iTRAQ approach to investigate the impact of cigarette smoke, smoking cessation and switching to a prototypic modified risk tobacco product on the lung proteome of C57BL/6 mice

T. Schneider[1]*, S. Dijon[1], A. Elamin[1], B. Phillips[2], Emilija Veljkovic[1], B. Titz[1], F. Martin[1], N.V. Ivanov[1], J. Hoeng[1], M.C. Peitsch[1].

[1]Philip Morris International, Research & Development, Neuchâtel, Switzerland; [2]Philip Morris International, Research & Development, Singapore

Introduction

Reliable system-wide measurements are key to a meaningful systems-toxicology approach by which impact of toxicants in cells are evaluated (Hoeng et al., 2012). While still being challenging to setup, liquid-chromatography mass-spectrometry (LC-MS) based quantitative proteomics may lead to a further understanding of systems response and complement the widely used transcriptomics data.

In the context of a life-long animal study aiming at characterizing the impact of cigarette smoke in mouse lung and the effect of cessation and switching to a prototypic Modified Risk Tobacco Product (pMRTP), we established an isobaric tags for absolute and relative quantification (iTRAQ®)-based pipeline aiming at capturing a wide variety of proteins. The arms of this animal study involving C57/Bl6 mice were exposure to: (i) fresh-air only, (ii) conventional cigarette smoke (CS) from 3R4F cigarettes (from University of Kentucky), (iii) CS and switch to a pMRTP, (iv) CS and switch to fresh air (cessation) and (v) pMRTP only.

The aim of the study was:

(i) To setup a robust iTRAQ-based proteomics approach for quantifying abundance changes in mouse lung.

(ii) To evaluate the potential of an iTRAQ worflow in a systems-toxicology approach for product assessment, in particular in the context of novel modified risk tobacco products (MRTPs) assessment using *in vivo* model systems.

(iii) To identify regulated proteins and biological processes in mouse lungs in response to cigarette-smoke, cessation , switching to an pMRTP and pMRTP only.

MATERIALS & METHODS

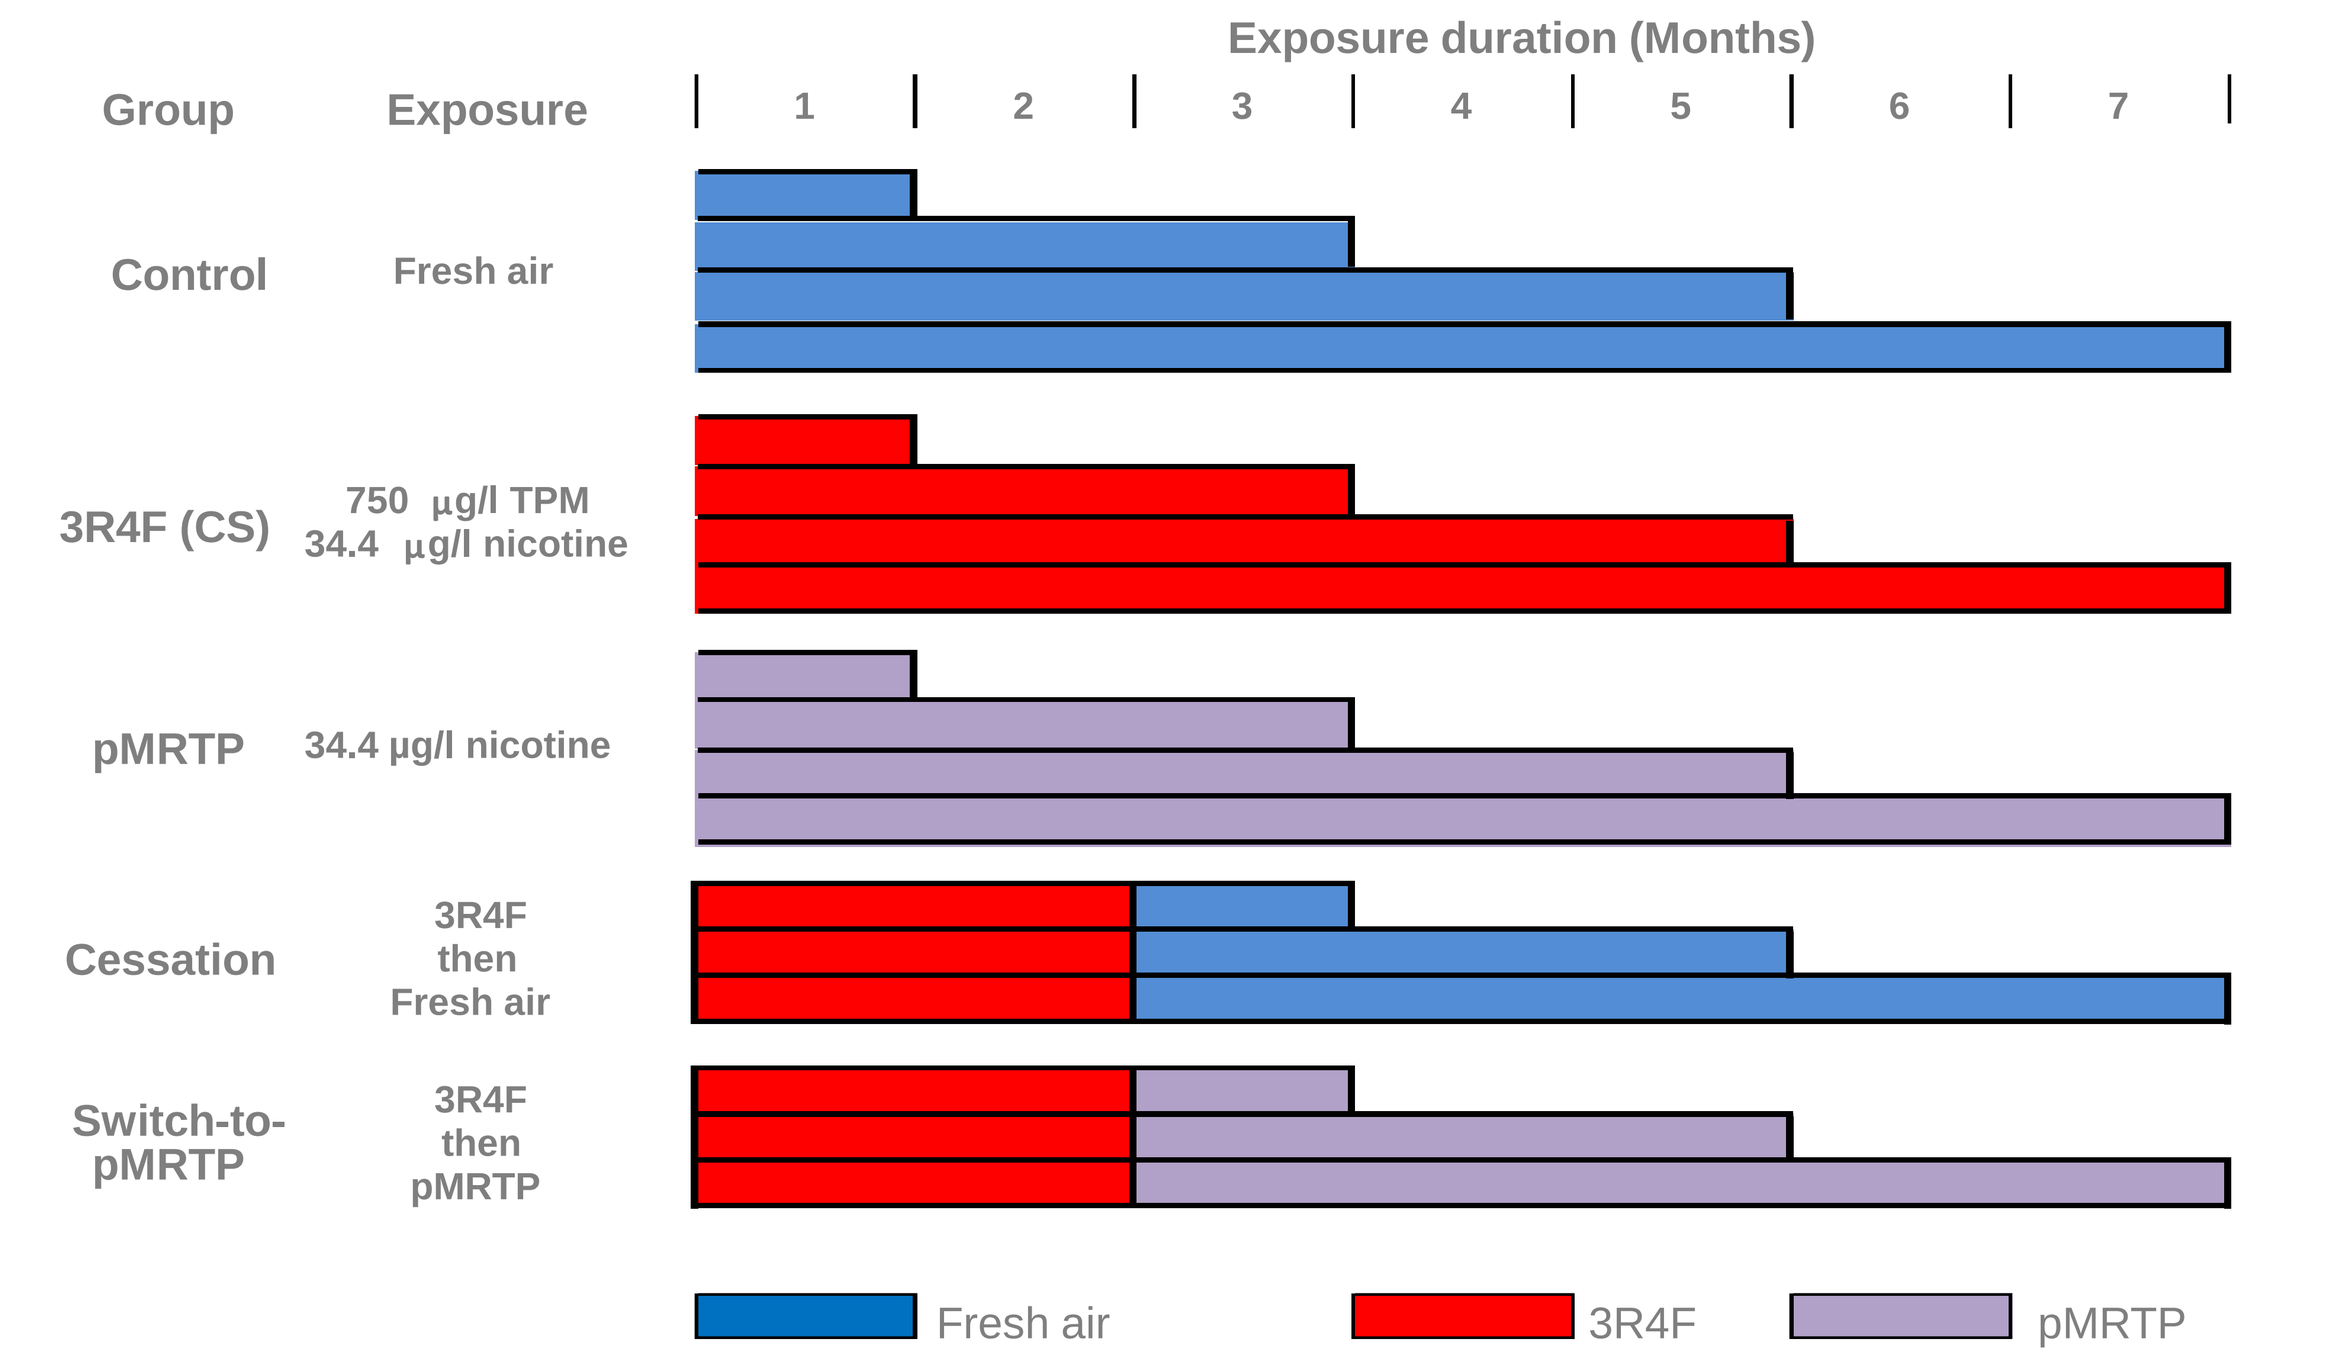
Exposure duration (Months)
Group
Exposure
1 2 3 4 5 6 7
Control
Fresh air
3R4F (CS)
750 µg/l TPM
34.4 µg/l nicotine
pMRTP
34.4 µg/l nicotine
Cessation
3R4F then Fresh air
Switch-to-pMRTP
3R4F then pMRTP
Fresh air
3R4F
pMRTP

