Organotypic tissue cultures as models to study the impact of cigarette smoke on human upper and lower respiratory tract

413

M. Talikka[1], R. Kostadinova[1], Y. Xiang[1], D. Kuehn[1], S. Majeed[1], C. Mathis[1], S. Frenzel[1], A. Sewer[1], N. Ivanov[1], F. Martin[1], A.W. Hayes[2], J. Hoeng[1], and M.C. Peitsch[1]
[1]Philip Morris International R&D, Neuchâtel, Switzerland
[2] Spherix Consulting, Rockville, MD, USA

Introduction

Cigarette smoke (CS) is etiologically linked to fatal respiratory diseases. To enable the development of modified risk tobacco products (MRTPs) to reduce the risk of smoking related diseases, there is a need to understand the mechanisms whereby CS causes disease. As an alternative to animal experimentation, the principles of 3 R's necessitate the establishment of reliable in vitro models, which may also provide a better predictability of exposure response, when derived from human cells. Among the in vitro models, the three-dimensional organotypic pseudo-stratified culture systems better mimic the morphological, physiological, and molecular attributes of human respiratory tract than primary or immortalized cells cultured as a submerged monolayer.

The bronchus and lung parenchyma are the primary sites that manifest smoking related respiratory diseases, however, nasal epithelium has been proposed as a surrogate tissue to study smoking effects on the respiratory tract (1,2).

Previously, we showed that the transcriptomic responses were very similar in CS-exposed bronchial and nasal organotypic tissue cultures when analyzed in the context of a network model representing xenobiotic metabolism. In addition, the perturbation of the xenobiotic metabolism network was similar in organotypic in vitro cultures and nasal and bronchial brushings from smokers (3).

Here, we further characterize the response of nasal and bronchial organotypic cultures to whole mainstream CS.

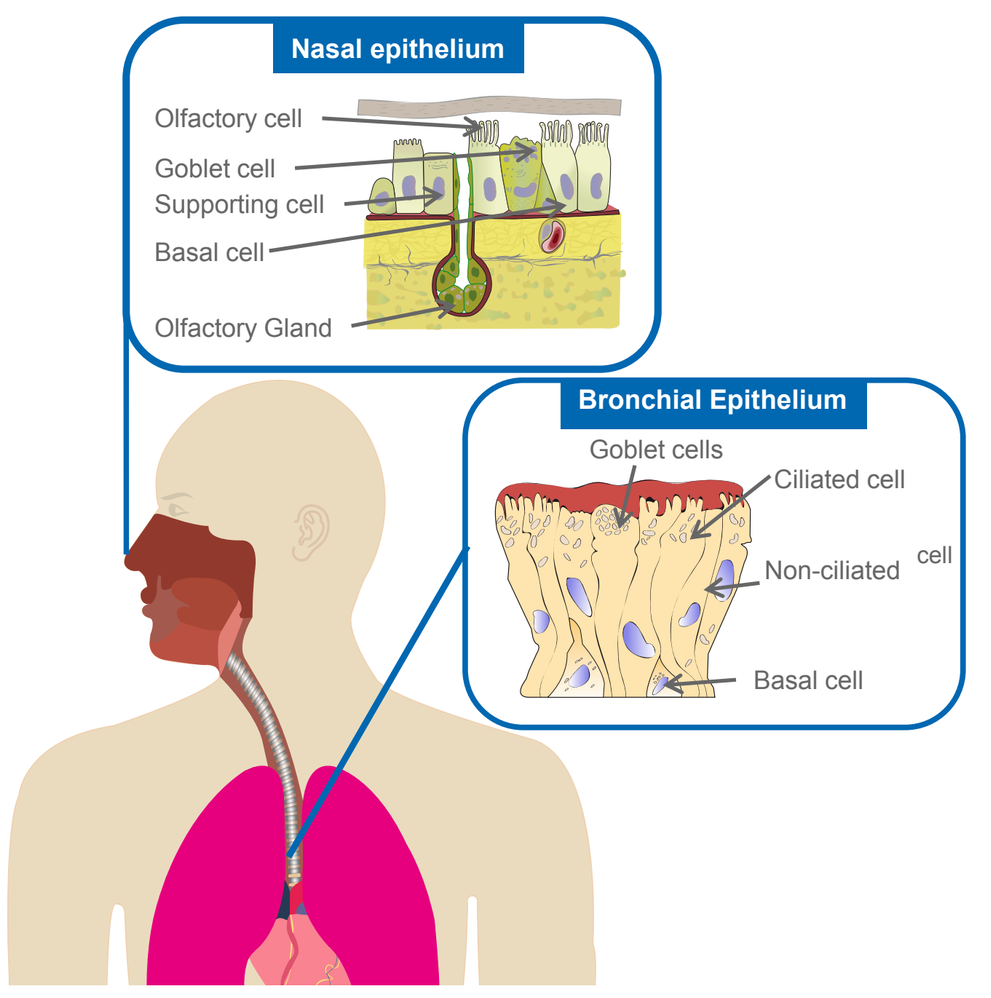
Nasal epithelium
Olfactory cell
Goblet cell
Supporting cell
Basal cell
Olfactory Gland
Bronchial Epithelium
Goblet cells
Ciliated cell
Non-ciliated cell
Basal cell

Figure 1. Organotypic tissue cultures were derived from human primary nasal and bronchial epithelial cells

