Six-Month Systems Toxicology Inhalation/Cessation Study in ApoE$^{-/-}$ Mice to Investigate Cardiovascular and Respiratory Exposure Effects of Two Reduced-Risk Products Compared with Conventional Cigarettes

Justyna Szostak[1], Blaine Phillips[2], Emmanuel Guedj[1], Ee Tsin Wong[2], Bjoern Titz[1], Florian Martin[1], Grégory Vuillaume[1], Patrice Leroy[1], Ansgar Buettner[3], Nikolai V. Ivanov[1], Patrick Vanscheeuwijck[1], Manuel C. Peitsch[1], and Julia Hoeng[1]

[1] PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland
[2] PMI R&D, Philip Morris International Research Laboratories Pte. Ltd., Science Park II, Singapore
[3] Histovia GmbH, Schöne Aussicht 5, D-51491 Overath, Germany

Introduction and Objectives

Cigarette smoking is the main risk factor for the development and progression of a series of diseases, including cardiovascular disease (CVD) (1-4) and chronic obstructive pulmonary disease (COPD) (5). Suitable animal models play an important role in understanding smoke-induced pathogenesis. This study examined the development of hallmarks of CVD in ApoE$^{-/-}$ mice exposed either to cigarette smoke (CS) from a 3R4F reference cigarette or to aerosol from two heated tobacco products, the Tobacco Heating System (THS) 2.2 and the Carbon-Heated Tobacco Product (CHTP) 1.2, over a six-month period. In addition to chronic exposure regimes, a comparison of exposure cessation or switching to CHTP 1.2 after three months of exposure to CS was performed. Assessment of effects within this system toxicology study leveraged a battery of assays: physiological, morphological, and molecular analysis. The interpretation of various endpoints enables understanding of the biological effects of cessation/switching as compared with continued smoking.

Study Design and Endpoints

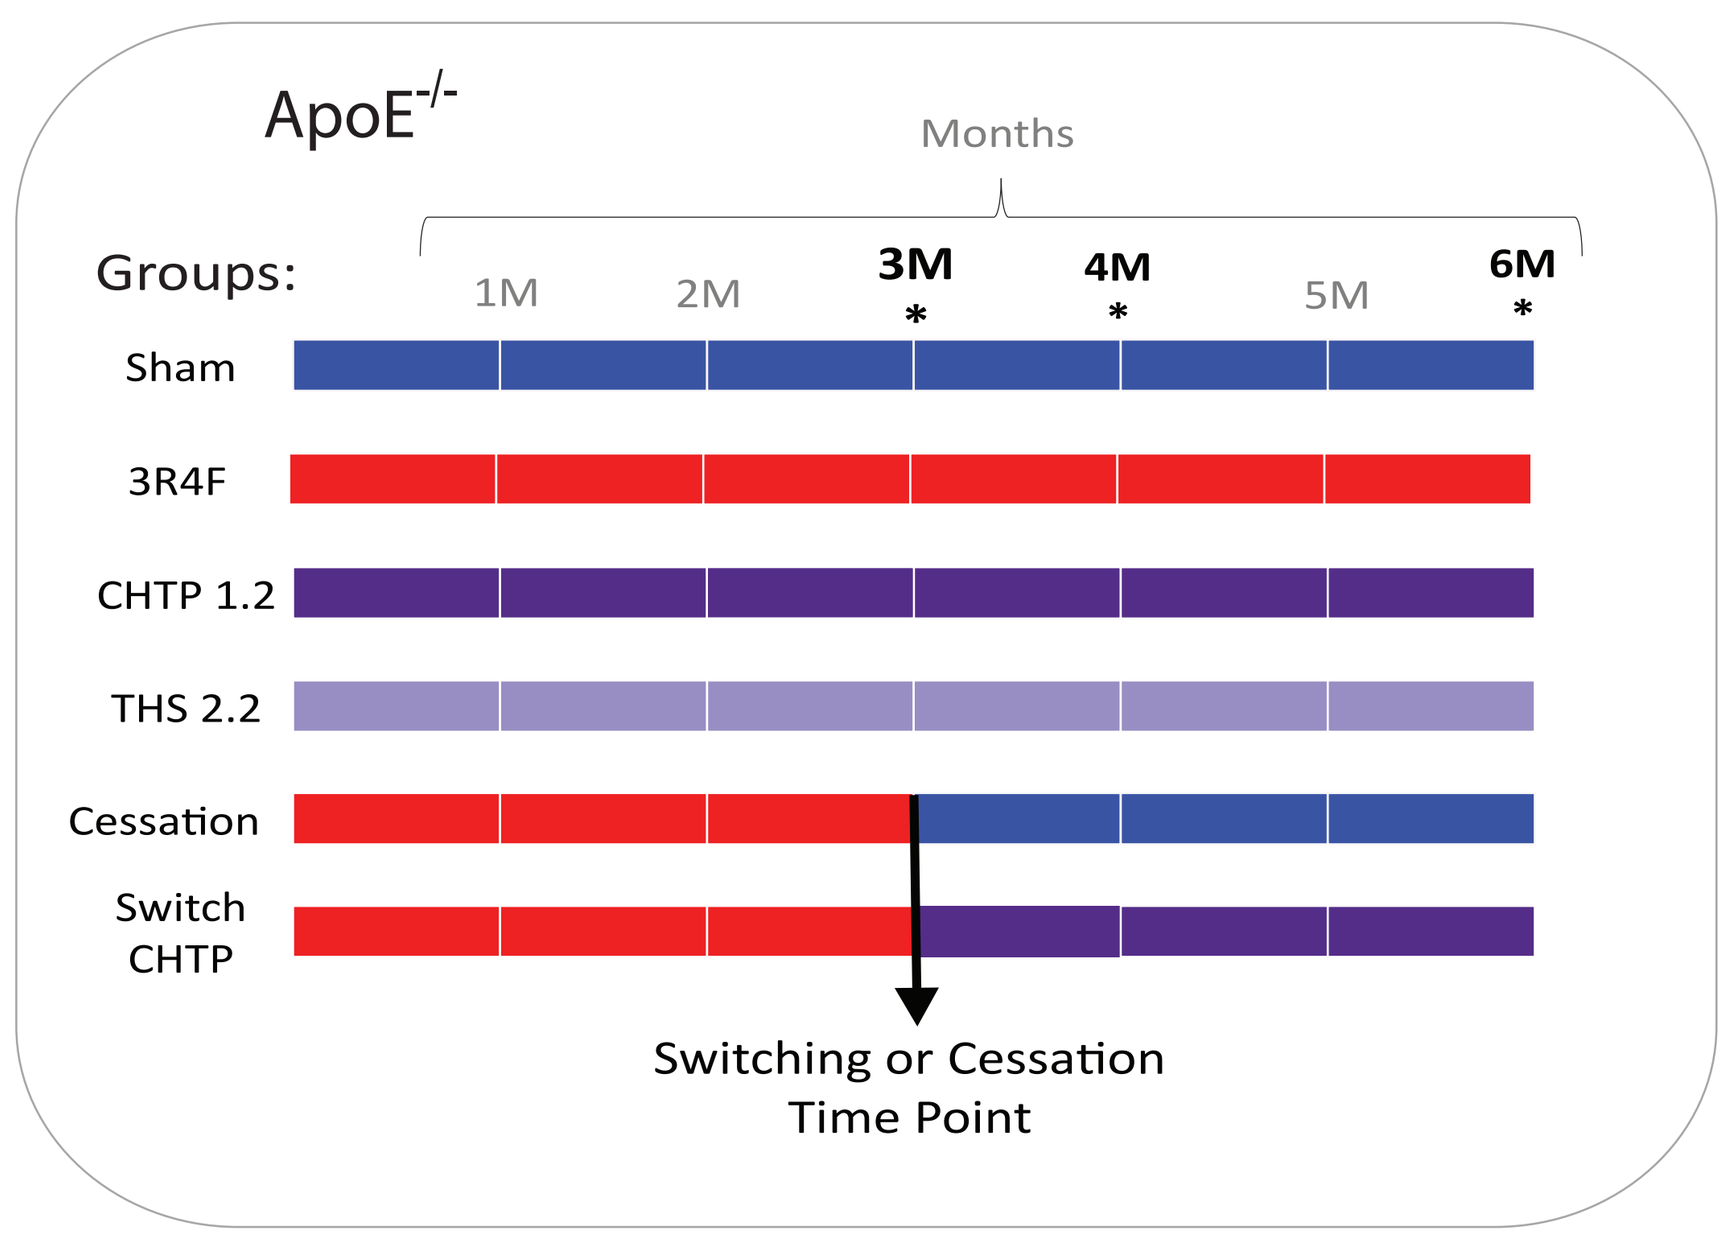
ApoE-/-
Months
Groups:
1M 2M 3M 4M 5M 6M
Sham
3R4F
CHTP 1.2
THS 2.2
Cessation
Switch CHTP
Switching or Cessation Time Point

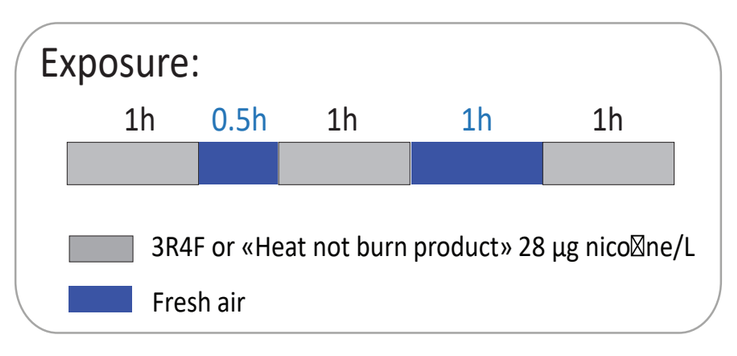
Exposure:
1h 0.5h 1h 1h 1h
3R4F or «Heat not burn product» 28 µg nicotine/L
Fresh air

* Time point for transcriptomics, proteomics, lipidomis, and genomics analysis

ApoE$^{-/-}$ mice were exposed to air (control group) or to CHTP 1.2 or THS 2.2 aerosol and to 3R4F CS at a concentration of 28.0 µg nicotine/L (equivalent to 600 µg total particulate matter/L) for six months.