Figure 1: Study design with different exposure groups and durations. 3R4F cigarettes were smoked according to the Health Canada Intense Puffing Regime. Nicotine concentrations of 3R4F and pMRTP treatments were matched.

Eight to 10 week old mice were exposed to aerosols for 5 days a week and 4 h per day. The different exposure groups and durations are indicated in **Figure 1.** Protein from lung tissue was extracted according to **Figure 2.** An LC-MS based quantitative proteomics approach using iTRAQ® was performed in order to detect changes in protein expression levels between the different groups (experiments were run in six biological replicates). Acquired data were quality controlled and analyzed to identify differentially expressed proteins by in-house developed pipelines (**Figure 3 and 4**).

.

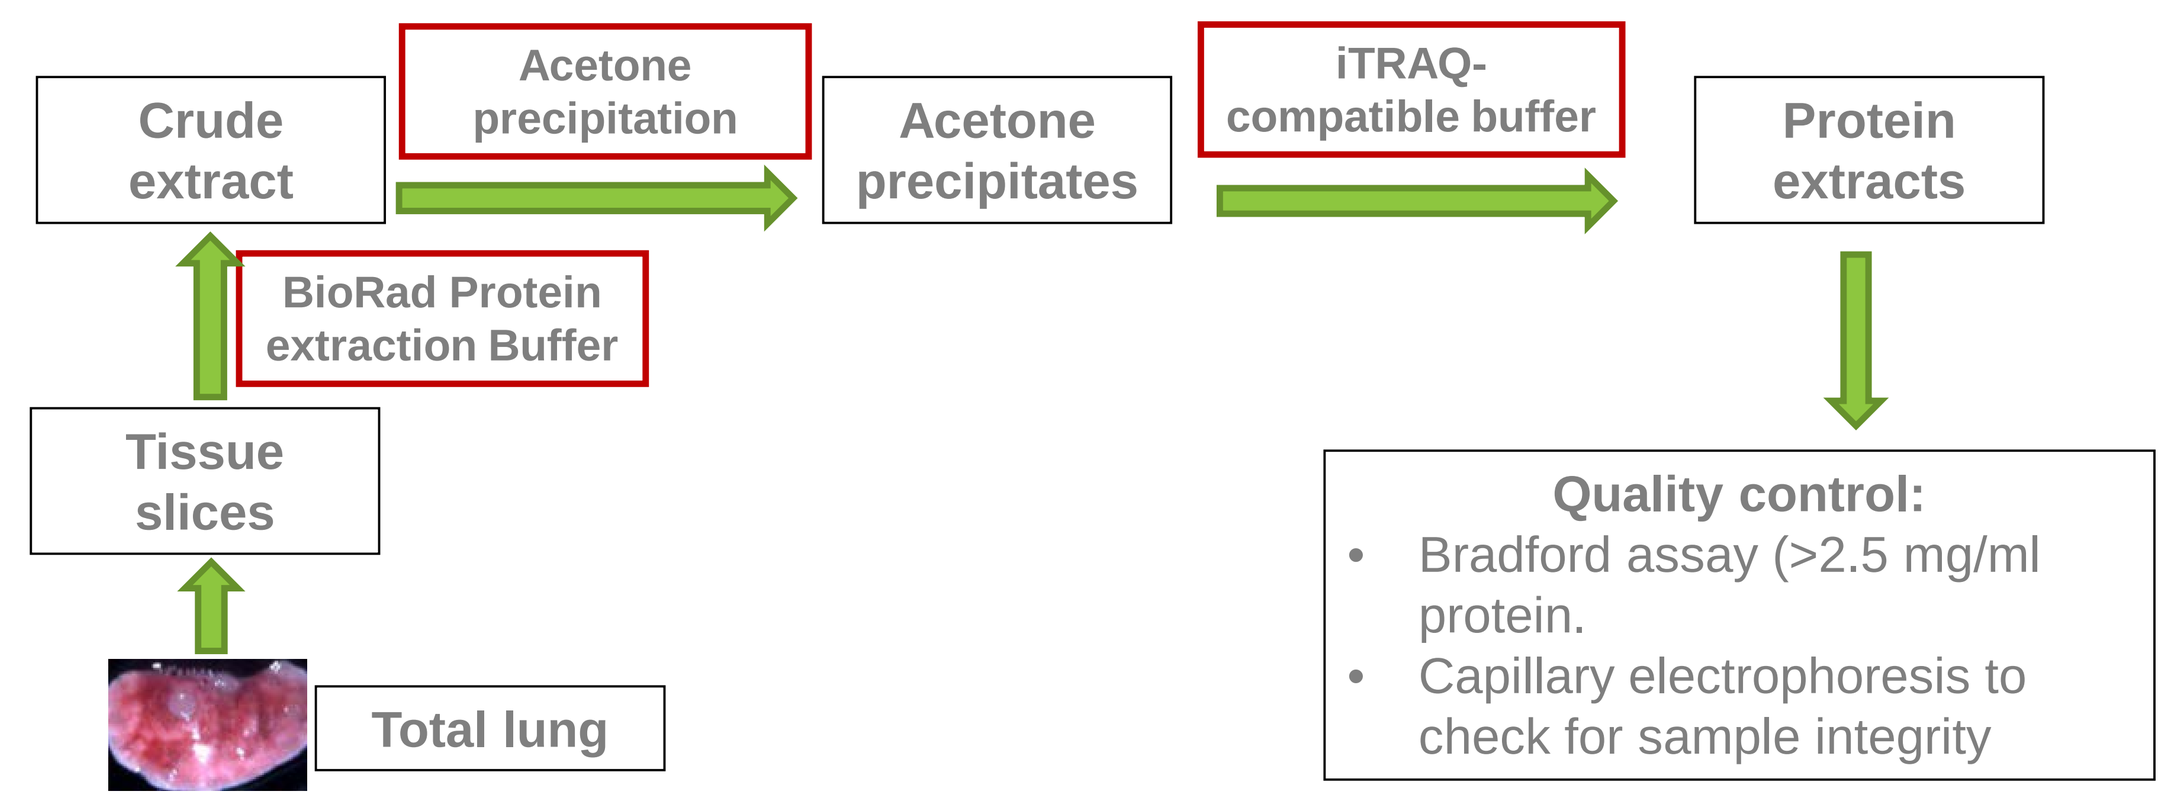
Crude extract
Acetone precipitation
Acetone precipitates
iTRAQ-compatible buffer
Protein extracts
BioRad Protein extraction Buffer
Tissue slices
Total lung
Quality control:
Bradford assay (>2.5 mg/ml protein.
Capillary electrophoresis to check for sample integrity