Exposure Design and Quality Controls

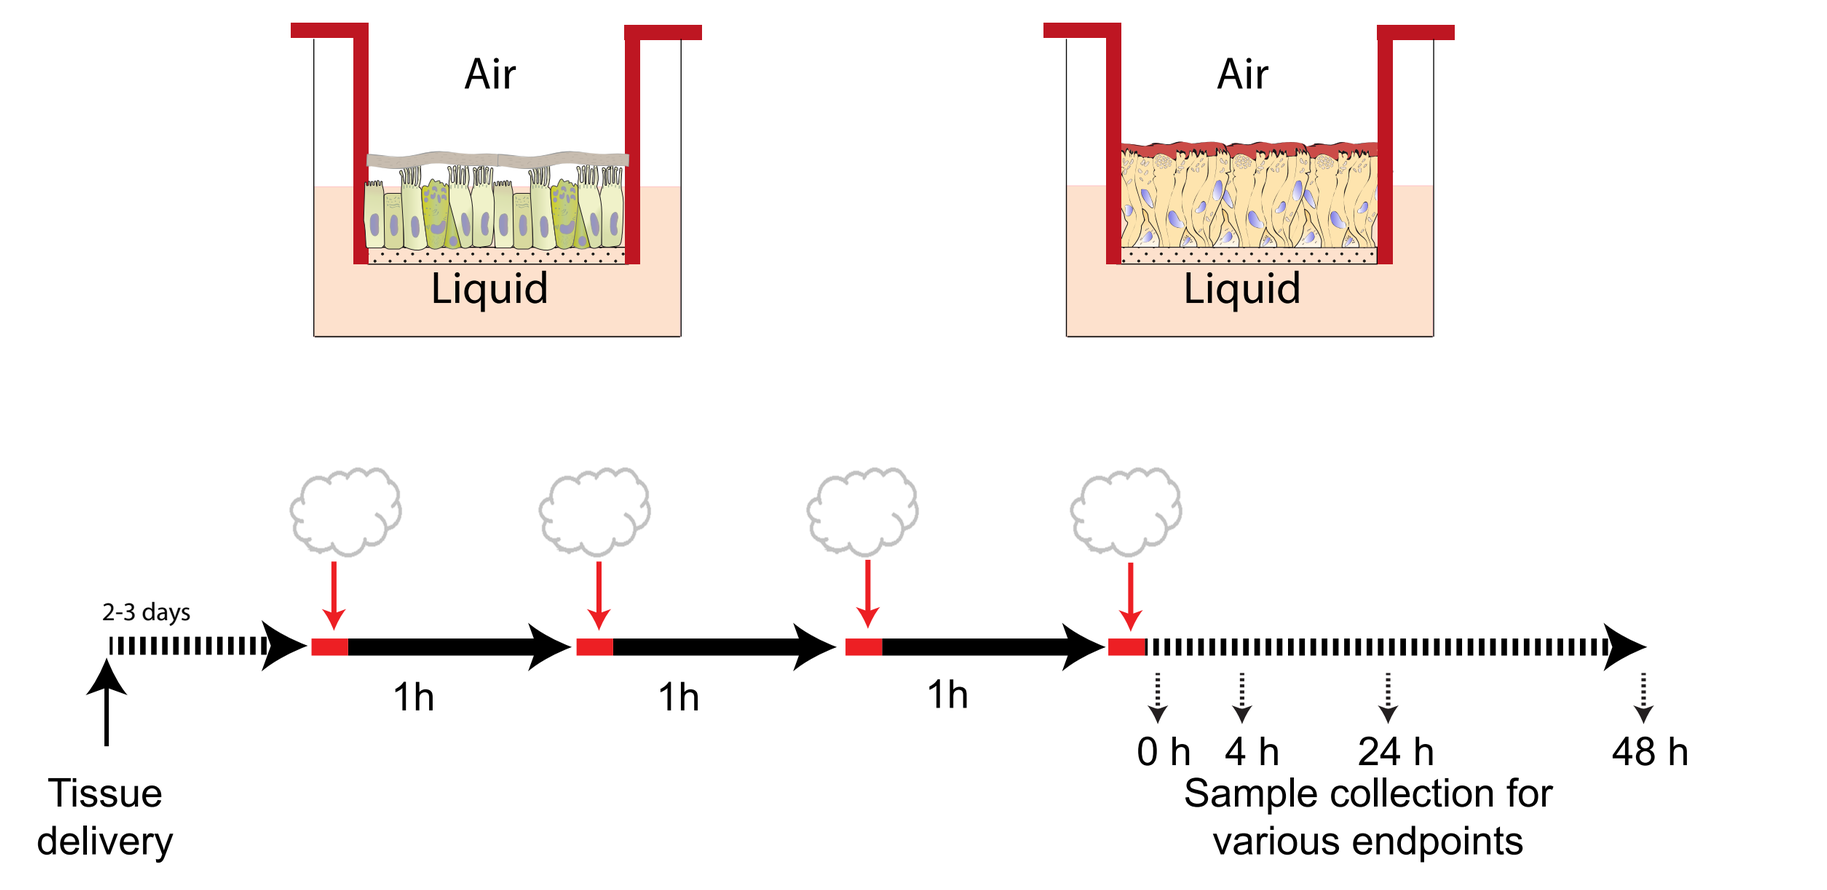
Air
Liquid
Air
Liquid
2-3 days
1h
1h
1h
Tissue delivery
0 h 4 h 24 h 48 h
Sample collection for various endpoints

Figure 2. Organotypic nasal and bronchial tissue cultures were exposed repeatedly to mainstream CS (3R4F) at air-liquid interface.

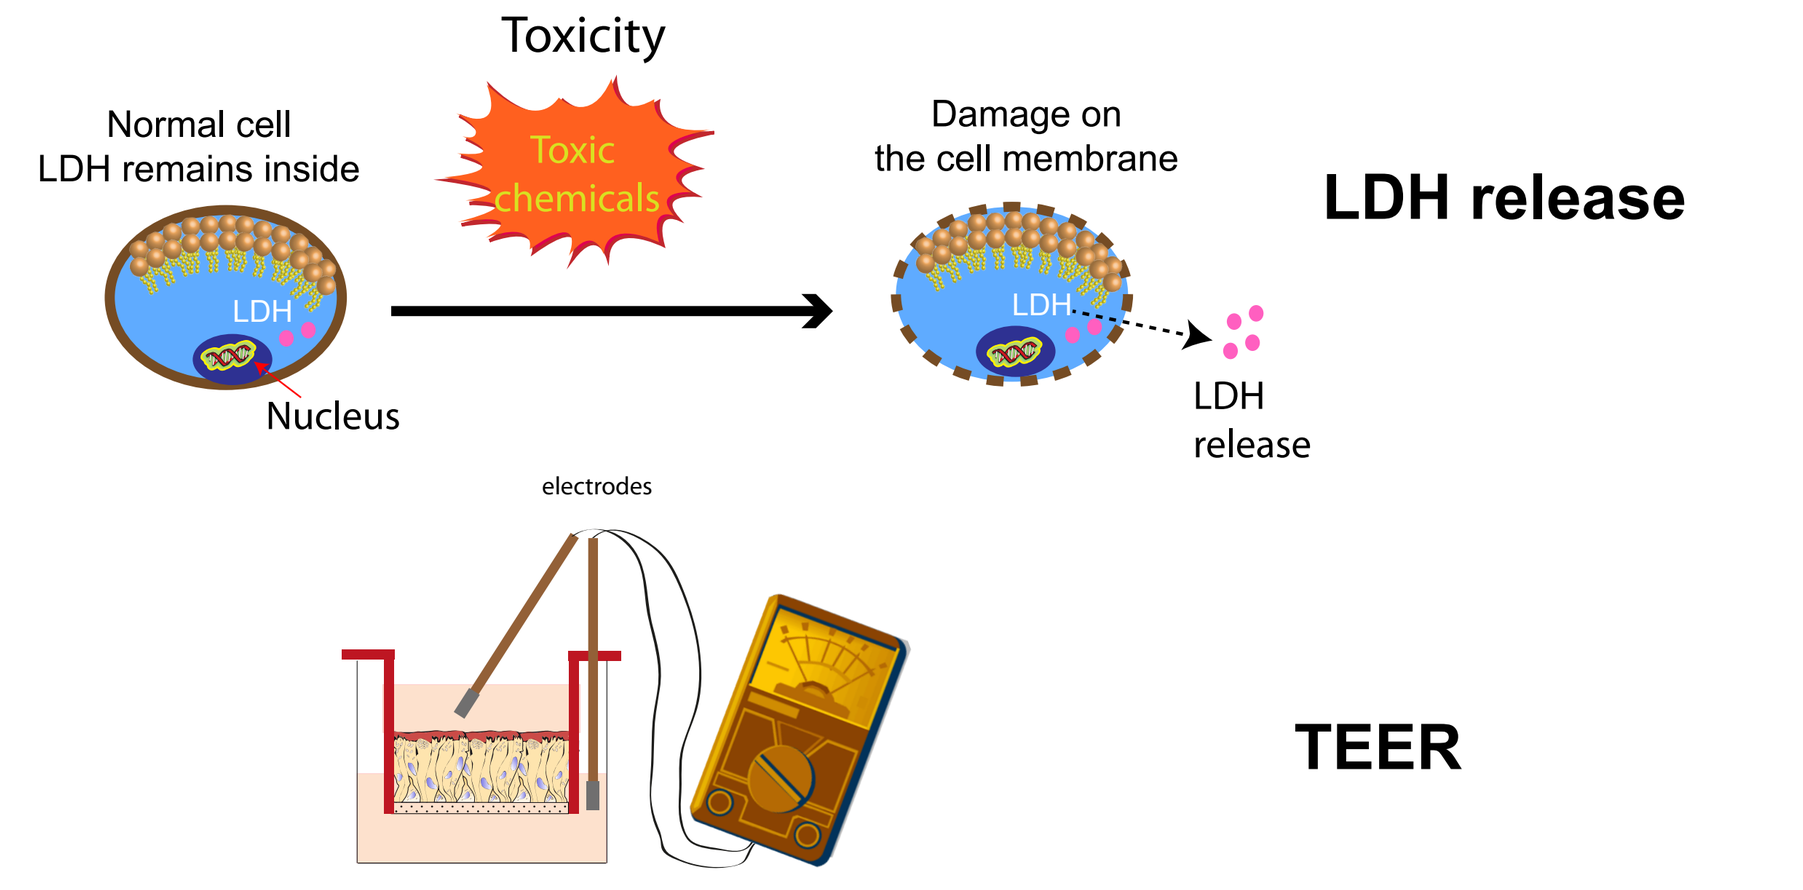
Toxicity
Normal cell
LDH remains inside
Toxic chemicals
Damage on the cell membrane
LDH release
LDH
Nucleus
LDH release
electrodes
TEER

Figure 3. Cytotoxicity and tissue integrity were assessed by measuring lactate dehydrogenase (LDH) release and transepithelial electrical resistance (TEER), respectively.

