Characterization of clones derived from human bronchial epithelial cells after long-term treatment with TPM from reference cigarettes versus THS 2.2

Marco van der Toorn, Marie Sgandurra, Diego Marescotti, Manuel Peitsch, Julia Hoeng and Karsta Luettich

PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland (Part of Philip Morris International group of companies)

Abstract

The prevailing risk factor for lung cancer in humans is cigarette smoking. Lung cancer is a complex genetic disease characterized by genetic and epigenetic aberrations that are reflected in morphological and phenotypic changes of the bronchial and alveolar epithelium (1).

We successfully established an *in vitro* model to compare the effect of long-term exposure to total particulate matter (TPM) from 3R4F reference cigarettes with that from Tobacco Heating System 2.2 (THS 2.2) on cellular transformation (2). The study demonstrated that repeated exposure of BEAS-2B cells to THS 2.2 TPM, with a 20-fold higher concentration than 3R4F TPM, induced anchorage independence. Transformed cells were not observed following prolonged treatment of BEAS-2B cells with THS 2.2 TPM at the same or even at 5-fold higher concentration than with 3R4F TPM. Viable transformed cells (clones) were recovered from a single ancestor cell, making it possible to isolate and establish uniform cell populations. In total, 27 3R4F TPM-derived clones and 31 THS 2.2 TPM-derived clones were recovered.

We have established assay capabilities to characterize individual clones based on their growth properties and invasion phenotype. For example, population doubling time (PDT) was assessed using cell growth curves, expression of the EMT markers E-cadherin and vimentin was evaluated by antibody-based high-content screening, and invasion assays were run in xCelligence® CIM chambers. Metabolic activity of the clones was examined using Seahorse FX technology (Agilent, Santa Clara, CA, USA). In addition, Short Tandem Repeat (STR) profiling was performed by ATCC (Manassas, VA, USA) to verify the relatedness to the parental BEAS-2B cells.

Our data showed that all 3R4F TPM-derived clones had increased PDT, compared with 80% of THS 2.2 clones. Vimentin expression was increased in >90% of 3R4F as well as in all THS 2.2 TPM-derived clones. However, high expression of vimentin and low expression of E-cadherin did not correlate with the invasive capacity of the clones as measured by the invasion assay. Twenty-five of 27 3R4F and 29 of 31 THS 2.2 TPM-derived clones matched the STR profile of the parental BEAS-2B cells.

Future work will entail exome sequencing as well as *in vivo* xenograft studies to further elucidate the tumor-initiating and metastatic potential of the 3R4F and THS 2.2 TPM-derived clones.

Experimental Design

- BEAS-2B cells were cultured in cell culture flasks for 12 weeks with continuous treatment with 7.5 µg/mL TPM from the reference cigarette 3R4F or three doses of TPM from THS 2.2 (7.5, 37.5, and 150 µg/mL). Control cells treated with medium only or dimethyl sulfoxide (DMSO) were cultured separately in cell culture flasks.
- BEAS-2B cells treated with 7.5 µg/mL 3R4F TPM or 150 µg/mL THS 2.2 TPM for 12 weeks exhibited anchorage independence. Transformed cells were not observed following prolonged treatment of BEAS-2B cells with the same (7.5 µg/mL) or a 5-fold (37.5 µg/mL) higher concentration of THS 2.2 TPM.

