A Multi-Center, Multi-Regional, Study on Biological and Functional Changes in Healthy Adult Smokers during One Year of Continuous Smoking Abstinence

C.T. Tran, L. Felber Medlin, N. Lama, B. Taranu, E. Pierri, J. Ancerewicz, P. Picavet, G. Baker, C. Haziza, F. Lüdicke

PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland (Part of Philip Morris International group of companies)

Introduction and Objectives

Background

The harm from smoking mainly results from long-term exposure to Harmful and Potentially Harmful Constituents (HPHCs) contained in cigarette smoke generated by the combustion of tobacco. Smoking Cessation (SC) is the most effective way to reduce the harm and risk of smoking-related diseases to a relative risk level which may approach, over time, that of never-smokers. In most SC studies the main focus is on the successful quitting rate of the SC treatment being tested. However, only limited information on short- to long-term functional/biological changes following SC is available in the literature.

Main Objectives

The overall aim of this study was to assess over a one-year period of continuous smoking abstinence the reversibility of the harm related to smoking by assessing changes of Biomarkers of Exposure (BoExp) to HPHCs and Clinical Risk Endpoints (CREs) which are linked to pathophysiological pathways of smoking-related diseases. These BoExp and CREs were selected according to epidemiological evidence that the endpoints are associated with smoking-related diseases, and the expectation that these effects are reversed by smoking cessation over a period of time within the study duration.

BoExp to HPHCs Assessed	Clinical Risk Endpoints
Carbon Monoxide (CO) in exhaled breath Urinary Biomarkers: • NEQ • MHBMA • 3-HPMA • CEMA • Total 3-OH-B[a]P • Total 1-OHP • 3-HMPMA • Total NNN	Associated with cardiovascular diseases: • Lipid Metabolism: HDL-C, LDL-C, Apo A1, Apo B • Inflammation: WBC, hs-CRP, Homocysteine • Platelet function: Platelets, Fibrinogen, 11-DTX-B2 • Oxidative stress: 8-epi-PGF$_{2\alpha}$, MPO • Endothelial dysfunction: s-ICAM-1, Albumin • Metabolic Syndrome: HbA1c • Acute Cardiovascular effect: COHb Associated with respiratory diseases: • Spirometry (pre- and post-bronchodilator): FEV1 Associated with genotoxicity: • Total NNAL

Study Conduct

The study was approved by Independent Ethics Committees and Institutional Review Boards in all participating countries and was initiated in May 2015. The study was conducted according to the principles of ICH-GCP and registered on ClinicalTrials.gov (NCT02432729).

Harmful and potentially harmful constituents (Biomarker [Abbreviation]):
Carbon Monoxide (CO); Nicotine equivalents (NEQ = molar sum of free nicotine, nicotine-glucuronide, free cotinine, cotinine-glucuronide, free trans-3'-hydroxycotinine, trans-3'-hydroxycotinine-glucuronide); 1,3-Butadiene (Monohydroxybutenylmercapturic acid [MHBMA]); Acrolein (3-Hydroxypropylmercapturic acid [3-HPMA]); Acrylonitrile (2-Cyanoethylmercapturic acid [CEMA]); Benzo[a]pyrene (Total 3-hydroxybenzo(a)pyrene [Total 3-OH-B[a]P]); Pyrene (Total 1-Hydroxypyrene [Total 1-OHP]); Crotonaldehyde (3-Hydroxy-1-methylpropylmercapturic acid [3-HMPMA]); NNN (Total N-nitrosonornicotine [Total NNN]).