References

1) Sridhar, S. et al. 2008 Smoking-induced gene expression changes in the bronchial airway are reflected in nasal and buccal epithelium. BMC genomics 9(1):259.

2) Zhang, X., et al. 2010 Similarities and differences between smoking-related gene expression in nasal and bronchial epithelium. Physiological genomics 41(1):1-8.

3) Iskandar, A. R. et al. 2013 Systems approaches evaluating the perturbation of xenobiotic metabolism in response to cigarette smoke exposure in nasal and bronchial tissues. BioMed Research International 2013.

4) Gebel, S. et al. 2013 Construction of a Computable Network Model for DNA Damage, Autophagy, Cell Death, and Senescence. Bioinformatics and Biology Insights 7:97-117.

5) Schlage, W. K. et al. 2011 A computable cellular stress network model for non-diseased pulmonary and cardiovascular tissue. BMC Syst Biol 5:168.

6) Westra, J. W. et al. 2011 Construction of a Computable Cell Proliferation Network Focused on Non-Diseased Lung Cells. BMC Syst Biol 5(1):105.

7) Westra, J. W. et al. 2013 A Modular Cell-Type Focused Inflammatory Process Network Model for Non-Diseased Pulmonary Tissue. Bioinform Biol Insights 7:167-192.

8) The sbv Improver project team, et al. 2013 On Crowd-verification of Biological Networks. Bioinformatics and Biology Insights 7:307-325.

9) Hoeng, J. et al. 2012 A network-based approach to quantifying the impact of biologically active substances. Drug Discov Today 17(9-10):413-8.

10) Martin, F. et al. 2012 Assessment of network perturbation amplitude by applying high-throughput data to causal biological networks. BMC Syst Biol 6(1):54.

11) Thomson, T. M. et al. 2013 Quantitative assessment of biological impact using transcriptomic data and mechanistic network models. Toxicology and applied pharmacology 272(3):863-878.

Results

48 h post exposure time point

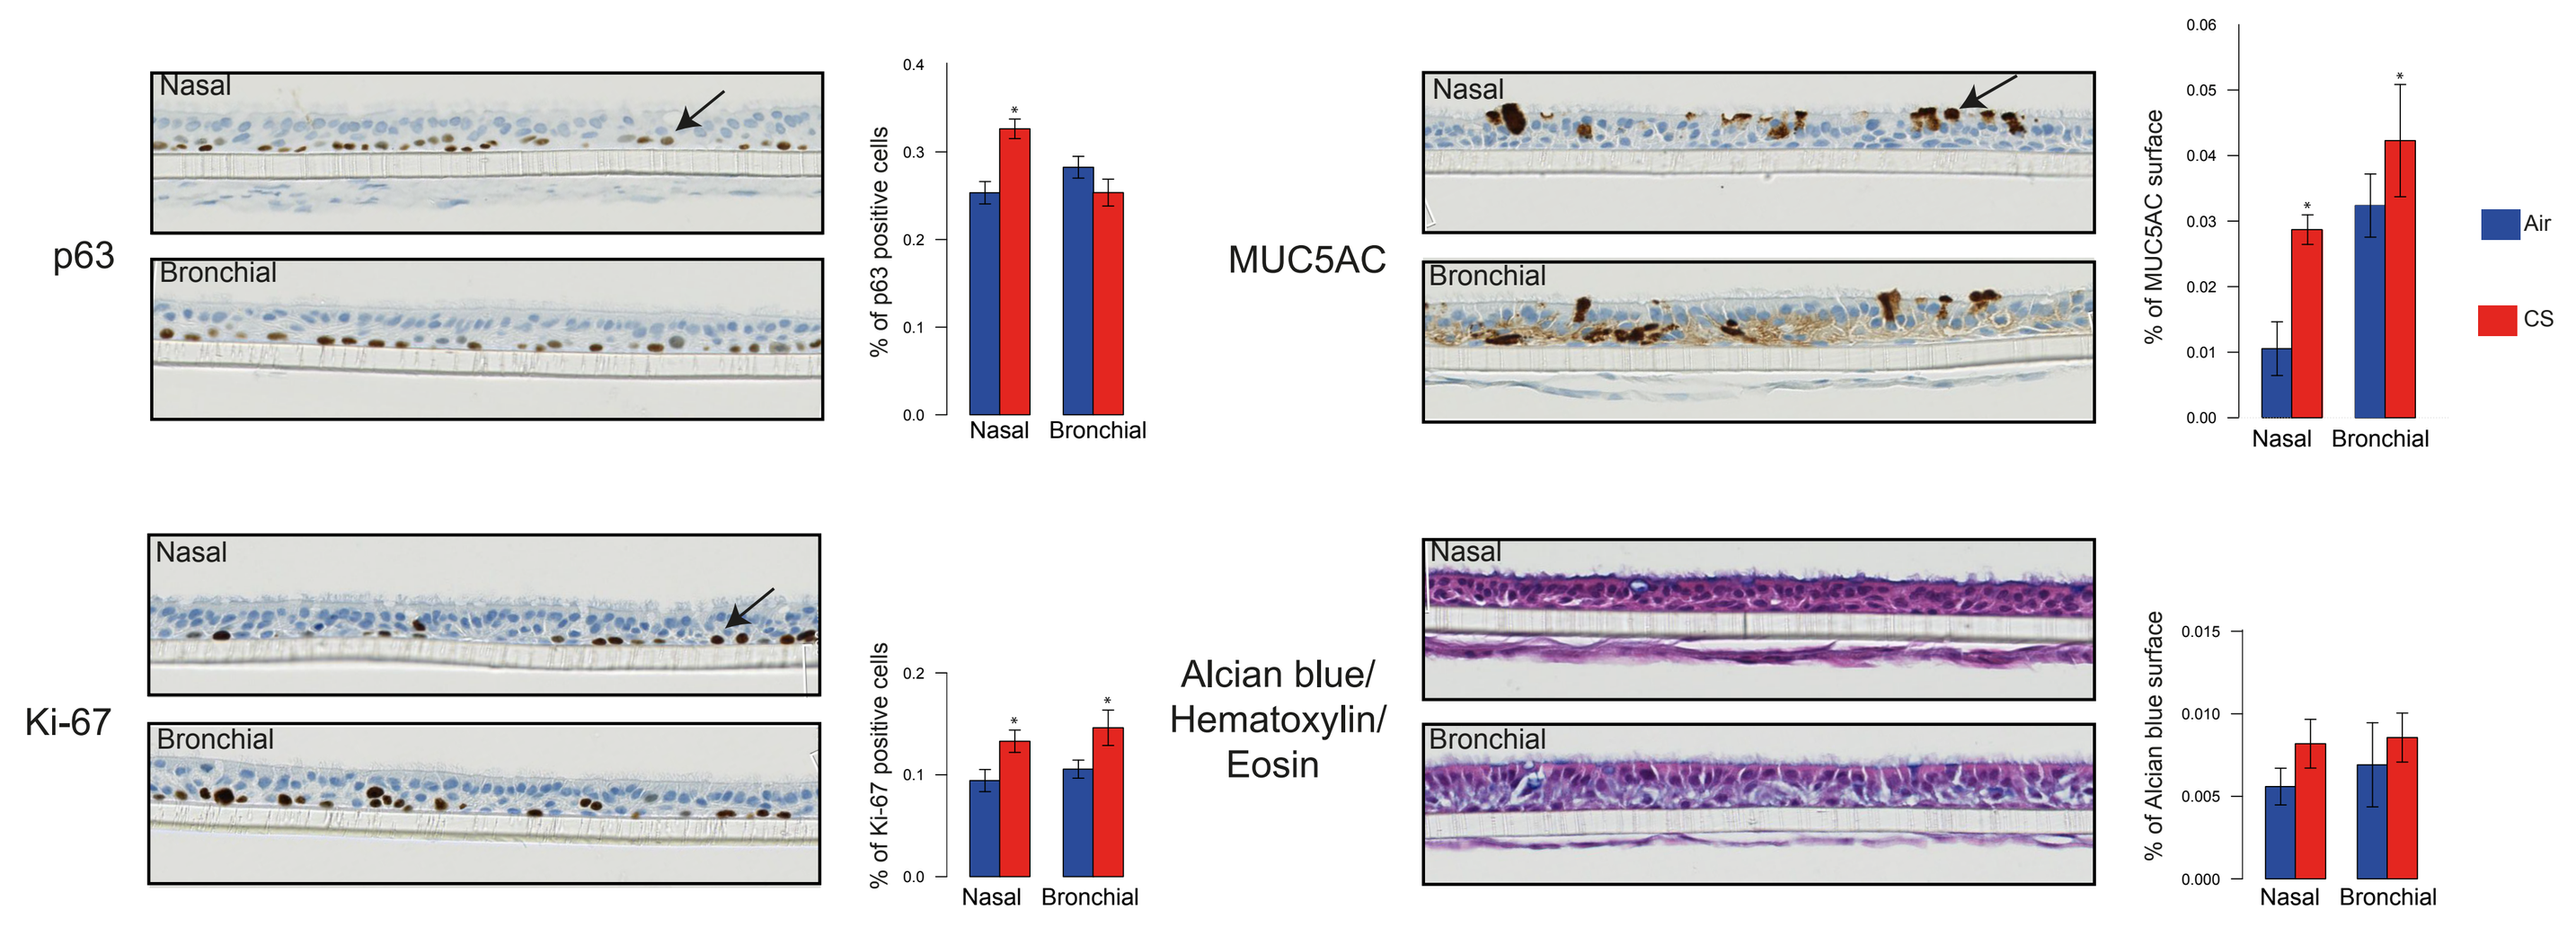
p63
Nasal
Bronchial
% of p63 positive cells
MUC5AC
% of MUC5AC surface
Air
CS
Ki-67
% of Ki-67 positive cells
Alcian blue/ Hematoxylin/ Eosin
% of Alcian blue surface

