Sharing in INTERVALS study details and data from the assessment of the impact of aerosol from the Carbon-Heated Tobacco Product (CHTP) 1.2, a potential modified risk tobacco product, on human organotypic gingival cultures.

Stéphanie Boué, Filippo Zanetti, Julia Hoeng, and Manuel C. Peitsch
PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

- **As part of a 21st century toxicology assessment framework, human gingival epithelia organotypic cultures were repeatedly exposed to nicotine-matching concentrations of Carbon-Heated Tobacco Product (CHTP) 1.2 aerosol or 3R4F cigarette smoke (CS), as well as a non-diluted (100%) CHTP 1.2 aerosol, and subsequently characterized.**
- **The results demonstrated the absence of cytotoxicity and reduction in pathophysiological alterations, toxicological marker proteins, and inflammatory mediators following exposure to CHTP 1.2 aerosol, as compared with 3R4F CS.**
- **Overall, repeated CHTP 1.2 aerosol exposures exerted a significantly lower impact than 3R4F CS on human gingival organotypic epithelial cultures.**
- **All results and data files, including transcriptomics results not included in this poster, are shared on the INTERVALS platform (www.intervals.science), developed to foster transparent sharing of assessment and mechanistic data relevant to tobacco harm reduction.**
- **INTERVALS allows scientists from academia and industry alike to share their results to contribute to tobacco harm reduction.**
- **The study can be accessed at:** www.intervals.science/studies/#/chtp-12-repeated-gingival-organotypic.

Background

- Cigarette smoke (CS) has been reported to increase predisposition to oral cancer and recognized as a risk factor for many conditions including periodontal diseases, gingivitis, and benign mucosal disorders (Johnson, 2000).
- Tobacco harm reduction through the development of Modified Risk Tobacco Products (MRTP), defined by the US FDA as “any tobacco product that is sold or distributed for use to reduce harm or the risk of tobacco-related disease associated with commercially marketed tobacco products”, provides a promising opportunity for adult smokers, who would otherwise continue cigarette smoking.
- Establishing a product’s potential as an MRTP requires scientific substantiation, including toxicity studies and measures of disease risk relative to that of cigarette smoking.
- Processes and/or platforms that encourage transparent sharing of data in a way that allows easy review and understanding should facilitate objective evaluation of the evidence (Combes 2015).
- INTERVALS™ (www.intervals.science) is an online platform developed by Philip Morris International R&D to enable independent data re-analysis and collaboration by sharing protocols, tools, and data from assessment studies (Boue 2017).
- In order to address reproducibility concerns, INTERVALS™ was built using the latest standards in data sharing and reproducible research to gather detailed information on the design and conduct of studies. This should enable an easy review of the methods and results as well as reuse of the data and generation of new hypotheses.
- The INTERVALS™ platform will allow researchers to find all relevant information on studies, detailed protocols, and, most importantly, interoperable data files in a single platform to allow independent re-analysis of key findings, meta-analyses, and efficient data reuse.
- INTERVALS™ will also encourage communication by enabling constructive feedback on studies and protocols and will foster education by providing reference texts and media on diverse topics relevant to tobacco harm reduction.
- Strengthened by community and peer-review features, INTERVALS™ aims to enable the necessary dialogue between industry, independent reviewers, the public health community, and regulatory agencies that can validate the harm reduction potential of these products.

INTERVALS
ADVANCING SCIENCE FOR A SMOKE-FREE WORLD

STUDIES

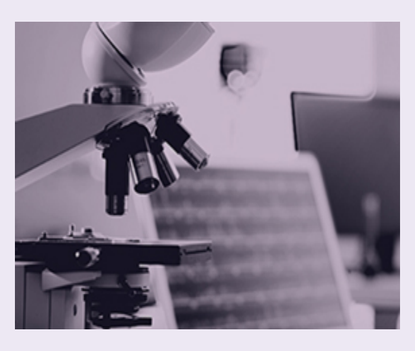

- Rationale and details of study design
- Detailed results
- Contact information of the reference scientist
- Links to publications
- Categorization

DATASETS

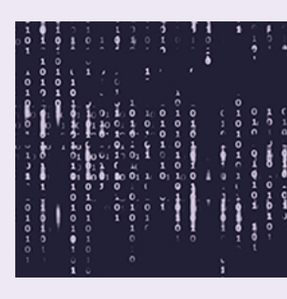

- Flexible data format incl. rich metadata
- Regulatory format (CDISC) & ISA-TAB accepted
- Analytics under development
- Categorization

PROTOCOLS

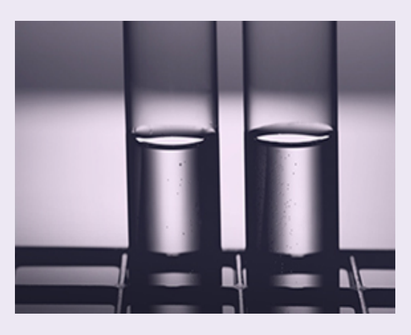

- Short description
- Step-by-step instructions
- Versioning
- Categorization

DISEASES & PATHWAYS

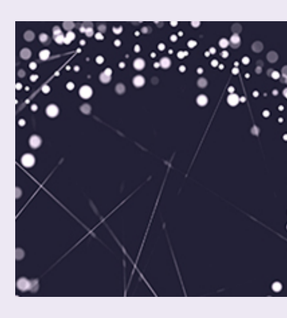

- Explore studies and results by pathway or disease of interest
- Gain mechanistic understanding to formulate new hypotheses

Study Design & Methods

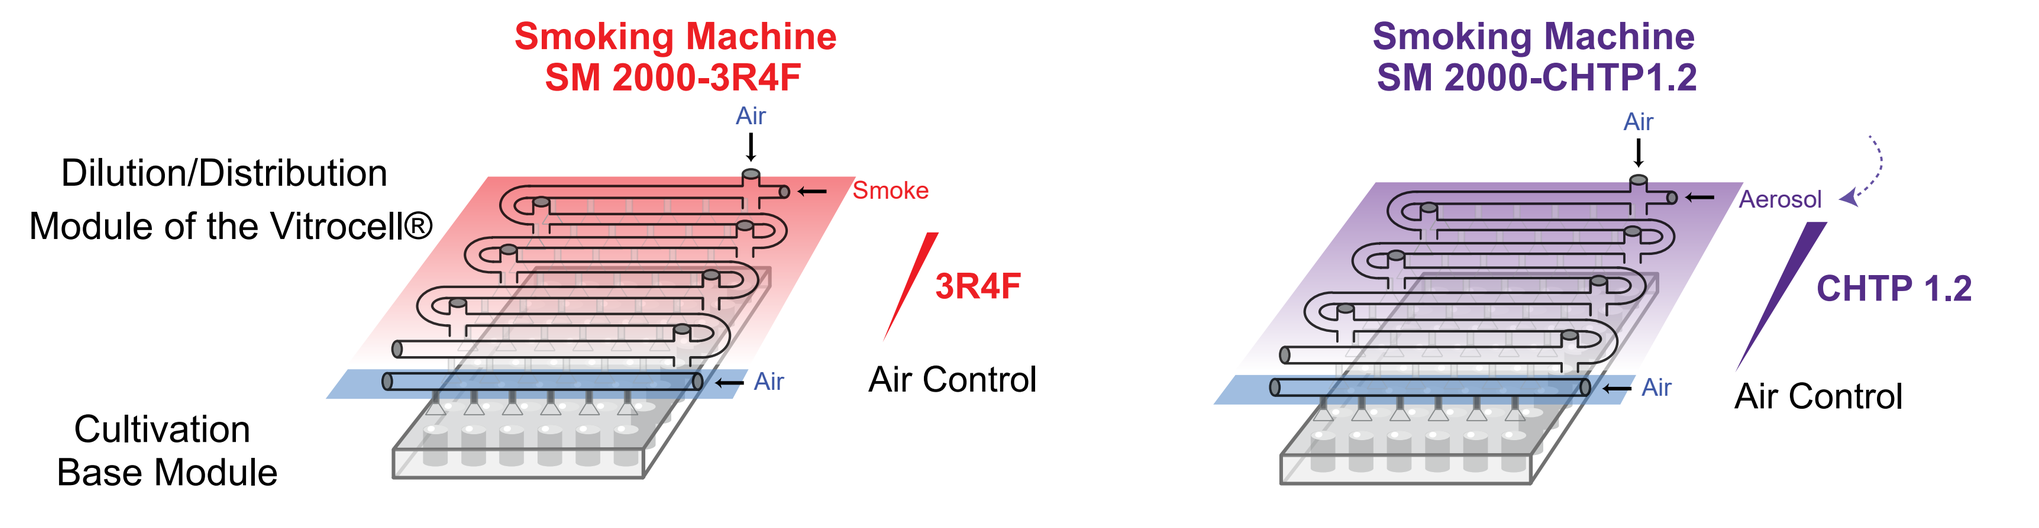
Smoking Machine SM 2000-3R4F
Dilution/Distribution Module of the Vitrocell®
Air
Smoke
3R4F
Air
Air Control
Cultivation Base Module
Smoking Machine SM 2000-CHTP1.2
Air
Aerosol
CHTP 1.2
Air
Air Control