Figure 2: Workflow for the extraction of proteins from mouse lungs

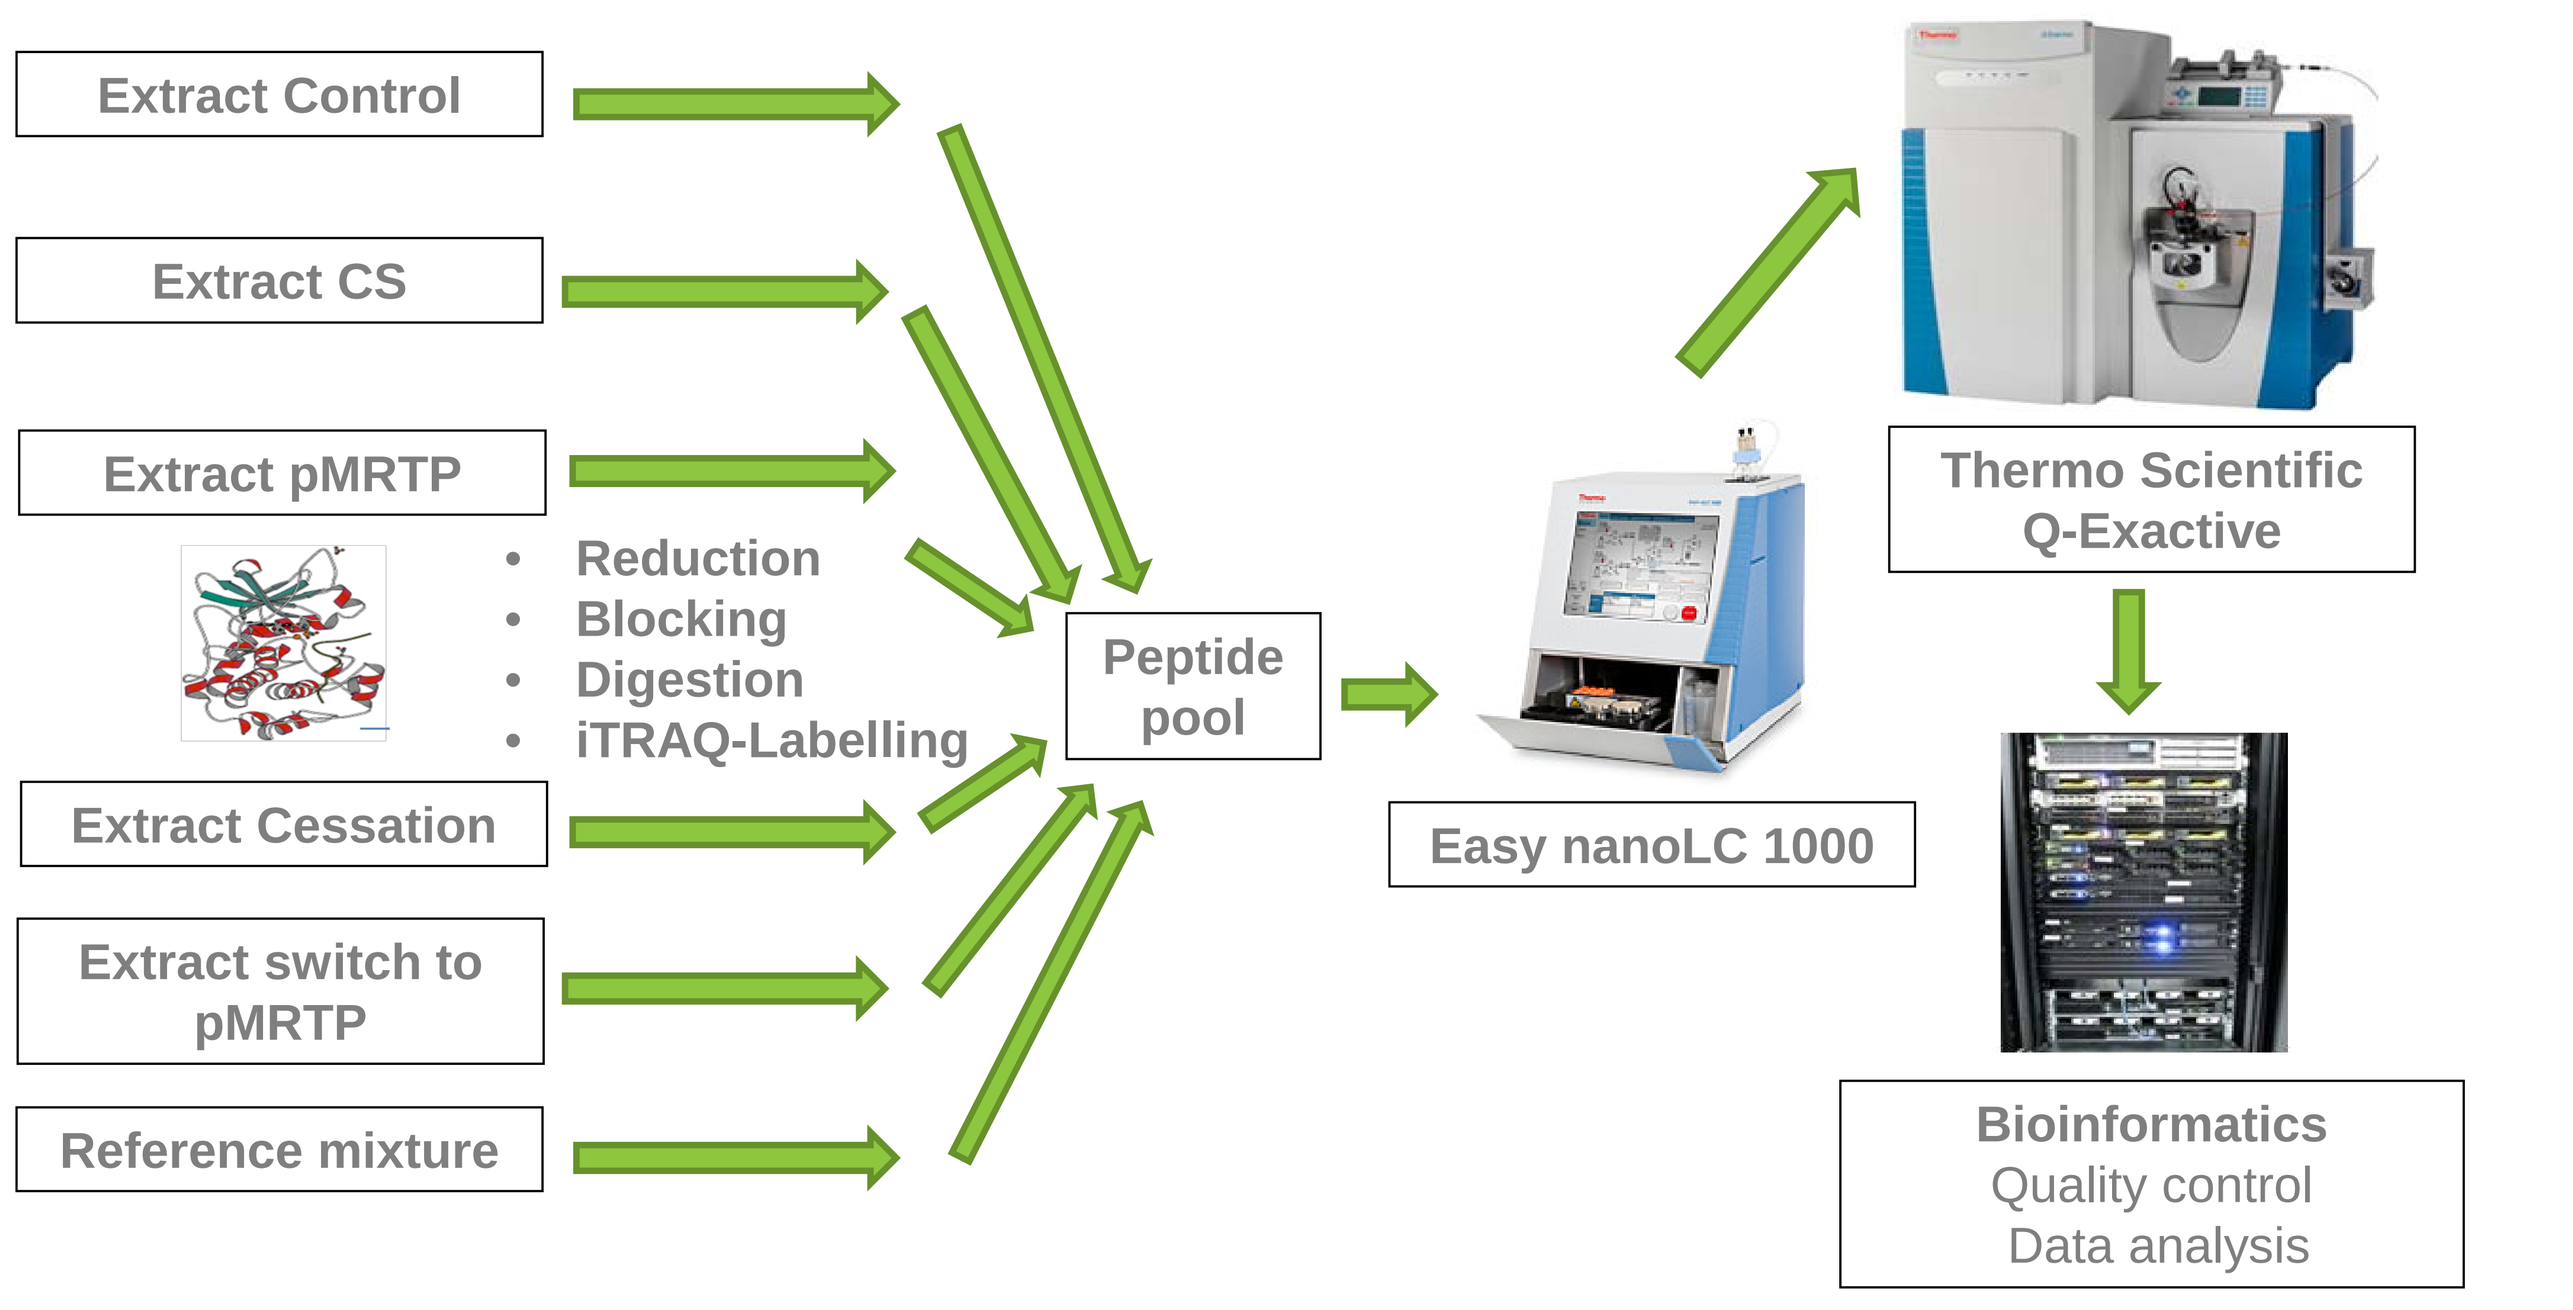
Extract Control
Extract CS
Extract pMRTP
Reduction
Blocking
Digestion
iTRAQ-Labelling
Extract Cessation
Extract switch to pMRTP
Reference mixture
Peptide pool
Easy nanoLC 1000
Thermo Scientific Q-Exactive
Bioinformatics
Quality control
Data analysis

Figure 3: iTRAQ workflow used for the identification of differentially expressed proteins.