Clinical Risk Endpoints (Abbreviation):
High density lipoprotein cholesterol (HDL-C); Low density lipoprotein cholesterol (LDL-C); Apolipoprotein A1 (Apo A1); Apolipoprotein B (Apo B); White blood cell count (WBC); High sensitivity C-reactive protein (hs-CRP); 11-dehydrothromboxane B2 (11-DTX-B2); 8-epi-prostaglandin F2alpha (8-epi-PGF$_{2\alpha}$); Myeloperoxidase (MPO); Soluble intercellular adhesion molecule-1 (sICAM-1); Glycosylated hemoglobin (HbA1c); Carboxyhemoglobin (COHb); Forced expiratory volume in 1 second (FEV1); Total 4-[methylnitrosamino]-1-[3-pyridyl]-1-butanol (Total NNAL).

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

Methods

Design

This was a multi-region, multi-center (43 sites), ambulatory study conducted in the US, UK, Poland, Germany, and Japan, in healthy adult smokers who were willing to quit smoking and were asked to continuously abstain from smoking during a 52-week (1-year) period in an ambulatory setting followed by a 28-day Safety Follow-Up period.

To support the subjects to stop smoking, Nicotine Replacement Therapy (NRT) was provided at subjects' request and used as per country label for up to 3 months. Additionally, smoking cessation support, including counseling and behavioral support, was provided throughout the study and upon subjects' request.

Participants

- Subjects understood the information provided for the study and signed the Informed Consent Form.
- Subjects were judged healthy by the Investigator.
- Subjects had no disorders or other conditions that would have jeopardized the subjects' safety or affect the validity of the study results as judged by the Investigator.
- Subjects were 30 to 65 years old (inclusive).
- Subjects smoked ≥10 commercially available cigarettes per day for the last 12 months and had been smoking for ≥ 10 years.
- Subjects had negative alcohol and drug tests.
- Female subjects were not pregnant or breast feeding.
- Subjects were willing to quit smoking within the next 30 days.
- Subjects accepted continuous smoking abstinence for 52 weeks.