Dose Range Finding Experiment

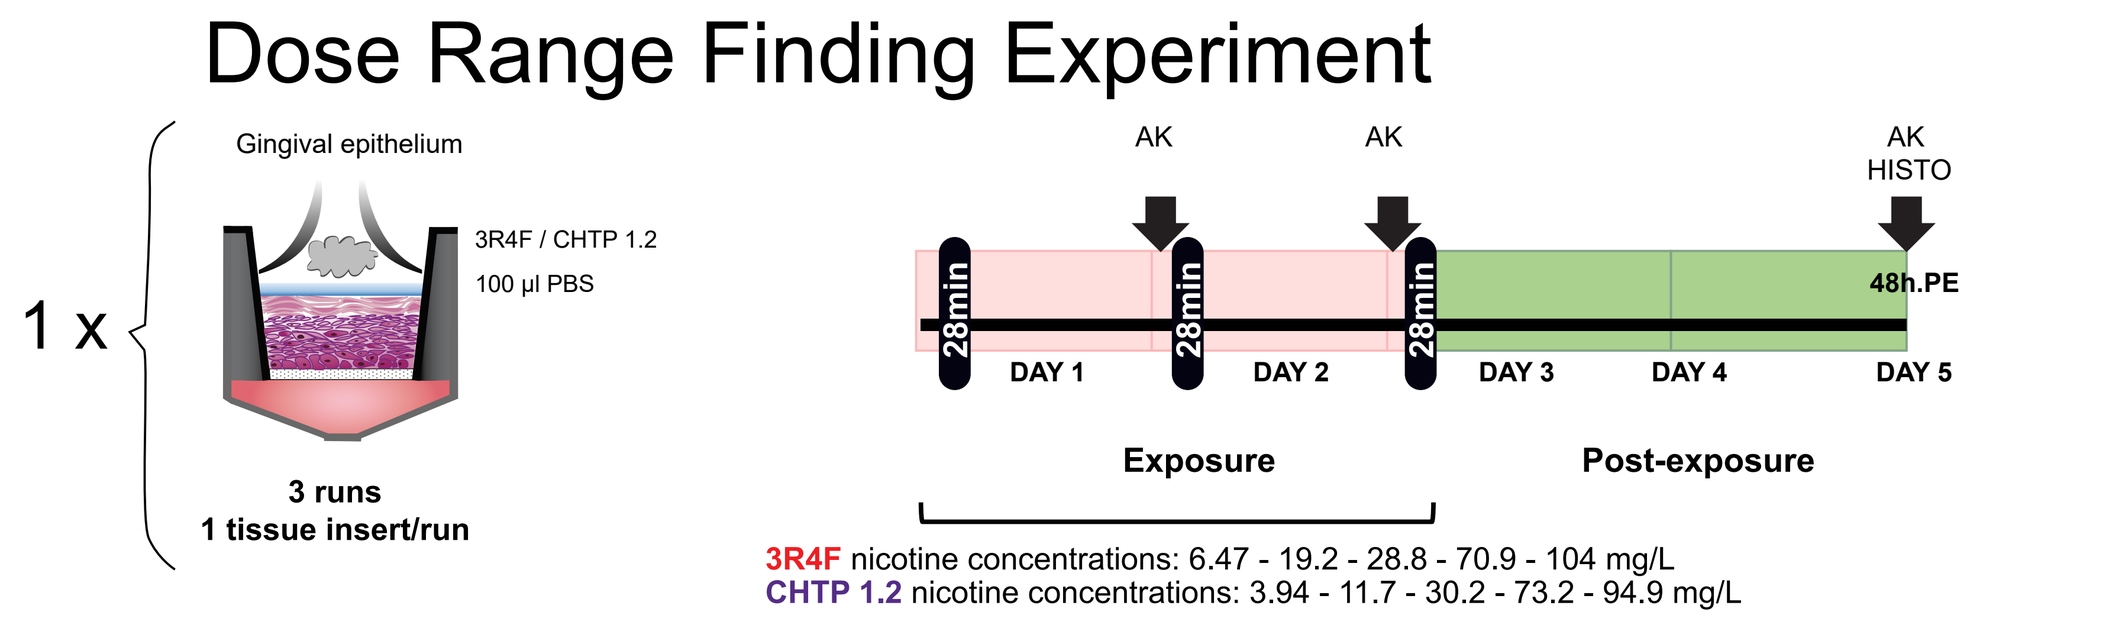
1 x
Gingival epithelium
3R4F / CHTP 1.2
100 µl PBS
3 runs
1 tissue insert/run
AK
AK
AK
HISTO
28min
28min
28min
48h.PE
DAY 1
DAY 2
DAY 3
DAY 4
DAY 5
Exposure
Post-exposure
3R4F nicotine concentrations: 6.47 - 19.2 - 28.8 - 70.9 - 104 mg/L
CHTP 1.2 nicotine concentrations: 3.94 - 11.7 - 30.2 - 73.2 - 94.9 mg/L