RESULTS

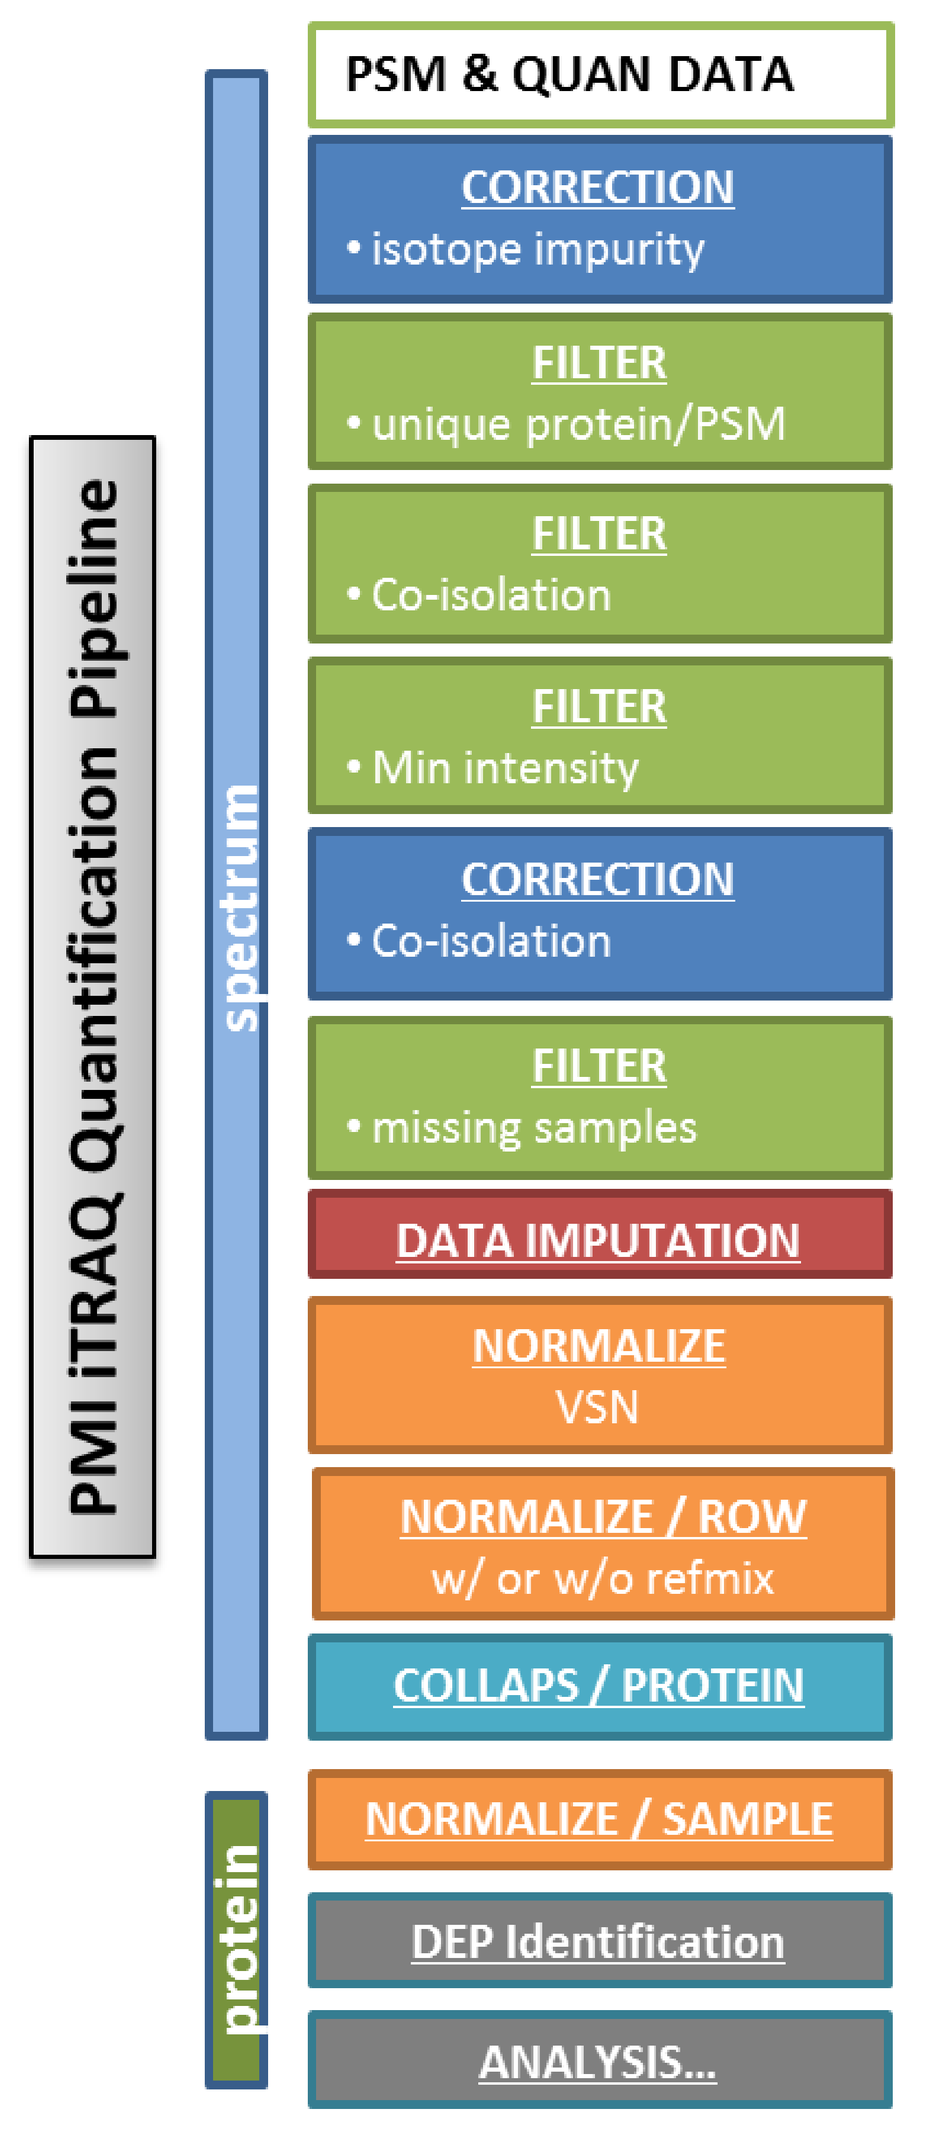
PMI iTRAQ Quantification Pipeline
spectrum
protein
PSM & QUAN DATA
CORRECTION
isotope impurity
FILTER
unique protein/PSM
FILTER
Co-isolation
FILTER
Min intensity
CORRECTION
Co-isolation
FILTER
missing samples
DATA IMPUTATION
NORMALIZE
VSN
NORMALIZE / ROW
w/ or w/o refmix
COLLAPS / PROTEIN
NORMALIZE / SAMPLE
DEP Identification
ANALYSIS...

Figure 4: Quantification pipeline for isobaric tag labeled samples. The pipeline is implemented in R. It allows full control over processing steps and settings. The workflow used allowed for sensitive and robust detection of differentially regulated proteins based on unique peptides only. DEP, differentially expressed protein; VSN, Variance stabilizing normalization (Arntzen et.al, 2011).

Table 1. Number of identified/quantified proteins for the different exposure durations. Proteins were assigned as quantified when quantitative information was present in ≥ 2/3 of the biological replicates analyzed.

Exposure duration (Month)	Identified proteins	Quantified proteins
1	4883	2615
3	4526	1980
5	4453	2298
7	4844	2220

