Evaluation of a small whole-body exposure chamber (sWBEC) for mice

Wei Teck Tan[1], Subash Krishnan[1], Blaine Phillips[1], Arkadiusz Kuczaj[2], Julia Hoeng[2], Patrick Vanscheeuwijck[2]

[1] PMI R&D, Philip Morris International Research Laboratories Pte. Ltd., Science Park II, Singapore
[2] PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

Introduction and objectives

New inhalable drugs are typically costly and available in limited quantities during the screening and efficacy-testing phase. They are, therefore, best evaluated by using inhalation chambers designed for a small number of animals. In this study, we selected whole-body exposure, as opposed to nose-only exposure, in order to avoid stress effects. A SCIREQ InExpose™ whole-body exposure chamber for mice was modified to improve its operational agility and enable characterization of the test atmosphere within the chamber during exposure.

The objective of this study was to determine the:

- spatial and temporal homogeneity of aerosol concentration within the sWBEC.
- particle size distribution of aerosols within the sWBEC.

System and principle of operation

SCIREQ InExpose™ whole-body exposure chamber for mice

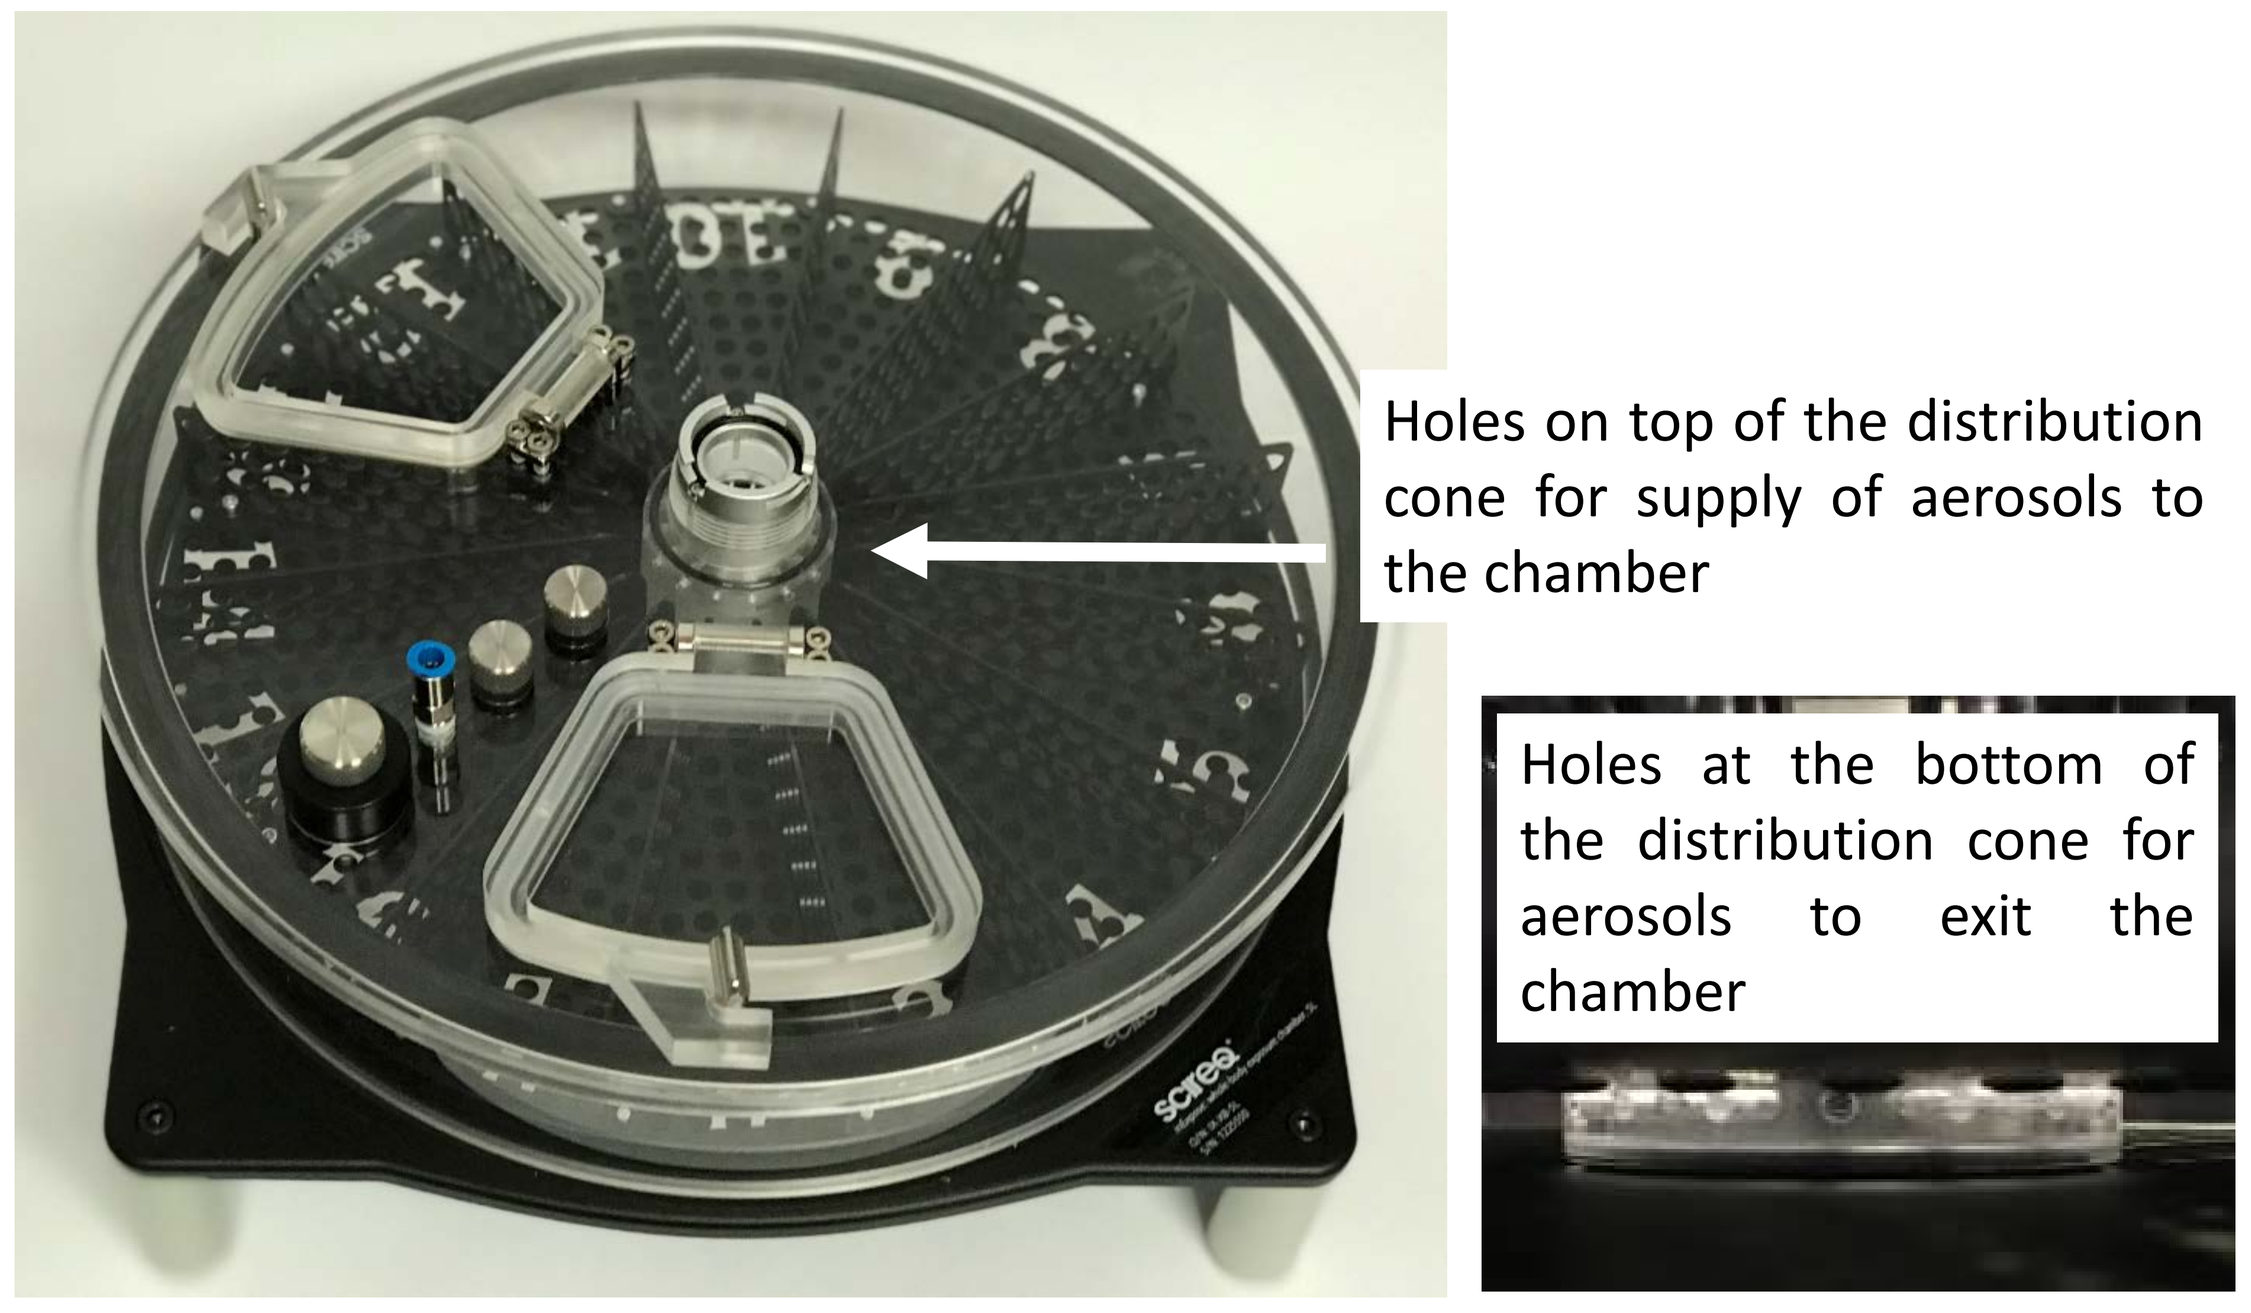
Holes on top of the distribution cone for supply of aerosols to the chamber
Holes at the bottom of the distribution cone for aerosols to exit the chamber

