POSTER
90

Reduced effects of aqueous aerosol extract from THS2.2, a candidate Modified Risk Tobacco Product on the adhesion of monocytic cells to human coronary arterial endothelial cells

Carine Poussin, Alexandra Laurent, Manuel C Peitsch, Julia Hoeng, Hector De Leon

Philip Morris International R&D, Quai Jeanrenaud 5, CH 2000, Neuchâtel, Switzerland

ABSTRACT

Smoking is a major risk factor for the development of cardiovascular diseases (1,2). Modified risk tobacco products (MRTP) are designed to reduce smoking-related health risks. The present study aimed to evaluate the impact of THS2.2, a candidate heat-not-burn technology-based MRTP, compared with a reference cigarette (3R4F), on the adhesion of monocytic cells to human coronary arterial endothelial cells (HCAECs), a critical stage in atherosclerosis development, using a functional *in vitro* adhesion assay combined with systems toxicology. HCAECs were treated for 4h with conditioned media of human monocytic mono mac 6 (MM6) cells preincubated with low or high concentrations of aqueous extracts from THS2.2 aerosol or 3R4F smoke for 2h (indirect treatment), unconditioned media (direct treatment), or fresh aqueous extract (fresh direct treatment). Previous results showed that aqueous 3R4F smoke extract induced the adhesion of MM6 cells to HCAECs *via* distinct direct and indirect concentration-dependent mechanisms (3). Leveraging the same experimental and computational framework, significant reduced effects of aqueous THS2.2 aerosol extract on MM6 cell-HCAEC adhesion were measured, also supported by markedly diminished molecular changes such as gene expression in both endothelial and monocytic cells. A shift towards 10 and 20 times higher concentrations of aqueous THS2.2 aerosol extract was required to observe similar effects as the ones measured with 3R4F in both fresh direct and indirect exposure modes, respectively. In conclusion, our *in vitro* systems toxicology investigations revealed reduced effects of THS2.2, a candidate MRTP, on monocytic cell-endothelial cell adhesion compared with a reference cigarette.

MATERIALS & METHODS

1. Cell exposure to 3R4F or THS2.2 s/abPBS (aqueous smoke/aerosol extract)

Conditioned-and unconditioned-media preparation
MM6 cells (2 million/mL) were starved in MM6 starvation medium (RPMI1640 with 0.5% FBS) for 2h and, then exposed for 2h to 3R4F or THS2.2 sbPBS (or PBS). Medium without MM6 cells underwent identical incubation conditions. Both conditioned- and unconditioned-media were collected and frozen down.

Treatment of HCAECs
- Indirect (I) and Direct (D) treatments: 24h-starved (0.1% FBS instead of 2%) HCAECs were treated with thawed conditioned- and unconditioned-media for 4h.
- Fresh Direct (FD) treatment: 24h-starved HCAECs were exposed to freshly generated 3R4F or THS2.2 sbPBS (or PBS) for 4h.

HCAECs and MM6 lysates were collected and stored at -80°C for RNA extraction.

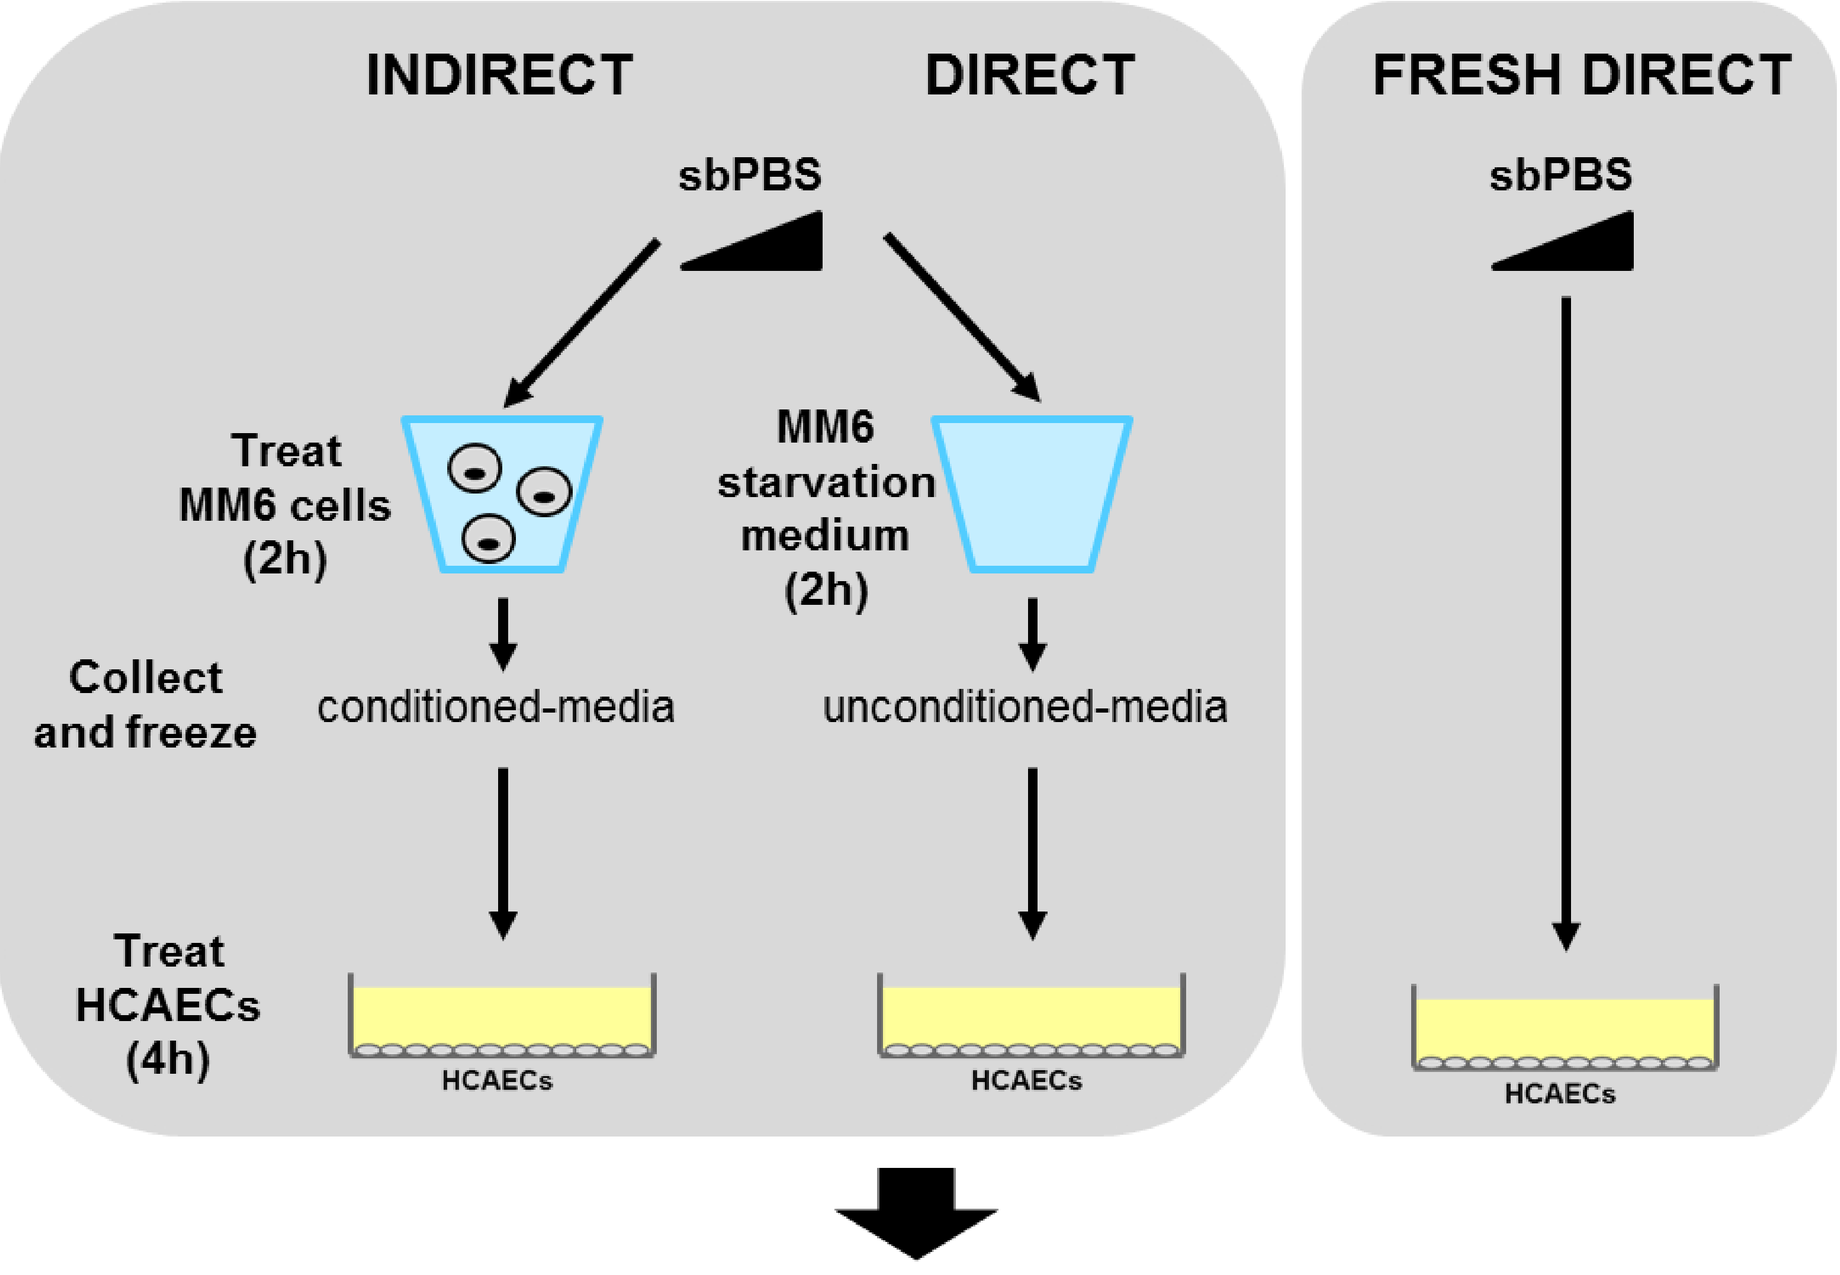
INDIRECT
DIRECT
FRESH DIRECT
sbPBS
sbPBS
Treat MM6 cells (2h)
MM6 starvation medium (2h)
Collect and freeze
conditioned-media
unconditioned-media
Treat HCAECs (4h)
HCAECs
HCAECs
HCAECs