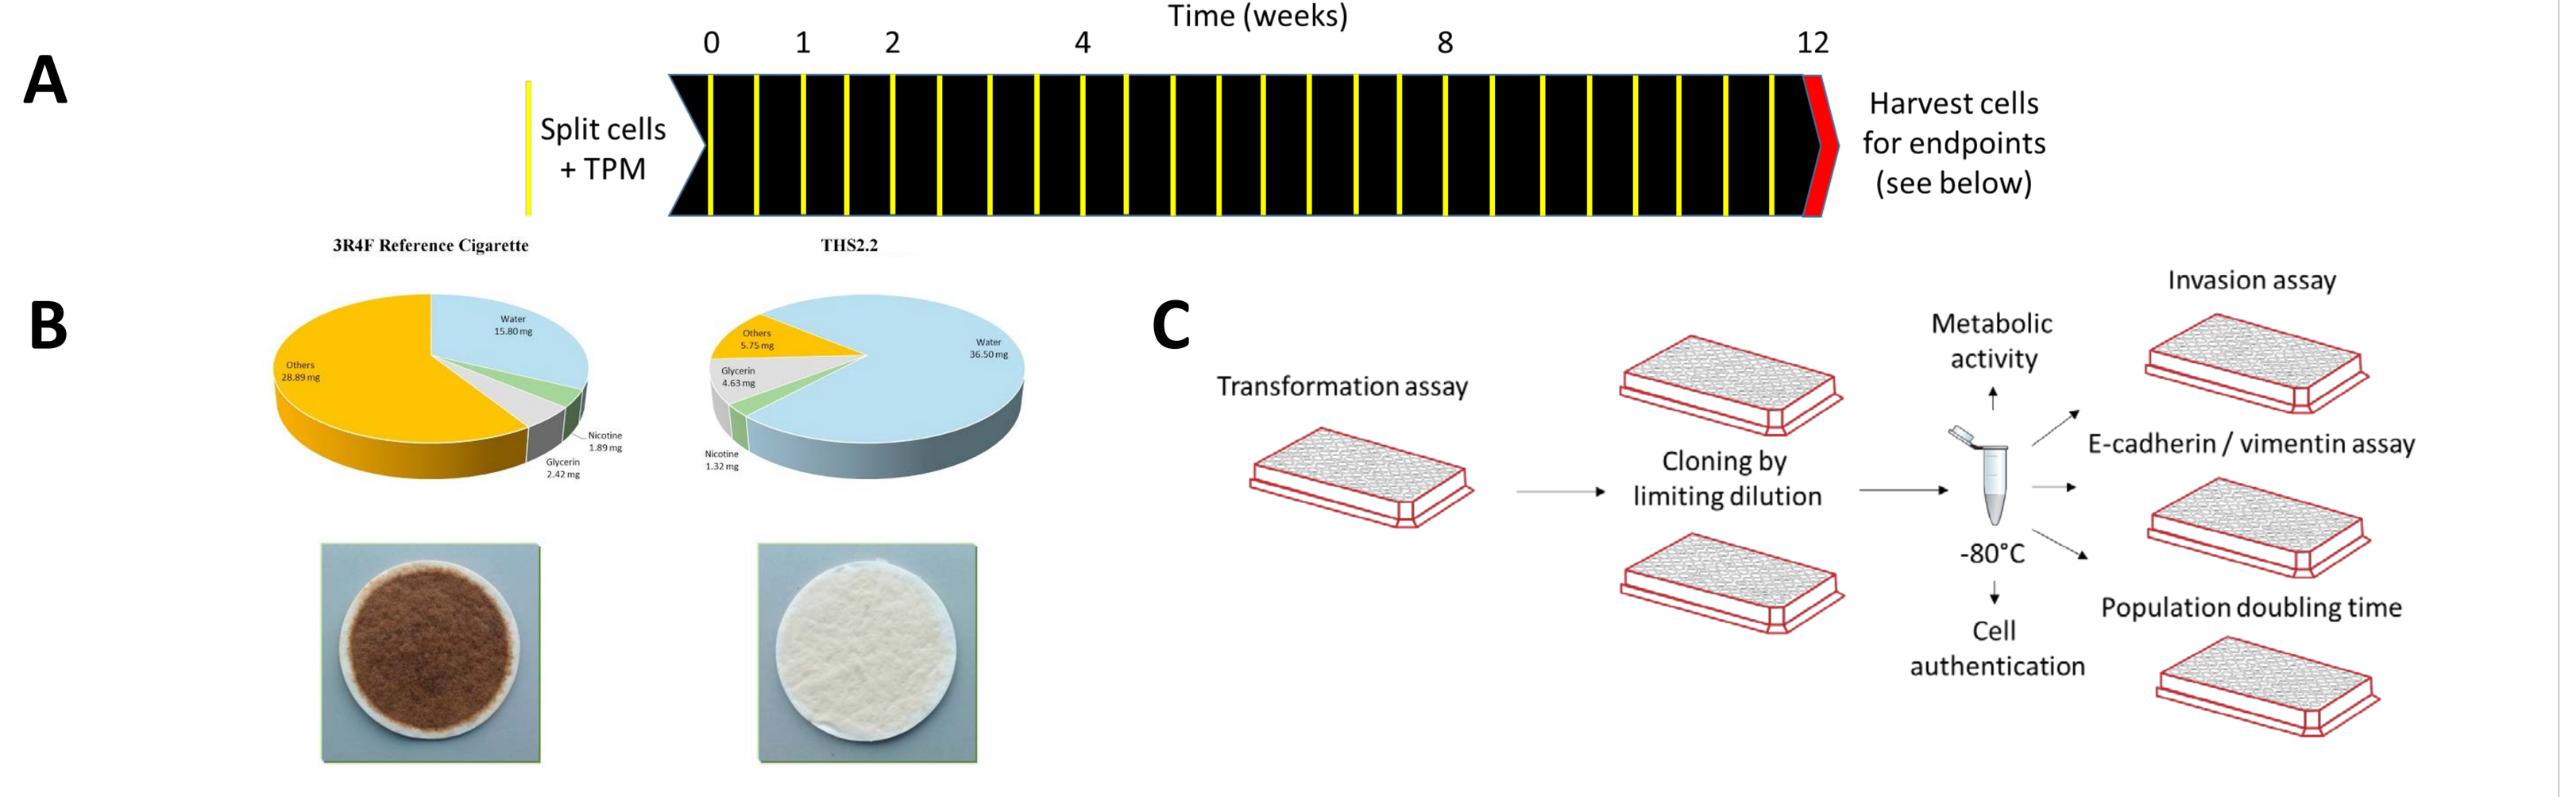
A
Time (weeks)
0
1
2
4
8
12
Split cells + TPM
Harvest cells for endpoints (see below)
B
3R4F Reference Cigarette
THS2.2
C
Transformation assay
Cloning by limiting dilution
Metabolic activity
-80°C
Cell authentication
Invasion assay
E-cadherin / vimentin assay
Population doubling time

Figure 1. *Schematic of experimental design for long-term exposure of BEAS-2B cells to TPM from reference cigarette 3R4F and THS 2.2.* ***A)*** *Twelve-week exposure of BEAS-2B cells. Yellow lines represent splitting of the cells and refreshment of TPM treatment. Red line represents harvesting of cells for further analysis, including soft agar assay.* ***B)*** *Cigarette TPM composition compared with that obtained from THS 2.2 aerosol, generated according to the HCI machine-smoking conditions. The photographs of the Cambridge glass-fiber filter pads after the collection of cigarette smoke (left) and THS 2.2 aerosol (right) are also shown (3).* ***C)*** *Schematic overview of individual clone characterization based on their invasion phenotype and growth properties.*