Figure 4. Representative sections of nasal (top) and bronchial (bottom) cultures 48 h after the last CS exposure. Arrows indicate the positively stained cells. The bar graphs show the quantification of the staining as a percentage (median ± standard error of the median) of all nuclei in the tissue sections for p63 and Ki-67 (percentage of stained nuclei), and the area occupied by positive cells for MUC5AC and Alcian blue (percentage of stained area). * indicates a statistically significant difference between the median of the CS and air exposed cultures for the same tissue. The Wilcoxon rank-sum test calculated the significance level as 5%.

0 100 200 300 µm

CS exposure resulted in a small increase in the numbers of basal and proliferating cells.
The impact of CS on the Muc5Ac positive cells was more pronounced in the nasal cultures.

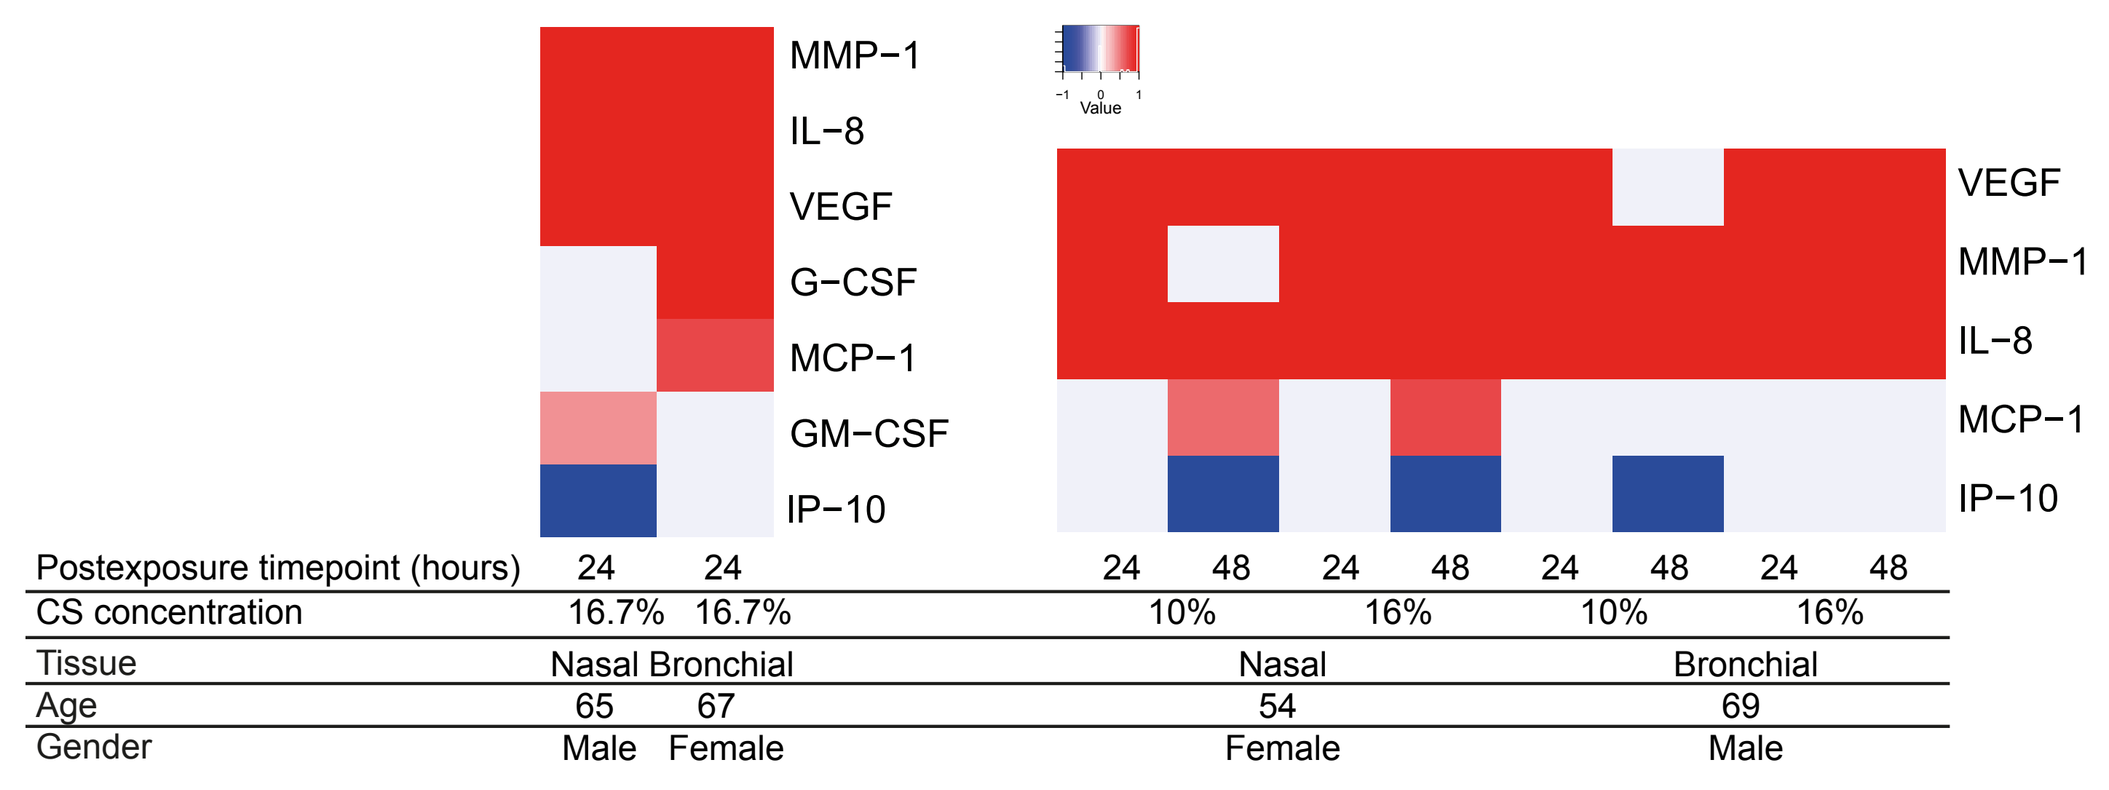
MMP−1
IL−8
VEGF
G−CSF
MCP−1
GM−CSF
IP−10
Value
VEGF
MMP−1
IL−8
MCP−1
IP−10
Postexposure timepoint (hours) 24 24 24 48 24 48 24 48 24 48
CS concentration 16.7% 16.7% 10% 16% 10% 16%
Tissue Nasal Bronchial Nasal Bronchial
Age 65 67 54 69
Gender Male Female Female Male