➢ **Adhesion assay (functional endpoint)**
➢ **Transcriptomics (molecular endpoints)**

2. Adhesion Assay

Untreated MM6 cells and 4h-treated HCAECs were nuclear-stained for 15min with Draq5 and Hoechst fluorescent dyes, respectively and then incubated together for 45min. After cell fixing (formaldehyde 4%; 15min) and washing, remaining adherent MM6 cells and HCAECs were counted using a Cellomics ArrayScan instrument. The adhesion rate (AR) was calculated as follows: AR=(MM6 cell count/HCAECs cell count) X 100.

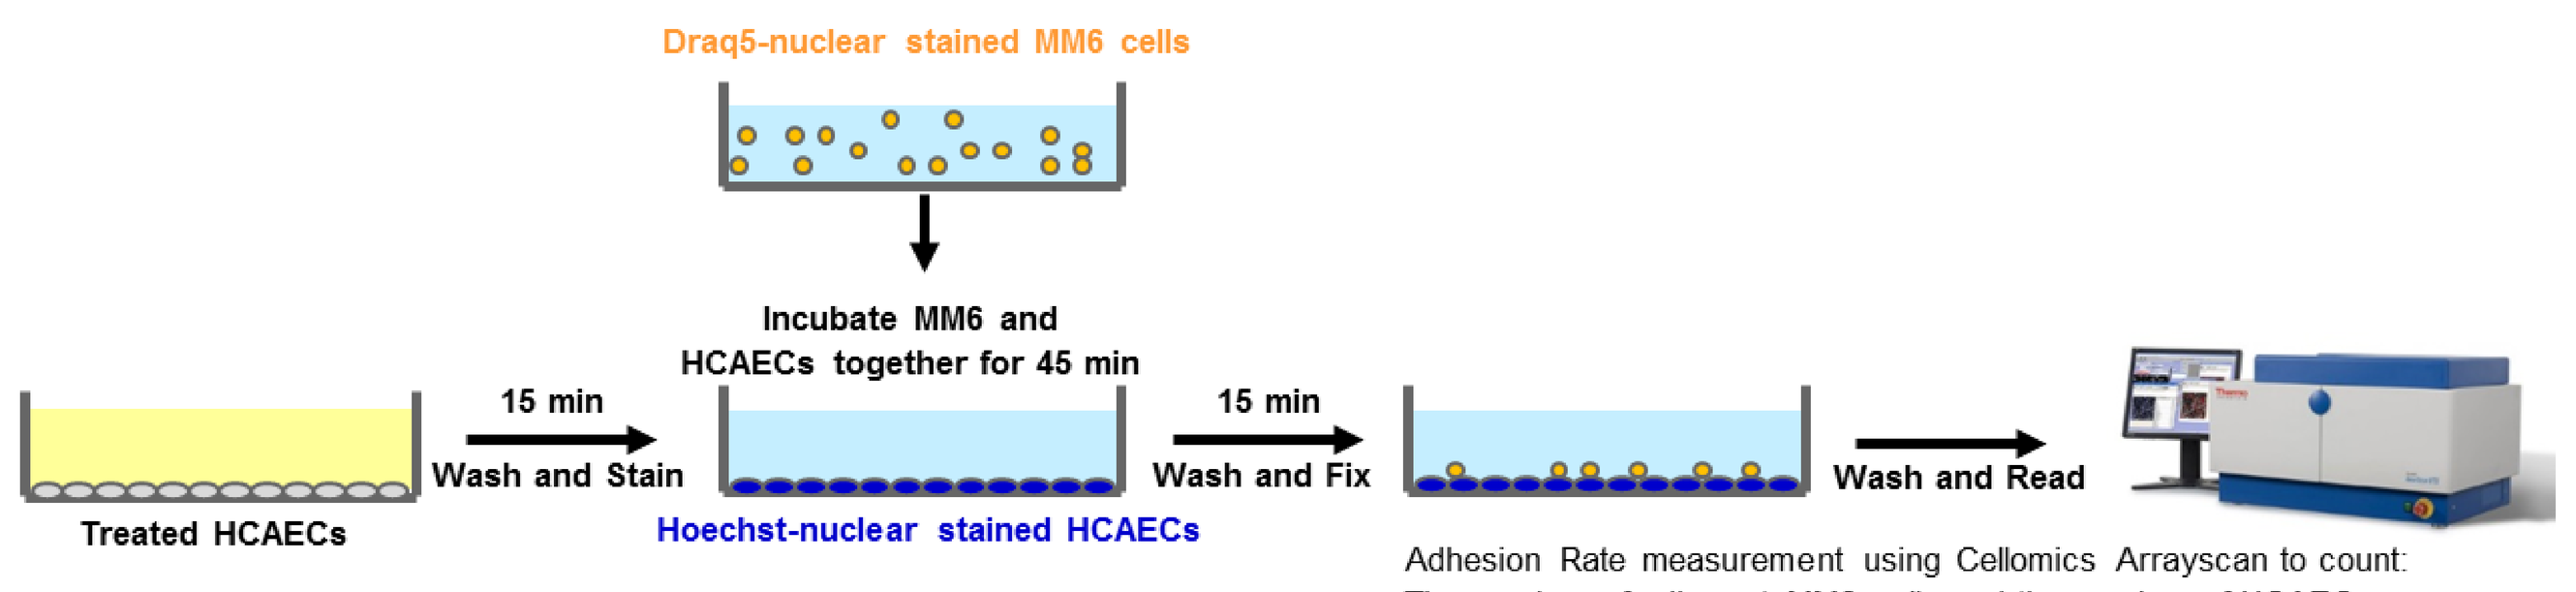
Draq5-nuclear stained MM6 cells
Incubate MM6 and HCAECs together for 45 min
15 min
Wash and Stain
15 min
Wash and Fix
Wash and Read
Treated HCAECs
Hoechst-nuclear stained HCAECs
Adhesion Rate measurement using Cellomics Arrayscan to count:
The number of adherent MM6 cells and the number of HCAECs

3. Other endpoints

Cell viability: MM6 and HCAECs viability was determined using a resazurin assay (Sigma-Aldrich).
Inflammatory mediators: A panel of 45-biomarkers was measured in conditioned-media (MM6 supernatants) by Myriad-RBM (Austin, TX, USA) using the Human InflammationMAP® v. 1.0 kit.
Transcriptomics: mRNA extracted from MM6 and HCAEC cell lysates was analysed in our transcriptomics laboratory using Affymetrix GeneChip® Human Genome U133 Plus 2.0 Array.