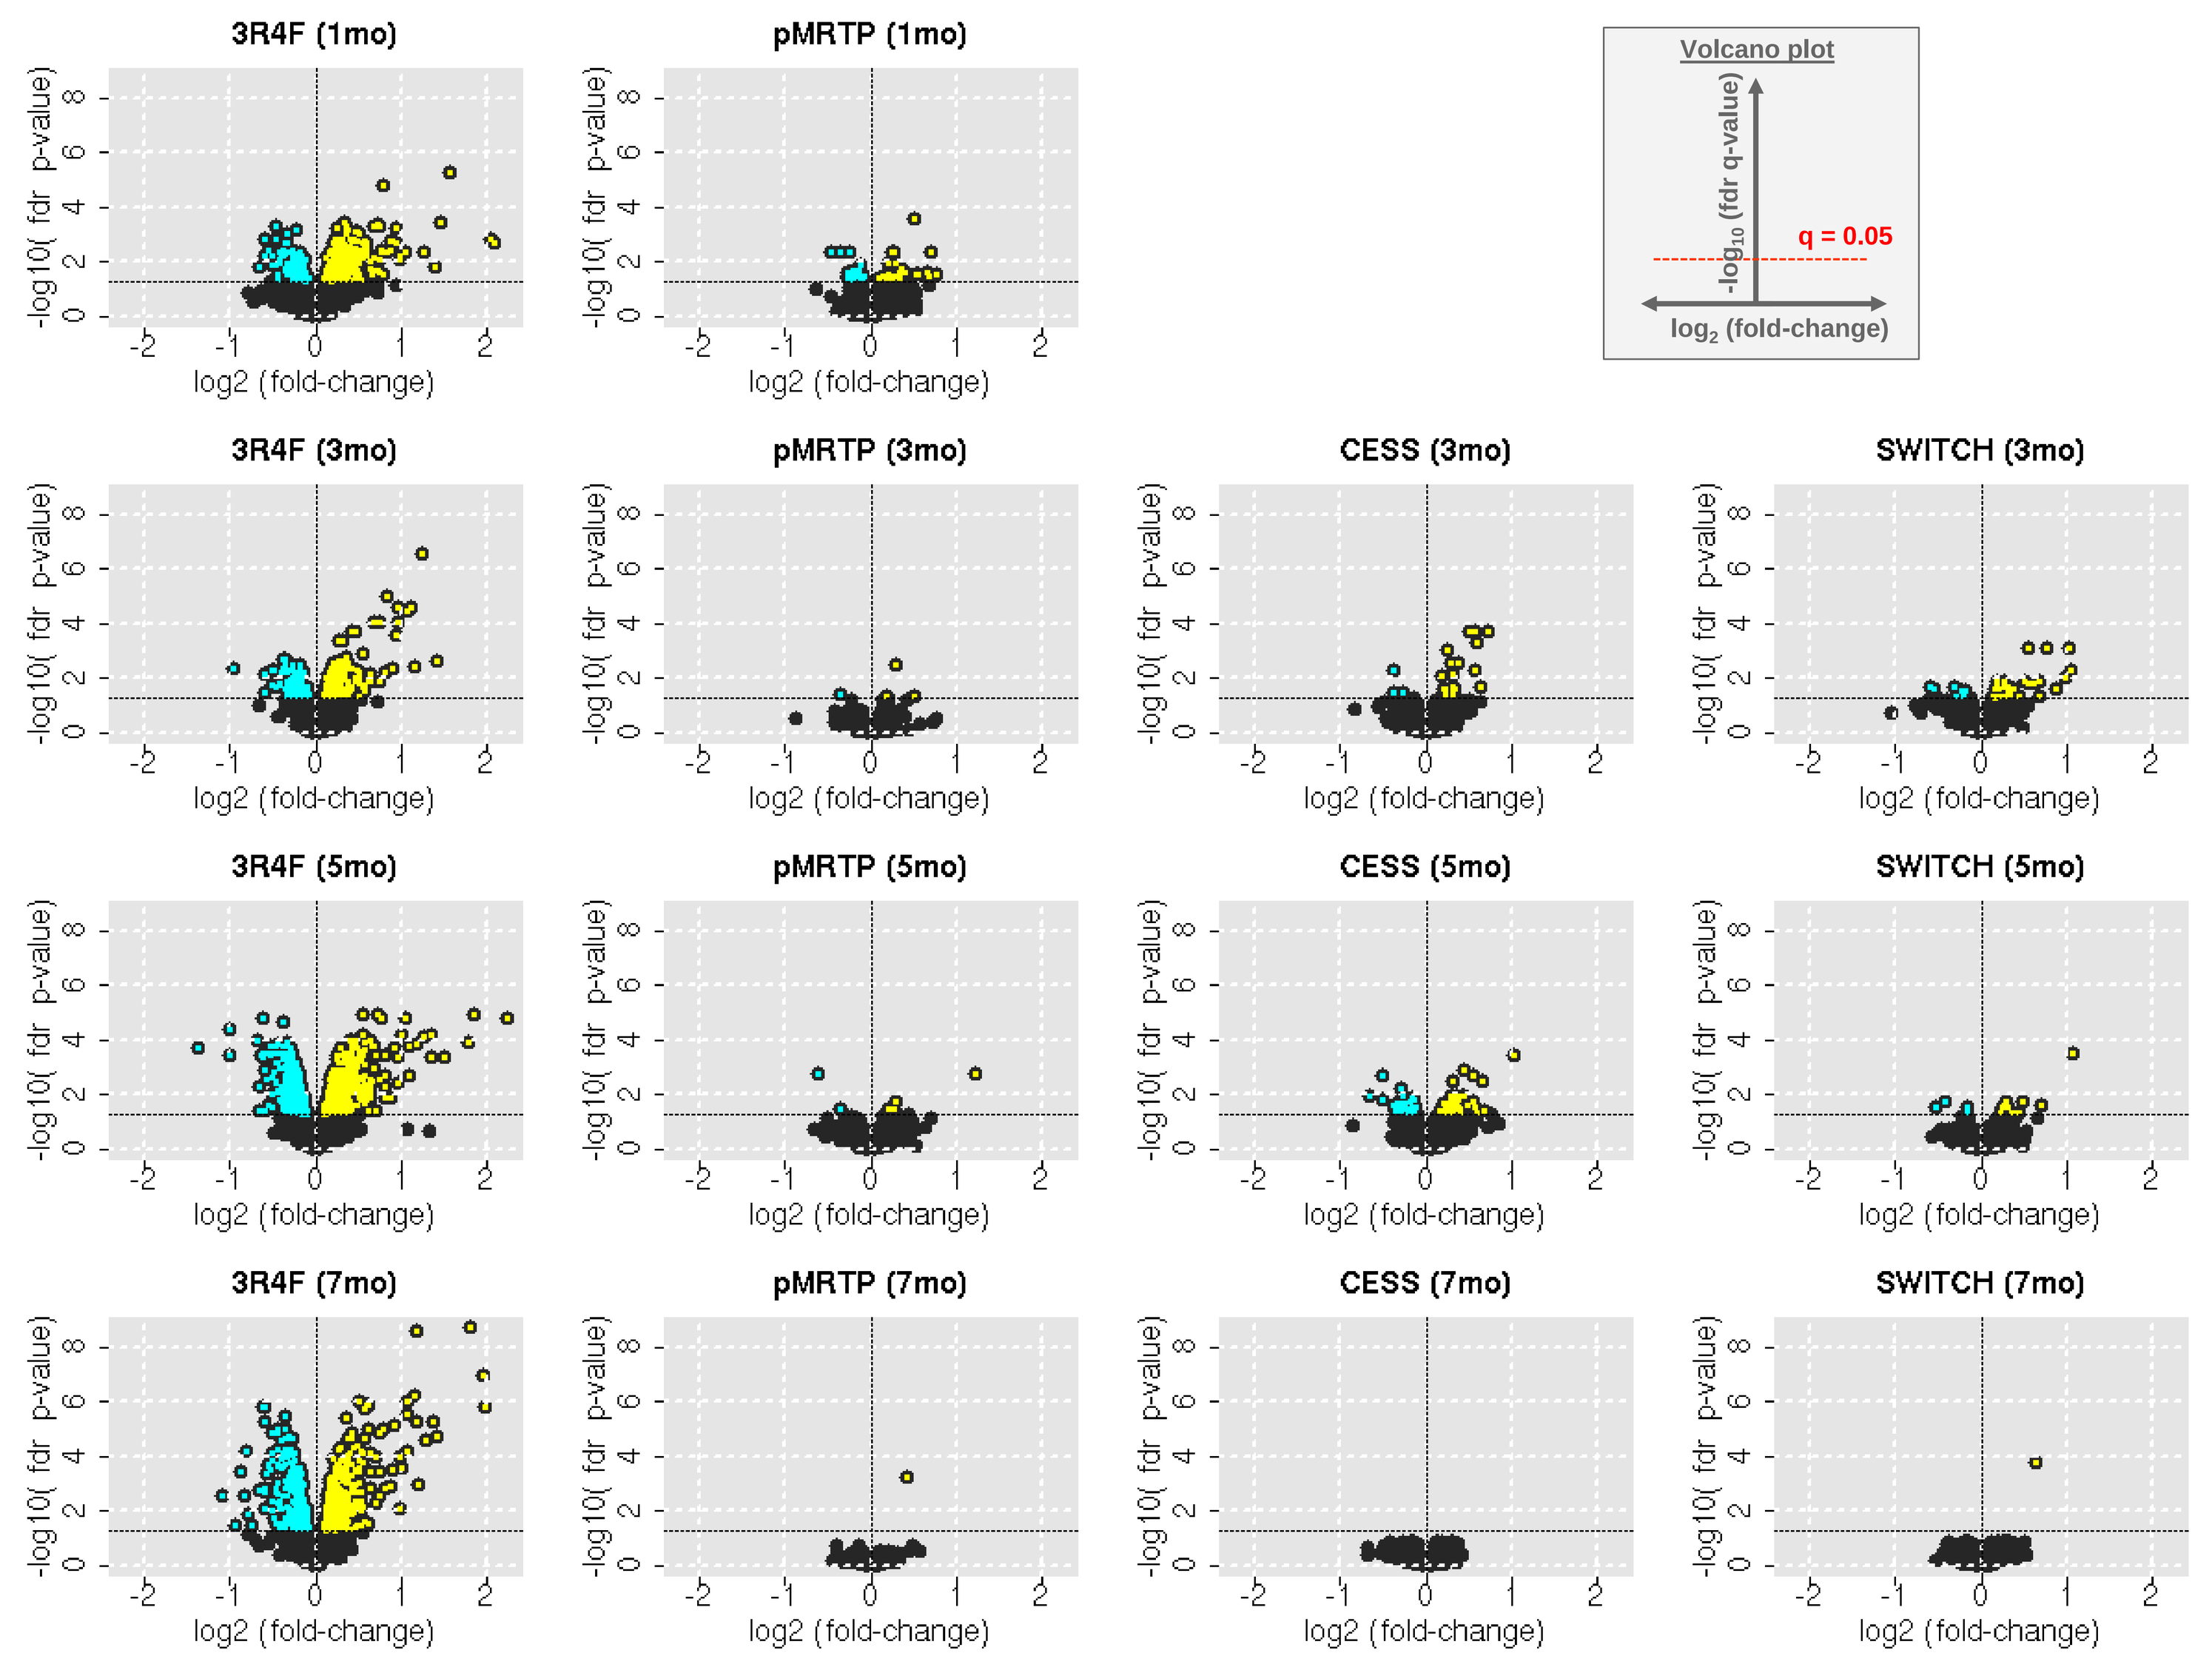
3R4F (1mo)
pMRTP (1mo)
Volcano plot
q = 0.05
3R4F (3mo)
pMRTP (3mo)
CESS (3mo)
SWITCH (3mo)
3R4F (5mo)
pMRTP (5mo)
CESS (5mo)
SWITCH (5mo)
3R4F (7mo)
pMRTP (7mo)
CESS (7mo)
SWITCH (7mo)
-log10(fdr p-value)
log2 (fold-change)

Figure 5: Differentially expressed proteins in mouse lung tissue exposed to cigarette smoke (3R4F), pMRTP, cessation (cess) or switching to pMRTP (switch).
Volcano plots show the $\log_2$ fold-change and the $-\log_{10}$ FDR-adjusted p-value for each quantified protein and each comparison. Each exposure condition is compared to the fresh-air exposed control. Differentially expressed proteins with a false discovery rate (FDR)-adjusted p-value < 0.05 are marked (up-regulated = yellow, down-regulated = cyan). Numbers in brackets indicate exposure duration. Mo, month.

