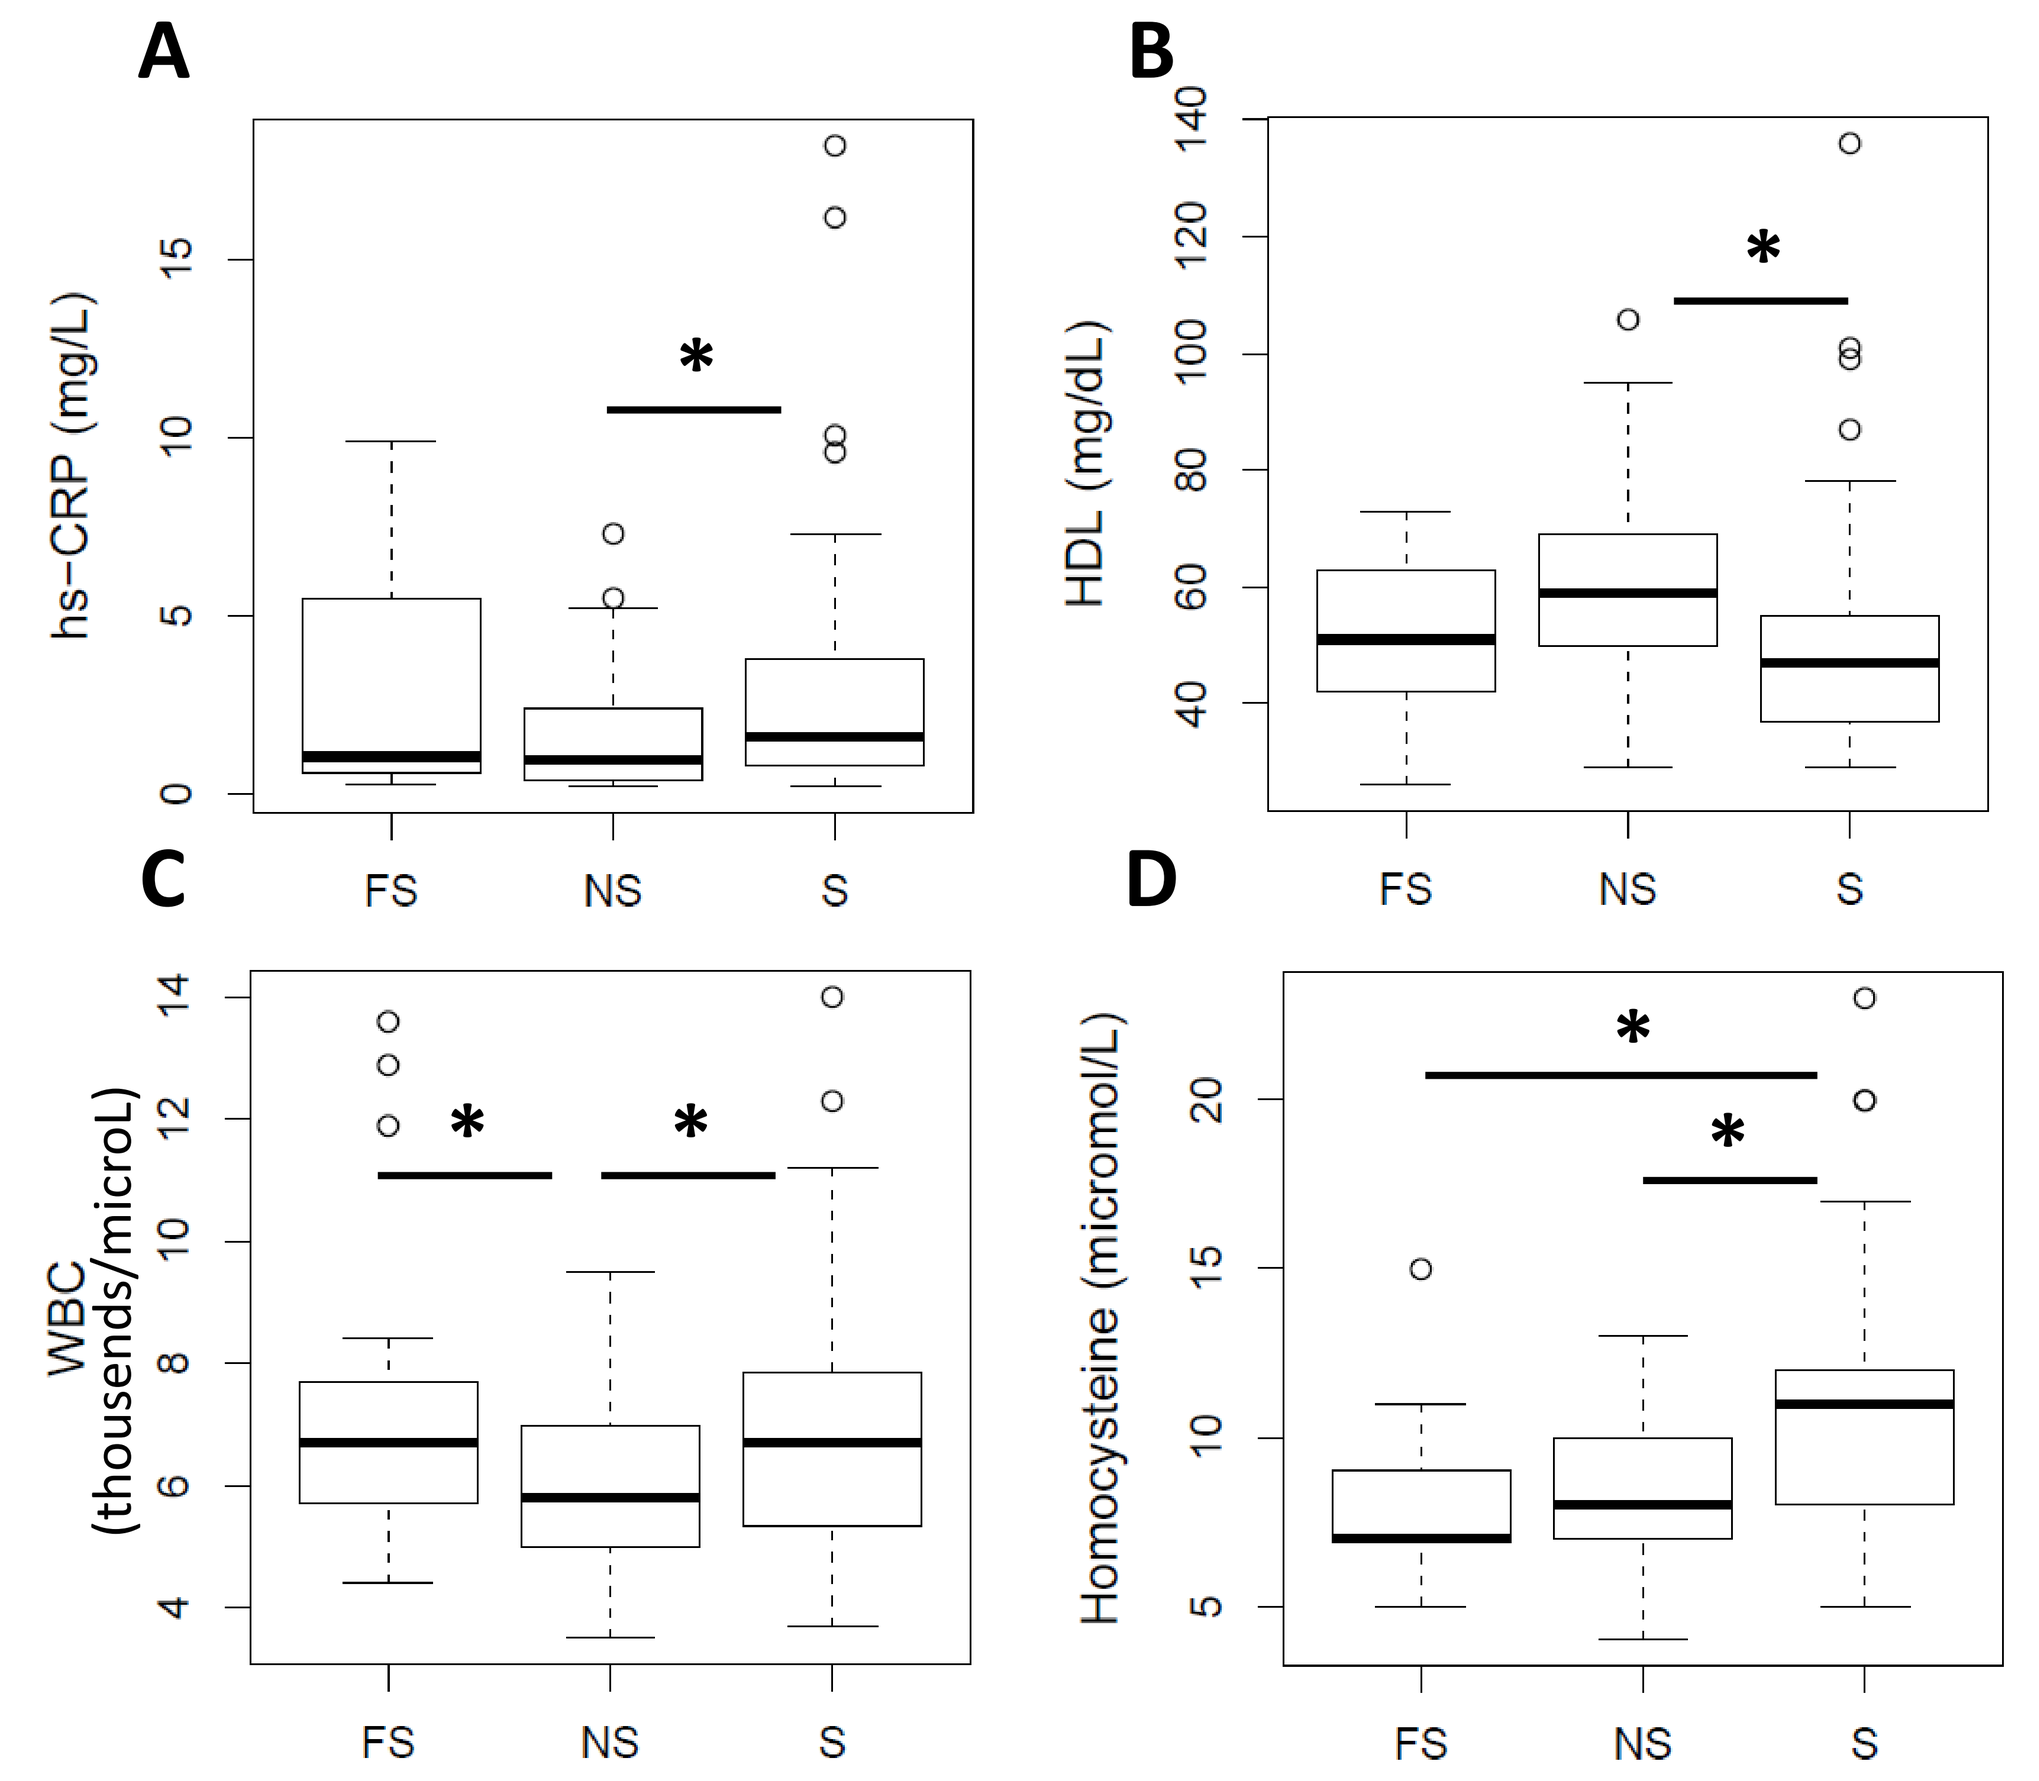
A
hs−CRP (mg/L)
B
HDL (mg/dL)
C
WBC (thousends/microL)
D
Homocysteine (micromol/L)
FS
NS
S
*