Additionally, the impact of smoking cessation or switching to CHTP 1.2 aerosol exposure after three months of 3R4F CS exposure was assessed. Blood lipid profiling, histopathological evaluation, computed tomography scans, and transcriptomic analysis of thoracic aorta and heart ventricle were performed to investigate the impact of CHTP 1.2 aerosol and CS exposure on the cardiovascular system.

Animals were observed on a daily basis, body weight progression was monitored twice per week, and exposure and uptake parameters (including nicotine metabolites in urine) were measured three times during the study.

Dissections were performed after two, three, four, and six months of exposure.

Characterization of Aerosol: 3R4F, CHTP 1.2, and THS 2.2

Characterization of aerosol particles

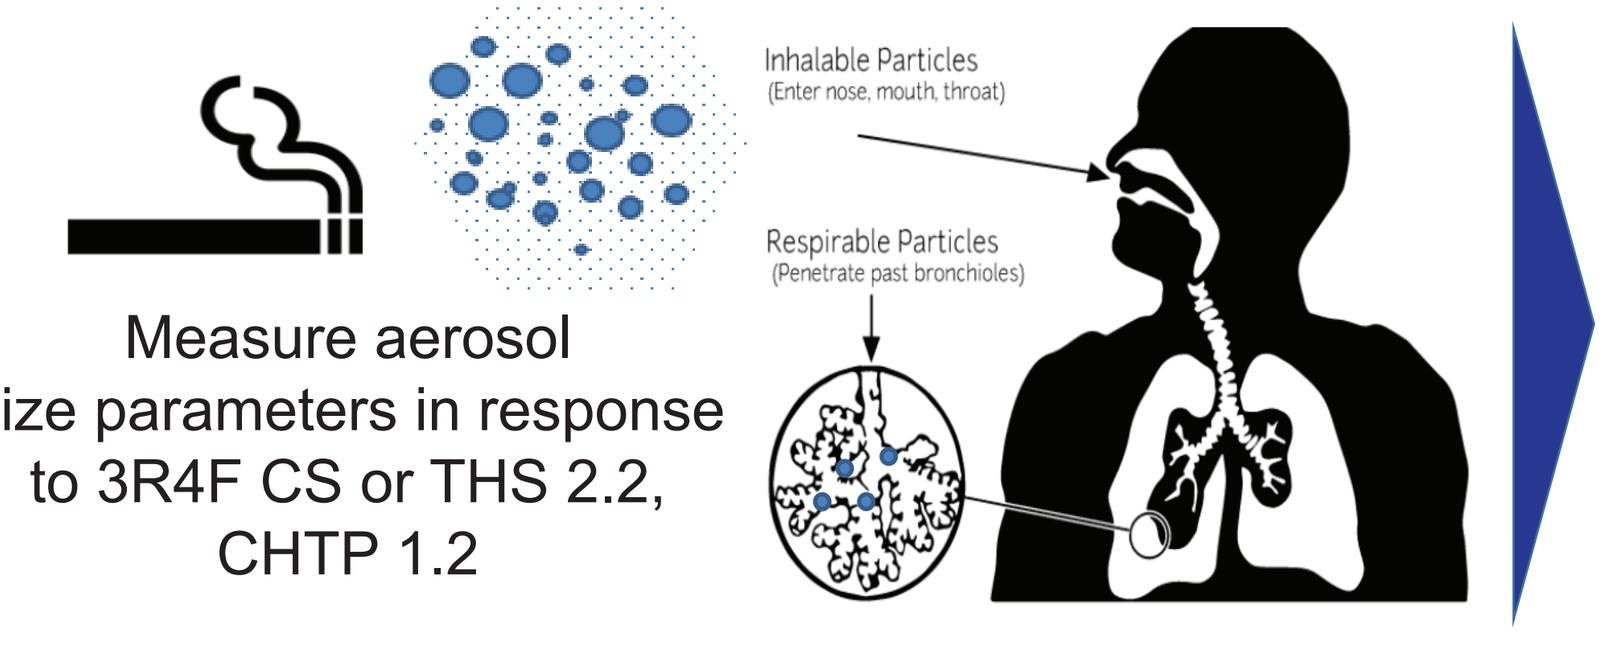
Inhalable Particles (Enter nose, mouth, throat)
Respirable Particles (Penetrate past bronchioles)
Measure aerosol size parameters in response to 3R4F CS or THS 2.2, CHTP 1.2

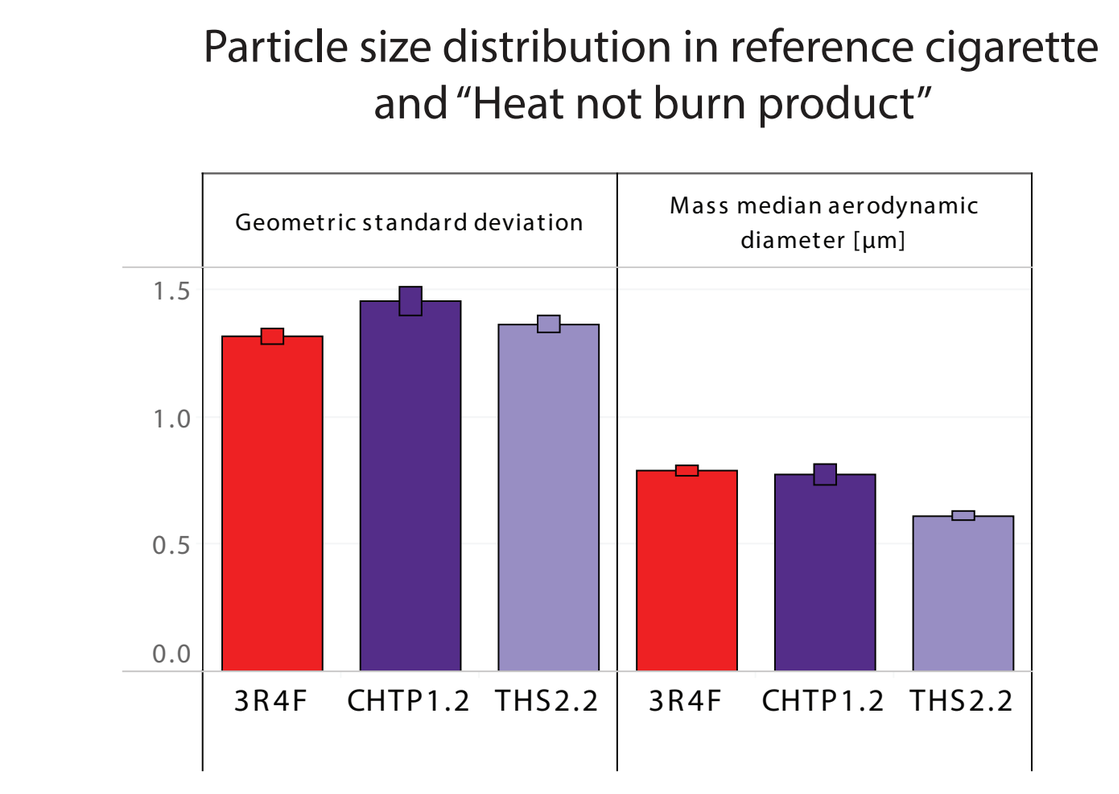
Particle size distribution in reference cigarette and "Heat not burn product"
Geometric standard deviation
Mass median aerodynamic diameter [µm]
3R4F CHTP1.2 THS2.2 3R4F CHTP1.2 THS2.2

The controlled tobacco-heating products (CHTP 1.2, THS 2.2) present aerosol particles comparable to conventional burning tobacco products (3R4F).