4. Computational analysis

Transcriptomics data were processed using GCRMA R package. Pairwise differential gene expression (systems response profile, SRP) analysis comparing 3R4F or THS2.2 sbPBS vs PBS for each exposure-condition type (I, D, FD) was computed with limma R package. Relative biological impact factor (RBIF) analysis was conducted for each SRP using two-layer causal network models representative of different biological processes (4,5). The approach enabled to identify significantly perturbed biological networks, and to quantify their respective contribution to the overall biological impact of the treatment on cells.

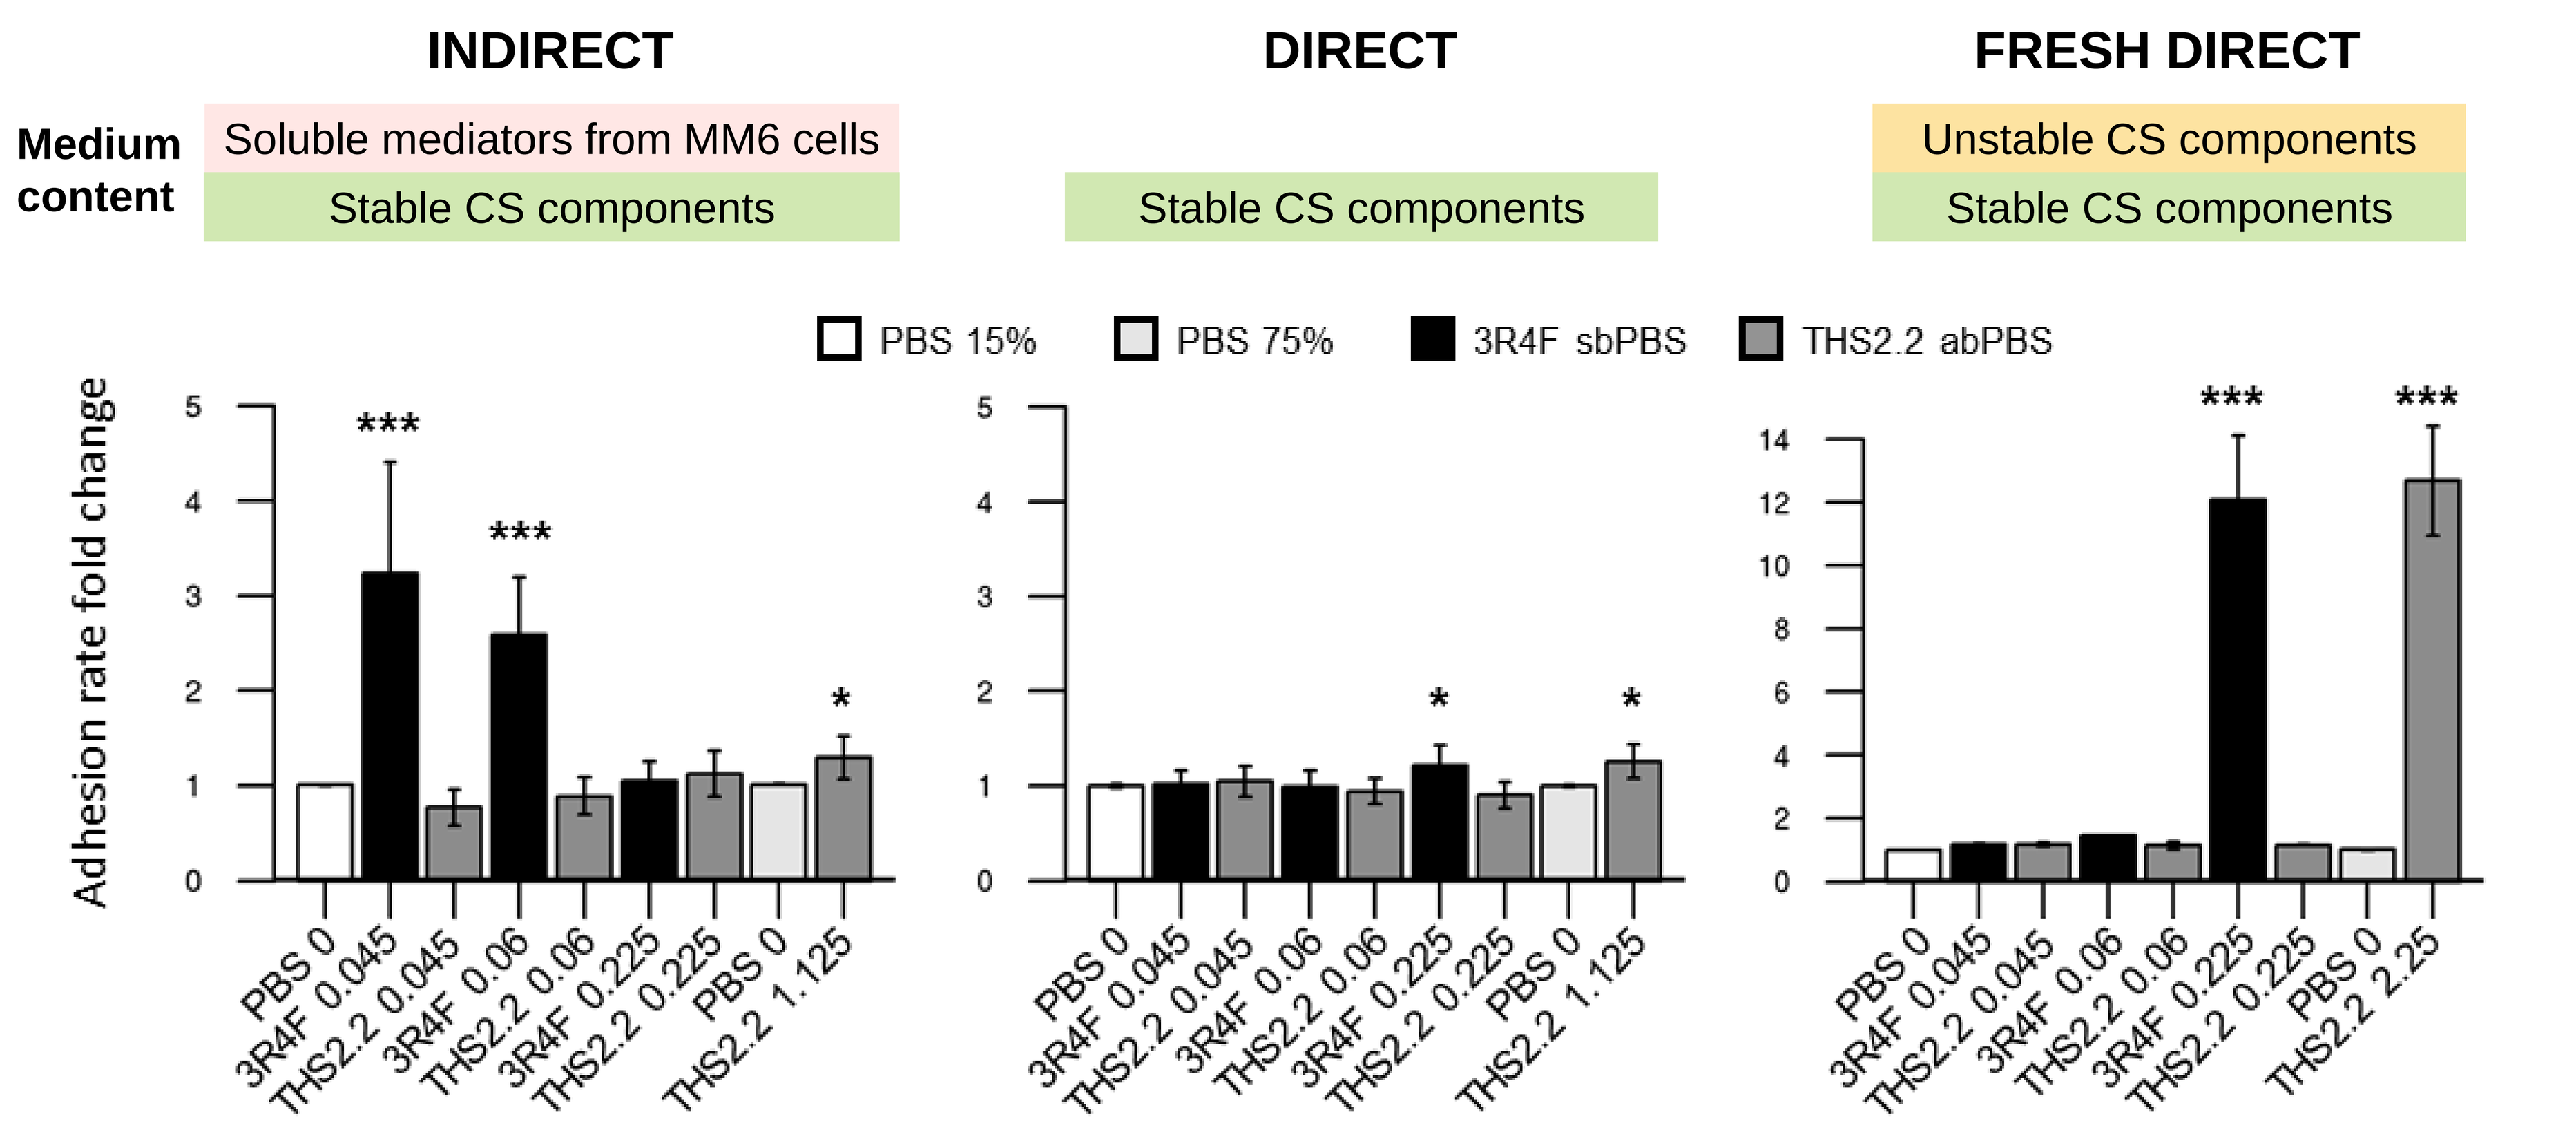
INDIRECT
DIRECT
FRESH DIRECT
Medium content
Soluble mediators from MM6 cells
Stable CS components
Stable CS components
Unstable CS components
Stable CS components
PBS 15%
PBS 75%
3R4F sbPBS
THS2.2 abPBS
Adhesion rate fold change