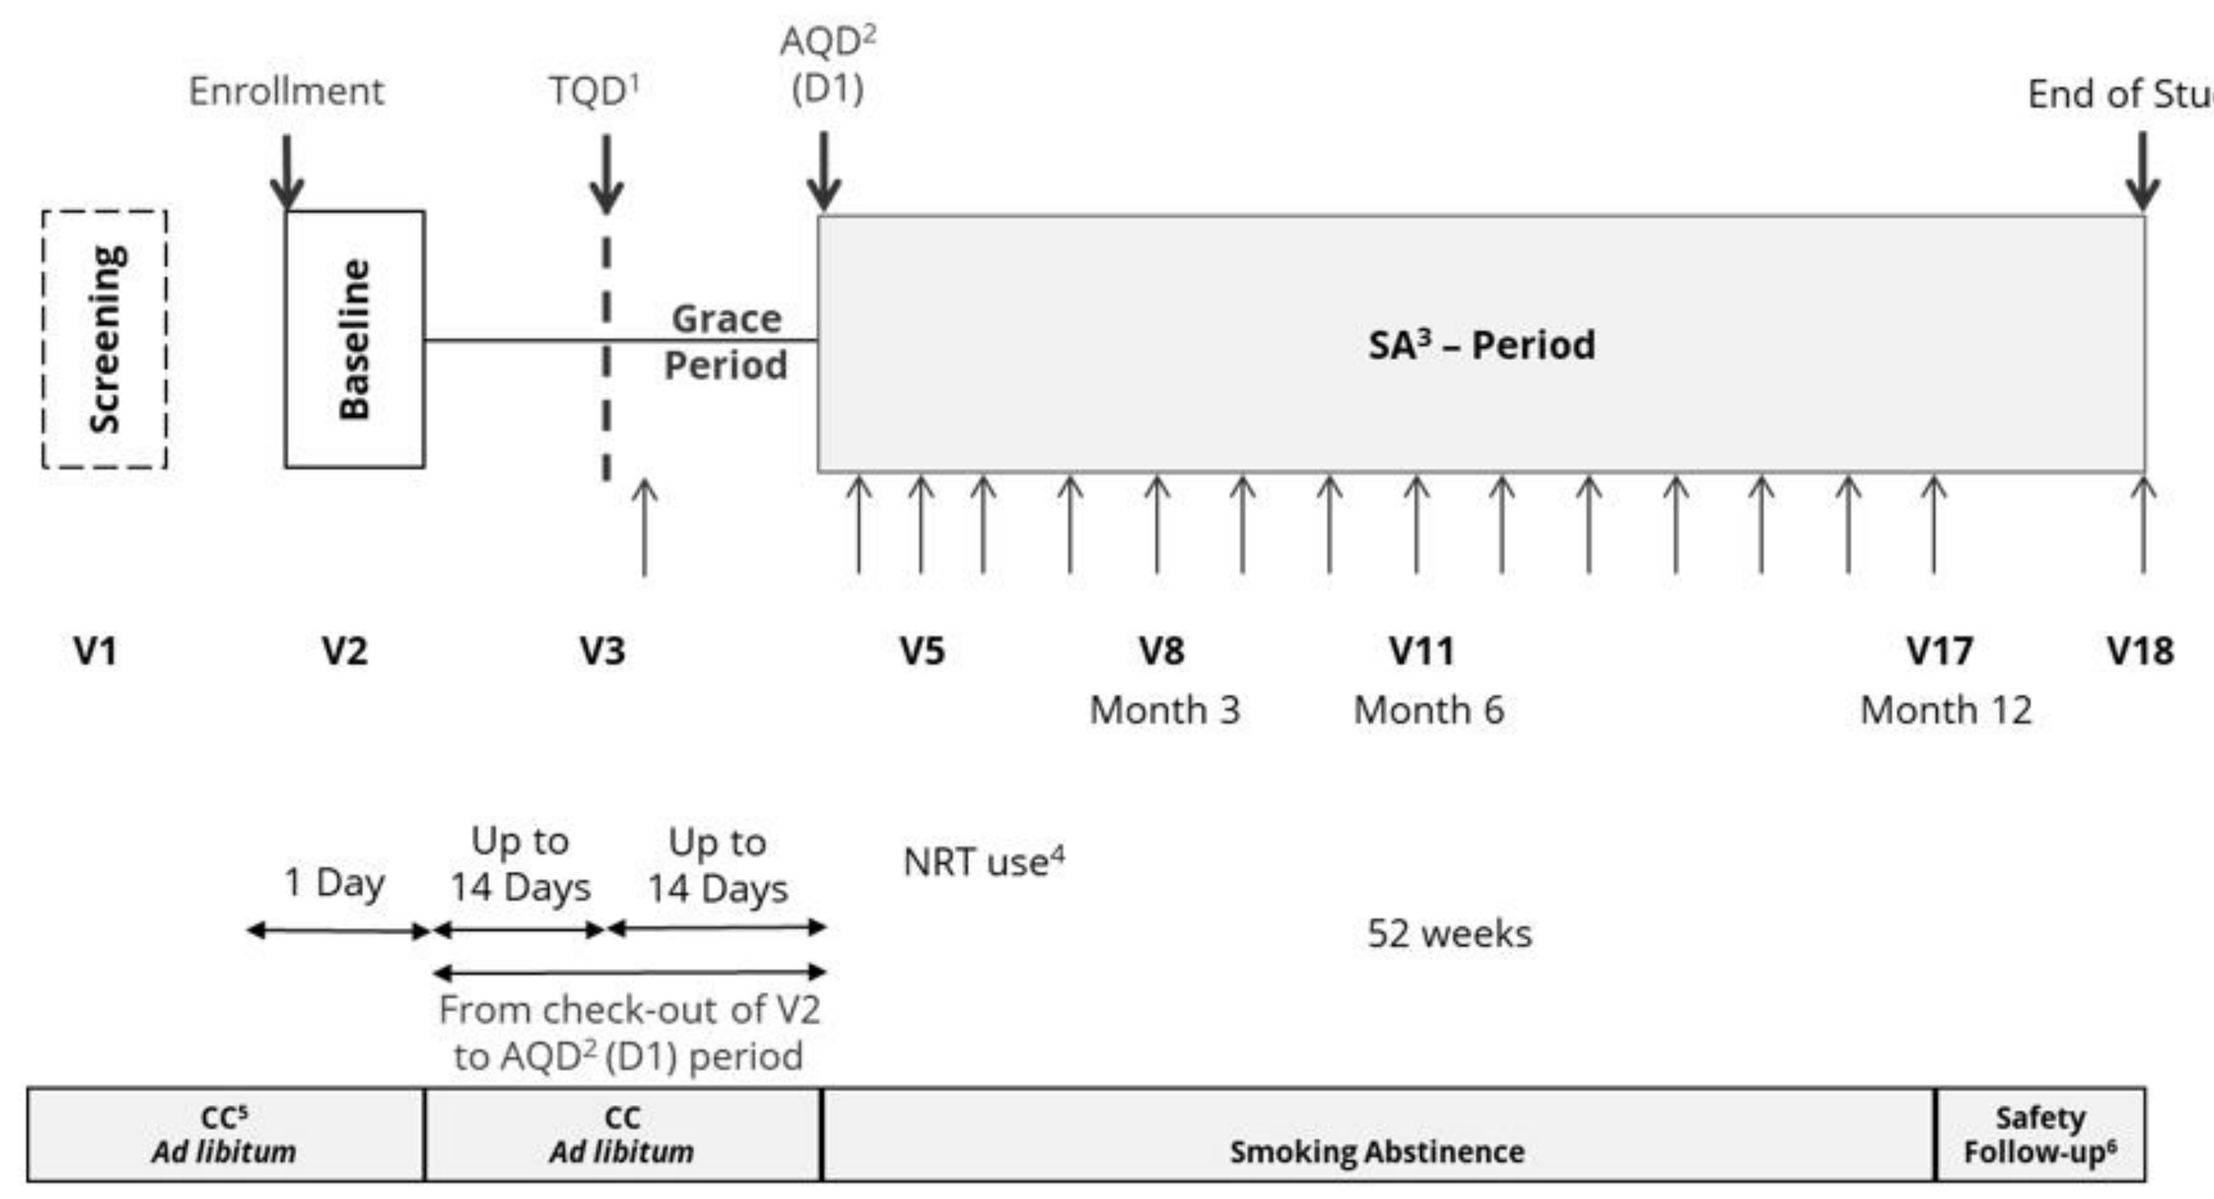
Enrollment
TQD1
AQD2 (D1)
End of Study
Screening
Baseline
Grace Period
SA3 – Period
V1
V2
V3
V5
V8 Month 3
V11 Month 6
V17 Month 12
V18
1 Day
Up to 14 Days
Up to 14 Days
NRT use4
52 weeks
From check-out of V2 to AQD2 (D1) period
CC5 Ad libitum
CC Ad libitum
Smoking Abstinence
Safety Follow-up6