HPHC yield in CHTP 1.2 relative to 3R4F mainstream smoke

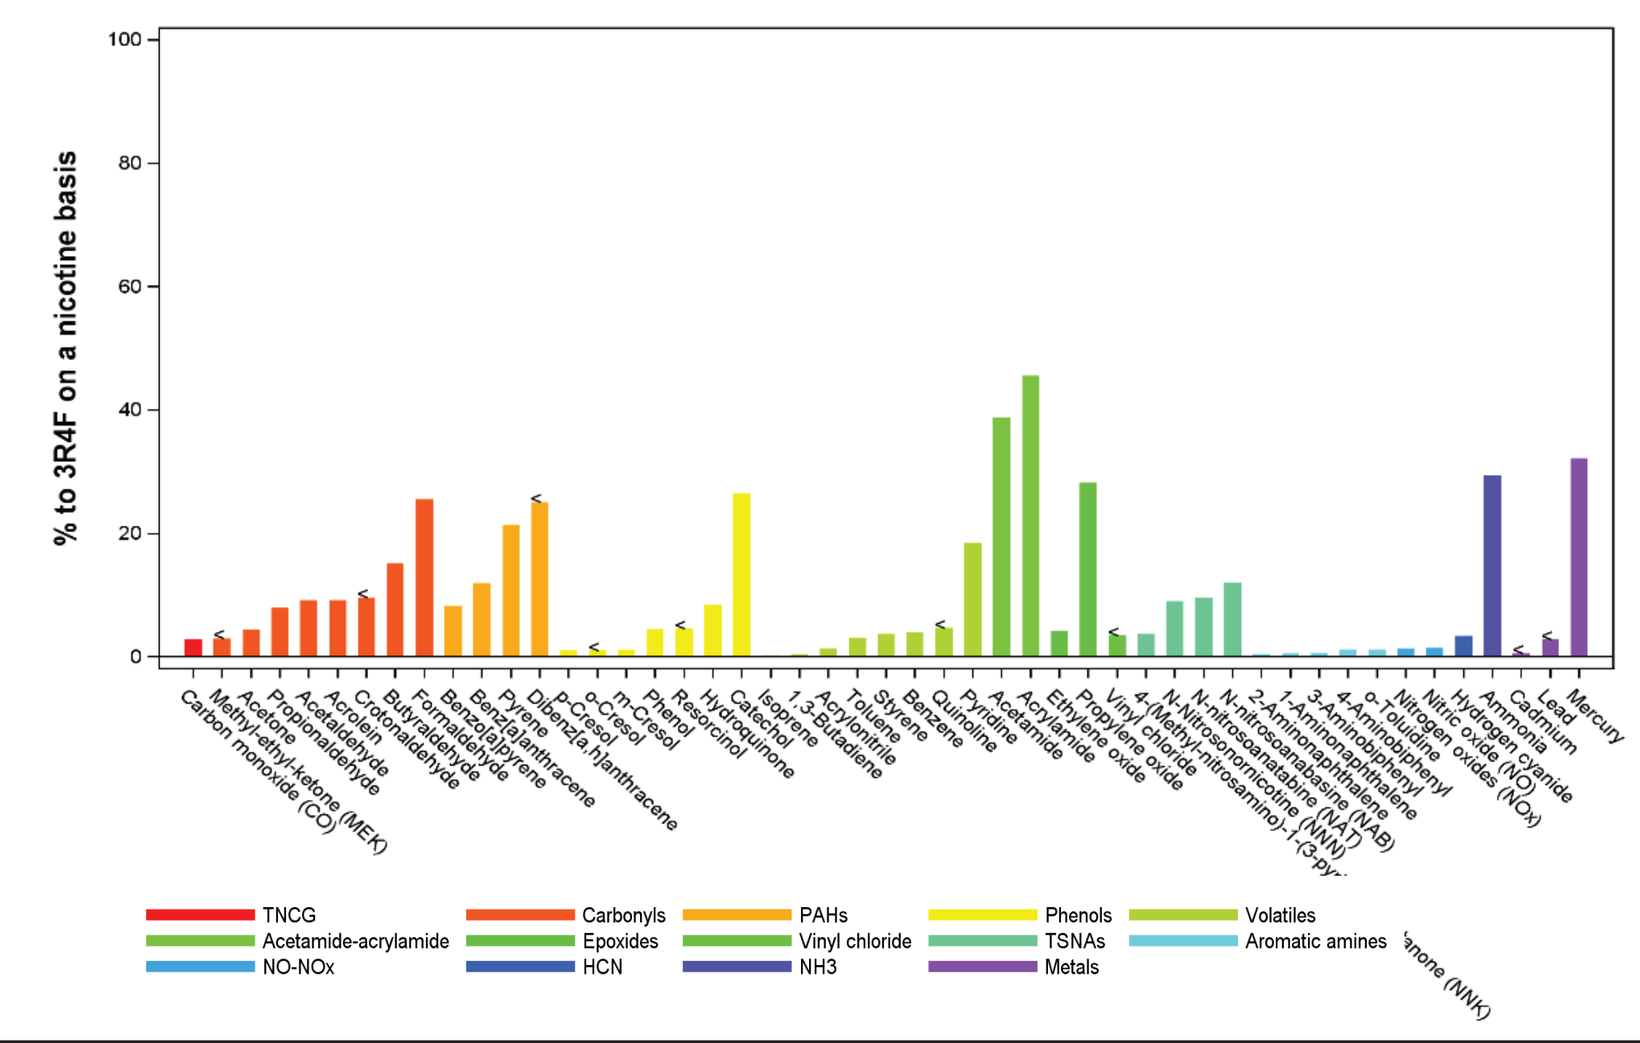
% to 3R4F on a nicotine basis
TNCG
Carbonyls
PAHs
Phenols
Volatiles
Acetamide-acrylamide
Epoxides
Vinyl chloride
TSNAs
Aromatic amines
NO-NOx
HCN
NH3
Metals

The controlled tobacco-heating approach of CHTP 1.2 reduces the delivery of harmful smoke constituents as compared with conventional burning tobacco products, as shown for 49 priority toxicants (6).

Biomarkers of Aerosol Exposure and Uptake

Biomarkers of CS exposure as well as nicotine exposure were measured in urine

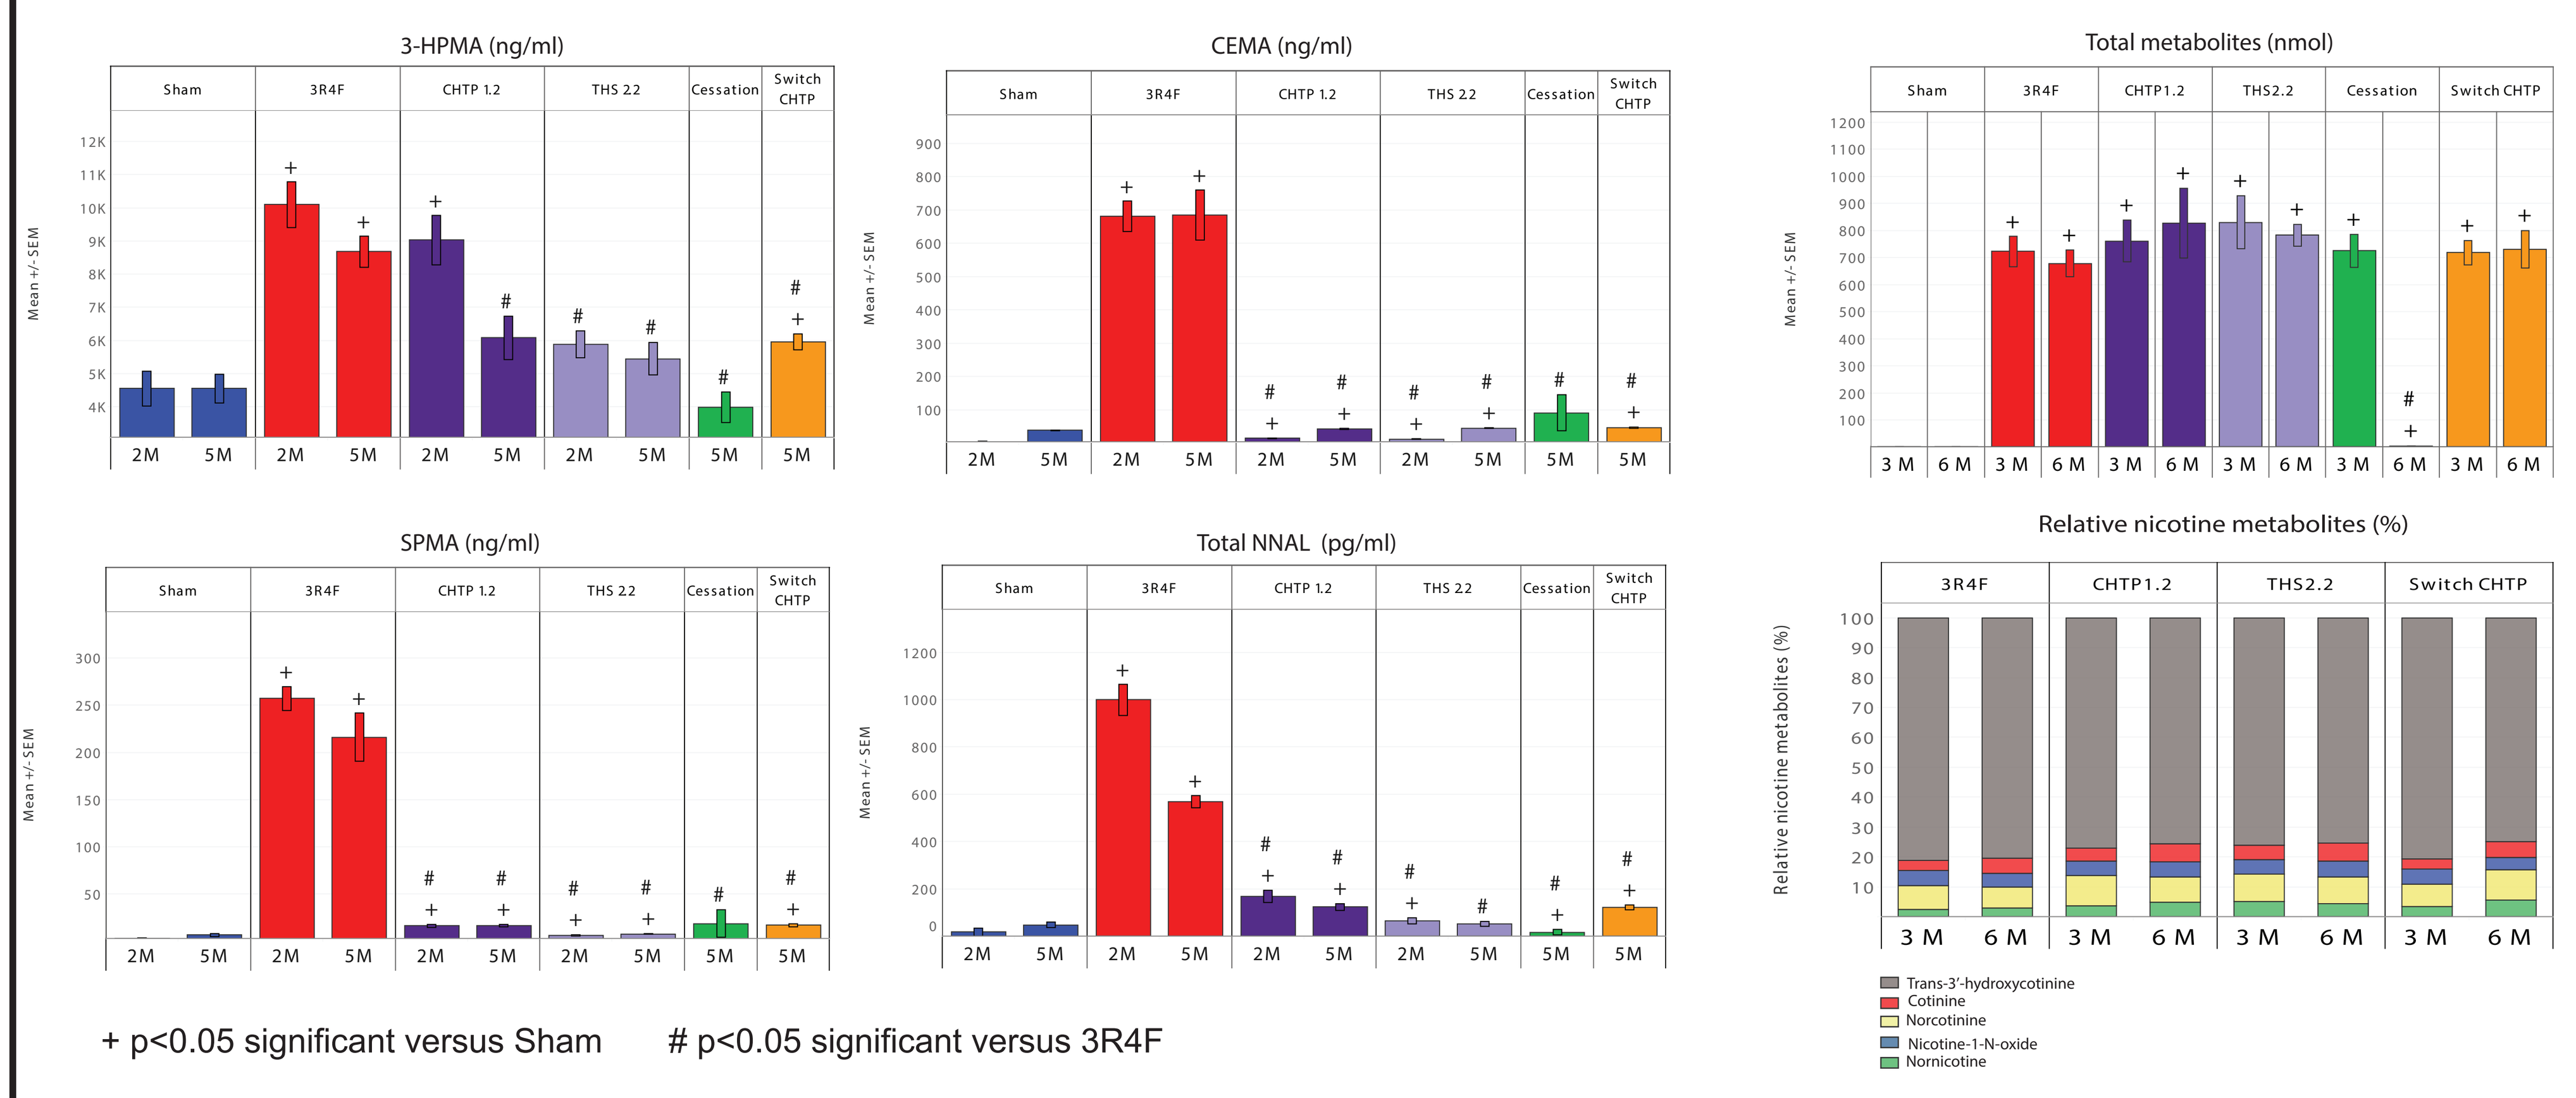
3-HPMA (ng/ml)
CEMA (ng/ml)
Total metabolites (nmol)
SPMA (ng/ml)
Total NNAL (pg/ml)
Relative nicotine metabolites (%)
Sham
3R4F
CHTP 1.2
THS 2.2
Cessation
Switch CHTP
Mean +/- SEM
Trans-3'-hydroxycotinine
Cotinine
Norcotinine
Nicotine-1-N-oxide
Nornicotine