Experimental Phase

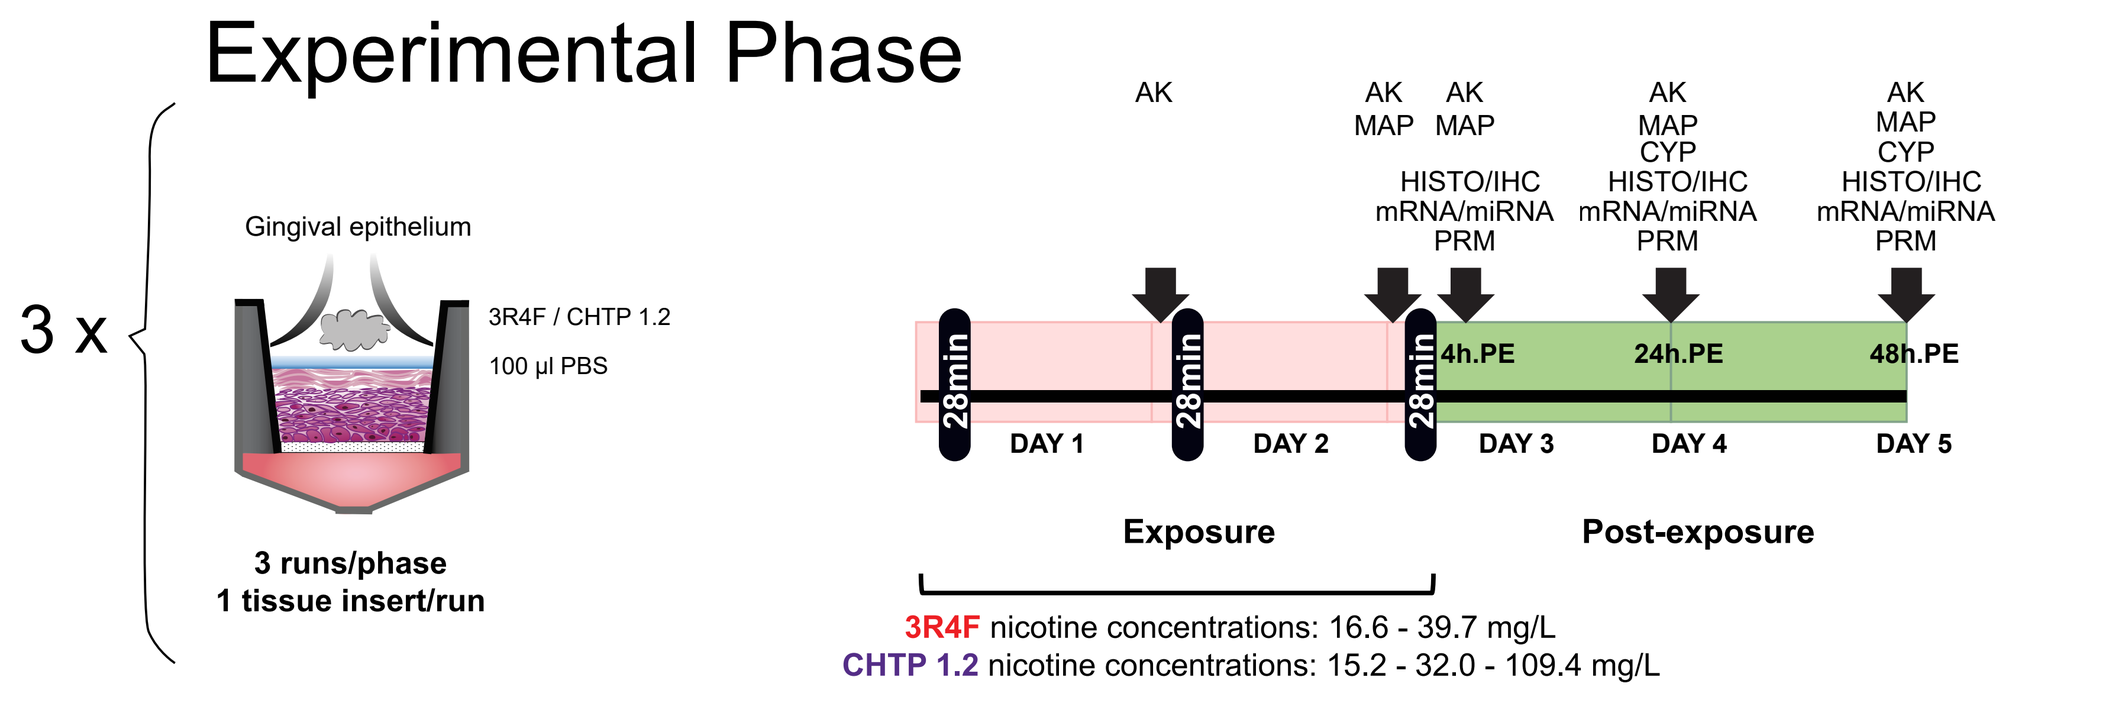
3 x
Gingival epithelium
3R4F / CHTP 1.2
100 µl PBS
3 runs/phase
1 tissue insert/run
AK
AK MAP
AK MAP HISTO/IHC mRNA/miRNA PRM
AK MAP CYP HISTO/IHC mRNA/miRNA PRM
AK MAP CYP HISTO/IHC mRNA/miRNA PRM
28min
28min
28min
4h.PE
24h.PE
48h.PE
DAY 1
DAY 2
DAY 3
DAY 4
DAY 5
Exposure
Post-exposure
3R4F nicotine concentrations: 16.6 - 39.7 mg/L
CHTP 1.2 nicotine concentrations: 15.2 - 32.0 - 109.4 mg/L

- Human gingival organotypic epithelial cultures (EpiGingival™) were derived from a 46-year-old non-smoker male donor (MatTek corp., Ashland, MA, USA).
- Samples were exposed at the air-liquid interface in the Vitrocell® 24/48 exposure system.
- Adenylate Kinase (AK)-based cytotoxicity was measured in the basolateral media using the ToxiLight™ bioassay kit (Lonza, Rockland, MA, USA).
- Morphology of the cultures was evaluated in Hematoxylin & Eosin (HE)-stained tissue sections.
- The concentrations of released inflammatory mediators were measured in the basolateral medium using a Luminex®-based technology (Luminex, Austin, TX, USA).
- Transcriptomics data were analyzed in the context of hierarchically structured network models describing the molecular mechanisms underlying essential biological processes in non-diseased respiratory cells (Hoeng, 2012). Those results are summarized in (Zanetti, 2018) and available on INTERVALS.

Key references

Boue, S. et al. Supporting evidence-based analysis for modified risk tobacco products through a toxicology data-sharing infrastructure. F1000Research 6, 12, doi:10.12688/f1000research.10493.2, http://www.ncbi.nlm.nih.gov/pubmed/29123642, (2017).

Combes, R. D. & Balls, M. A critical assessment of the scientific basis, and implementation, of regulations for the safety assessment and marketing of innovative tobacco-related products. Alternatives to laboratory animals : ATLA 43, 251-290, http://www.ncbi.nlm.nih.gov/pubmed/26375889, (2015).

Johnson, N. W. & Bain, C. A. Tobacco and oral disease. EU-Working Group on Tobacco and Oral Health. British dental journal 189, 200-206, http://www.nature.com/bdj/journal/v189/n4/pdf/4800721a.pdf, (2000).

Hoeng, J. et al. A network-based approach to quantifying the impact of biologically active substances. Drug discovery today 17, 413-418, doi:10.1016/j.drudis.2011.11.008, http://www.ncbi.nlm.nih.gov/pubmed/22155224, (2012).

Zanetti, F. et al. Assessment of the impact of aerosol from a potential modified risk tobacco product compared with cigarette smoke on human organotypic oral epithelial cultures under different exposure regimens. Food and chemical toxicology : an international journal published for the British Industrial Biological Research Association 115, 148-169, doi:10.1016/j.fct.2018.02.062, http://www.ncbi.nlm.nih.gov/pubmed/29505817, (2018).

Additional relevant references can be found on INTERVALS.science

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

Results

Key results as published on INTERVALS™ are given below.

Nicotine concentration

Nicotine concentration measured in the Vitrocell® 24/48 exposure system during the dose range finding experiment and each experimental phase confirms delivery of smoke and aerosol consistent with the study design.