1 Target Quit Date (TQD) is within 1-14 days of check-out of V2
2 Actual Quit Date (AQD) is within 14 days of TQD (grace period with occasional CC use)
3 Smoking Abstinence (SA)
4 Use of Nicotine Replacement Therapy (NRT) will be only allowed for up to 3 months (+2 weeks) after the start date of NRT. NRT may be started at any time between the TQD and 1 week after the AQD.
5 Conventional Cigarettes (CC)
6 Follow-Up Phone Call will be done 4 weeks after V17.

Figure 1: Study Design

Results

Demographics

A total of 1,185 smokers willing to quit smoking were enrolled, and 436 successfully completed the study after one year (analysis ongoing).

Interim Data Extraction (6 months)

Raw data were extracted from a subset of 348 subjects, of which 272 were continuously smoking abstinent until Month 6 based on the following criteria: self-reported use of tobacco/nicotine containing products, CO breath test ≤ 10 ppm, cotinine test < 100 ng/mL in spot urine (Month 5 onwards), free cotinine concentration < 50 ng/mL in 24-hour-urine (at Month 6).

Baseline Characteristics	Abstinent Subjects at Month 6 (N=272)
Male (n; %)	122 (44.9)
Female (n; %)	150 (55.1)
Age (years; Mean [SD])	43.3 (9.13)
BMI (kg/m^2; Mean [SD])	25.8 (3.69)
Caucasian (n; %)	230 (84.6)
Not Caucasian (n; %)	42 (15.4)
Smoking intensity over the past year (cig/day; Mean [SD])	17.0 (5.54)
Smoking duration (years; Mean [SD])	22.5 (8.71)