Cell Recovery from the Transformation Assay and Re-plating: Isolation and recovery of colony-forming BEAS-2B cells from the transformation assay were performed according to the manufacturer's instructions for the CytoSelect™ 96-well Cell Transformation Assay (Cell Biolabs, San Diego, CA, USA). Successfully isolated colonies were grown in the bronchial epithelial cell growth medium, BEGM™ (Lonza, Basel Switzerland) on collagen I-coated culture flasks at 37°C/5% CO2 until reaching 75–80% confluence. To obtain a monoclonal BEAS-2B cell population, suspensions of colony-forming BEAS-2B cells were prepared in 100 mL medium containing 100 or 10 viable BEAS-2B cells. The cell suspensions were dispensed into 96-well plates (100 µL/well) at a density of 0.1 and 1 cell/mL. The plates were incubated for 14 days without changing the medium. During this time, the plates were monitored for growth of isolated clones. Cells were collected from wells containing single clones and cryopreserved for further analysis.

PDT: Population doubling time (PDT) was calculated using the linear portion of the growth curve by this equation: $t = H \times \ln 2 / \ln(c2/c1)$, where t is time, H is the duration, and c is the number of cells.

Cell Authentication: Parental BEAS-2B, BEAS-2B cells treated with vehicle for 12 weeks, and single clones were sent to the ATCC for cell authentication (ATCC Manassas, Virginia, USA).

Cell Invasion: The rate of cell invasion was monitored in real time using cell invasion migration (CIM) plates and a real-time cell analyzer dual-plate (RTCA-DP) xCELLigence® instrument (Bucher Biotec). The electrode surface of the CIM plate was coated with collagen A for 1 hour. The membrane of the CIM plate was coated overnight with Geltrex® Ready-To-Use as an extracellular matrix. Parental BEAS-2B, BEAS-2B cells treated with vehicle for 12 weeks, and single clones were grown overnight in bronchial epithelial basal medium (BEBM™). The next day, 40,000 cells were seeded in each well of the upper chamber in BEBM™. The lower chambers were filled with BEBM™ containing 4% FCS or no serum as the negative control. Changes in the cell index (impedance), which reflects cell invasion, were recorded in real-time and analyzed using RTCA software v2.0 (Bucher Biotec).

Vimentin and E-cadherin Expression: Parental BEAS-2B, BEAS-2B cells treated with vehicle for 12 weeks, and single clones were seeded in black collagen I-coated, clear bottom 96-well tissue culture plates (BD, Allschwil, Switzerland) at a density of 12,000 cells/well. The next day, cells were stained for E-cadherin (HECD-1; AbCam, Cambridge, UK) and vimentin (ab45939; AbCam) expression. Vehicle-treated cells (12 weeks) were used as control. Following staining of the cells, fluorescence data were acquired with a Thermo Fisher Cellomics® ArrayScanVTI High Content Screening platform (Thermo Fisher Scientific Inc., Waltham, MA, USA) and vHCS view software (Thermo Fisher Scientific Inc.). Twenty fields were imaged per well using a ×10 wide field objective.

Metabolic Activity: The single clones derived from 3R4F and THS 2.2 TPM exposure as well as the parental BEAS-2B and the DMSO-treated cells were processed according to the manufacturer's instructions (Agilent, Santa Clara, CA, USA) to assess the metabolic phenotype, using the Seahorse Cell Energy Phenotype Test Kit. Data produced were then exported to Excel for analysis.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

Results

Cell authentication

- STR analysis was used to verify the parental BEAS-2B cells and evaluate the relatedness of the derived clones to the parental cells.
- Twenty-five of 27 3R4F and 29 of 31 THS 2.2 TPM-derived clones matched the STR profile of the parental BEAS-2B cells.

Table 1*. Percentage of shared short tandem repeats on loci between individual clones and parental BEAS-2B cell line. For comparison purposes, the following cut-off values were applied: profile is an exact match with the parental cell line [95% - 100%], profile is related to the parental cell line [80% - 95%], profile is not considered to be related to the parental cell line [80% - 0%].*

Loci shared with BEAS-2B	3R4F TPM-derived clones	THS2.2 TPM-derived clones
[95-100%]	25/27	29/31
[80-95%]	2/27	0/31
[0-80%]	0/27	0/31

Population doubling time

- Population doubling time (PDT) was used to predict how fast parental BEAS-2B cells and individual clones grow.
- Almost all 3R4F TPM-derived clones had increased PDT compared with ~80% of THS 2.2 clones.

Table 2*. PDT of individual clones and parental BEAS-2B cell line. For comparison purposes, the following cut-off values were applied: <24 h, 24 – 48 h, and > 48 h PDT.*

Population doubling time	Parental BEAS-2B	3R4F TPM-derived clones	THS2.2 TPM-derived clones
[0 – 24 h]	-	0/27	1/31
[24 – 48 h]	1	0/27	7/31
[48h – 72 h]	-	26/27	23/31

Cell Invasion

- Cell Invasion was monitored in real time to study the intravasation/extravasation properties of BEAS-2B clones.
- 3R4F TPM-derived clones were less invasive than the THS 2.2 TPM-derived clones.