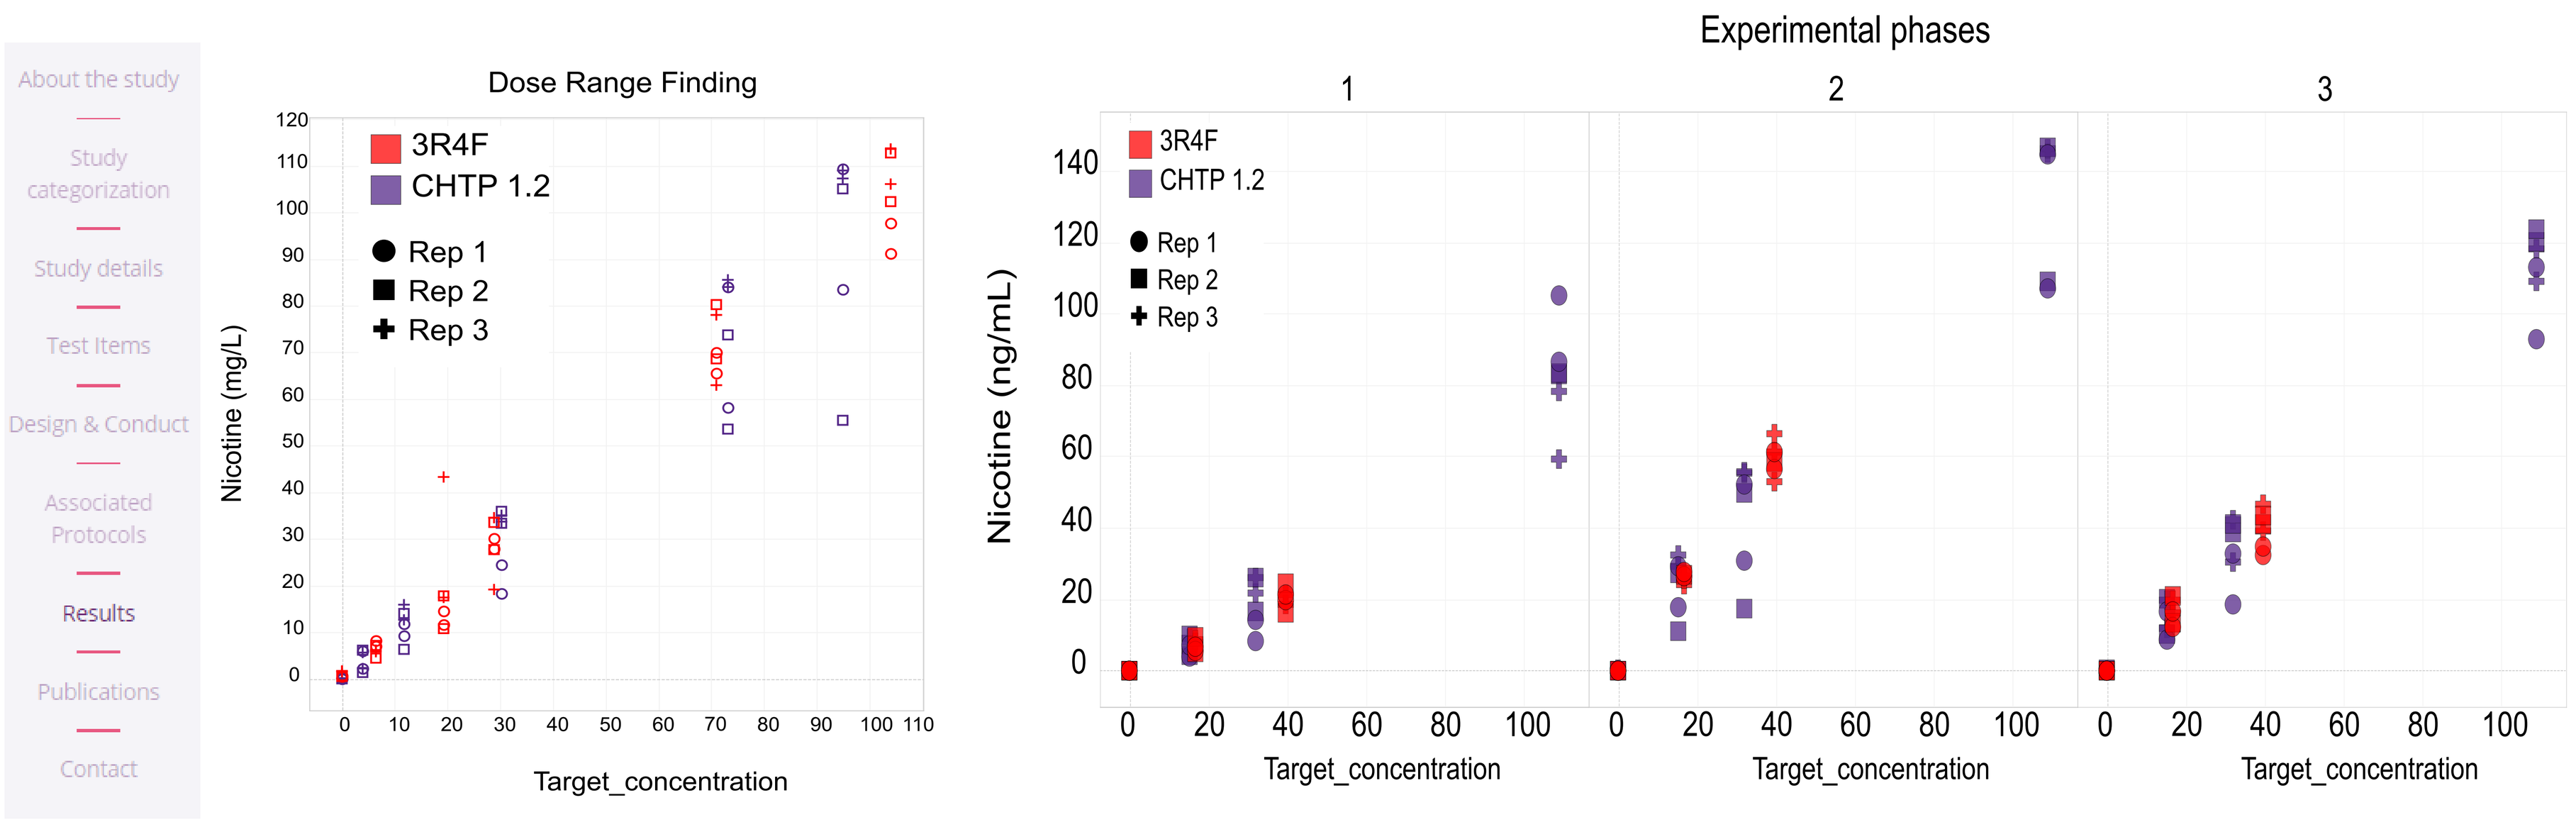
About the study
Study categorization
Study details
Test Items
Design & Conduct
Associated Protocols
Results
Publications
Contact
Dose Range Finding
3R4F
CHTP 1.2
Rep 1
Rep 2
Rep 3
Nicotine (mg/L)
Target_concentration
Experimental phases
1
2
3
Nicotine (ng/mL)
Target_concentration

Cytotoxicity (AK assay)

The cytotoxicity levels of the CHTP 1.2 aerosol-exposed cultures were not significantly different from those of the air controls, independently of aerosol concentration or collection time point.