Results

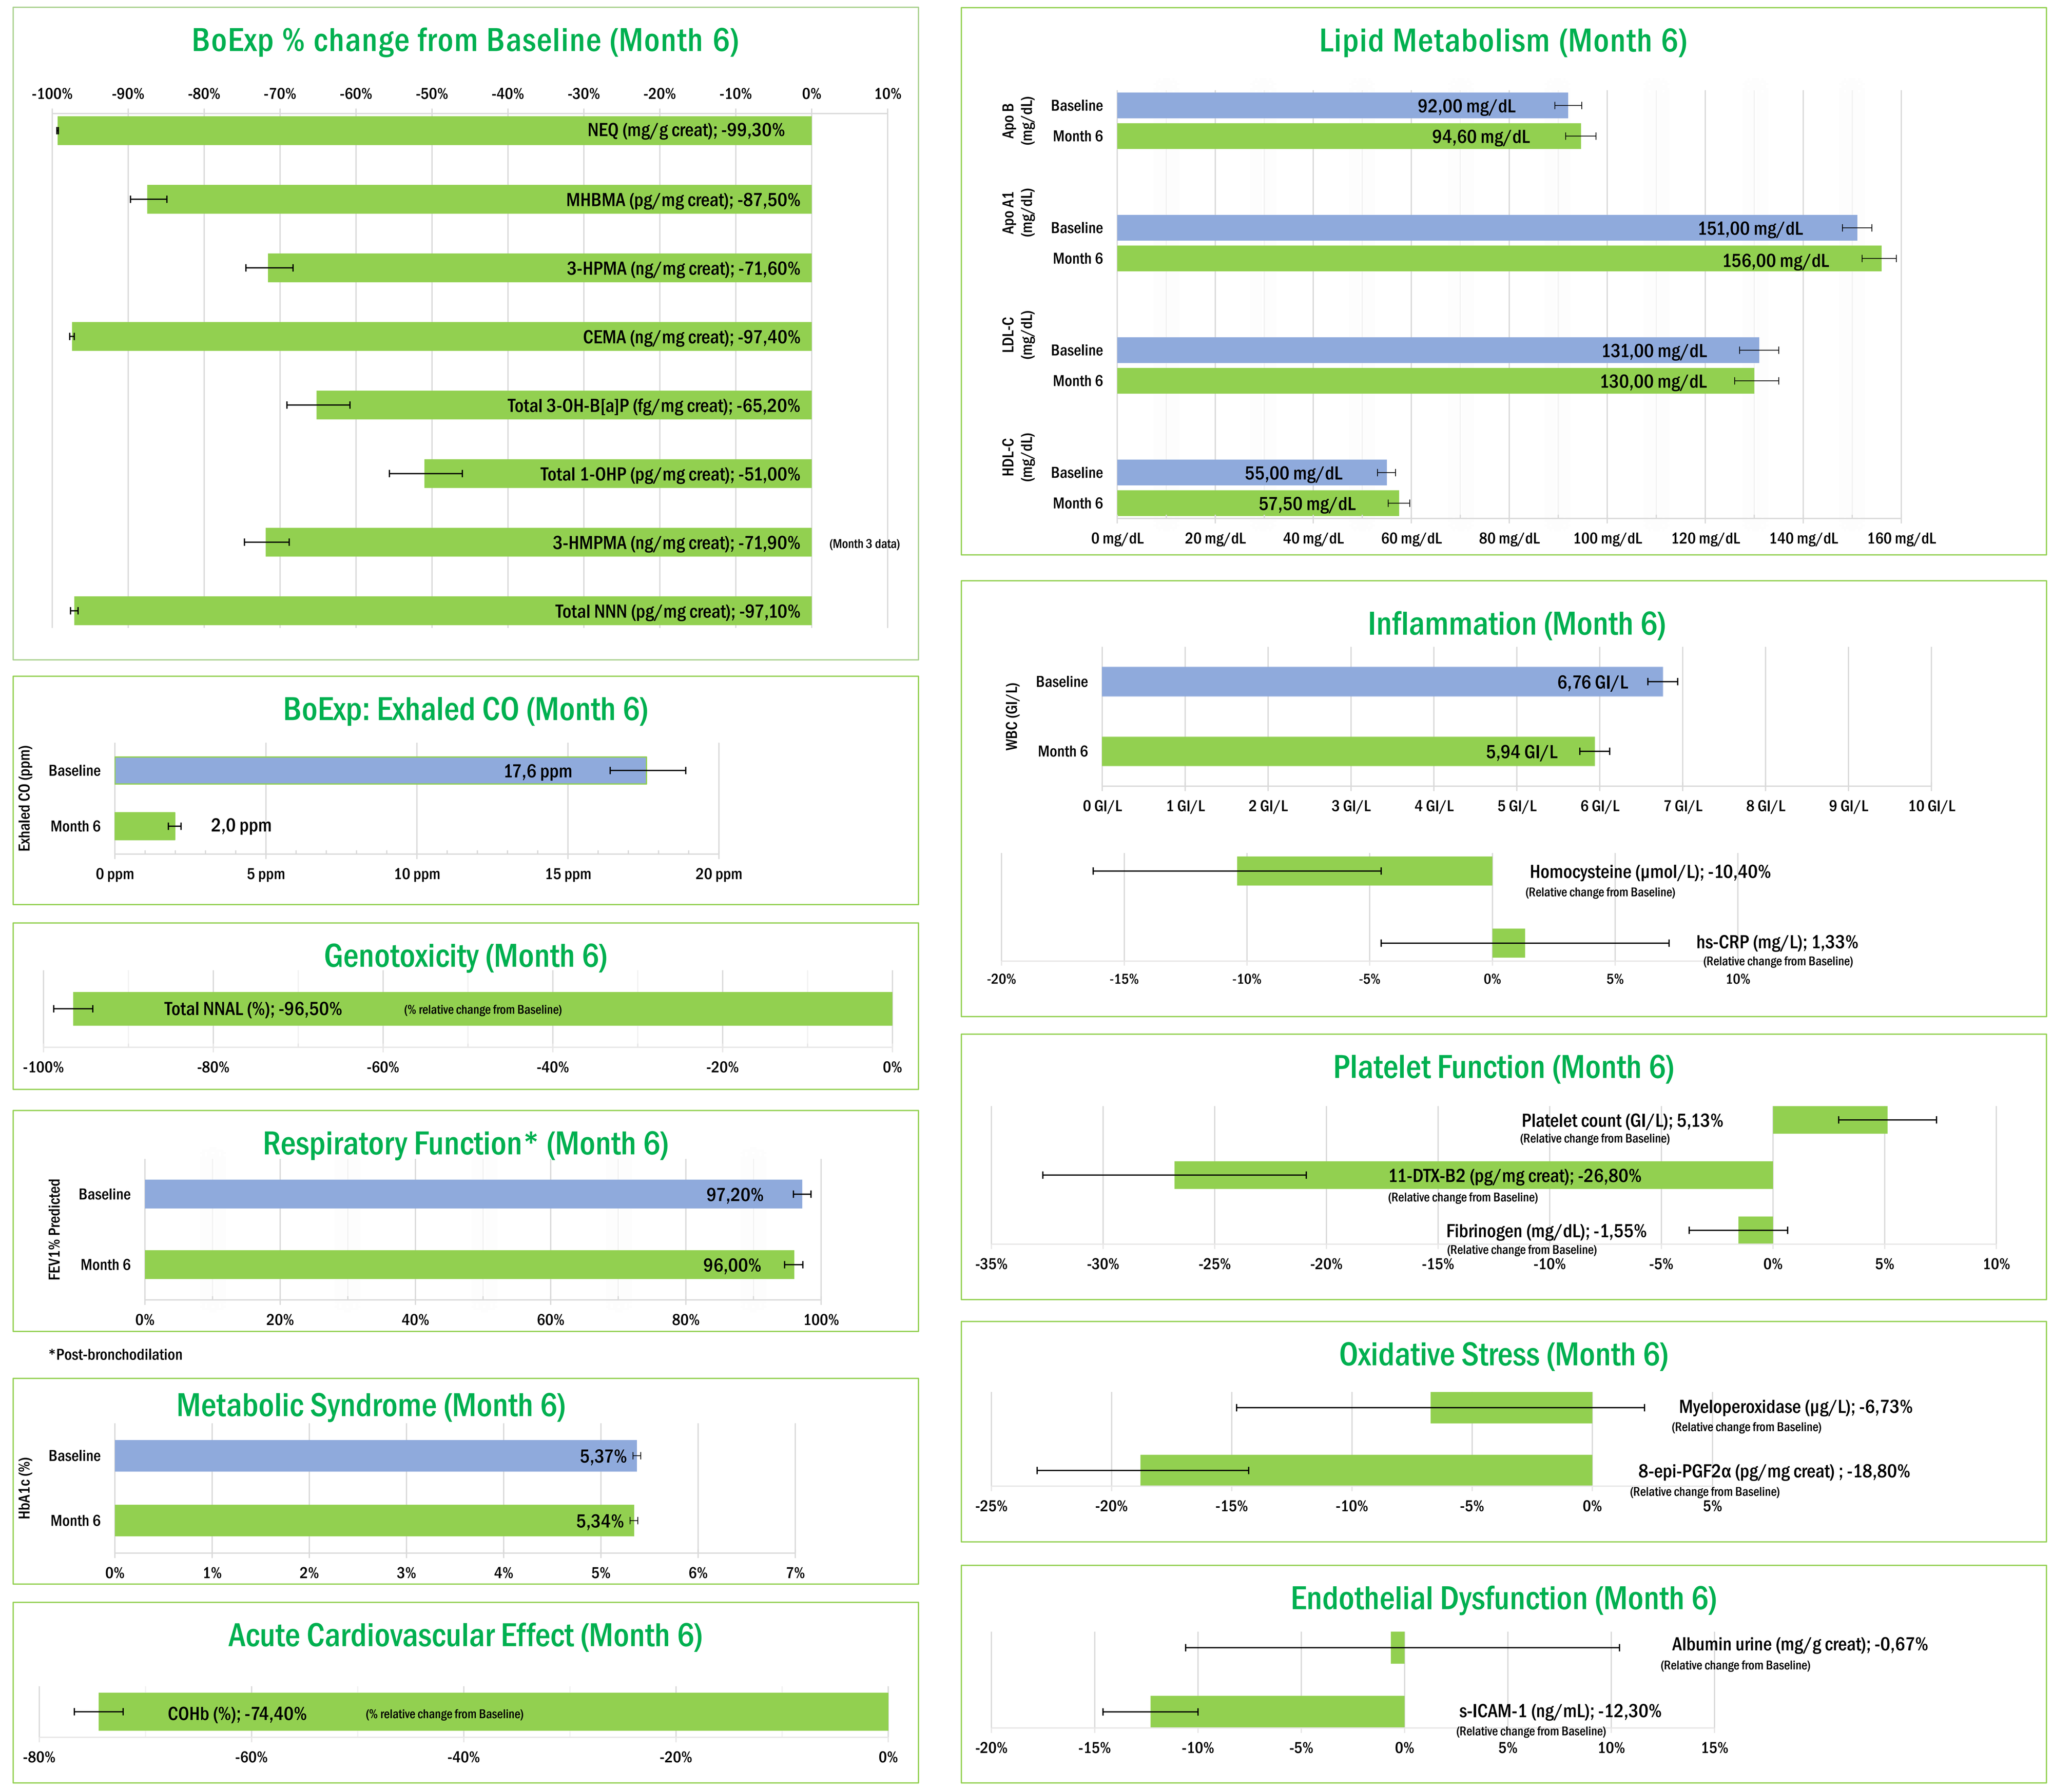
BoExp % change from Baseline (Month 6)
NEQ (mg/g creat); -99,30%
MHBMA (pg/mg creat); -87,50%
3-HPMA (ng/mg creat); -71,60%
CEMA (ng/mg creat); -97,40%
Total 3-OH-B[a]P (fg/mg creat); -65,20%
Total 1-OHP (pg/mg creat); -51,00%
3-HMPMA (ng/mg creat); -71,90%
(Month 3 data)
Total NNN (pg/mg creat); -97,10%
BoExp: Exhaled CO (Month 6)
Exhaled CO (ppm)
Baseline 17,6 ppm
Month 6 2,0 ppm
Genotoxicity (Month 6)
Total NNAL (%); -96,50%
(% relative change from Baseline)
Respiratory Function* (Month 6)
FEV1% Predicted
Baseline 97,20%
Month 6 96,00%
*Post-bronchodilation
Metabolic Syndrome (Month 6)
HbA1c (%)
Baseline 5,37%
Month 6 5,34%
Acute Cardiovascular Effect (Month 6)
COHb (%); -74,40%
(% relative change from Baseline)
Lipid Metabolism (Month 6)