Figure 5. Heatmaps of the fold changes of inflammatory markers from two independent studies. Columns represent different post-exposure time points, CS concentrations, tissues, and donor characteristics as labeled under the heatmap. Fold changes were obtained by taking the log2 of the cytokine abundance ratio between the CS and sham exposure group for every tissue. Welch's t-test was performed to test the null hypothesis that cytokine abundance in CS and sham exposure groups are the same. Fold change was set at zero for p-values > 0.05. Blue and red indicate statistically significant negative and positive fold changes, respectively.

CS exposure resulted in the reproducible accumulation of MMP-1, IL-8, and VEGF in the culture medium of both nasal and bronchial tissue cultures and attenuated the secretion of interferon gamma-induced protein (IP-10) by nasal cultures.

The accumulation of IL-10, IL-1A, IL-1B, IL-6, MIP-1A, MMP-9, eotaxin, and RANTES in the culture medium was not impacted (not shown) by CS exposure and the impact of CS on the secretion of MCP-1 G-CSF, and GM-CSF was not reproducible at this post-exposure time point.

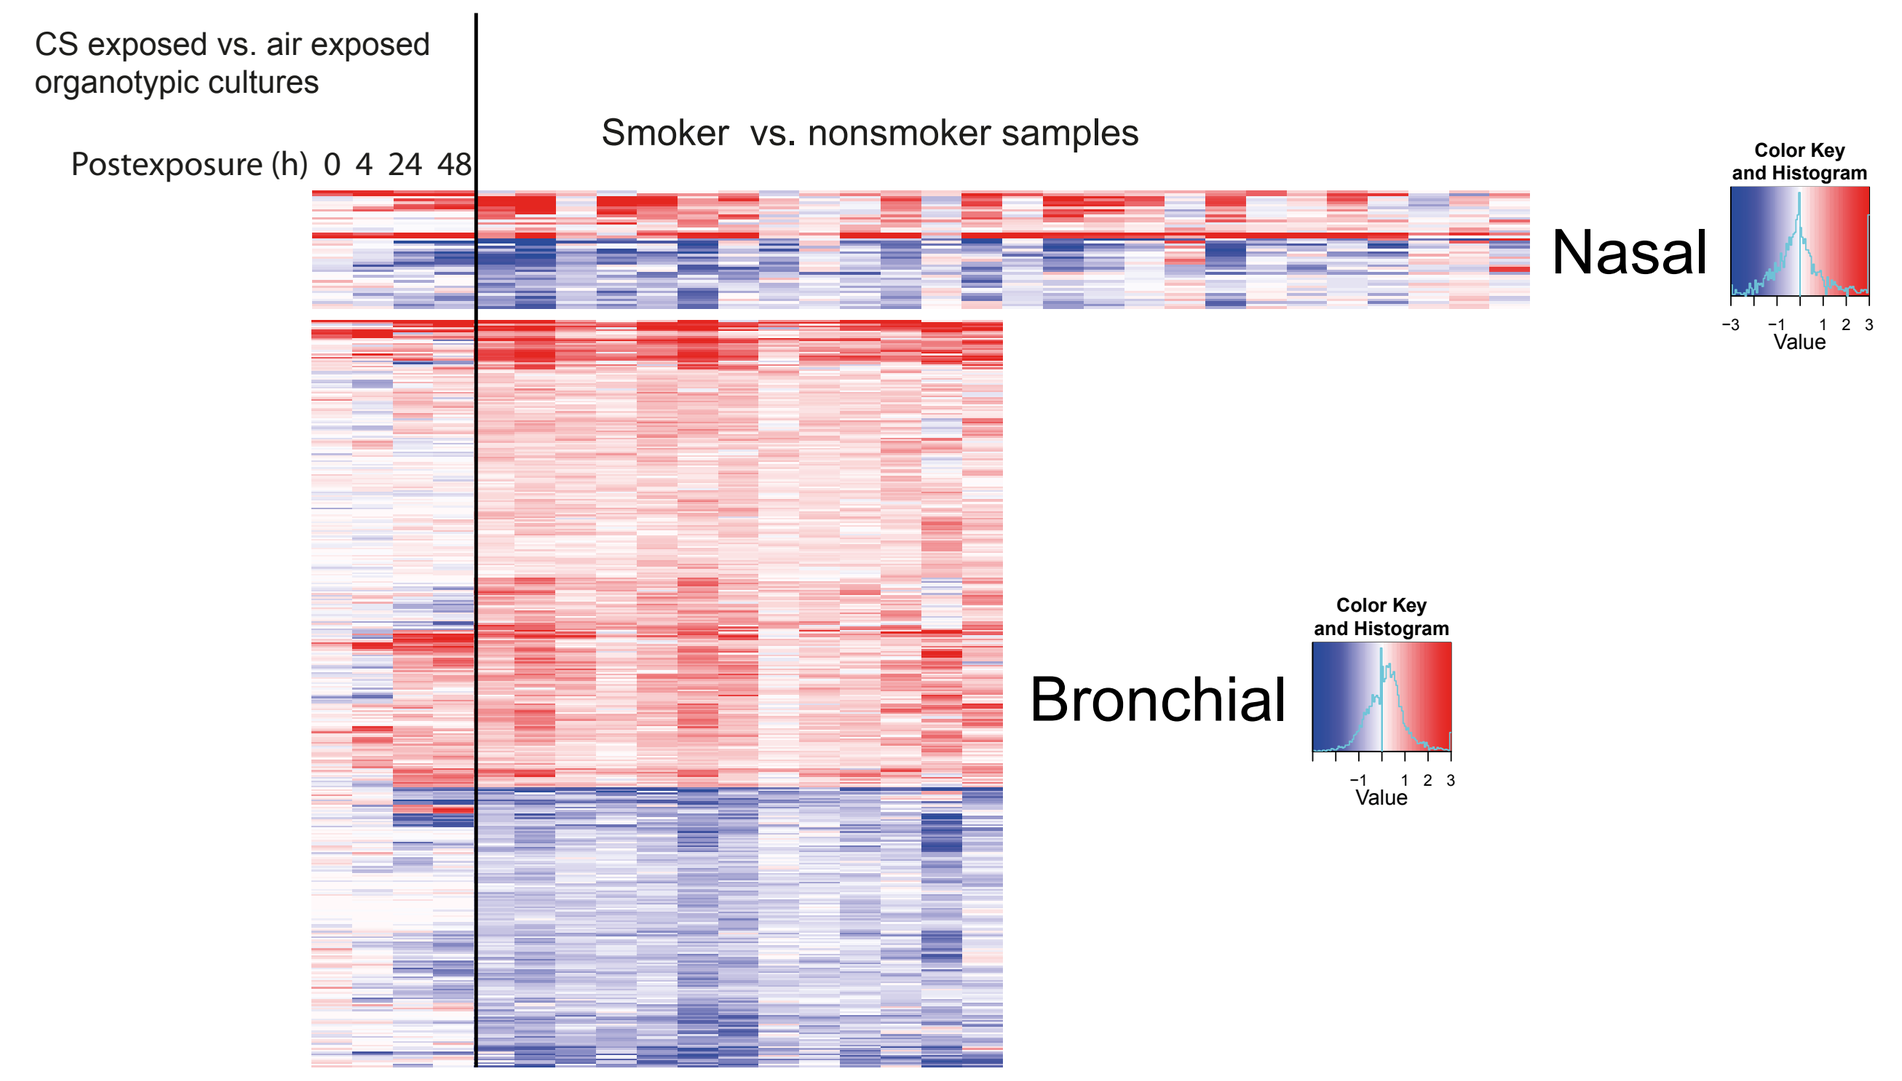
CS exposed vs. air exposed organotypic cultures
Postexposure (h) 0 4 24 48
Smoker vs. nonsmoker samples
Nasal
Color Key and Histogram
Value
Bronchial
Color Key and Histogram
Value