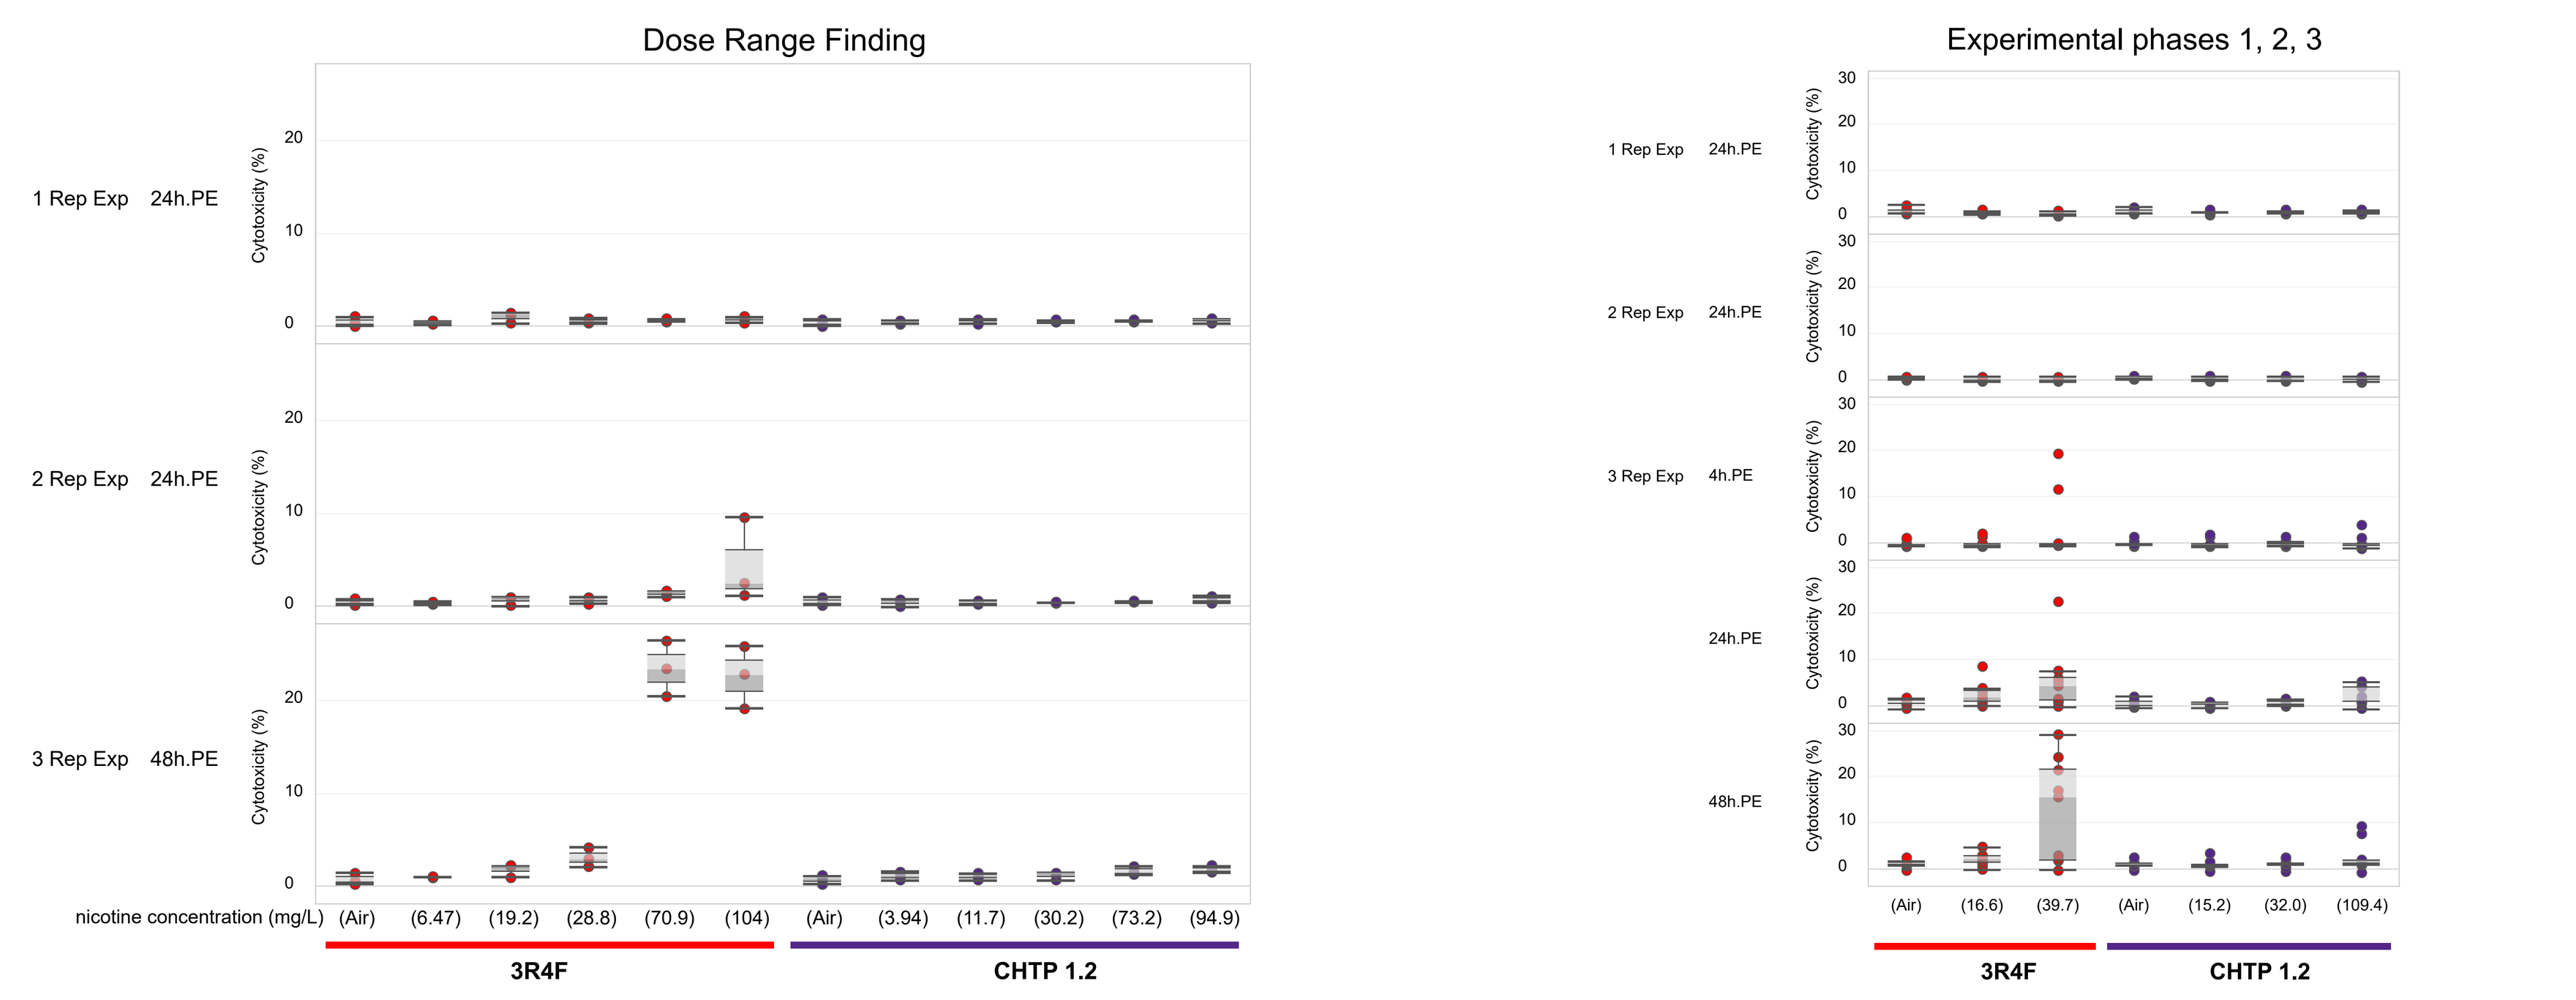
Dose Range Finding
1 Rep Exp 24h.PE
2 Rep Exp 24h.PE
3 Rep Exp 48h.PE
Cytotoxicity (%)
nicotine concentration (mg/L) (Air) (6.47) (19.2) (28.8) (70.9) (104) (Air) (3.94) (11.7) (30.2) (73.2) (94.9)
3R4F
CHTP 1.2
Experimental phases 1, 2, 3
1 Rep Exp 24h.PE
2 Rep Exp 24h.PE
3 Rep Exp 4h.PE
24h.PE
48h.PE
(Air) (16.6) (39.7) (Air) (15.2) (32.0) (109.4)
3R4F
CHTP 1.2

Histological analysis

Exposure to 3R4F CS caused marked signs of damage, leading to increased cell alterations, atrophy, apoptosis, hypergranulosis, parakeratosis, suprabasal splitting, and epithelial splitting compared with the air controls.