+ p<0.05 significant versus Sham # p<0.05 significant versus 3R4F

The controlled tobacco-heating approaches of CHTP 1.2 and THS 2.2 reduce the delivery of harmful smoke constituents, such as SPMA, CEMA, and COHb, as compared with conventional burning tobacco products (3R4F). Levels of total metabolites and nicotine metabolites (Trans-3-hydrocotinine, Cotinine,Nicotine-1-N-oxide, Norcotinine, Nornicotine) are similar in CHTP 1.2-, THS 2.2- and 3R4F- exposed animals.

Respiratory Disease Endpoints

Lung tissue inflammation

The absolute number of inflammatory cells, as determined by flow cytometry-based analysis of free lung cells, was dramatically increased as early as two months following 3R4F smoke exposure. Inflammatory cells largely reverted back toward levels obtained with continuous exposure to fresh air within one month after cessation or switching.

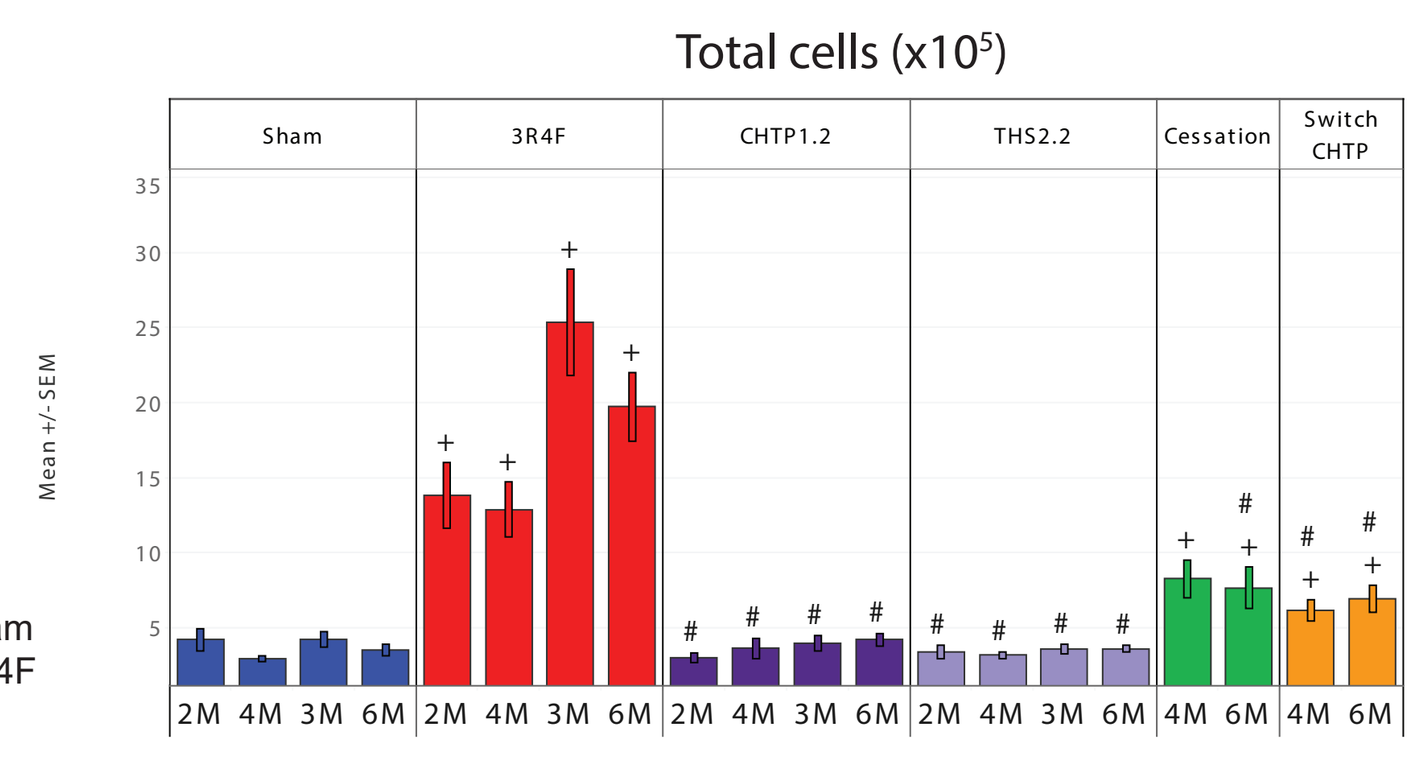
Total cells (x10⁵)
Sham
3R4F
CHTP1.2
THS2.2
Cessation
Switch CHTP
Mean +/- SEM

+ p<0.05 significant versus Sham
\# p<0.05 significant versus 3R4F

Lung function

Exposure of mice to 3R4F CS resulted in a leftward- and upward-trending shift in the P-V loops (Month 6) as compared with the results obtained with fresh air-exposed animals . Switching resulted in a return to a P-V loop profile of the fresh air group. Increased lung volume was similarly observed in the 3R4F-exposed mice at necropsy . Chronic (six months) exposure to aerosols from the heat-not-burn products (CHTP 1.2, THS 2.2) did not have any significant impact on either the P-V loop results or the lung volumes.