Table 3. *Real-time cell invasion of individual clones and parental BEAS-2B cell line. For comparison purposes, the following cut-off values were applied: invasion speed <0.05, 0.05 – 0.15, and > 0.15 1/h.*

Invasion speed (1/h)	Parental BEAS-2B	3R4F TPM-derived clones	THS2.2 TPM-derived clones
[<0.05]	1	12/27	9/31
[0.05 – 0.15]	-	13/27	12/31
[>0.15]	-	2/27	8/31

Expression of epithelial-mesenchymal transition markers

- Epithelial–mesenchymal transition is implicated in fostering a microenvironment capable of promoting tumor formation and is characterized by the suppression of epithelial (e.g., E-cadherin) and the induction of mesenchymal (e.g., vimentin) cell markers.
- More than 70% of the 3R4F and THS 2.2 TPM-derived clones were mesenchymal in phenotype.

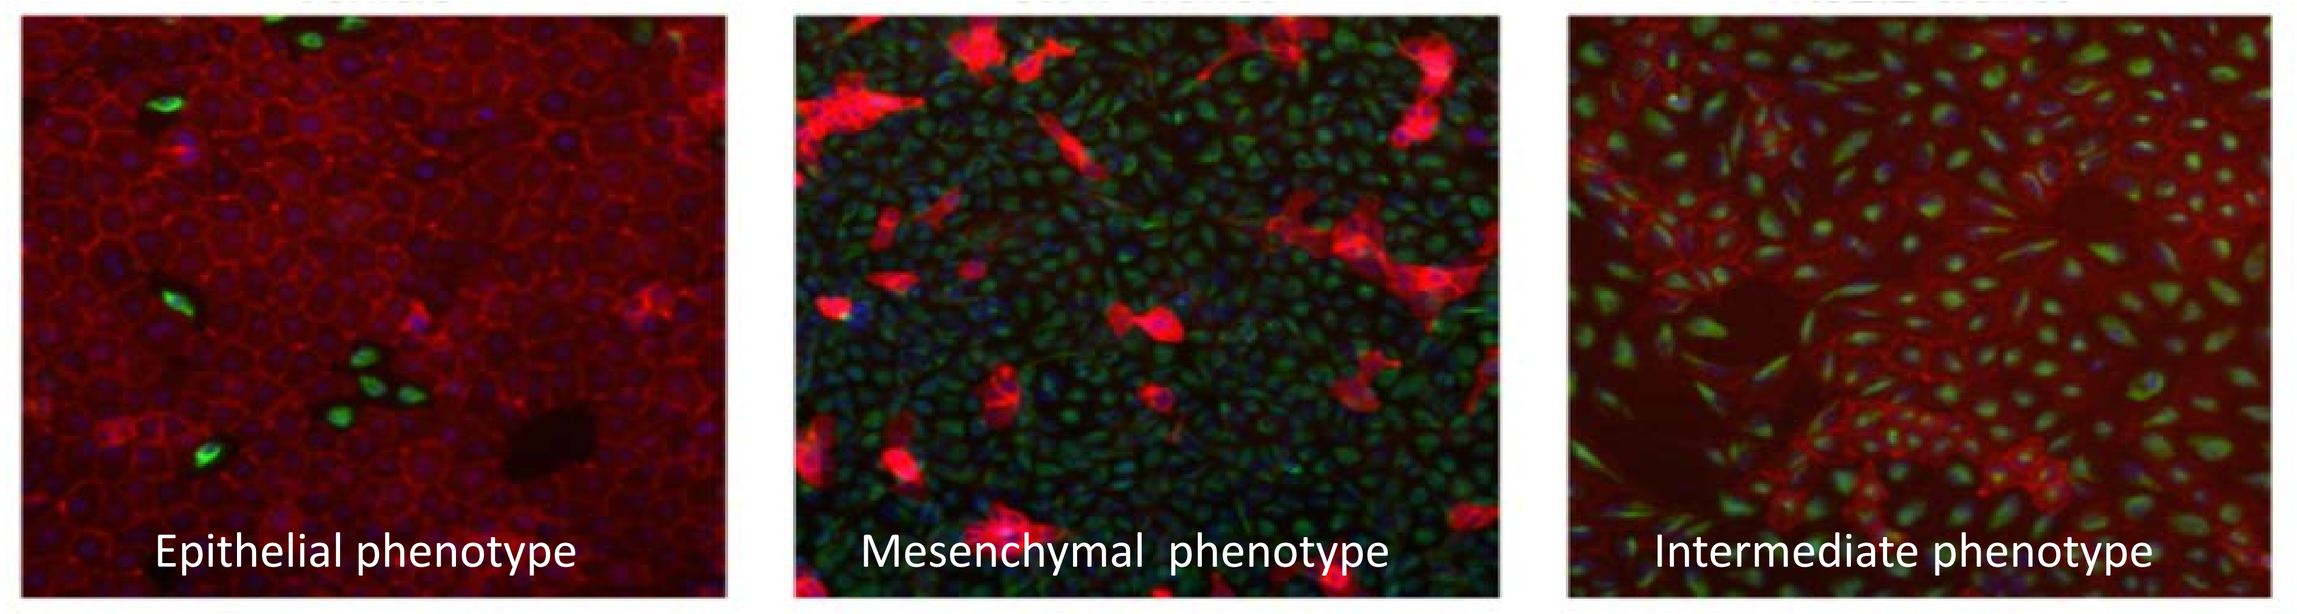
Epithelial phenotype
Mesenchymal phenotype
Intermediate phenotype