CHTP 1.2 aerosol-exposed cultures exhibited changes of the same findings but much less marked, even following exposure to the highest concentration (109.4 mg/L).

4 h Post-exposure

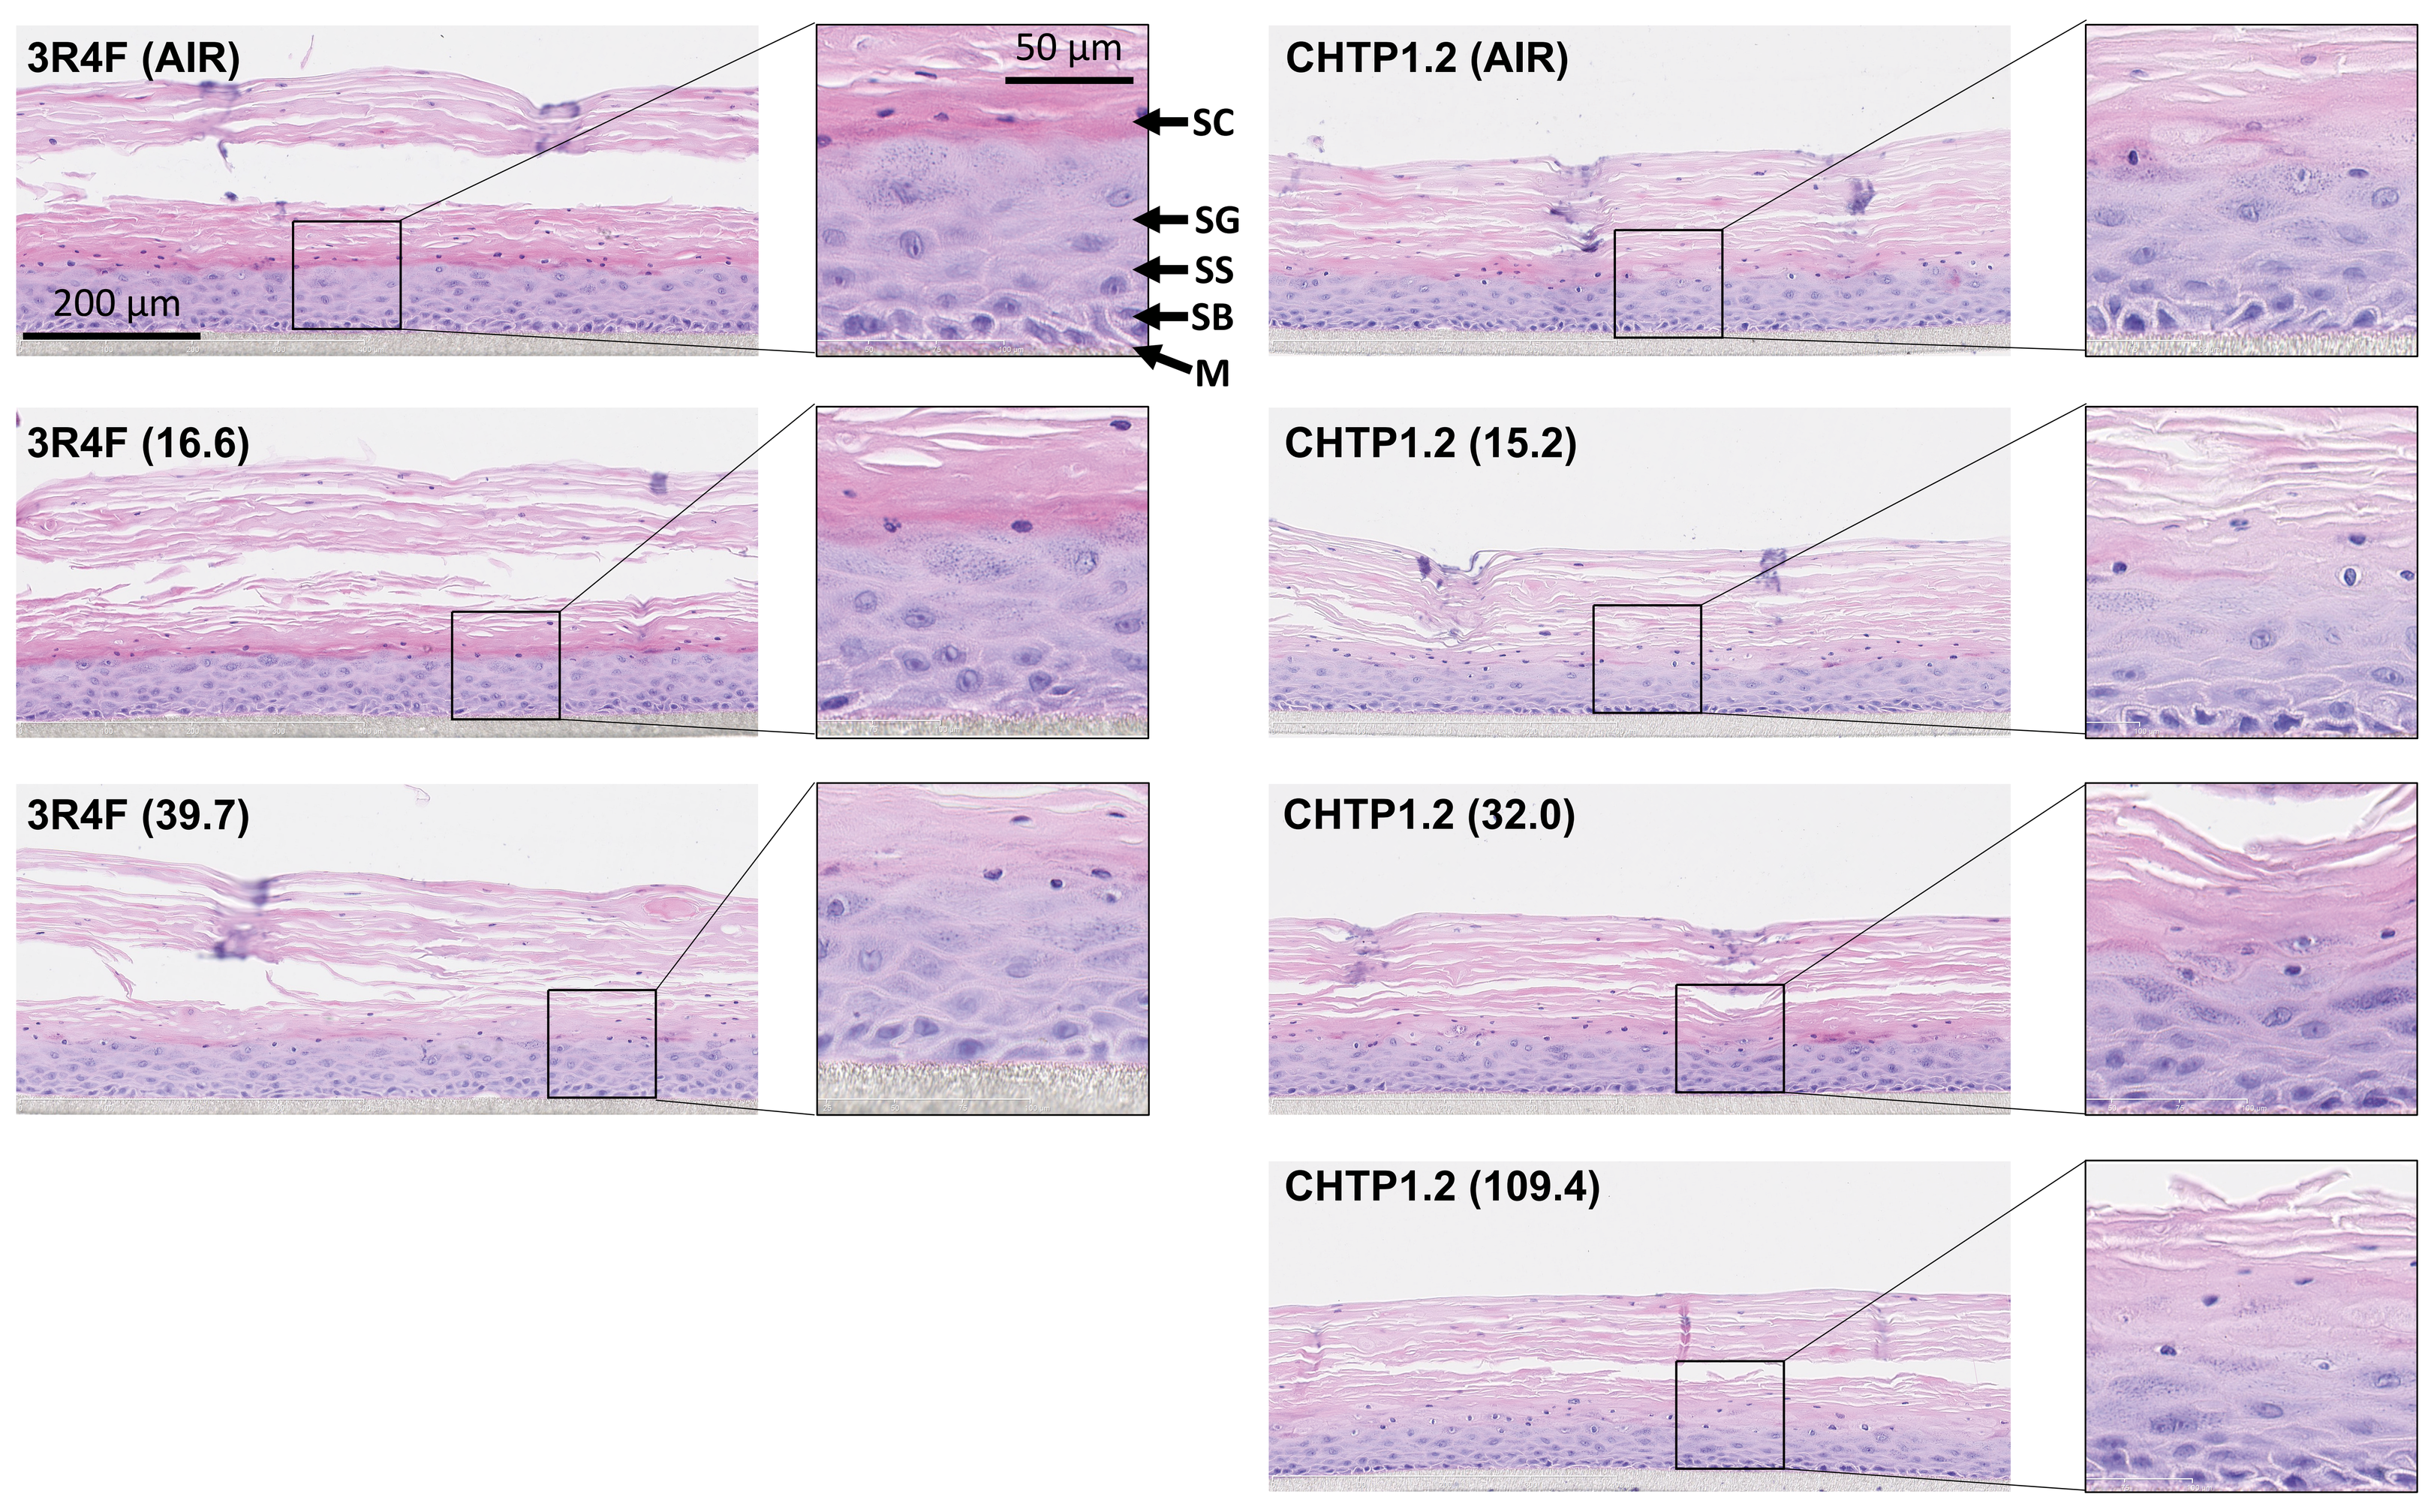
3R4F (AIR)
50 µm
SC
SG
SS
SB
M
200 µm
CHTP1.2 (AIR)
3R4F (16.6)
CHTP1.2 (15.2)
3R4F (39.7)
CHTP1.2 (32.0)
CHTP1.2 (109.4)