*
*
*

PBS 0
3R4F 0.045
THS2.2 0.045
3R4F 0.06
THS2.2 0.06
3R4F 0.225
THS2.2 0.225
PBS 0
THS2.2 1.125
THS2.2 2.25

*Figure 1: Effects of THS2.2 abPBS and 3R4F sbPBS on the adhesion of MM6 cells to HCAECs following indirect, direct, and fresh direct treatments of HCAECs. Bar charts represent fold changes of the adhesion rate relative to respective vehicle controls. The adhesion rate reflects the number of adherent MM6 cells relative to the total number of HCAECs counted in the same well multiplied by 100. Data are presented as the mean ± SEM; N=2–3 independent experiments (n=3–6 replicates). *p≤0.05, ***p≤0.001 vs. 0 puffs/ml (PBS 15% or 75%).*

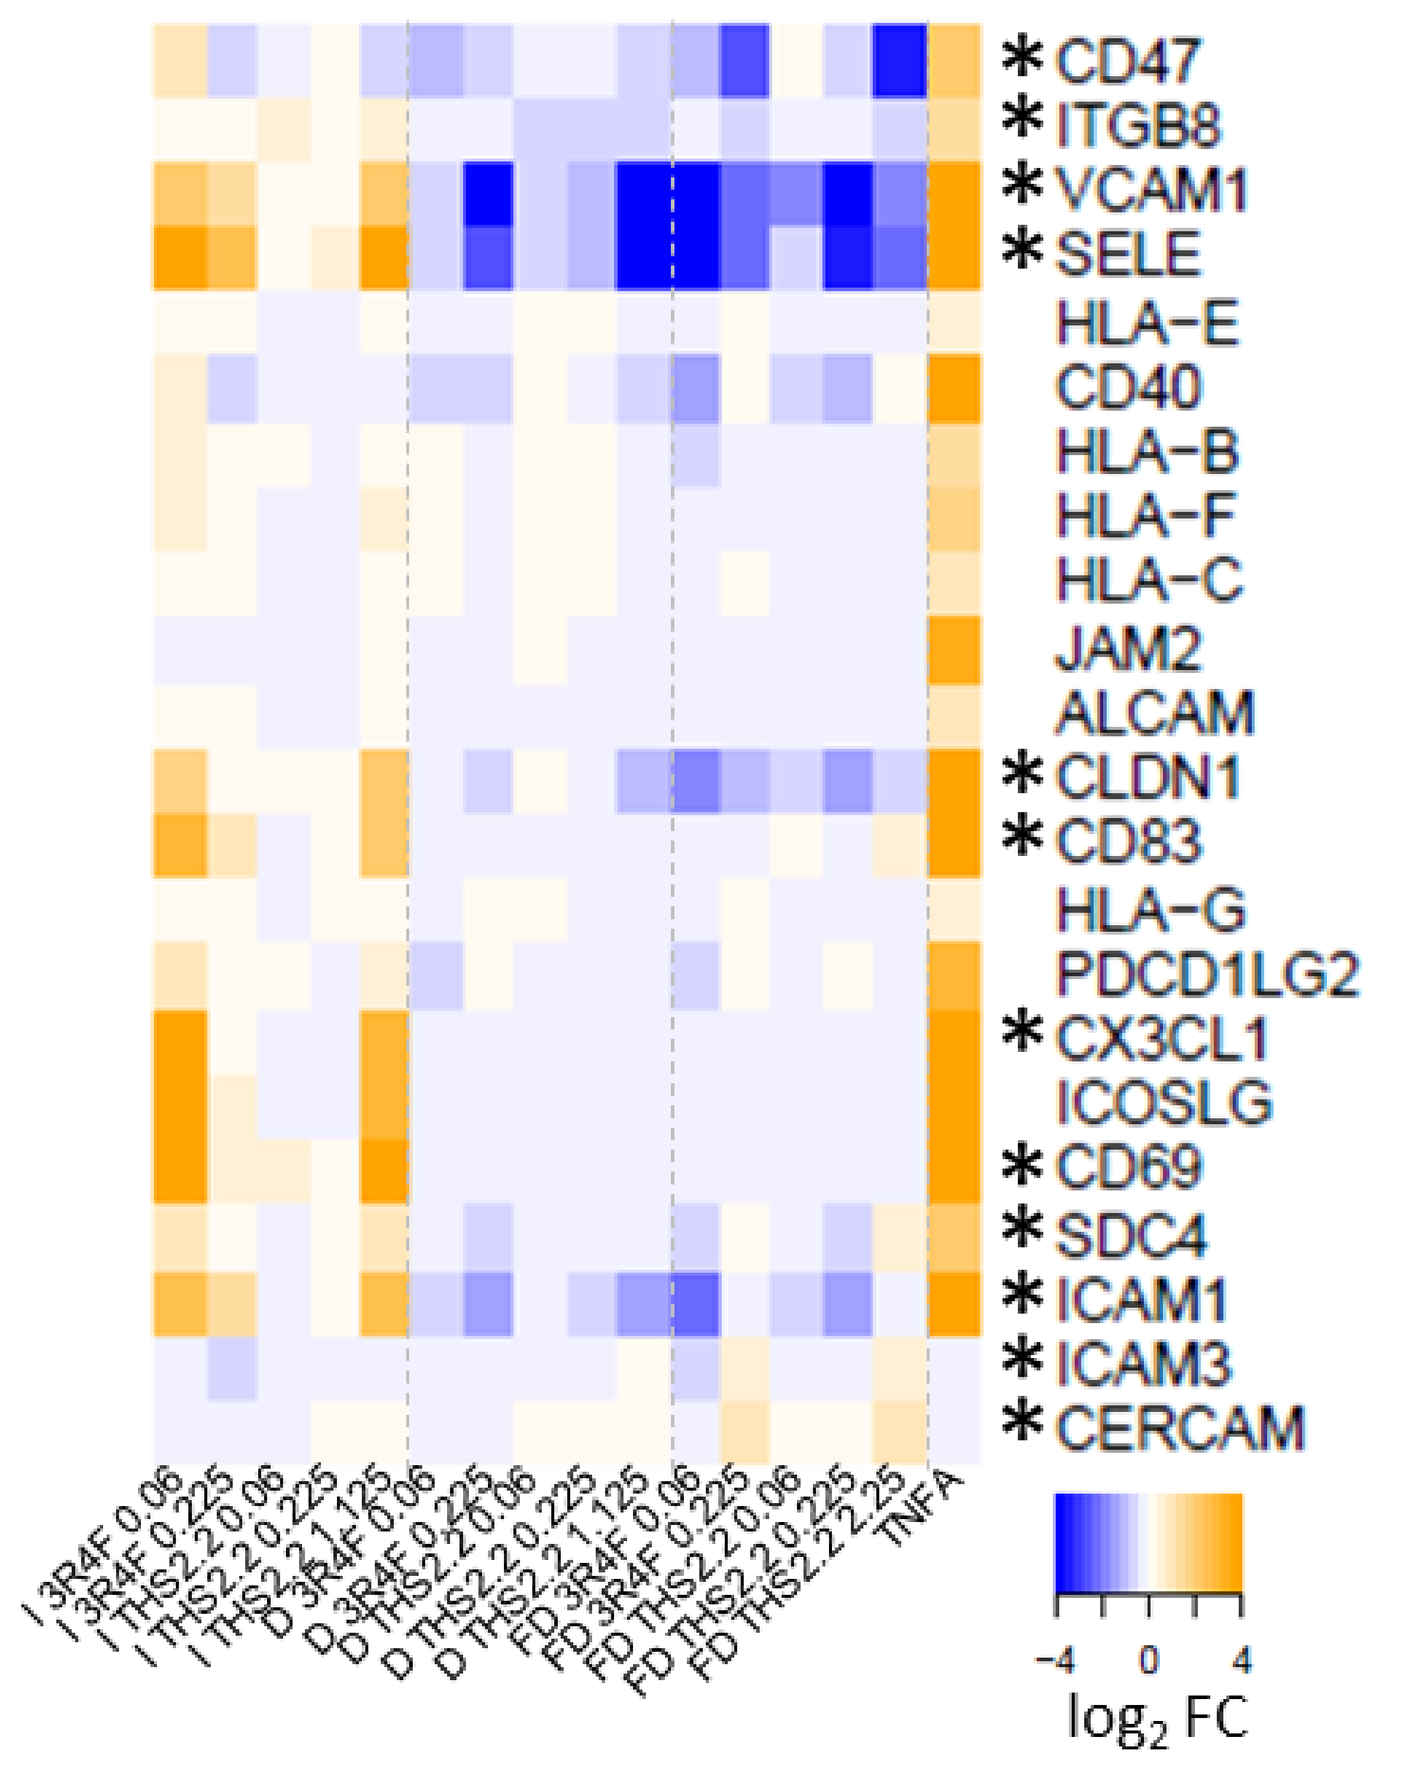
*CD47
*ITGB8
*VCAM1
*SELE
HLA-E
CD40
HLA-B
HLA-F
HLA-C
JAM2
ALCAM
*CLDN1
*CD83
HLA-G
PDCD1LG2
*CX3CL1
ICOSLG
*CD69
*SDC4
*ICAM1
*ICAM3
*CERCAM
TNFA
log2 FC