Figure 2. *Detection of different epithelial-mesenchymal transition phenotypes using vimentin and E-cadherin markers and high-content imaging.*

EMT phenotype	Parental BEAS-2B	3R4F TPM-derived clones	THS2.2 TPM derived-clones
[Epithelial]	1	1/27	0/31
[Intermediate]	-	7/27	8/31
[Mesenchymal]	-	19/27	22/31

Table 4. *Detection of different epithelial-mesenchymal transition phenotypes. For comparison purposes, the following cut-off values were applied: <25%, 25–75%, and >75% (vimentin); >95 mean fluorescence intensity (MFI), 50–95 MFI, and <50 MFI (E-cadherin).*

Metabolic activity

- Metabolic potential of individual BEAS-2B clones to meet an energy demand via respiration and/or glycolysis was examined on the Seahorse platform.
- Approximately 80% of 3R4F TPM-derived clones meet their energy demands by both respiration and glycolysis. About 50% and 80% of THS 2.2 TPM-derived clones meet their energy demands by respiration and glycolysis, respectively.

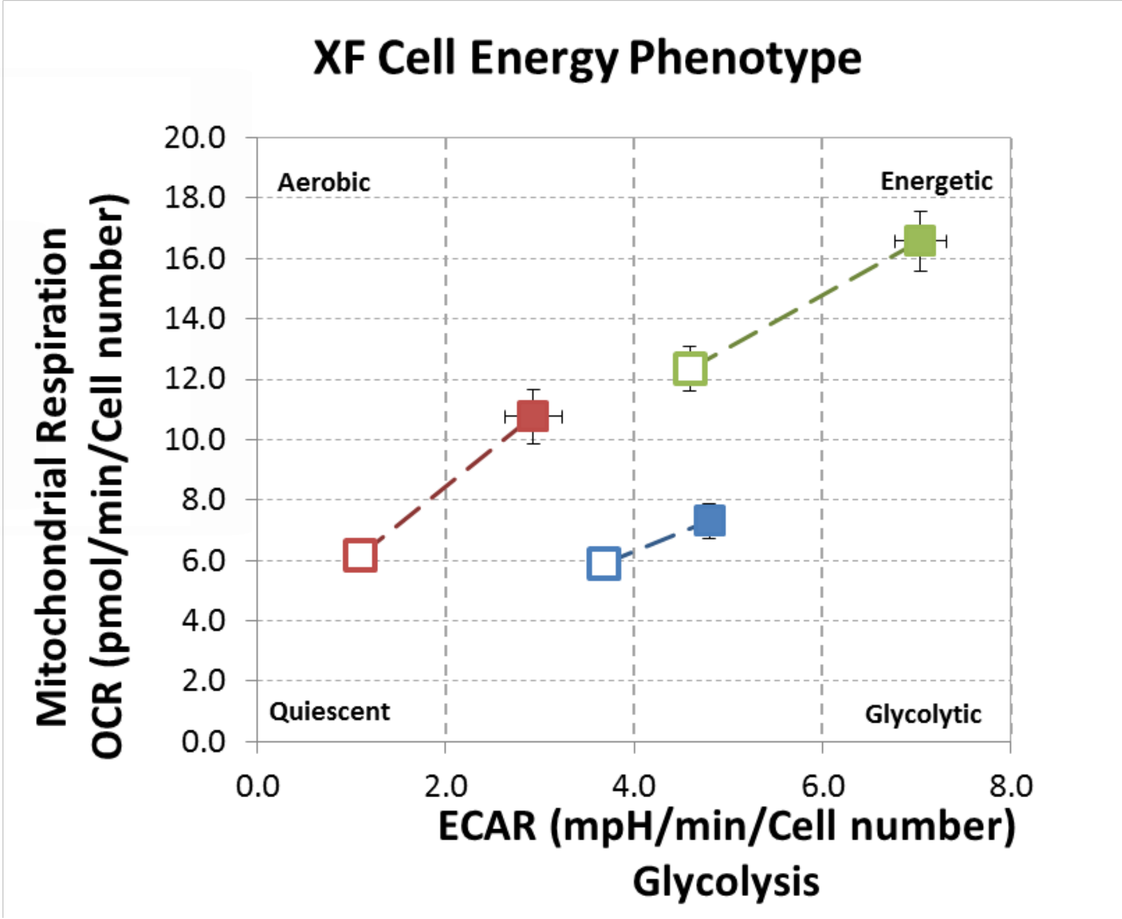
XF Cell Energy Phenotype
Mitochondrial Respiration
OCR (pmol/min/Cell number)
ECAR (mpH/min/Cell number)
Glycolysis
Aerobic
Energetic
Quiescent
Glycolytic
20.0
18.0
16.0
14.0
12.0
10.0
8.0
6.0
4.0
2.0
0.0
0.0
2.0
4.0
6.0
8.0

Figure 3. *Representative normalized metabolic phenotype profile of two clones (red and green) and parental BEAS-2B cells (blue). The baseline phenotype profile shows differences in both glycolysis (ECAR) and mitochondrial respiration (OCR). Open square: baseline conditions. Closed square: stressed conditions.*