Apo B (mg/dL) Baseline 92,00 mg/dL
Month 6 94,60 mg/dL
Apo A1 (mg/dL) Baseline 151,00 mg/dL
Month 6 156,00 mg/dL
LDL-C (mg/dL) Baseline 131,00 mg/dL
Month 6 130,00 mg/dL
HDL-C (mg/dL) Baseline 55,00 mg/dL
Month 6 57,50 mg/dL
Inflammation (Month 6)
WBC (GI/L)
Baseline 6,76 GI/L
Month 6 5,94 GI/L
Homocysteine (µmol/L); -10,40%
(Relative change from Baseline)
hs-CRP (mg/L); 1,33%
(Relative change from Baseline)
Platelet Function (Month 6)
Platelet count (GI/L); 5,13%
(Relative change from Baseline)
11-DTX-B2 (pg/mg creat); -26,80%
(Relative change from Baseline)
Fibrinogen (mg/dL); -1,55%
(Relative change from Baseline)
Oxidative Stress (Month 6)
Myeloperoxidase (µg/L); -6,73%
(Relative change from Baseline)
8-epi-PGF2α (pg/mg creat) ; -18,80%
(Relative change from Baseline)
Endothelial Dysfunction (Month 6)
Albumin urine (mg/g creat); -0,67%
(Relative change from Baseline)
s-ICAM-1 (ng/mL); -12,30%
(Relative change from Baseline)

Figure 2: Results

Conclusions

The 6-month interim study results indicate that continuously stopping smoking leads to a substantial reduction in exposure to HPHCs, subsequently resulting in favorable changes in CREs reflecting multiple mechanisms and biological functions including lipid metabolism, inflammation or oxidative stress. All of these changes are likely to be contributing to the reduction of the risk of developing smoking-related diseases.

SRNT 24th Annual Meeting, Baltimore, Maryland, USA

February 21-24, 2018

Competing Financial Interest

The research described in this poster was sponsored by Philip Morris International.