Figure 2. The concentrations of a panel of biomarkers of potential harm were determined in subjects' plasma, serum, or whole blood by the URMC Clinical Laboratories. Examples of analytes measured are provided here with (A) high sensitivity C-reactive protein (hs-CRP), (B) high density lipoprotein (HDL), (C) white blood cell (WBC) count and (D) homocysteine showing differential levels between S, NS and FS groups. (unpaired t-test, two-tailed; *p≤0.05).

Platelet-Monocyte Interactions

A – PMC gating by FACS | **B – PMC quantification**

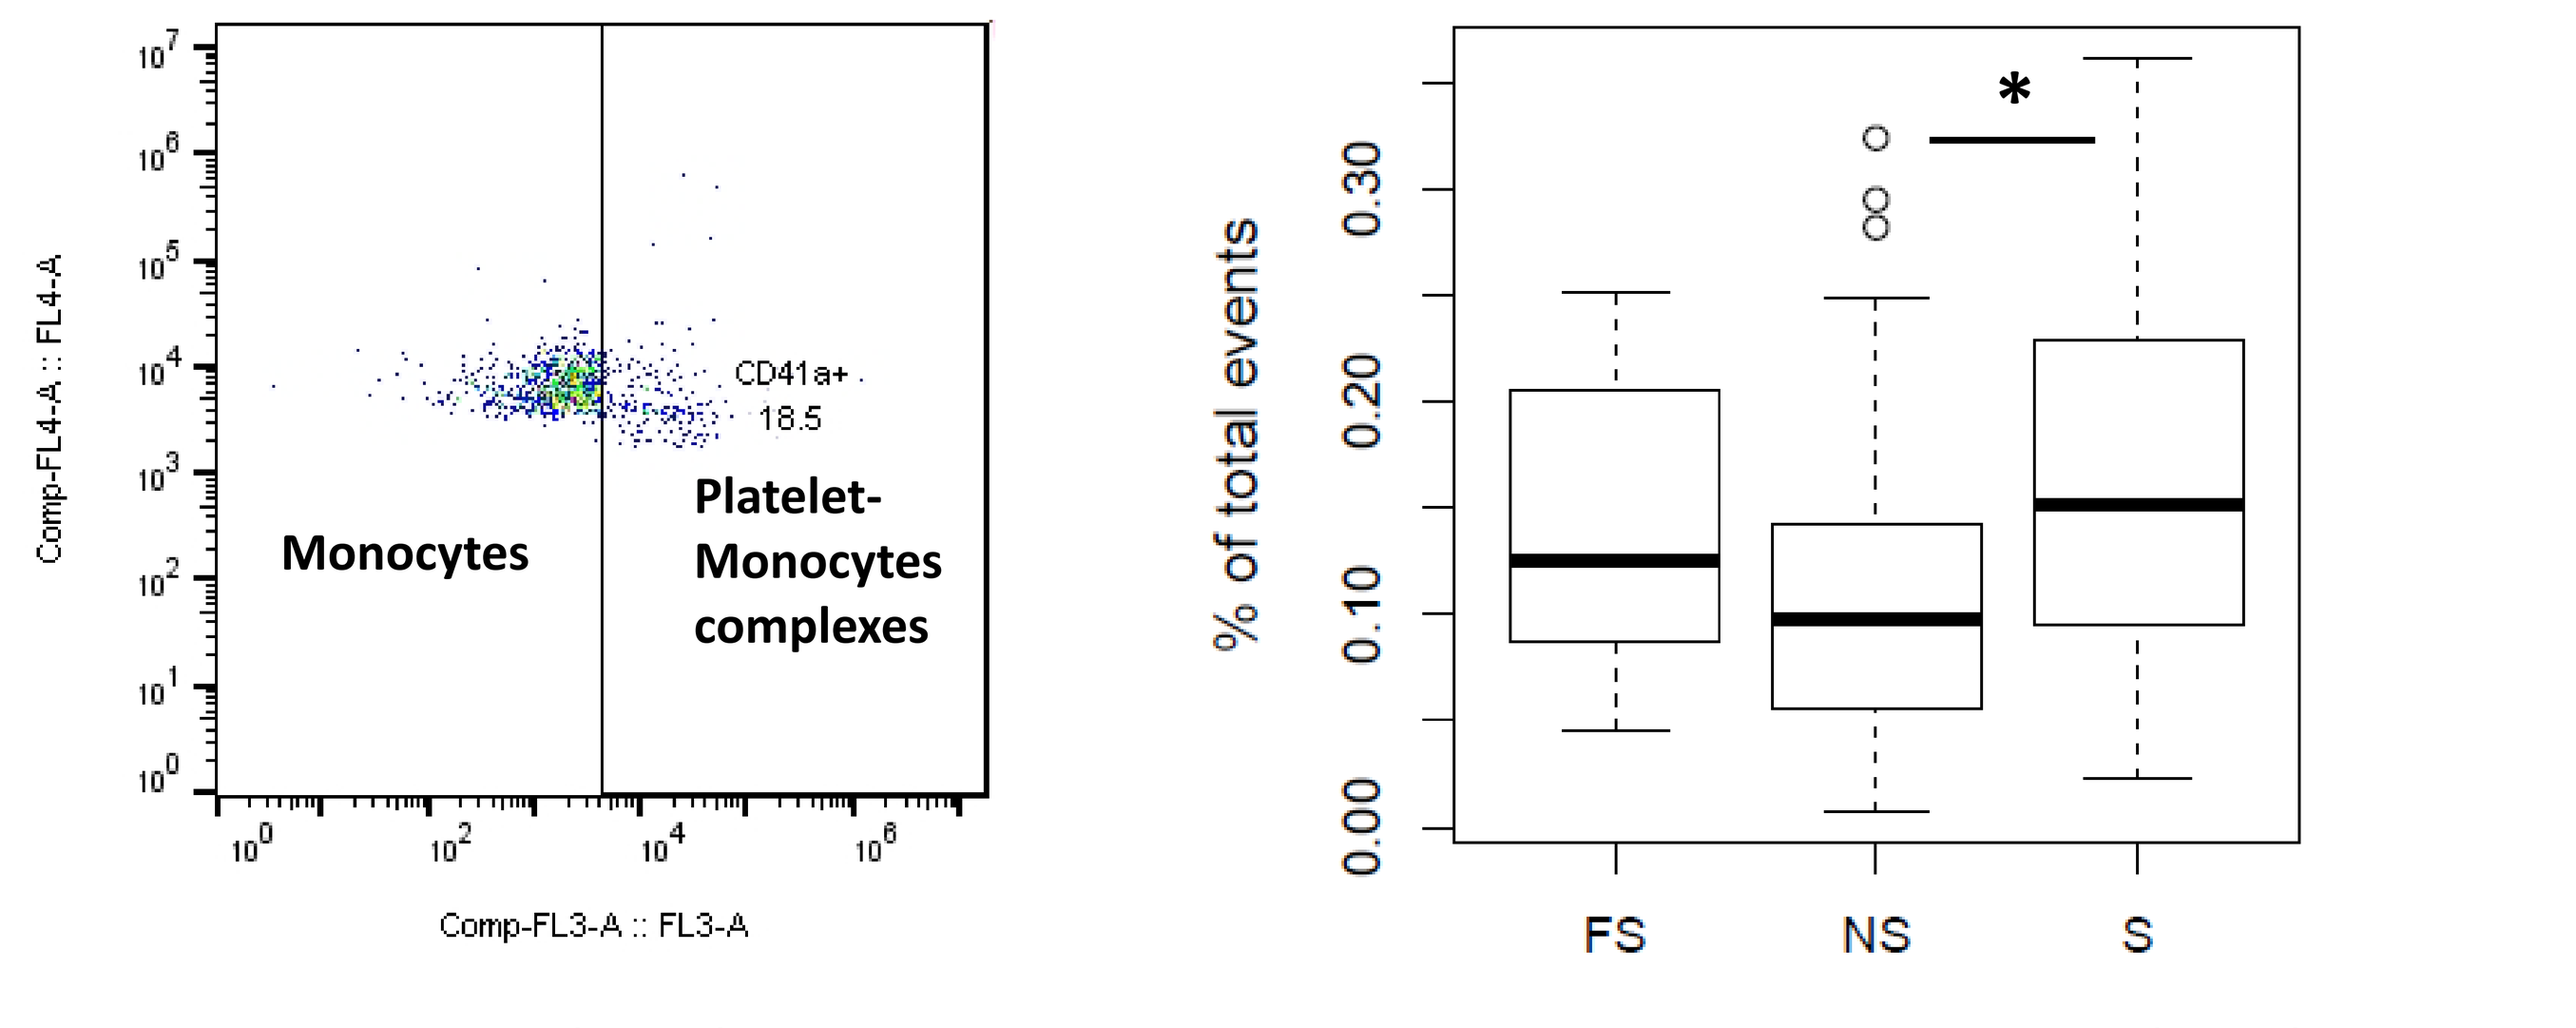
Comp-FL4-A :: FL4-A
Comp-FL3-A :: FL3-A
CD41a+
18.5
Monocytes
Platelet-Monocytes complexes
% of total events
FS
NS
S
*