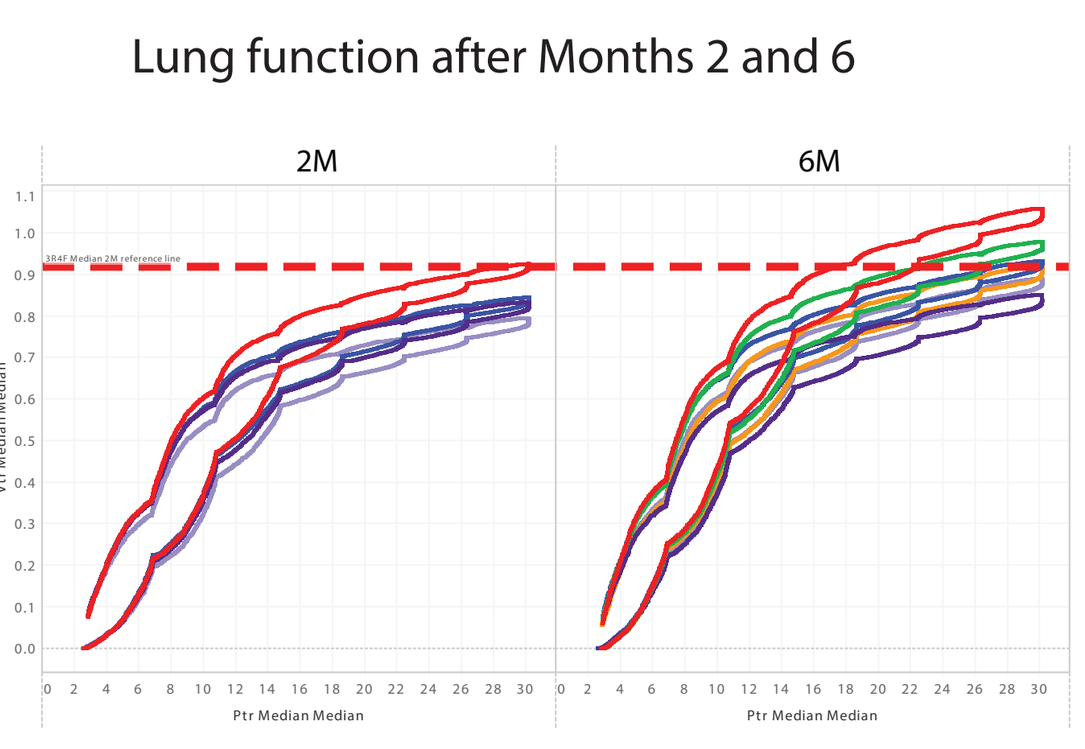
Lung function after Months 2 and 6
2M
6M
Ptr Median Median

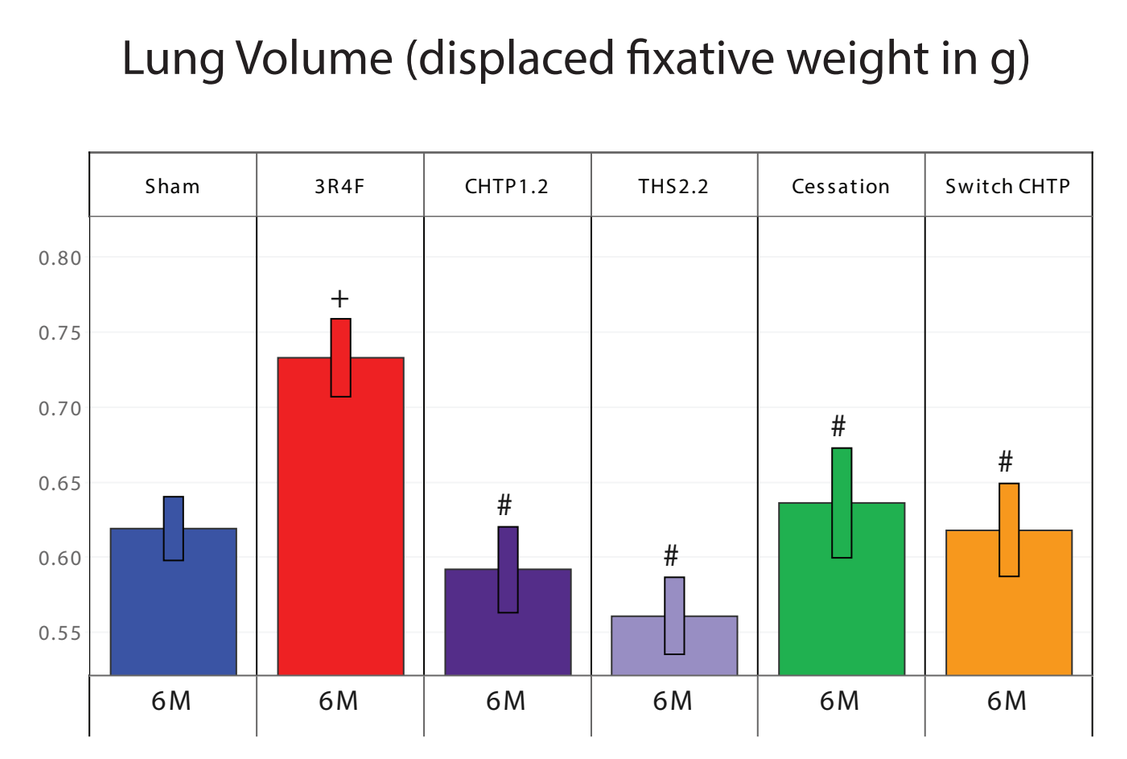
Lung Volume (displaced fixative weight in g)
Sham
3R4F
CHTP1.2
THS2.2
Cessation
Switch CHTP
Mean +/- SEM

+ p<0.05 significant versus Sham # p<0.05 significant versus 3R4F

Lung transcriptomics analysis

This causal biological network enrichment measured the total perturbation of the system using the relative biological impact factor (BIF) (RBIF) for all exposed groups at the three-, four-, and six-month time points for lung.

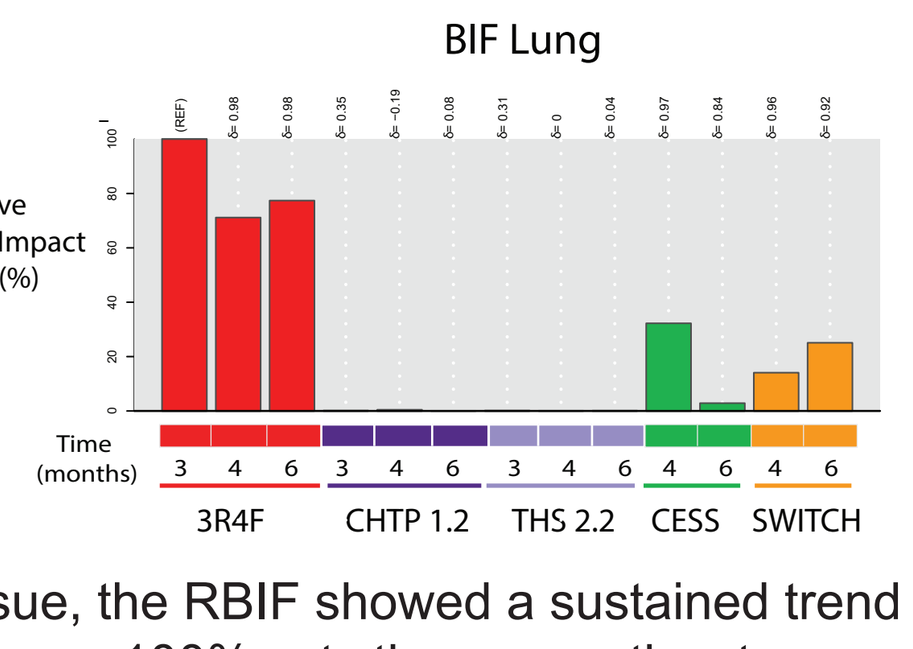
BIF Lung
Relative Biological Impact Factor (%)
Time (months)
3 4 6 3 4 6 3 4 6 4 6 4 6
3R4F
CHTP 1.2
THS 2.2
CESS
SWITCH

In lung tissue, the RBIF showed a sustained trend in response to 3R4F exposure (from 100% at three months to approximately 75% at six months).The RBIFs for the CHTP 1.2 and THS 2.2 groups remained close to zero for lung. The RBIF also tended to decrease following cessation or switching; while the RBIF decreased to zero for cessation, the switching process maintained the RBIF between 10% and 25%.