Table 5*. Percentage of metabolic potential of individual clones and parental BEAS-2B cell line. For comparison purposes, the following cut-off values were applied: respiration (OCR) is a match with the parental cell line [<125%], respiration is increased compared to the parental cell line [125% - 150%], respiration is highly increased compared to the parental cell line [>150%]; glycolysis (ECAR) is a match with the parental cell line [<125%], glycolysis is increased compared to the parental cell line [125% - 150%], glycolysis is highly increased compared to the parental cell line [>150%]. OCR=Oxygen Consumption Rate, ECAR=Extracellular Acidification Rate*

Metabolic potential (% baseline OCR)	Parental BEAS-2B	3R4F-TPM derived clones	THS2.2-TPM derived clones
[<125]	1	5/27	12/31
[125 - 150]	-	21/27	12/31
[>150]	-	0/27	4/31
Metabolic potential (% baseline ECAR)	**Parental BEAS-2B**	**3R4F-TPM derived clones**	**THS2.2-TPM derived clones**
[<125]	1	4/27	4/31
[125 - 175]	-	13/27	17/31
[>175]	-	9/27	7/31

Summary

- More then 70% of the 3R4F and 60% of the THS 2.2 TPM-derived clones exhibit more than five out of seven phenotypic differences when compared with parental BEAS-2B cells. Only one 3R4F clone exhibits seven out of seven phenotypic differences when compared with parental BEAS-2B cells.