Figure 3. Flow cytometry (FACS) was used to measure platelet monocyte complexes (PMC), a marker of increased levels of vascular inflammation. Three conjugated antibodies (CD45, white blood cells; CD14, monocytes; CD41a, platelets) were used to stain fixed whole blood (RBC's were lysed after fixing). After eliminating possible double-events, samples were gated to exclude CD45 negative cells. Using the CD45+ population, the gate was determined for CD45+/CD14+ (monocytes). (A) PMCs were identified by gating on the CD45+/CD14+/CD41+ population. (B) PMCs are significantly higher in cigarette smokers (CS) vs never (NS), as determined by unpaired t-test, two-tailed. *p≤0.05.

Summary

The recruitment is ~80% complete, with more than 10,000 samples collected to date. Samples that are not analyzed immediately are biobanked at -80ºC, and all samples and data are managed using a customized research data integration and analytical database. Initial results indicated expected changes (2,3,4) for biomarkers of exposure, showing an increase of the concentrations of nicotine and its metabolites, and carboxyhemoglobin in S compared with NS and FS. These measures provide quantitative estimates of the uptake of selective smoke compounds by the population of S compared with NS and FS recruited in the study (2). Changes in levels of biomarkers of potential harm from smoking, such as hs-CRP, HDL, WBC and homocysteine, may be associated with increased health risk (2). An augmentation of hs-CRP concentration, WBC and platelet-monocyte interactions in blood of S compared with NS and FS reflects an increase of systemic inflammation in S (2,5,6). Further analysis considering demographic and smoking history covariates complemented with large-scale "omics" data will enable to identify new mechanisms and markers of vascular dysregulation associated with smoking, and understand the impact of smoking cessation on them.

References & Acknowlegements

1. Rigotti NA et al. (2013), Eur Heart J, 34 (42):3259-67
2. Saxena K et al. (2017), Biomarkers, 22 (5):403-12
3. Ludicke F et al. (2016), Nicotine Tob Res, 18 (7):1606-13
4. Haziza C et al. (2017), Data Brief, 10:283-93
5. Harding SA et al. (2004), Circulation, 109 (16):1926-9
6. Passacquale G et al. (2011), PloS One, 6 (10):e25595

We would like to thank Pauline Betsch, Christophe Maier and Cedric Montandon for project and financial management of the study at PMI as well as Jeanne Holden-Wiltse and her team for the processing, storage and management of data in a database at the University of Rochester.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

EAS, Lisbon, Portugal

5-8 May 2018

Competing Financial Interest
The research described in this poster was funded by Philip Morris International