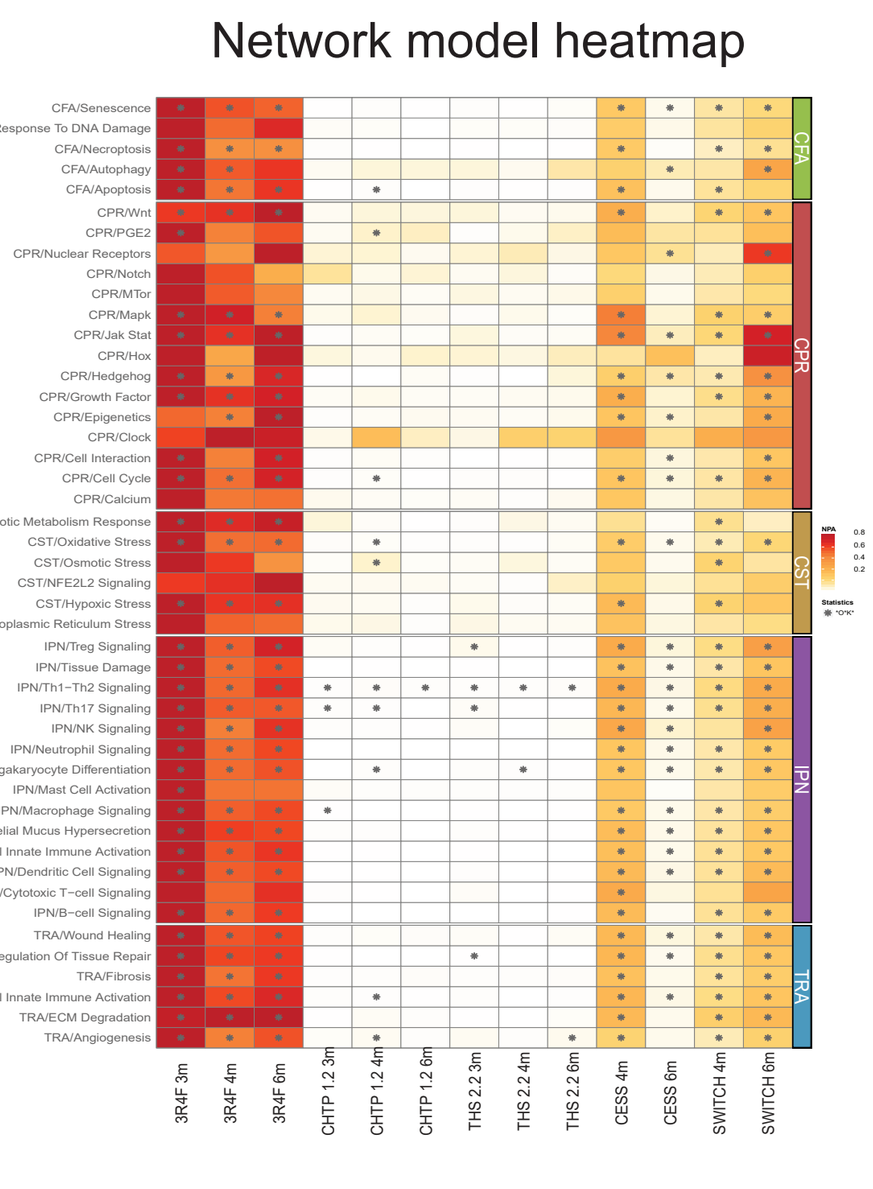
Network model heatmap
CFA
CPR
CST
IPN
TRA

The network model heatmap represents the decomposition of the calculated BIF from the transcriptomics data into its mechanistic components.

The aggregation of separate networks under the RBIF per network category demonstrate that processes such as cell fate and apoptosis (CFA), cell proliferation (CPR), cell stress (CST), inflammatory responses (IPN), and tissue repair and angiogenesis (TRA) were strongly perturbed in the 3R4F-exposed group, mostly in the lung after three months, and were either absent or only very weakly perturbed in groups exposed to CHTP 1.2 or THS 2.2 over the six-month study period.

In the cessation and switching groups, the heatmap intensity coloring for each network was strongly reduced in the lung, reconfirming that cessation and switching resulted in lower perturbation than 3R4F CS exposure.

Cardiovascular Disease Endpoints

Aortic arch plaque progression measurements

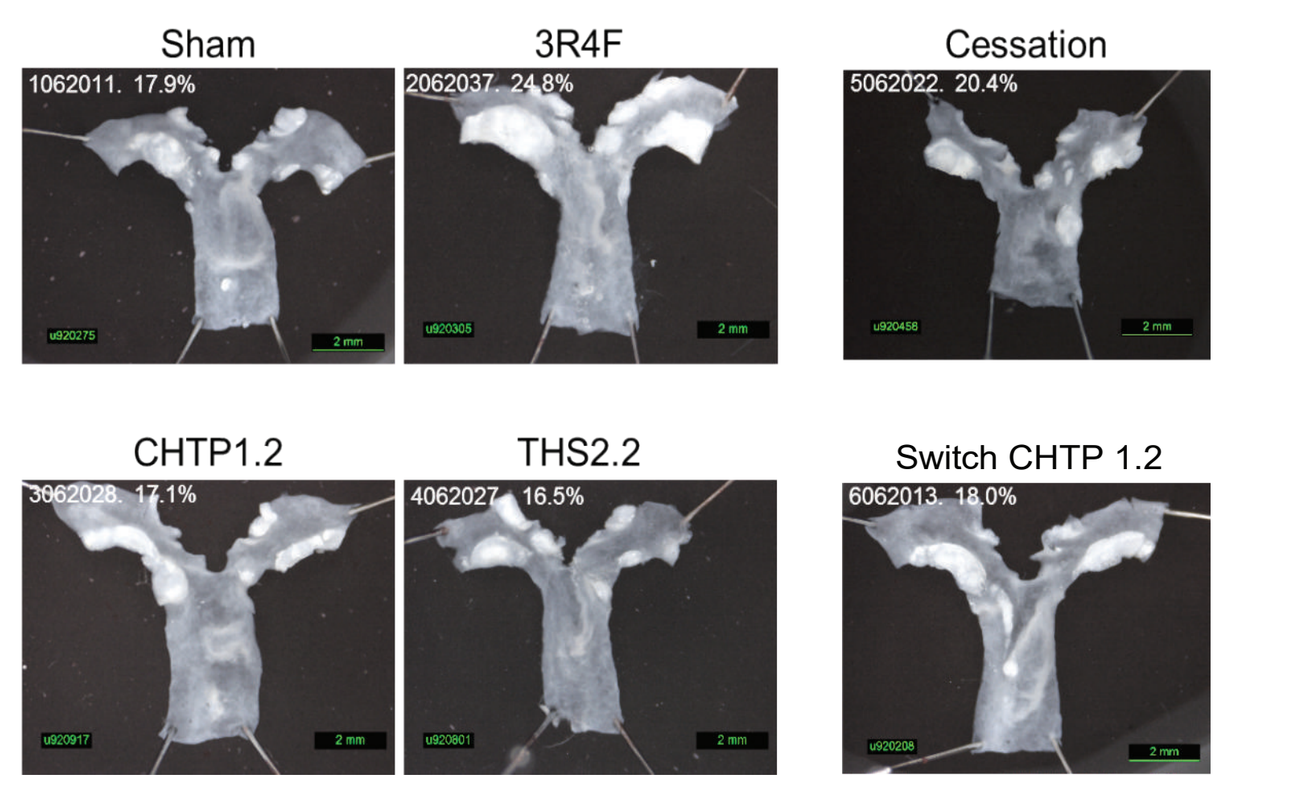
Sham
3R4F
Cessation
CHTP1.2
THS2.2
Switch CHTP 1.2
1062011. 17.9%
2062037. 24.8%
5062022. 20.4%
3062028. 17.1%
4062027. 16.5%
6062013. 18.0%

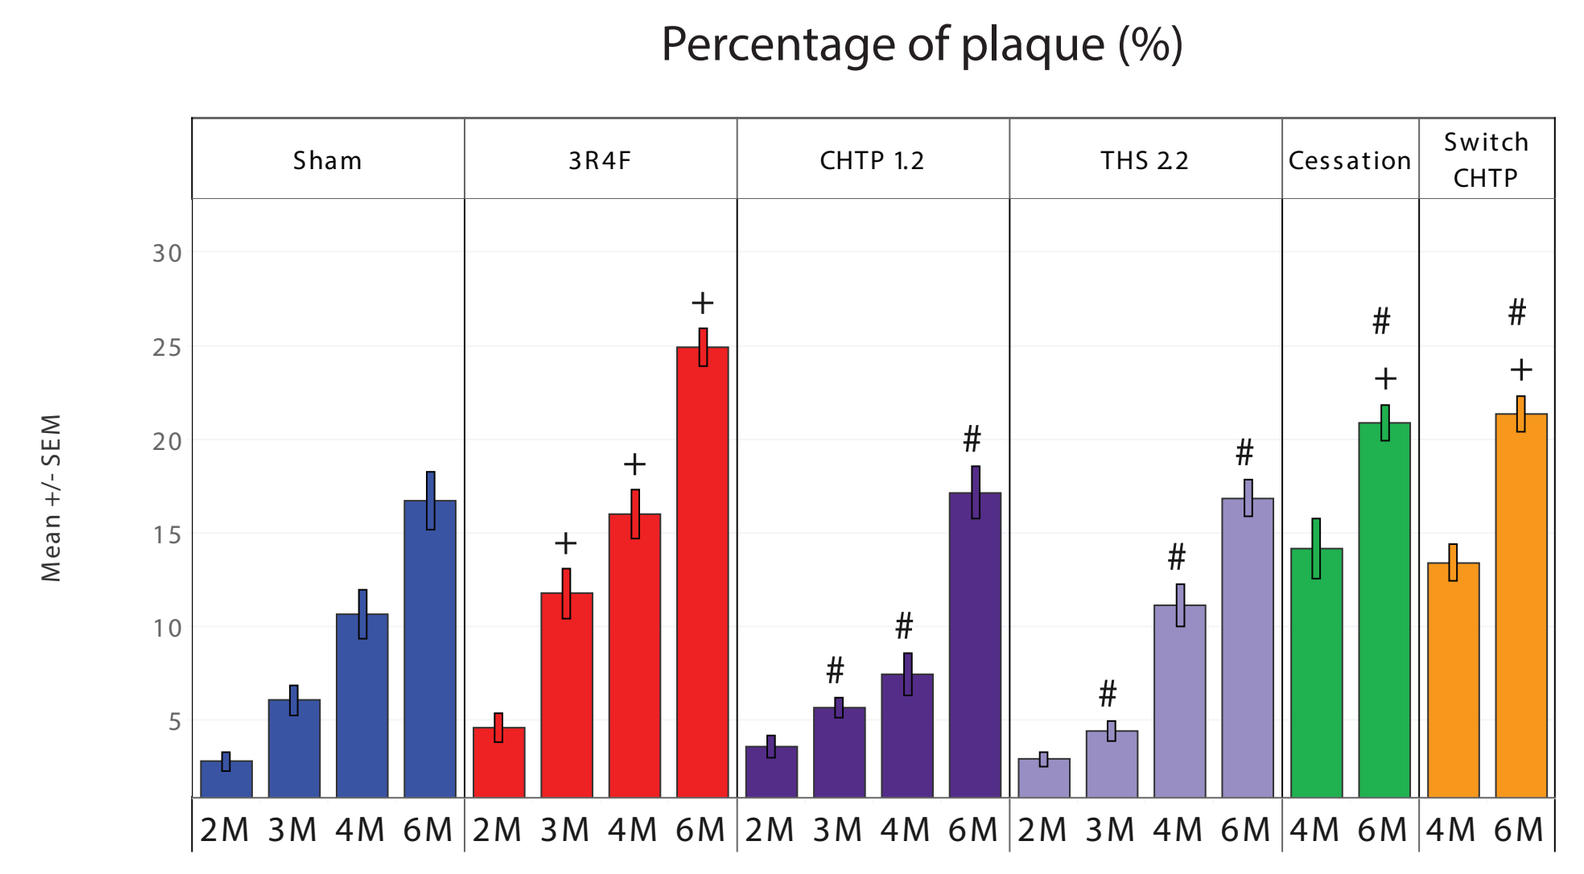
Percentage of plaque (%)
Sham
3R4F
CHTP 1.2
THS 2.2
Cessation
Switch CHTP
Mean +/- SEM