Multianalyte profiling (MAP)

Smaller alterations in the concentrations of inflammatory mediators were observed following CHTP 1.2 aerosol than 3R4F CS exposures at the comparable concentrations (16.6 vs. 15.2 and 39.7 vs. 32.0 mg/L).

4 h Post-exposure

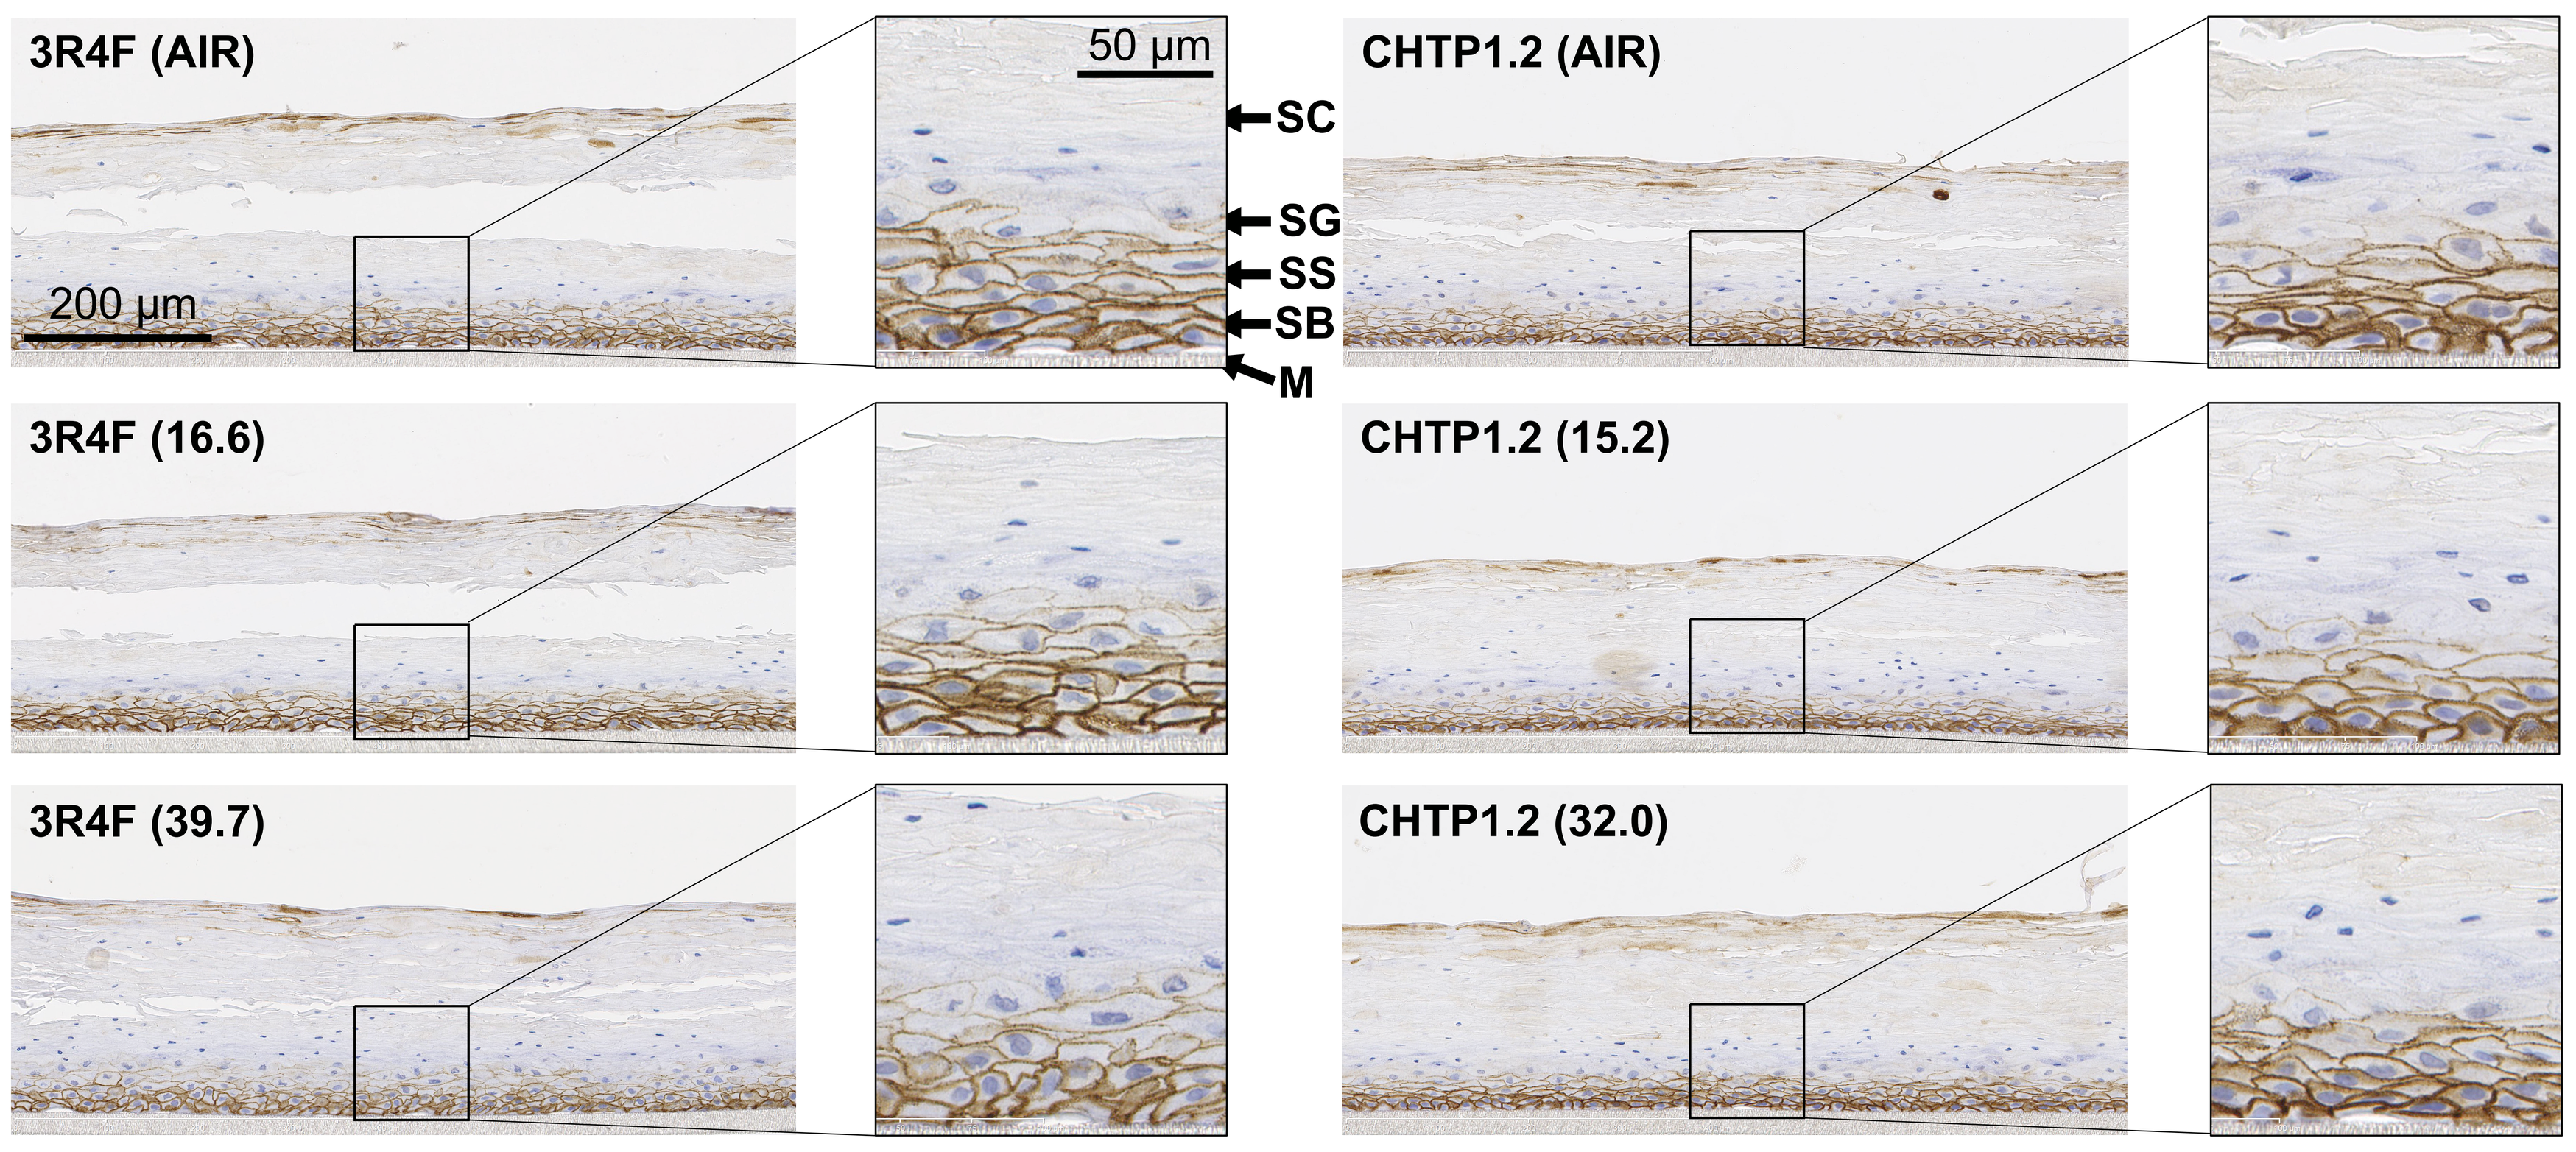
3R4F (AIR)
50 µm
SC
SG
SS
SB
M
200 µm
CHTP1.2 (AIR)
3R4F (16.6)
CHTP1.2 (15.2)
3R4F (39.7)
CHTP1.2 (32.0)

Competing Financial Interest
The research described in this poster was sponsored by Philip Morris Products SA