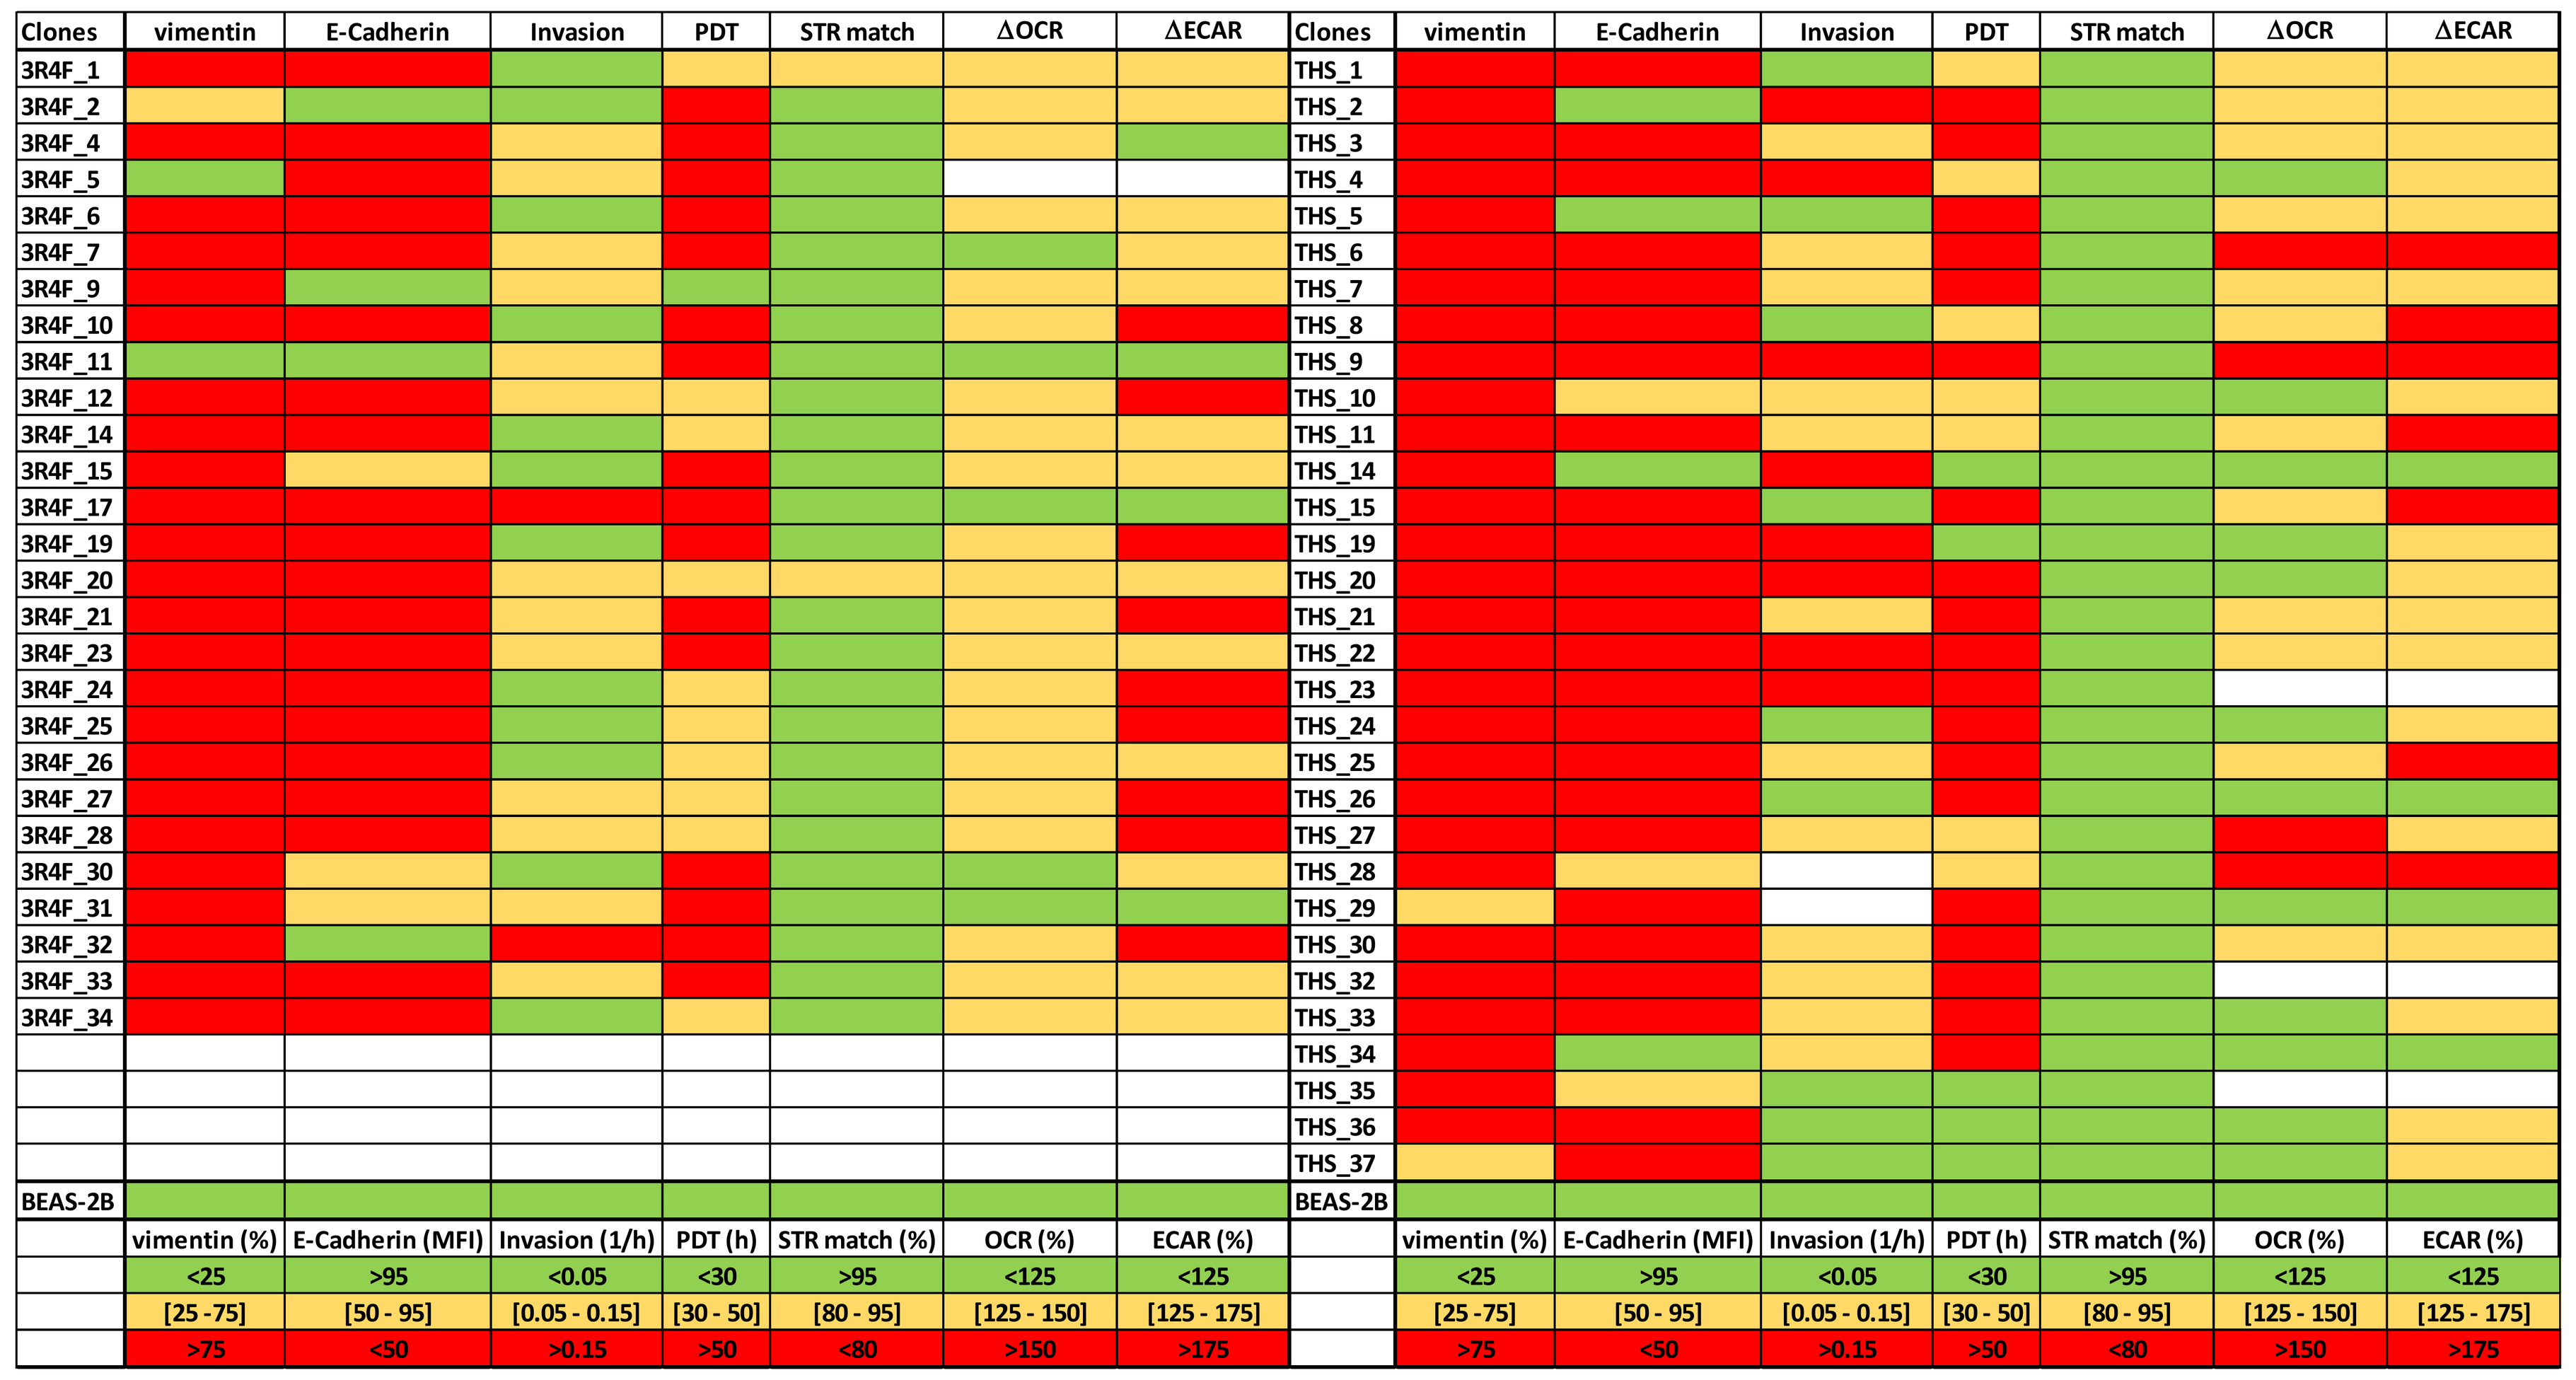
Clones
vimentin
E-Cadherin
Invasion
PDT
STR match
ΔOCR
ΔECAR
3R4F_1
3R4F_2
3R4F_4
3R4F_5
3R4F_6
3R4F_7
3R4F_9
3R4F_10
3R4F_11
3R4F_12
3R4F_14
3R4F_15
3R4F_17
3R4F_19
3R4F_20
3R4F_21
3R4F_23
3R4F_24
3R4F_25
3R4F_26
3R4F_27
3R4F_28
3R4F_30
3R4F_31
3R4F_32
3R4F_33
3R4F_34
BEAS-2B
THS_1
THS_2
THS_3
THS_4
THS_5
THS_6
THS_7
THS_8
THS_9
THS_10
THS_11
THS_14
THS_15
THS_19
THS_20
THS_21
THS_22
THS_23
THS_24
THS_25
THS_26
THS_27
THS_28
THS_29
THS_30
THS_32
THS_33
THS_34
THS_35
THS_36
THS_37
vimentin (%)
E-Cadherin (MFI)
Invasion (1/h)
PDT (h)
STR match (%)
OCR (%)
ECAR (%)
<25
>95
<0.05
<30
>95
<125
<125
[25 -75]
[50 - 95]
[0.05 - 0.15]
[30 - 50]
[80 - 95]
[125 - 150]
[125 - 175]
>75
<50
>0.15
>50
<80
>150
>175