+ p<0.05 significant versus Sham
\# p<0.05 significant versus 3R4F

Exposure to 3R4F CS resulted in increased plaque formation in the aortic arch of ApoE$^{-/-}$ mice compared with the sham exposure, starting from Month 3. Cessation or switching to CHTP 1.2 resulted in a slowing of the plaque formation, as the plaque area in these groups was trending lower than the continuous 3R4F-exposed group in Month 4 and continuing to Month 6, where it was significantly lower.

Even three months after 3R4F exposure, the cessation and switching groups did not return to baseline (continuous fresh air) plaque area levels. There was no difference in plaque area in animals exposed to CHTP 1.2 or THS 2.2 for six months compared with the fresh air-treated animals.

Heart left ventricle thickness

Heart left ventricle thickness was measured after six months of exposure. The 3R4F-exposed animals had higher left ventricular thickness compared with sham animals at six months of exposure. There was no difference in left ventricle thickness in animals exposed to CHTP 1.2 or THS 2.2 as well as cessation or switch.

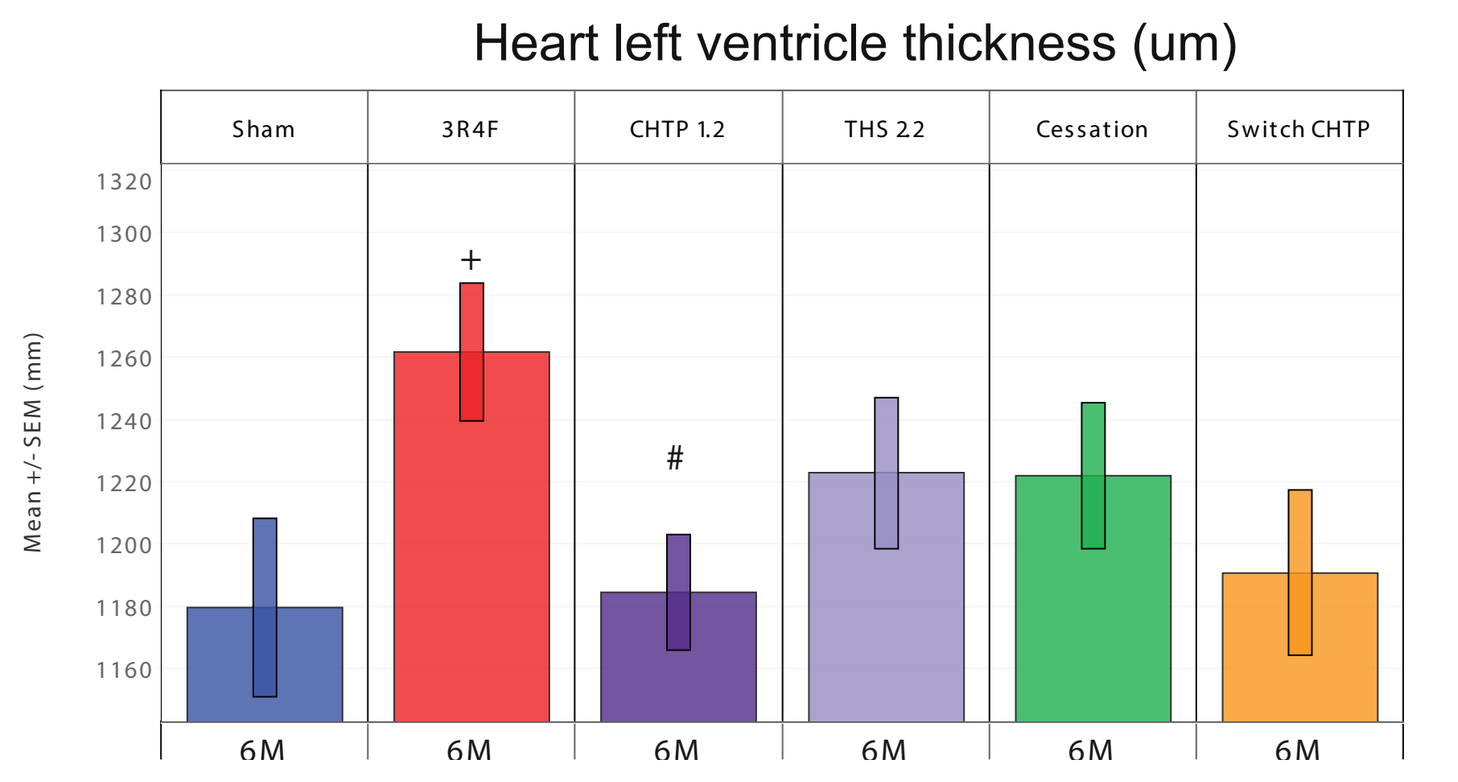
Heart left ventricle thickness (um)
Sham
3R4F
CHTP 1.2
THS 2.2
Cessation
Switch CHTP
Mean +/- SEM (mm)