Figure 3: Heatmap of adhesion molecule encoded-gene expression fold changes (as log_2 scale) comparing the effect of THS2.2 abPBS or 3R4F sbPBS relative to PBS in different exposure-conditions (I: indirect, D: direct, FD: fresh direct).

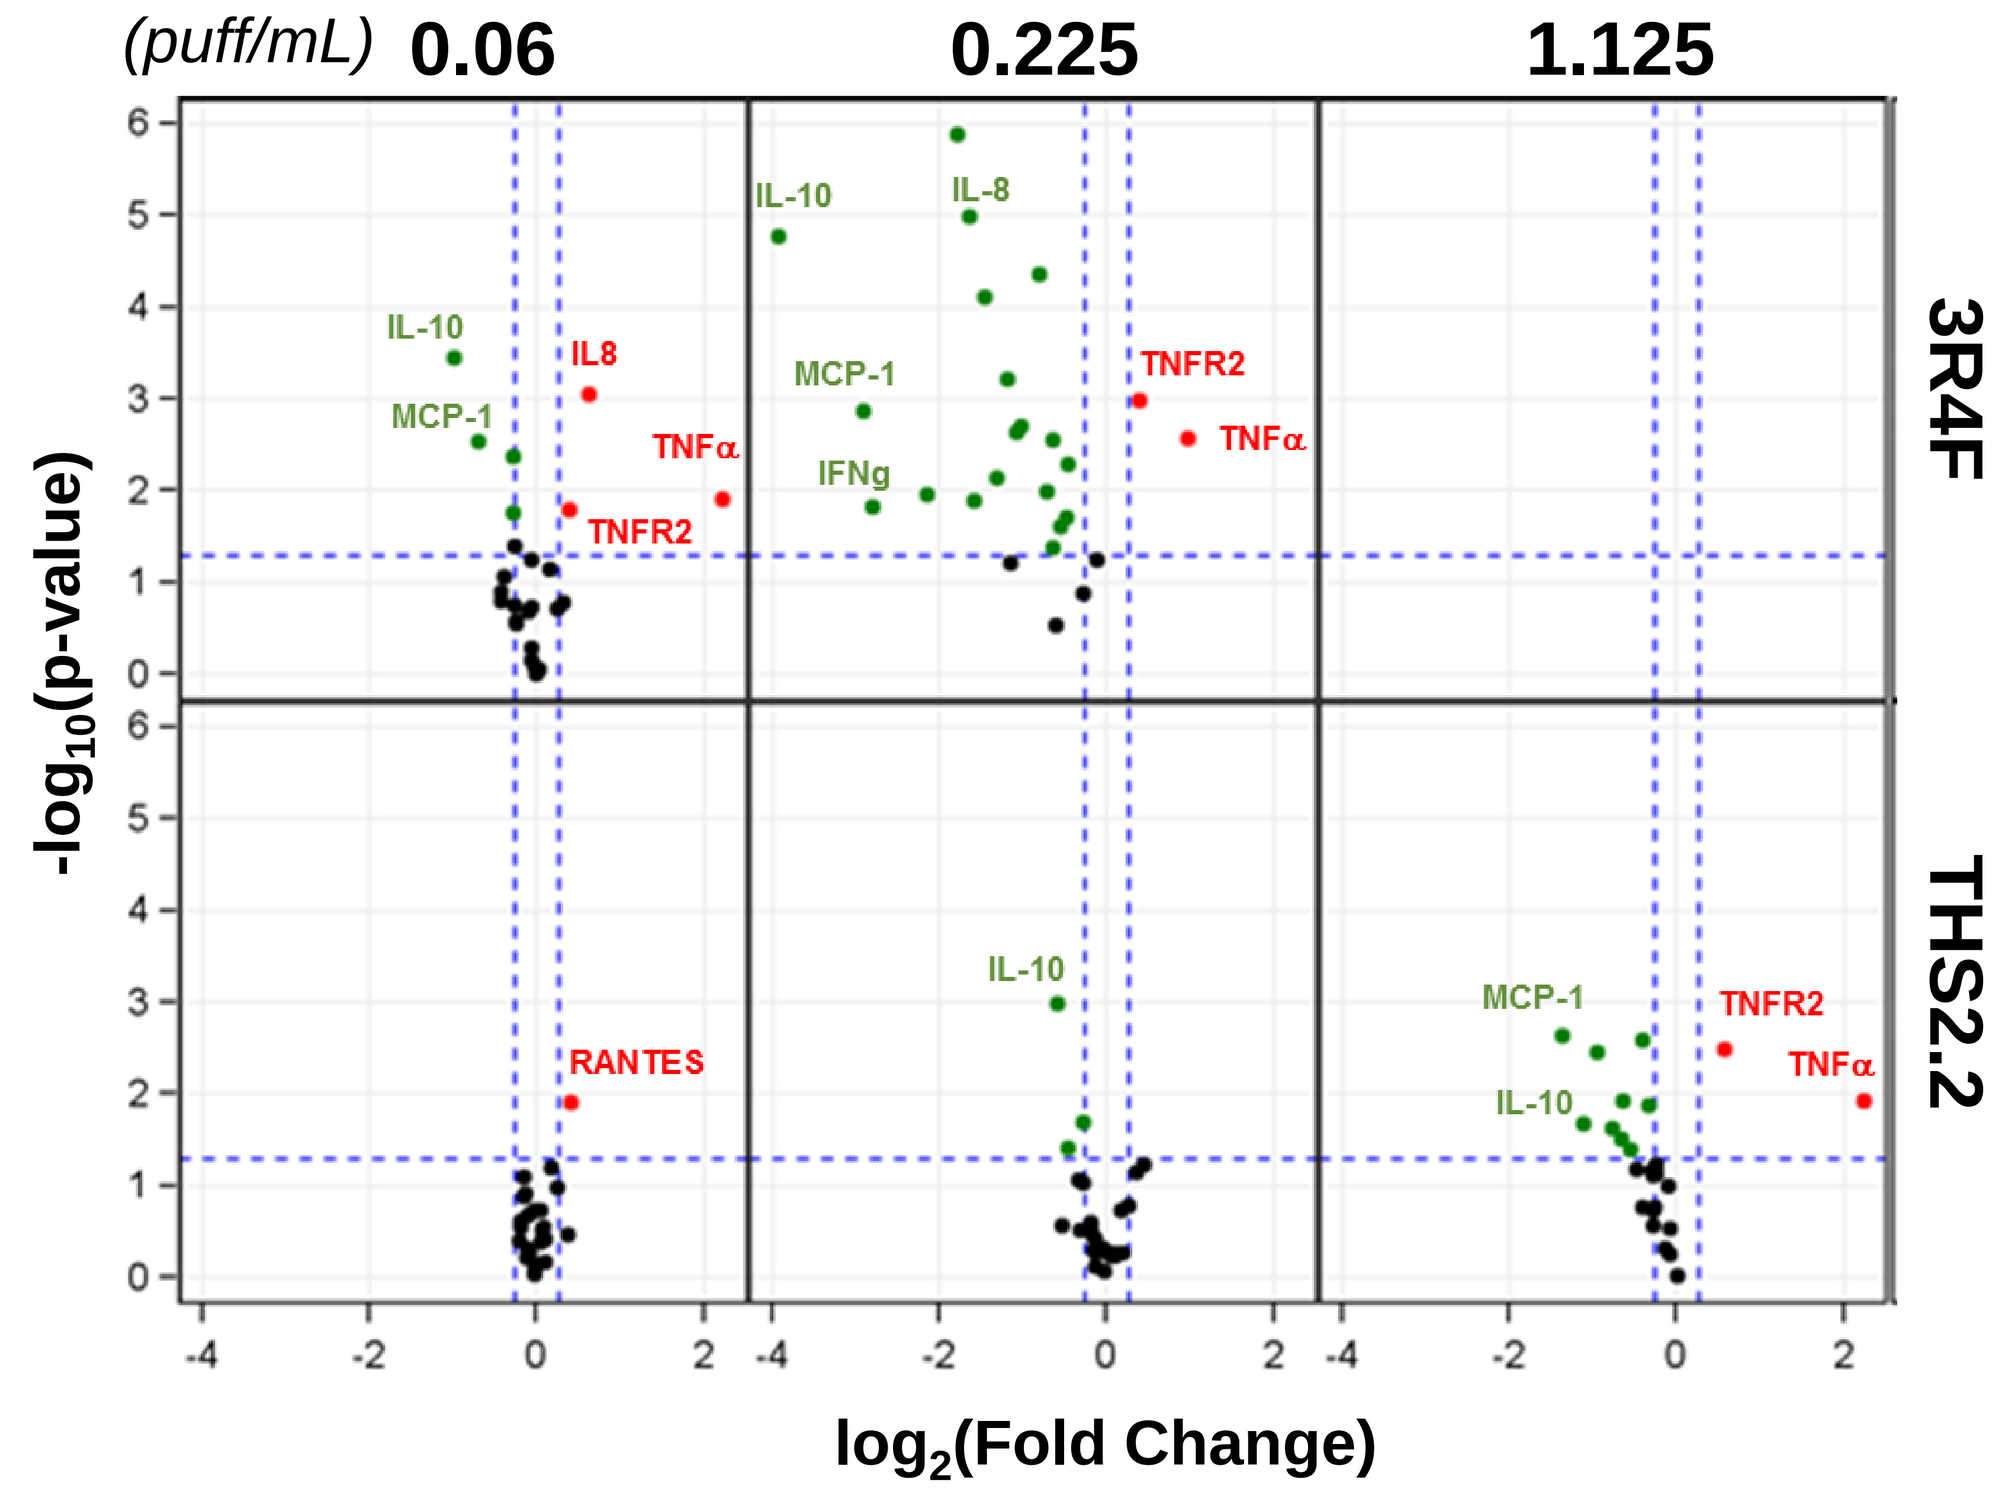
(puff/mL)
0.06
0.225
1.125
3R4F
THS2.2
IL-10
MCP-1
IL8
TNFα
TNFR2
IL-8
IFNg
RANTES
$-log_{10}$(p-value)
log_2(Fold Change)

Figure 4: Measure of 45 inflammatory markers released by MM6 cells exposed to THS2.2 abPBS or 3R4F sbPBS. Fold changes (FC) between PBS-control and 3R4F or THS2.2 sbPBS treatment (x-axis) and associated p-values (y-axis) are visualized on volcano plots. Vertical and horizontal dotted lines correspond to FC and p-value thresholds of 1.2 and 0.05, respectively. Up- and down-regulated inflammatory mediators are colored in red and green, respectively. FC correspond to the mean of 3 supernatant replicates.

CONCLUSIONS

3R4F aqueous cigarette smoke extract promoted the adhesion of monocytic cells to human coronary endothelial cells in indirect and fresh direct exposure conditions *via* distinct mechanisms:

- **In indirect: the effect is observed at a low concentration (0.06 puff/mL) and is mediated through the activation of an inflammatory response in HCAECs (e.g. increase of the expression of adhesion molecules) promoted by a paracrine effect of 3R4F-treated MM6-derived soluble mediators (e.g. TNFα) present in conditioned-media.**
- **In fresh direct: the effect is observed at a high concentration (0.225 puff/mL) that also induces some toxicity and is mediated through an yet unknown mechanism promoted in HCAECs by unstable CS compounds still present in freshly generated sbPBS, however, decayed in unconditioned-media obtained with the direct protocol.**

At the same concentrations, no significant adhesion of MM6 cells to HCAECs, little changes in HCAEC and MM6 gene expression and in inflammatory protein marker release by sbPBS-treated MM6 cells are observed with THS2.2.

The concentrations of THS2.2 required to be increased by ~10 and 20 times to observe similar effects at functional and molecular levels to the ones observed with 3R4F sbPBS using fresh direct and indirect exposure protocols, respectively.