Figure 4*. Summary of the clone characterization. Green reflects the parental BEAS-2B endpoint value, yellow indicates a small difference, red represents a substantial difference compared with the parental BEAS-2B endpoint value, and white indicates a missing data point.*

Conclusions

- Transformed cells were not observed following prolonged treatment of BEAS-2B cells with the same or a 5-fold higher concentration of THS 2.2 TPM with respect to 3R4F TPM.
- All 3R4F TPM-derived clones had increased PDT but were less invasive compared to 20-fold higher THS 2.2 TPM-derived clones. More then 70% of the 3R4F and more then 60% of the 20-fold higher THS 2.2 TPM-derived clones had >5 out of 7 phenotypic differences when compared to parental BEAS-2B cells. Future work will entail exome sequencing and *in vivo* xenograft studies to further elucidate the tumor-initiating and metastatic potential of the clones.

References

(1) *Smith AL et al., (1996) Extensive areas of aneuploidy are present in the respiratory epithelium of lung cancer patients. British journal of cancer. 73: 203-209*

(2) *Toorn van der M et al., (2018) The Biological Effects of Long-Term exposure of Human Bronchial Epithelial Cells to Total Particulate Matter from a candidate Modified-Risk Tobacco Product. Toxicology in vitro. Under review*

(3) *Schaller et al., (2017) Evaluation of the Tobacco Heating System 2.2. Part 2: Chemical composition, genotoxicity, cytotoxicity, and physical properties of the aerosol. Regul Toxicol Pharmacol. Nov 30;81 Suppl 2:S27-S47.*

Society of Toxicology Meeting
San Antonio, TX, USA
March 11-15, 2018

Competing Financial Interest
Philip Morris International is the sole source of funding and sponsor of this research.