+ p<0.05 significant versus Sham
\# p<0.05 significant versus 3R4F

Heart left ventricle and aorta transcriptomics analysis

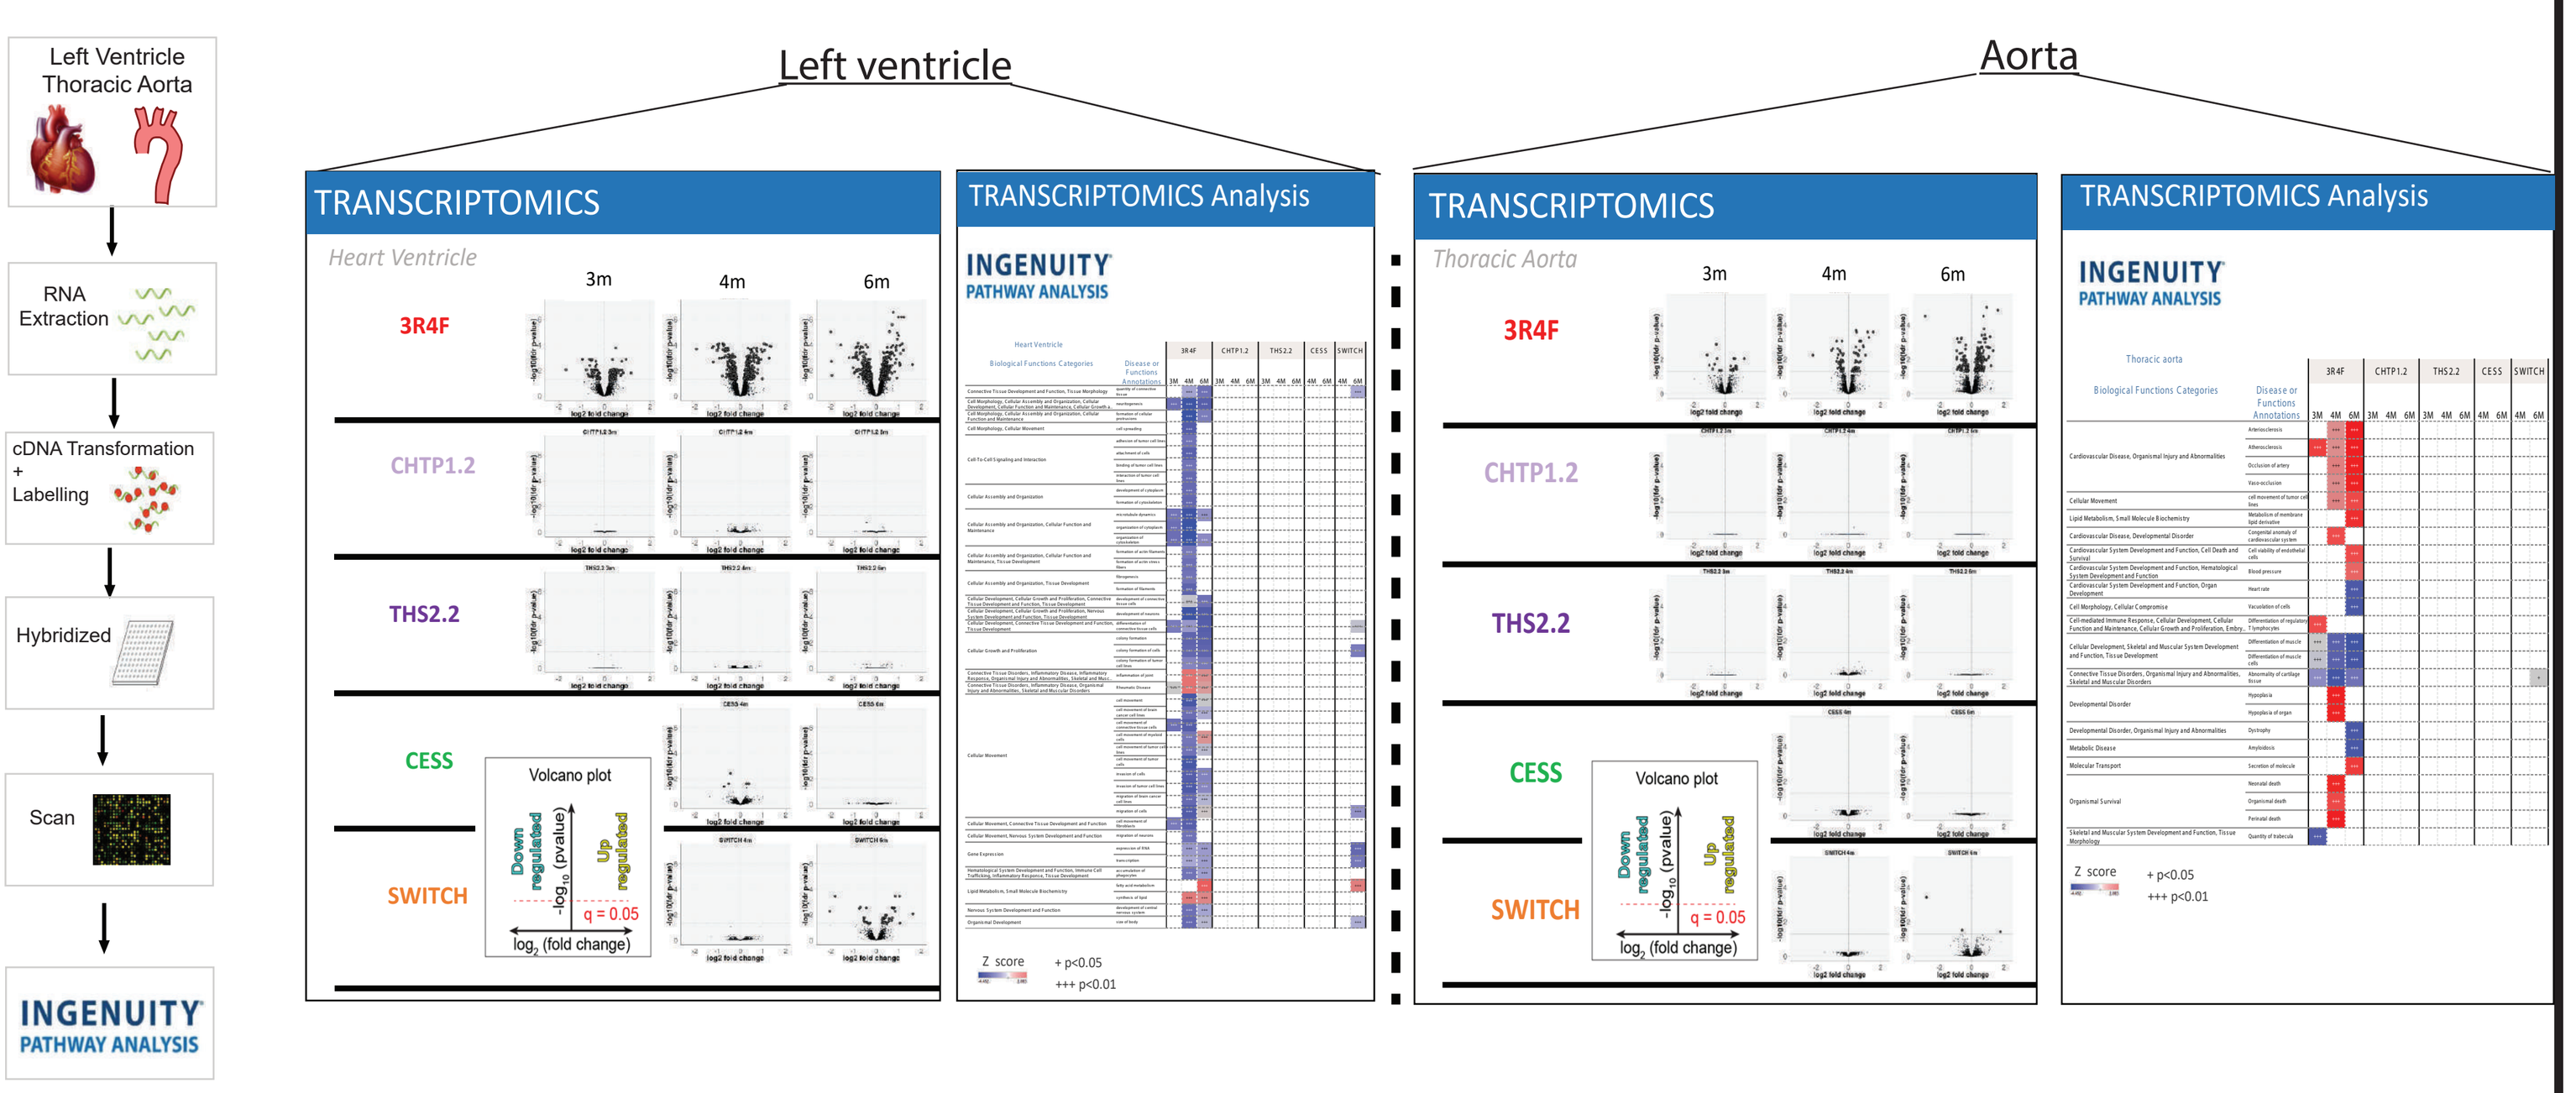
Left Ventricle Thoracic Aorta
RNA Extraction
cDNA Transformation + Labelling
Hybridized
Scan
INGENUITY PATHWAY ANALYSIS
Left ventricle
Aorta
TRANSCRIPTOMICS
TRANSCRIPTOMICS Analysis
Heart Ventricle
Thoracic Aorta
3R4F
CHTP1.2
THS2.2
CESS
SWITCH
Volcano plot

Exposure to 3R4F CS resulted in a time-dependent increase in the number of differentially expressed genes (compared to the sham-exposed mice) in the heart ventricle and the thoracic aorta. The most differentially expressed gene profile was observed in 3R4F at six months in both tissues. In mice exposed to the heat-not-burn (CHTP 1.2, THS 2.2) aerosol, no differentially expressed genes were detected compared with the sham groups at all time points evaluated in accordance with the false discovery rate<0.05. In the cessation and switching groups, the number of dysregulated genes is strongly decreased in comparison with those observed following 3R4F exposure.

Ingenuity Pathways Analysis® (IPA) of the transcriptomics results from the heart and the thoracic aorta demonstrated that mainly a 3R4F exposure-mediated effect was present in both tissues. Based on IPA of gene expression data, biological processes related to "cardiovascular system development and function," "occlusion of artery," "atherosclerosis," "connective tissue development and function, tissue morphology," and "cellular assembly and organization" were significantly affected by 3R4F CS exposure in the thoracic aorta and heart ventricle but not by exposure to aerosol from CHTP 1.2.

Cessation or switching decreased atherosclerotic plaques and restored transcriptomic profiles to profiles similar to those observed in air-exposed animals.

Conclusions

Exposure to 3R4F CS resulted in significant impact on respiratory and CVD parameters: atherosclerotic plaque progression, lung inflammation, lung function. Continuous exposure to heat-not-burn tobacco products (CHTP 1.2 and THS 2.2) resulted in a very small difference in all measured parameters related to CVD or COPD when compared with fresh air-exposed animals.The biological response to switching to CHTP 1.2 (after three months of 3R4F CS exposure) were similar to those observed in the cessation group across the spectrum of endpoints assessed and showed a generally positive effect with respect to continuous smoke exposure. Differential "omics" profiles associated with 3R4F exposure returned to nearly fresh air levels following switching to CHTP 1.2 or fresh air (cessation). These data collectively indicated a halting or regression of CVD parameters following switching from CS to CHTP 1.2 aerosol in ApoE$^{-/-}$ mice.

References

1. Aistrup GL, Balke CW, Wasserstrom JA. Arrhythmia triggers in heart failure: the smoking gun of [Ca2+]i dysregulation. Heart rhythm. 2011;8(11):1804-8.
2. Al Hariri M, Zibara K, Farhat W, Hashem Y, Soudani N, Al Ibrahim F, et al. Cigarette Smoking-Induced Cardiac Hypertrophy, Vascular Inflammation and Injury Are Attenuated by Antioxidant Supplementation in an Animal Model. Frontiers in pharmacology. 2016;7:397.
3. Bakhru A, Erlinger TP. Smoking cessation and cardiovascular disease risk factors: results from the Third National Health and Nutrition Examination Survey. PLoS medicine. 2005;2(6):e160.
4. Bleyer AJ, Shemanski LR, Burke GL, Hansen KJ, Appel RG. Tobacco, hypertension, and vascular disease: risk factors for renal functional decline in an older population. Kidney international. 2000;57(5):2072-9.
5. Boue S, De Leon H, Schlage WK, Peck MJ, Weiler H, Berges A, et al. Cigarette smoke induces molecular responses in respiratory tissues of ApoE$^{-/-}$ mice that are progressively deactivated upon cessation Toxicology. 2013;314:112 - 24.
6. Phillips BW, Schlage WK, Titz B, Kogel U, Sciuscio D, Martin F, et al. A 90-day OECD TG 413 rat inhalation study with systems toxicology endpoints demonstrates reduced exposure effects of the aerosol from the carbon heated tobacco product version 1.2 (CHTP 1.2) compared with cigarette smoke. I. Inhalation exposure, clinical pathology and histopathology. Food and chemical toxicology : an international journal published for the British Industrial Biological Research Association. 2018.

PMI SCIENCE PHILIP MORRIS INTERNATIONAL

Philip Morris International is the sole source of funding and sponsor of this project.

EUROTOX 2018
Brussels
2-5 September 2018