Figure 6. Comparison of cigarette smoke exposure response in in-vivo (GSE16008) (2) and in-vitro nasal and bronchial tissue cultures. Top panel: Grey and black bars denote sets of significantly up- and downregulated genes, respectively, that were significantly enriched in in-vitro organotypic bronchial tissue. NES: normalized enrichment score. ***False discovery rate (FDR) 0.001; **FDR 0.01; *FDR 0.05. Bottom panel: Gene expression fold changes in the in-vitro organotypic tissue cultures exposed to CS compared with air-exposed cultures are plotted side by side with gene expression fold changes from the in vivo study (GSE16008) obtained by taking the differentially expressed genes in smokers' samples compared to the average expression level from nonsmokers. Blue and red colors indicate negative and positive fold changes respectively.

Several differentially expressed genes in human smoker nasal and bronchial epithelial cells were also regulated in the organotypic cultures exposed to whole smoke.

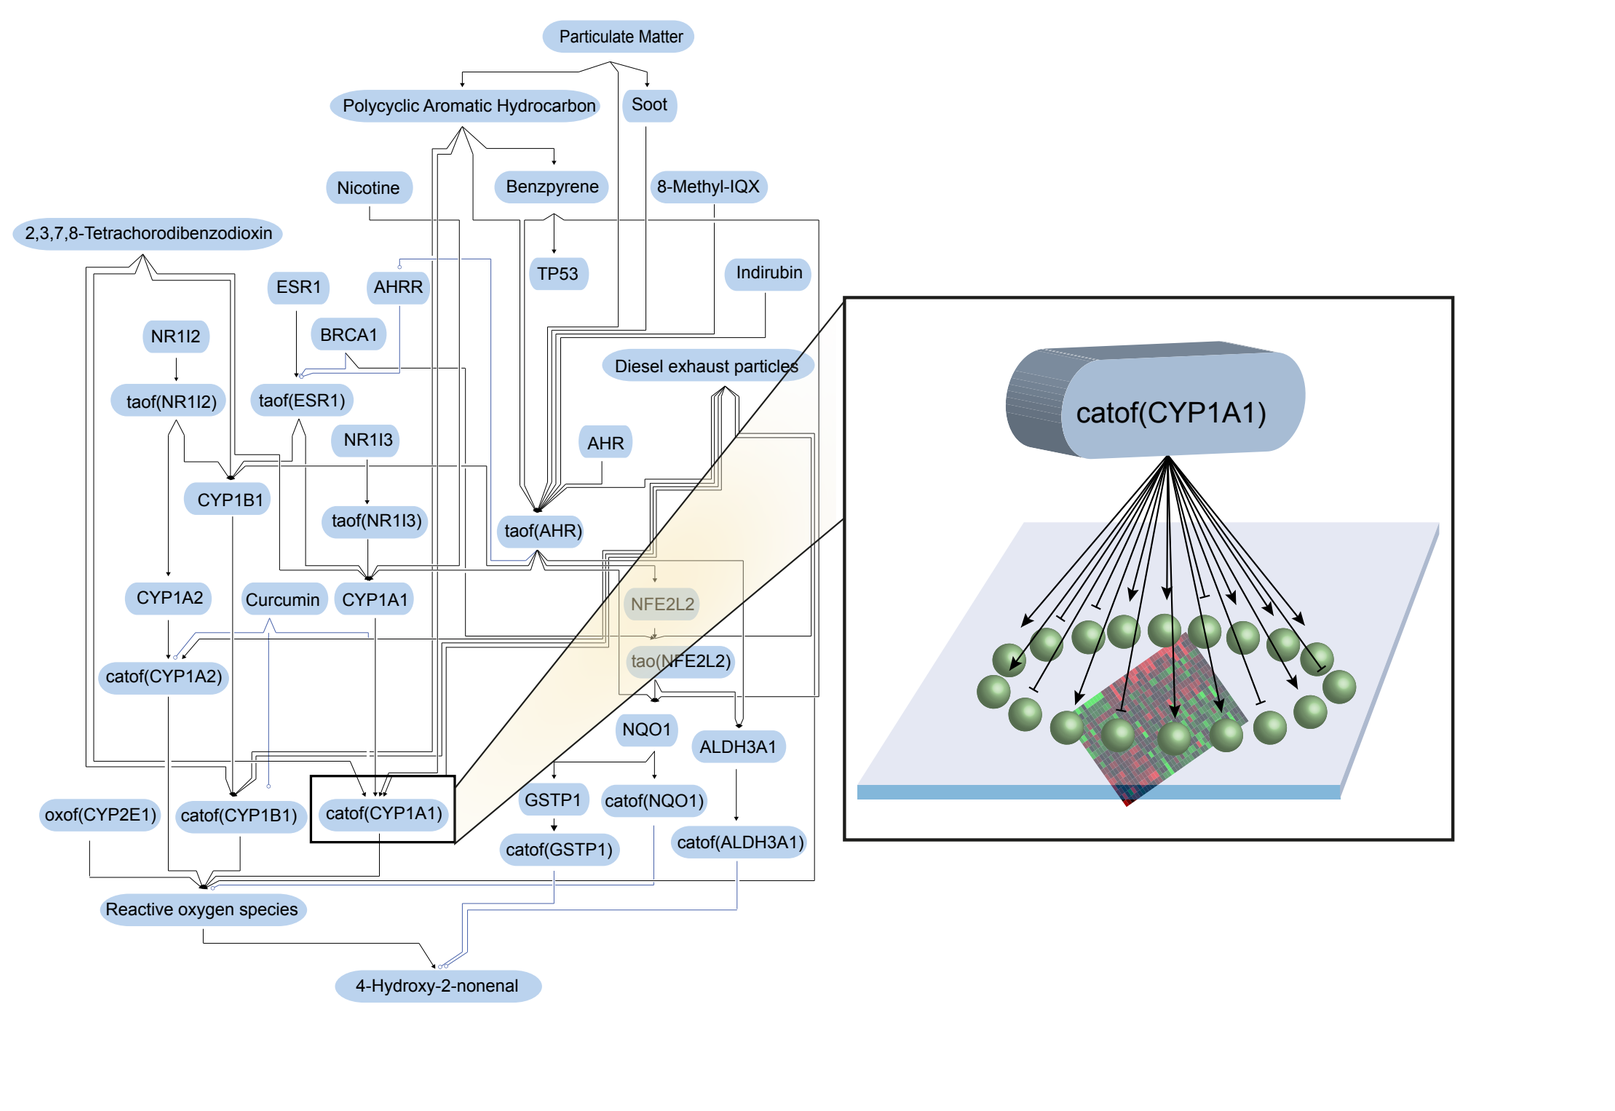
catof(CYP1A1)