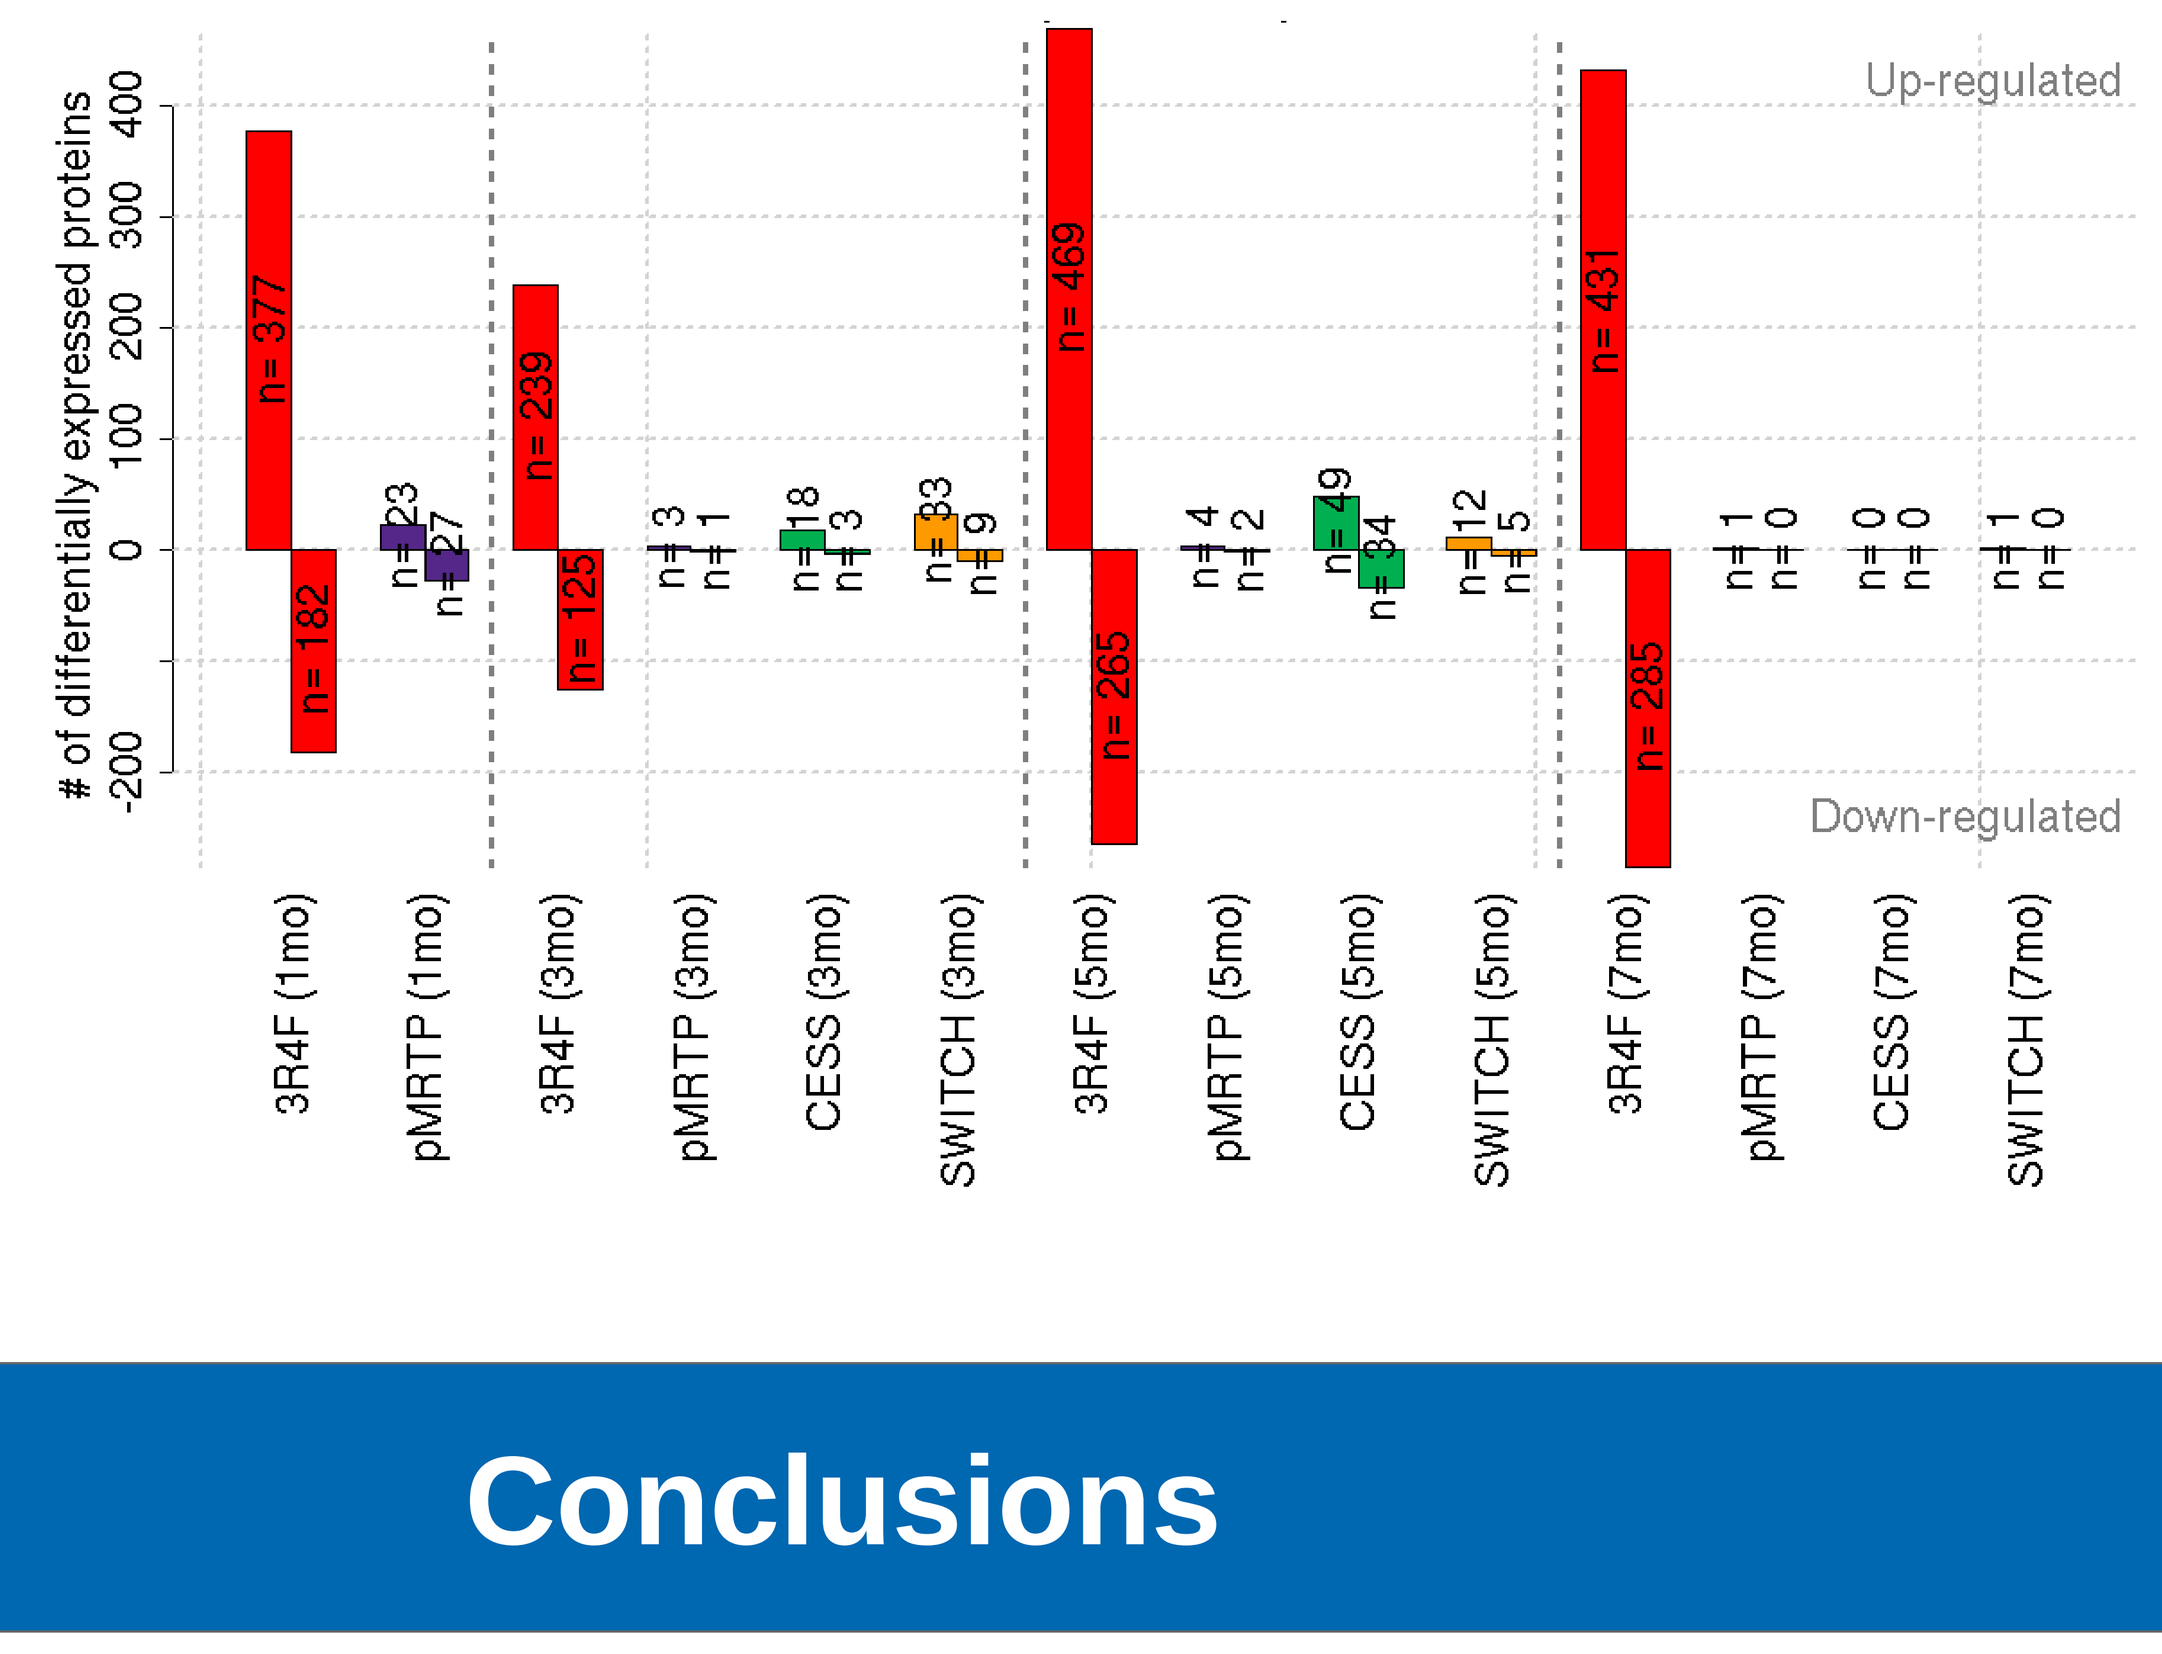
of differentially expressed proteins
Up-regulated
Down-regulated
n= 377
n= 182
n= 23
n= 27
n= 239
n= 125
n= 3
n= 1
n= 18
n= 3
n= 33
n= 9
n= 469
n= 265
n= 4
n= 2
n= 49
n= 34
n= 12
n= 5
n= 431
n= 285
n= 1
n= 0
n= 0
n= 0
n= 1
n= 0
3R4F (1mo)
pMRTP (1mo)
3R4F (3mo)
pMRTP (3mo)
CESS (3mo)
SWITCH (3mo)
3R4F (5mo)
pMRTP (5mo)
CESS (5mo)
SWITCH (5mo)
3R4F (7mo)
pMRTP (7mo)
CESS (7mo)
SWITCH (7mo)

Figure 6: Number of differentially expressed proteins in comparison to fresh-air control for each exposure treatment and exposure duration. Cess, cessation; mo, month; switch, switching to pMRTP . False discovery rate < 0.05.