RESULTS

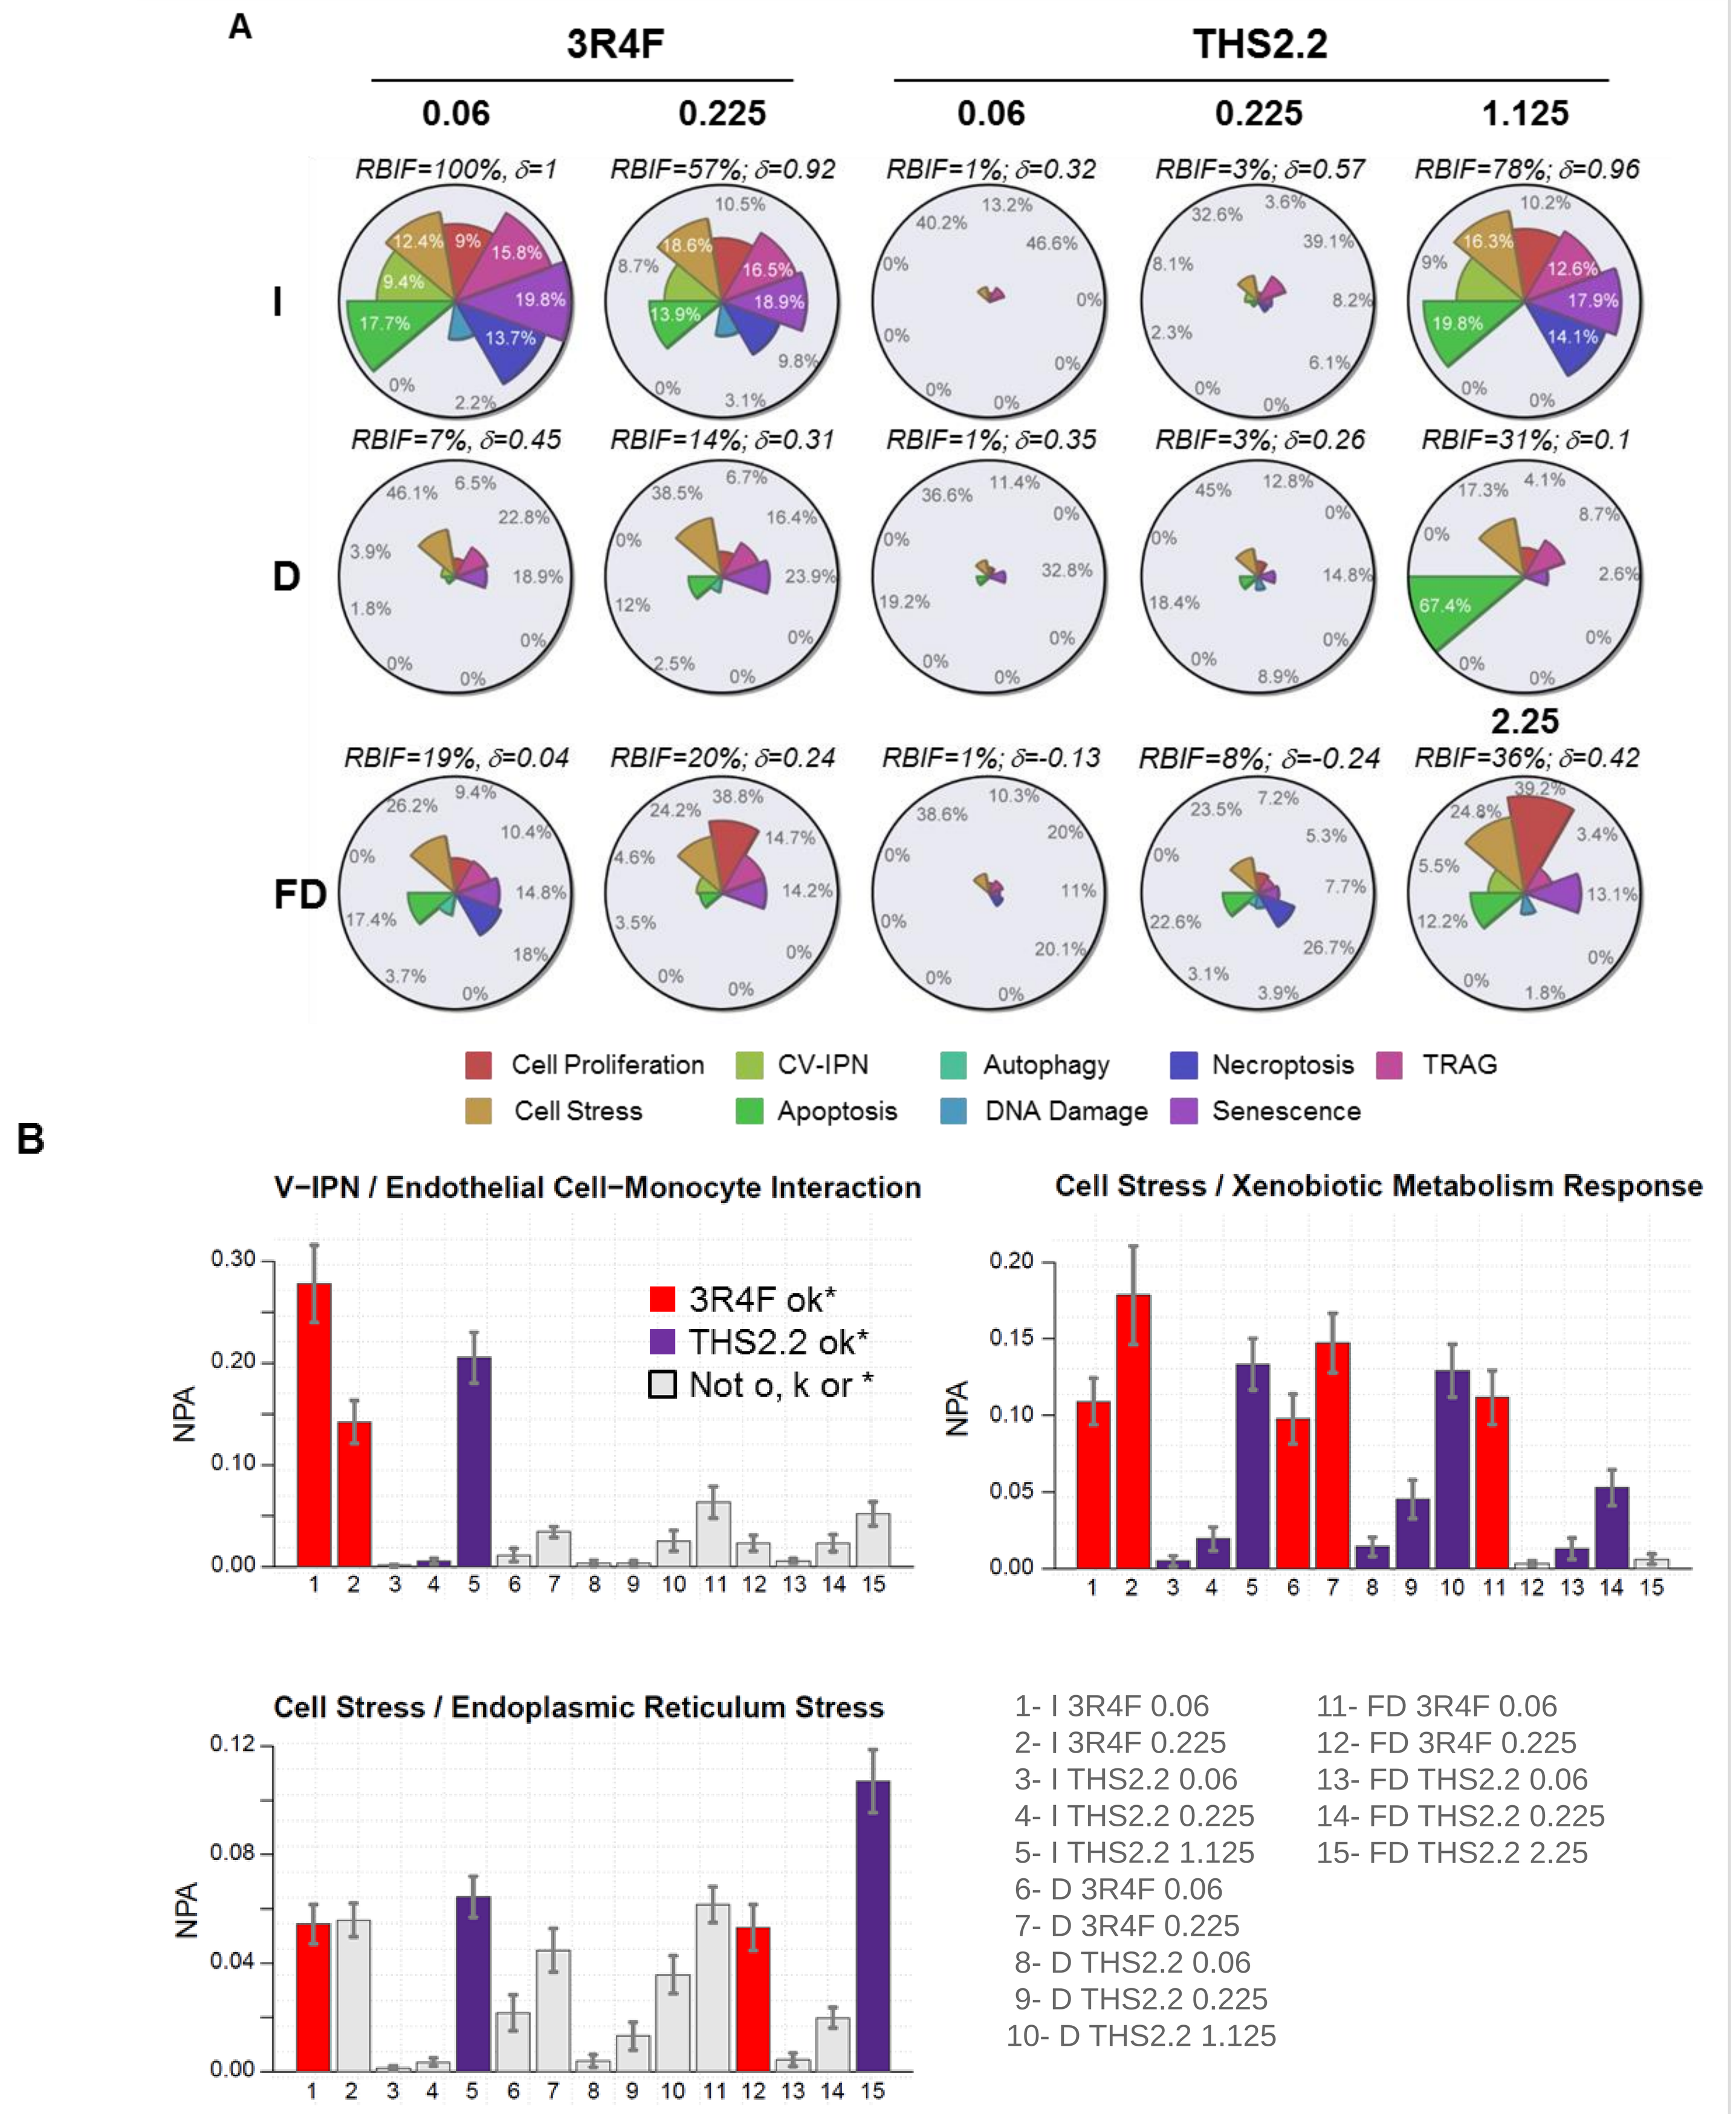
A
3R4F
THS2.2
0.06
0.225
0.06
0.225
1.125
2.25
I
D
FD
RBIF=100%, δ=1
RBIF=57%; δ=0.92
RBIF=1%; δ=0.32
RBIF=3%; δ=0.57
RBIF=78%; δ=0.96
RBIF=7%, δ=0.45
RBIF=14%; δ=0.31
RBIF=1%; δ=0.35
RBIF=3%; δ=0.26
RBIF=31%; δ=0.1
RBIF=19%, δ=0.04
RBIF=20%; δ=0.24
RBIF=1%; δ=-0.13
RBIF=8%; δ=-0.24
RBIF=36%; δ=0.42
Cell Proliferation
CV-IPN
Autophagy
Necroptosis
TRAG
Cell Stress
Apoptosis
DNA Damage
Senescence
B
V−IPN / Endothelial Cell−Monocyte Interaction
Cell Stress / Xenobiotic Metabolism Response
Cell Stress / Endoplasmic Reticulum Stress
NPA
3R4F ok*
THS2.2 ok*
Not o, k or *
1- I 3R4F 0.06
2- I 3R4F 0.225
3- I THS2.2 0.06
4- I THS2.2 0.225
5- I THS2.2 1.125
6- D 3R4F 0.06
7- D 3R4F 0.225
8- D THS2.2 0.06
9- D THS2.2 0.225
10- D THS2.2 1.125
11- FD 3R4F 0.06
12- FD 3R4F 0.225
13- FD THS2.2 0.06
14- FD THS2.2 0.225
15- FD THS2.2 2.25