Figure 7. A biological network model (above) comprises the functional layer of the network, which is connected by causal edges carrying the directional information. The blue ovals represent the activity of the backbone nodes (i.e., the functional layer) and the green balls represent the expression of genes (i.e., the transcriptional layer) (4-8).

Network scoring assesses the impact of treatment on specific backbone entities based on the differential gene expression of downstream genes that are known to be regulated by these mechanisms (9-11).The quantitative measure of this impact is called the Network Perturbation Amplitude (NPA).

The heatmap (right) shows the perturbation of the biological networks at each post exposure time point in nasal and bronchial organotypic tissue cultures in response to CS exposure. The biological NETWORKS and sub networks are labelled on the left with the number of nodes in each network. Shown are the results from two independent experiments conducted on the tissue cultures from different donors (donor characteristics shown on the bottom). Normalized NPA score takes into account both the magnitude of network perturbation and the Specificity statistics (asterisk).

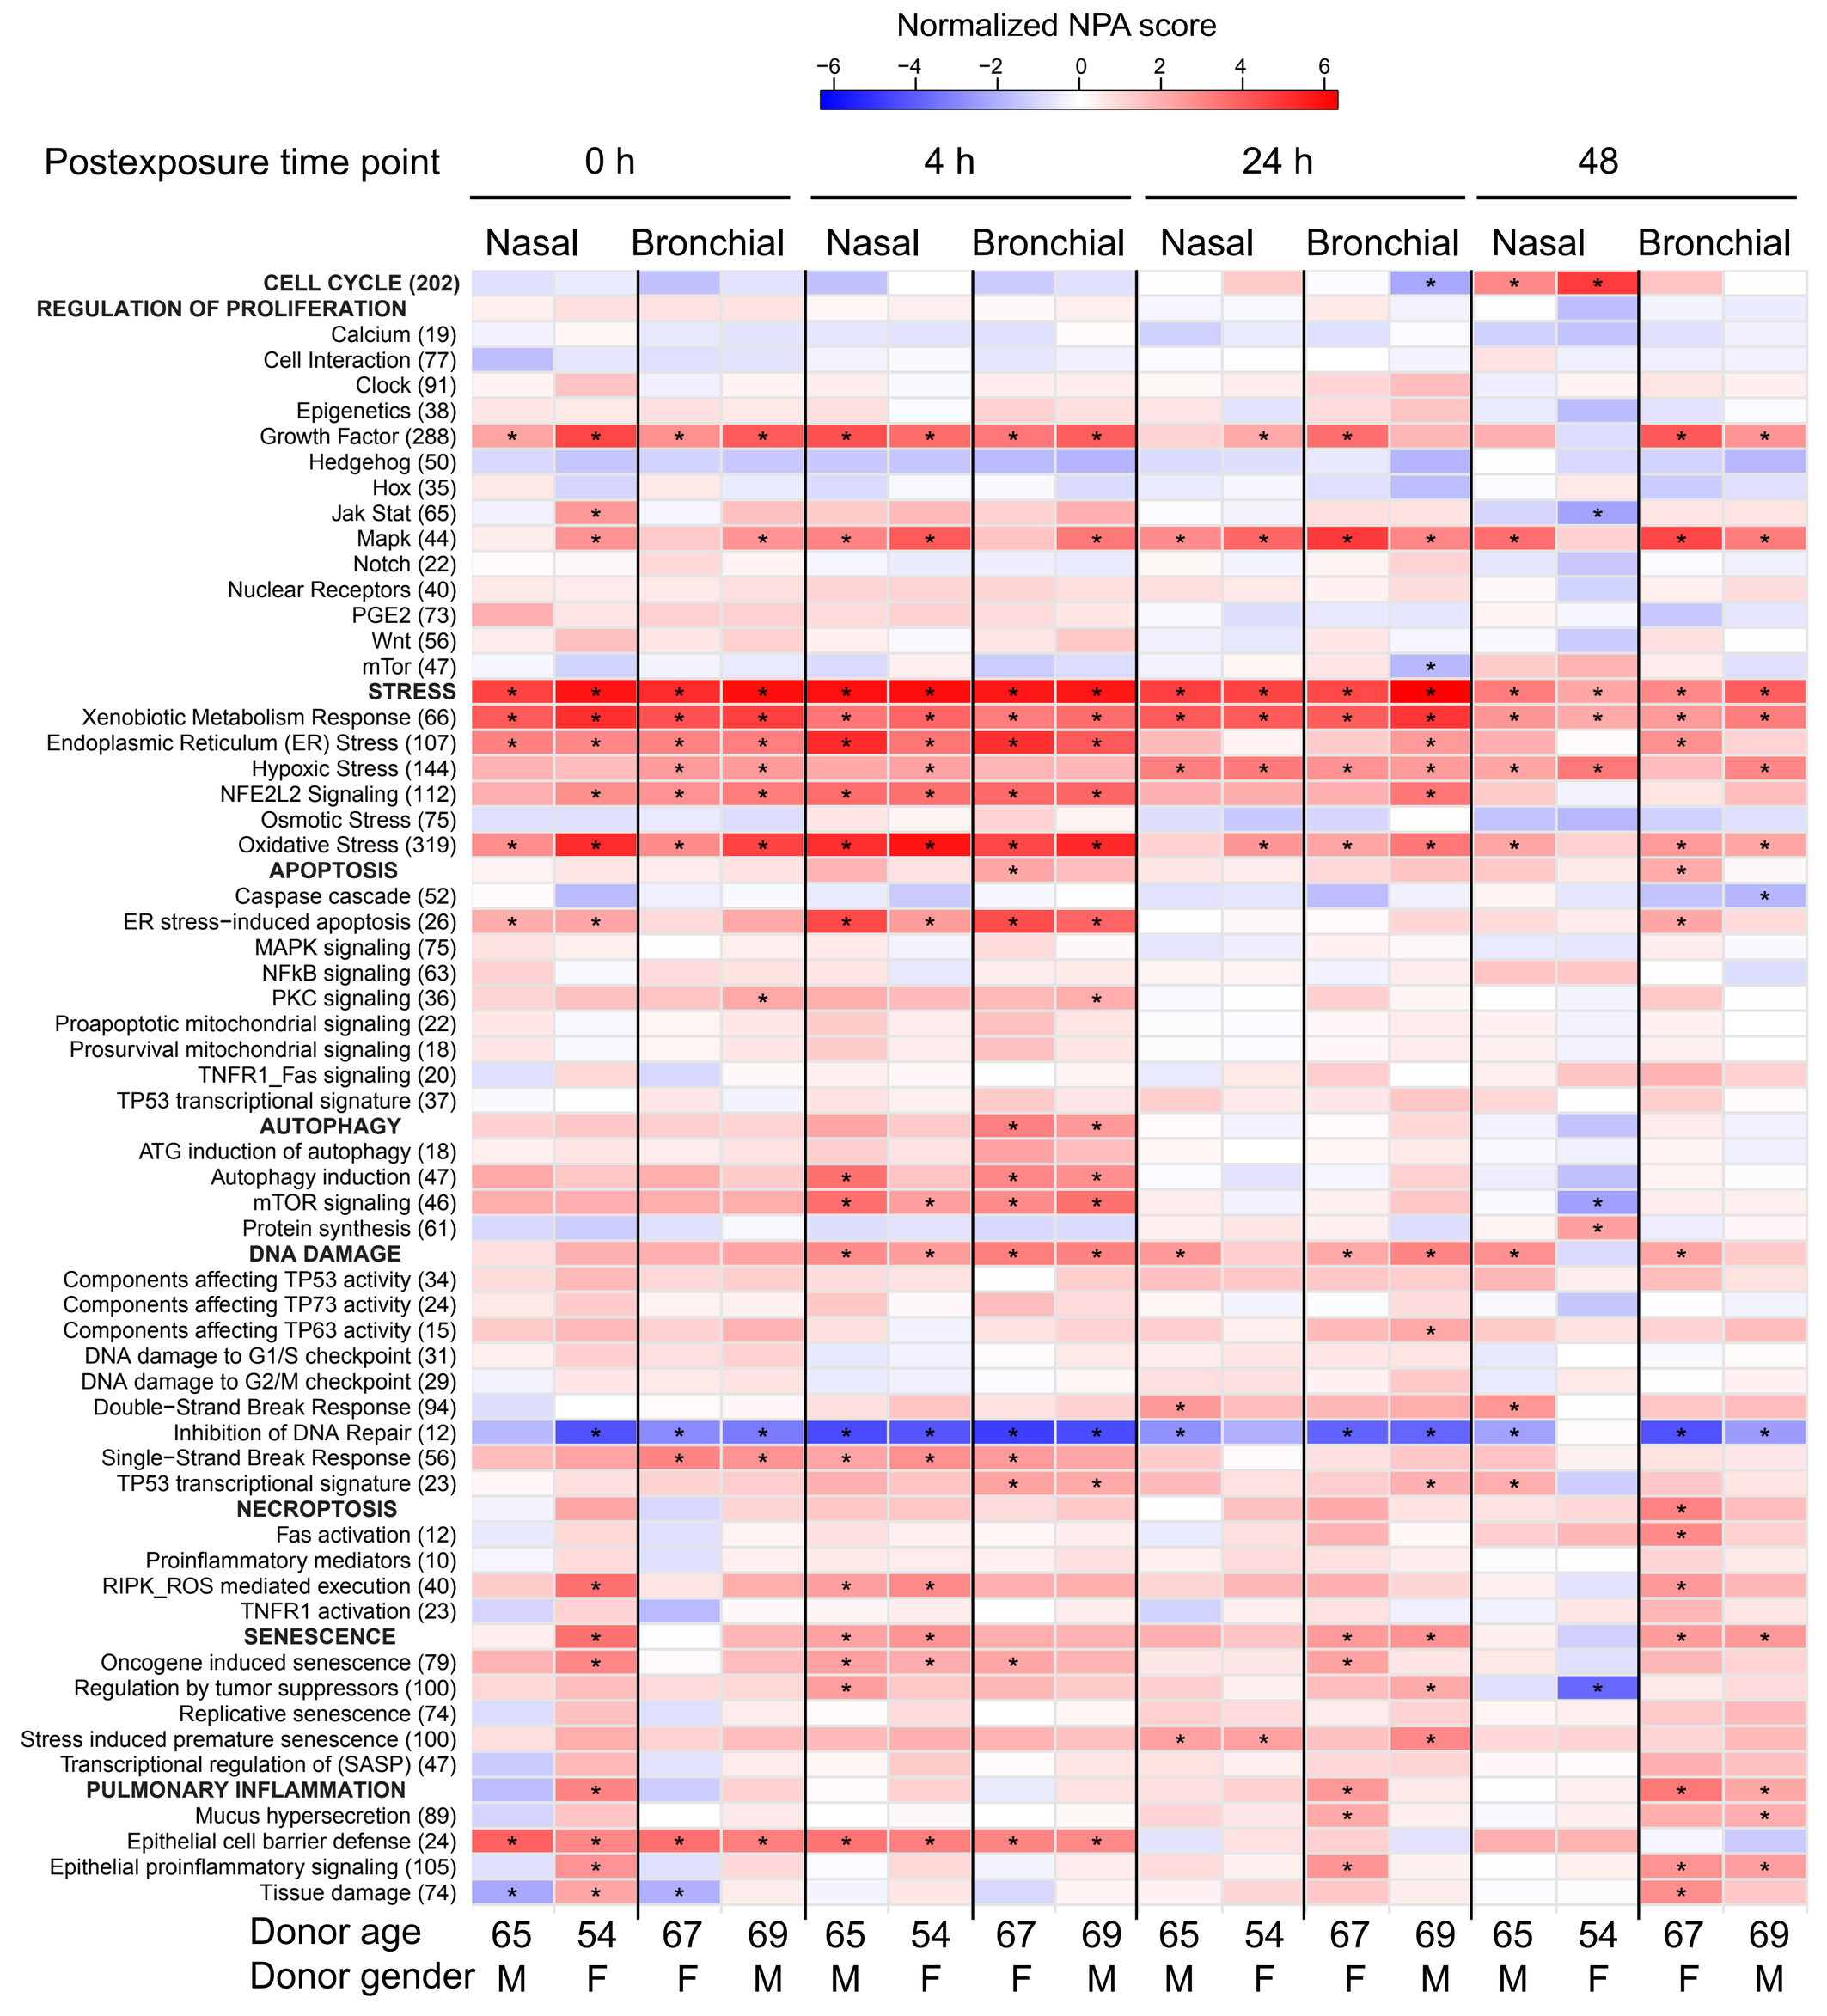
Normalized NPA score
−6 −4 −2 0 2 4 6
Postexposure time point 0 h 4 h 24 h 48
Nasal Bronchial Nasal Bronchial Nasal Bronchial Nasal Bronchial
CELL CYCLE (202)
REGULATION OF PROLIFERATION
STRESS
APOPTOSIS
AUTOPHAGY
DNA DAMAGE
NECROPTOSIS
SENESCENCE
PULMONARY INFLAMMATION
Donor age 65 54 67 69 65 54 67 69 65 54 67 69 65 54 67 69
Donor gender M F F M M F F M M F F M M F F M

The analysis of transcriptomic data in the context of biological network models showed that whole CS exposure had a clear impact on biological processes in both nasal and bronchial cultures:

1. The cellular stress response clearly dominated in both tissue cultures throughout the post exposure period.

2. Processes related to proliferation, cell fate, and inflammation were activated and DNA repair was inhibited in both nasal and bronchial tissues in response to CS exposure.

3. While cell cycle became activated in the nasal cultures at 48 hours after exposure, bronchial cultures reproducibly showed the activation of epithelial proinflammatory signaling at this post exposure time point.

Conclusions

Both nasal and bronchial organotypic cultures showed changes in inflammatory mediator release in response to repeated exposure to mainstream CS.

While the LDH release, TEER assay, and histology analysis did not indicate cytotoxicity in response to whole smoke exposure, the transcriptomic analysis revealed the activation of biological processes related to cellular stress, proliferation, inflammation, and cell fate.

Gene expression patterns in CS exposed organotypic tissue cultures closely resembled those observed in smoker nasal and bronchial epithelium.

The state-of-the art organotypic nasal and bronchial tissue cultures are a promising system to study inhalation toxicology; they provide an alternative to animal experimentation and cells grown on a monolayer.