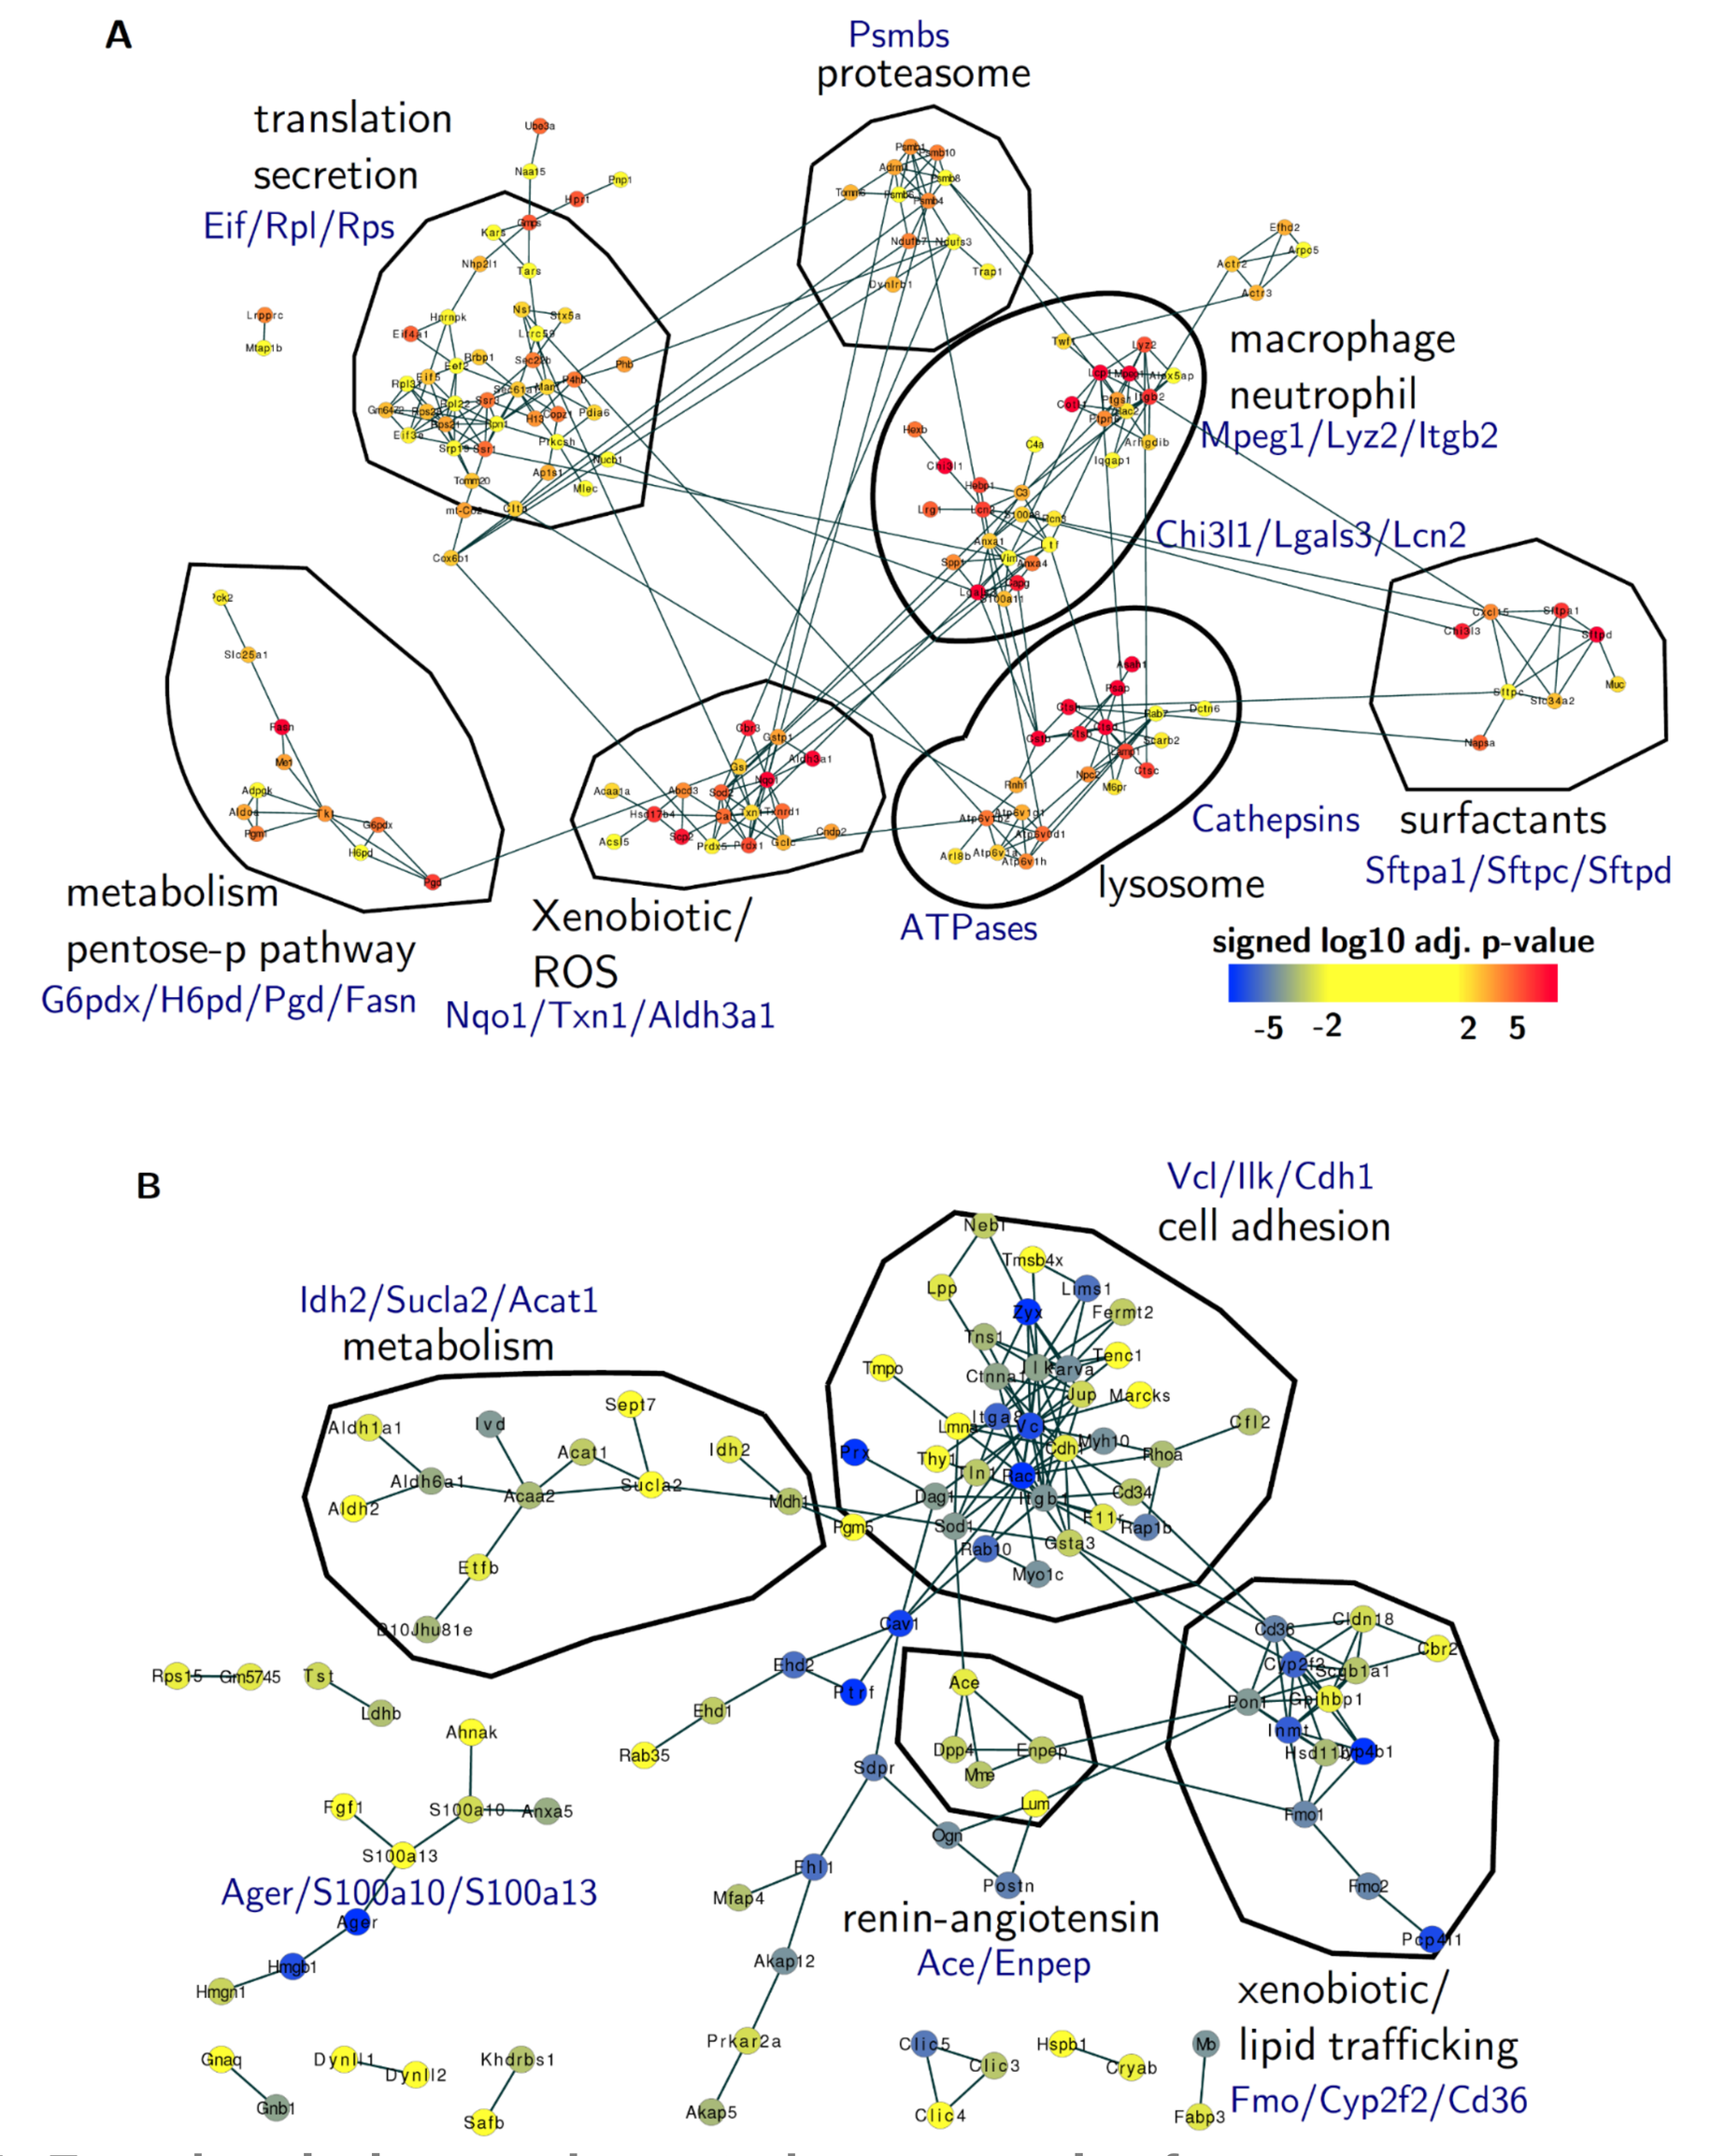
A
Psmbs
proteasome
translation
secretion
Eif/Rpl/Rps
macrophage
neutrophil
Mpeg1/Lyz2/Itgb2
Chi3l1/Lgals3/Lcn2
metabolism
pentose-p pathway
G6pdx/H6pd/Pgd/Fasn
Xenobiotic/
ROS
Nqo1/Txn1/Aldh3a1
ATPases
Cathepsins
lysosome
surfactants
Sftpa1/Sftpc/Sftpd
signed log10 adj. p-value
B
Vcl/Ilk/Cdh1
cell adhesion
Idh2/Sucla2/Acat1
metabolism
Ager/S100a10/S100a13
renin-angiotensin
Ace/Enpep
xenobiotic/
lipid trafficking
Fmo/Cyp2f2/Cd36

Figure 7: Functional clusters impacted as a result of exposure to conventional cigarette smoke. Functional protein networks of consistently up- (A) or down- (B) regulated proteins upon 5 and 7 months of 3R4F exposure highlight regulated biological functions. Each node in the network corresponds to a regulated protein, each edge to a functional protein link reported in the String database (Franceschini et al., 2013). Clusters of regulated protein functions were identified and functionally annotated. Node colors show the significance of regulation upon 5/7 months of cigarette smoke (3R4F) exposure (maximum signed $-\log_{10}$ adjusted p-value for both time points).