Figure 2. Computed biological impact factor and network perturbation amplitudes in HCAECs following indirect, direct and fresh direct treatments with THS2.2 abPBS and 3R4F sbPBS. (A) The NPA computed for each network leveraging transcriptomics data was aggregated as a single value termed the biological impact factor (BIF) that quantifies the overall perturbation of the system modeled in causal networks. For each SRP, the contribution of significantly perturbed biological networks to the overall BIF is represented by colored surfaces proportionally covering circular plots and indicated as a percentage. These colored surfaces are also comparable across SRPs. The BIF associated with the SRP that induces the maximum network perturbation was automatically set to 100% as the reference (Relative (R)BIFref). RBIF related to other SRPs was expressed as a percentage of the RBIFref. The delta (d, [-1,1]) value reflects the degree to which the underlying biology modeled in the networks is similarly perturbed compared with the reference SRP. (B) Bar charts show NPA scores for three sub-networks of interest. The NPA is considered to be significant when both O and K statistics reach significance at the 0.05 level as indicated by o and k, respectively, when the significance with respect to the experimental variation is below a threshold p-value of 0.05 (). Error bars represent the 95% confidence interval. I: indirect, D: direct, FD: fresh direct.*

REFERENCES

(1) Libby et al. (2011). Nature 473 (7347): 317
(2) Tsiara et al. (2003). Angiology 54 (5): 507
(3) Poussin et al. (2015). Toxicological Sciences 147 (2):370
(4) Thomson et al. (2013). Toxicol Appl Pharmacol 272(3):863
(5) Martin et al. (2014). BMC Bioinformatics 15:238

Abbreviations. **CS**: cigarette smoke; **MRTP**: modified risk tobacco product; **s/abPBS**: smoke/aerosol-bubbled phosphate buffered saline; **ICAM-1**: intercellular adhesion molecule 1; **VCAM-1**: vascular cell adhesion protein 1; **MM6**: mono mac 6 cells; **HCAECs**: human coronary arterial endothelial cells; **SD**: standard deviation; **RBIF**: relative biological impact factor. **CV-IPN**: cardiovascular-inflammatory processes network; **TRAG**: tissue repair and angiogenesis network; **SRP**: systems response profile.

Acknowledgements. We would like to thank Didier Goedertier and Claudius Pak for generating fresh aqueous extracts from THS2.2 and 3R4F for each experiment; Karine Braumer and Dariusz Peric for mRNA extraction; Emmanuel Guedj and Rémi Dulize for generating transcriptomics data; Dr Grégory Vuillaume for the statistical analysis of xMAP data; Dr Sam Ansari for submitting transcriptomics data to the Array Express repository database; The Aerosol Chemistry Analytical Laboratory and Quentin Dutertre for carbonyl and nicotine concentration determination, respectively; Dr Stefan Frentzel for generous support of laboratory resources. Drs Walter Schlage and Filippo Zanetti for scientific review.

PMI RESEARCH & DEVELOPMENT

ACT 36th Annual Meeting
Red Rock Resort Summerlin, Nevada November 8–11, 2015

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 13, **F**: +41 58 242 28 11, **W**: www.pmi.com