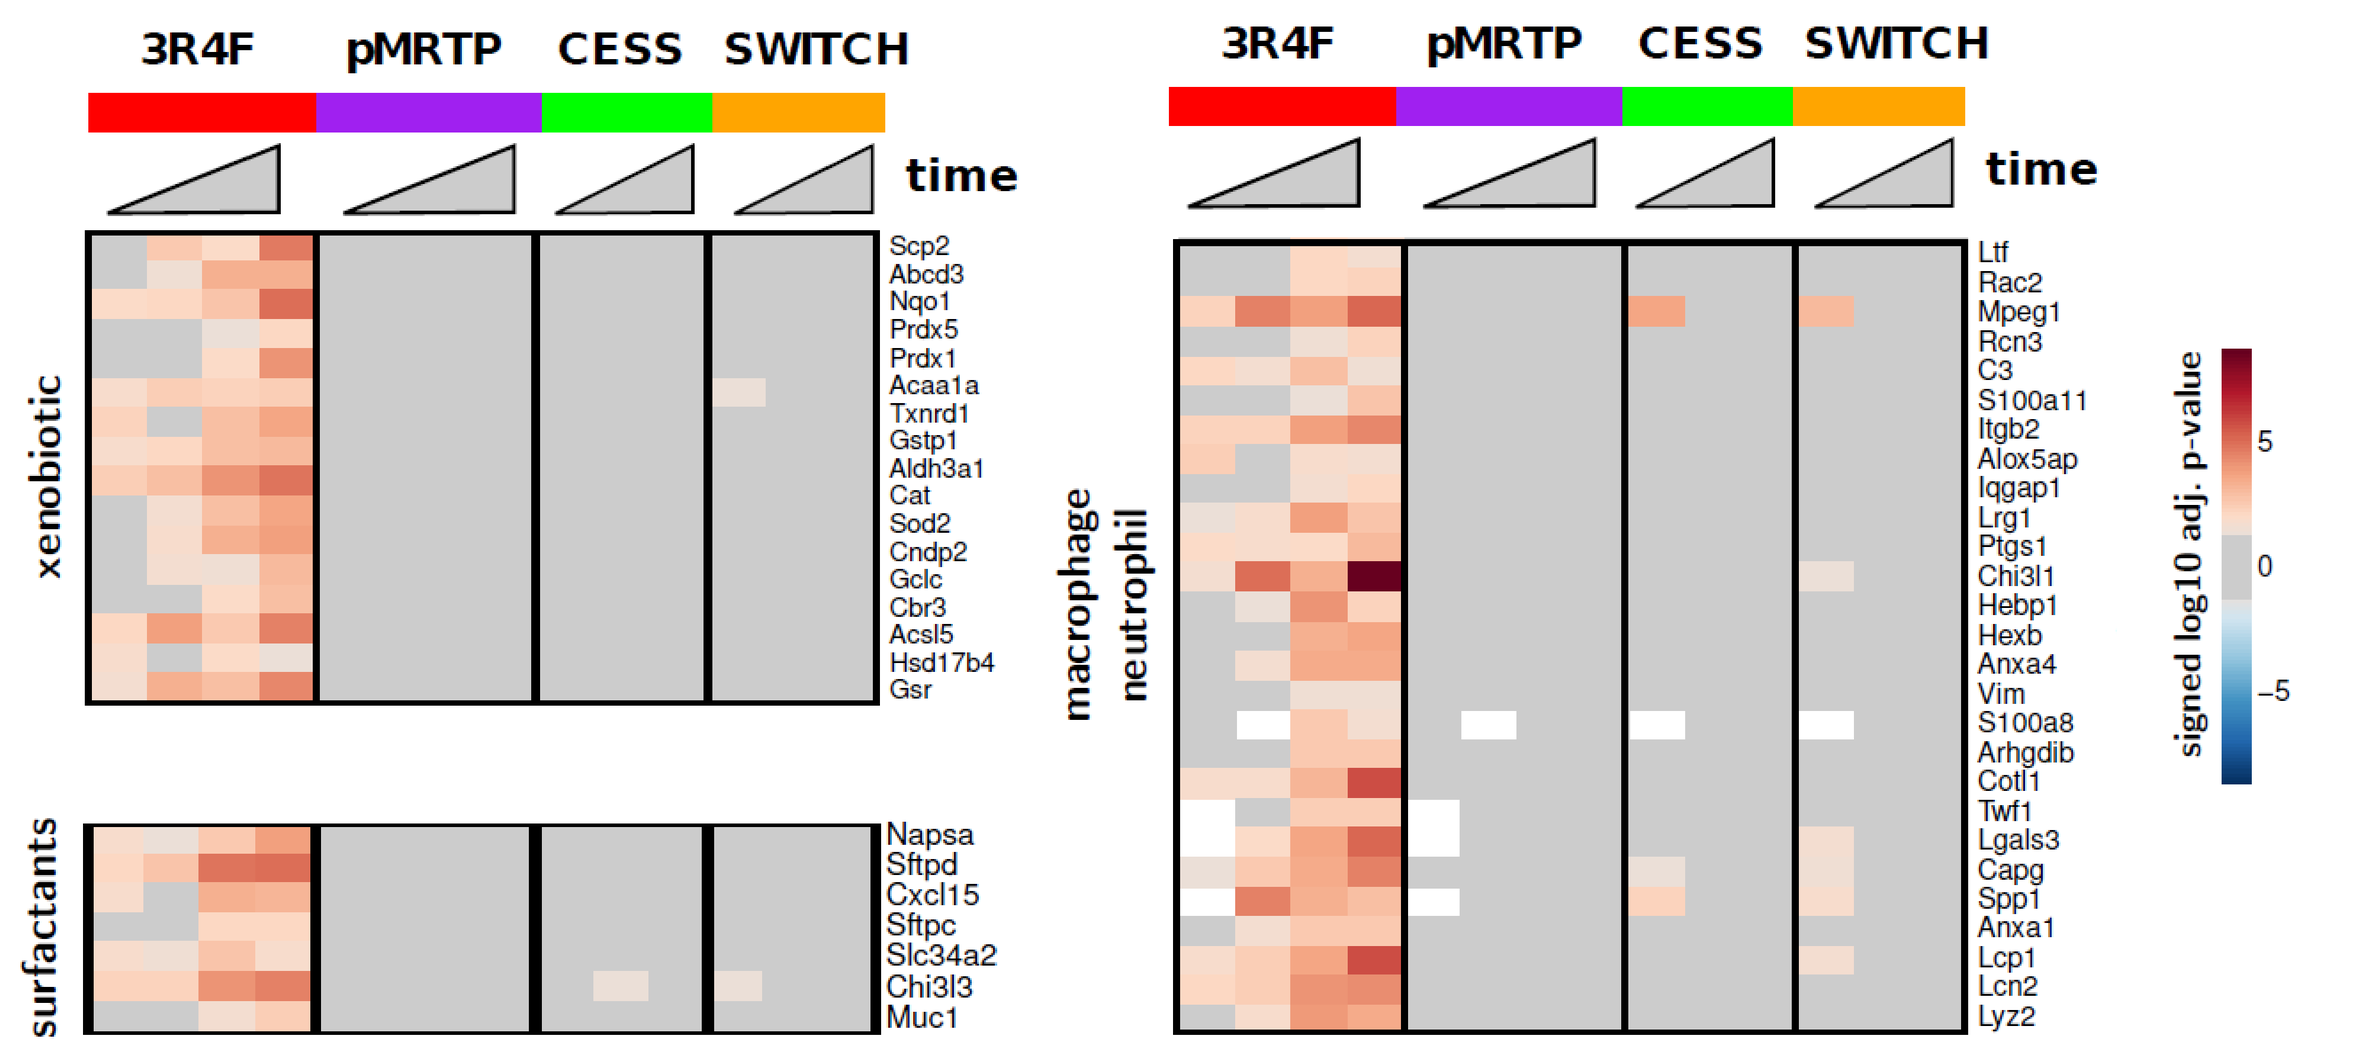
3R4F
pMRTP
CESS
SWITCH
time
xenobiotic
surfactants
macrophage neutrophil
signed log10 adj. p-value

Figure 8: Protein expression response for functional clusters impacted by cigarette smoke for all exposure durations. Heatmaps show the protein expression compared to fresh-air control for selected functional clusters from Fig. 7. The signed $-\log_{10}$ adjusted p-value is color-coded for each protein (row) and exposure condition (column) (grey = no significant deregulation, white = missing value). cess, cessation, switch, switching to pMRTP. Triangles below treatment groups indicate increasing exposure duration.

Conclusions

- An iTRAQ workflow involving in-house quality check and quantification pipelines was developed.
- Samples from mouse lungs led to a system response-profile involving more than two thousand identified proteins for each exposure duration (Table 1).
- Exposure to mainstream CS induced a system perturbation in a time dependent manner (Figure 5, 6 and 8) involving the regulation of biological functions such as xenobiotic metabolism, macrophage/neutrophil-related processes and surfactant homeostasis (Figure 7 and 8).
- Cessation and switching to a pMRTP aerosol resulted in a decrease of the number of differentially regulated proteins (Figure 6) and the reversal of the majority of the identified processes over time (Figure 8).
- The iTRAQ approach is a valuable technology for a systems-toxicology assessment of smoke-exposure response using *in vivo* models.

References

- Arntzen, M. O., et al. (2011), IsobariQ: software for isobaric quantitative proteomics using IPTL, iTRAQ, and TMT. J Proteome Res **10**(2): 913-920.
- Franceschini, A., et al. (2013), STRING v9.1: protein-protein interaction networks, with increased coverage and integration. Nucleic Acids Res **41** (Database issue): D808-815.
- Hoeng, J., et al. (2012), A network-based approach to quantifying the impact of biologically active substances. Drug Discovery Today **17**(9–10): 413-418.

PMI RESEARCH & DEVELOPMENT

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 13, **F**: +41 58 242 28 11, **W**: www.pmi.com

HUPO Conference, Madrid, Spain
October 5-8, 2014.