Figure 1. sWBEC with a modified cover to facilitate sample collection and animal loading/unloading.

The sWBEC has a volume of 5 L and 16 separated compartments where animals can be placed. Each separator is numbered for ease of identification in the event that tracking of animal position is required. Aerosols are supplied to the chamber through the 16 openings positioned at the top of the centrally located distribution cone, and they exit the chamber through another set of 16 openings positioned at the bottom of the distribution cone. Aerosols entering the chamber from the distribution cone impact the wall of each separated space, which causes the aerosols to disperse in the sWBEC and reduces their velocity towards the animals.

Aerosol generator: Blaustein Atomizing Module (BLAM)

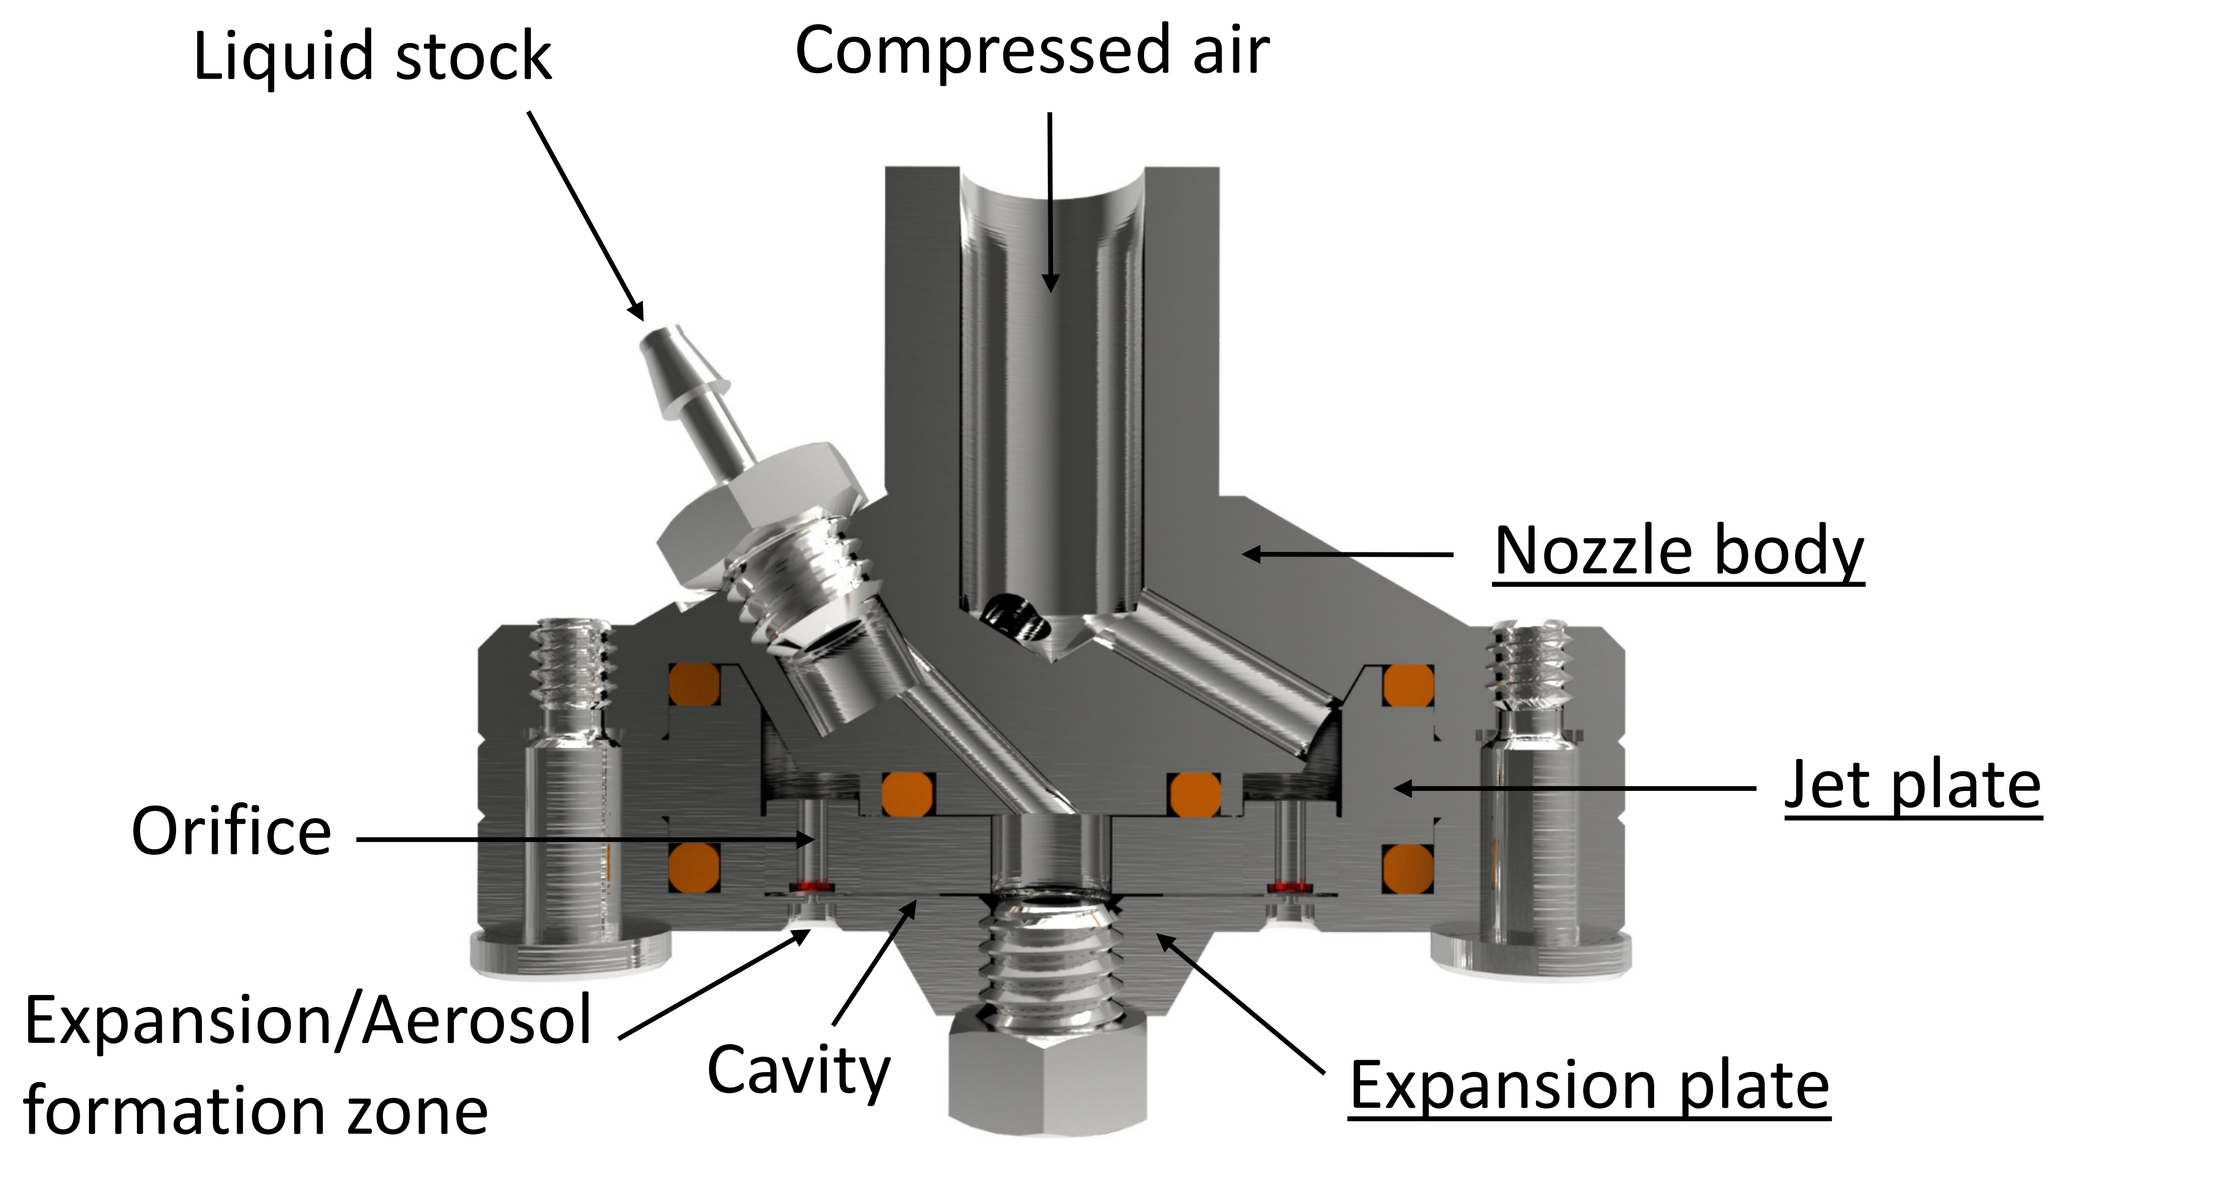
Liquid stock
Compressed air
Nozzle body
Jet plate
Orifice
Expansion/Aerosol formation zone
Cavity
Expansion plate

Figure 2. BLAM, vertical cross-sectional view.

A single-orifice BLAM (supplied by CH Technologies, NJ, USA) was used for aerosol generation. It consists of three main parts: the nozzle body, jet plate, and expansion plate (Figure 2). Compressed air coming in from the stem pushes through the orifices on the jet plate at a constant velocity. This creates a vacuum in the cavity between the jet and expansion plates, drawing the liquid stock solution into the orifices. As it expands upon exiting the orifices, the liquid is sheared into tiny droplets (i.e., converted to aerosols) by the compressed air. Any liquid that is not nebulized together with deposited aerosols will be collected at the bottom of the nebulization jar and drained. The collected liquid is not reused in order to ensure that the liquid nebulized by the BLAM has uniform temporal composition. This is important when using a multi-chemical stock solution, because differences in physicochemical properties between chemicals could result in differences in the fraction of each chemical being nebulized and deposited. Analysis of the collected liquid and comparison of chemical concentrations with the original mixture solution will provide insights into the phase-partitioning of the chemicals.

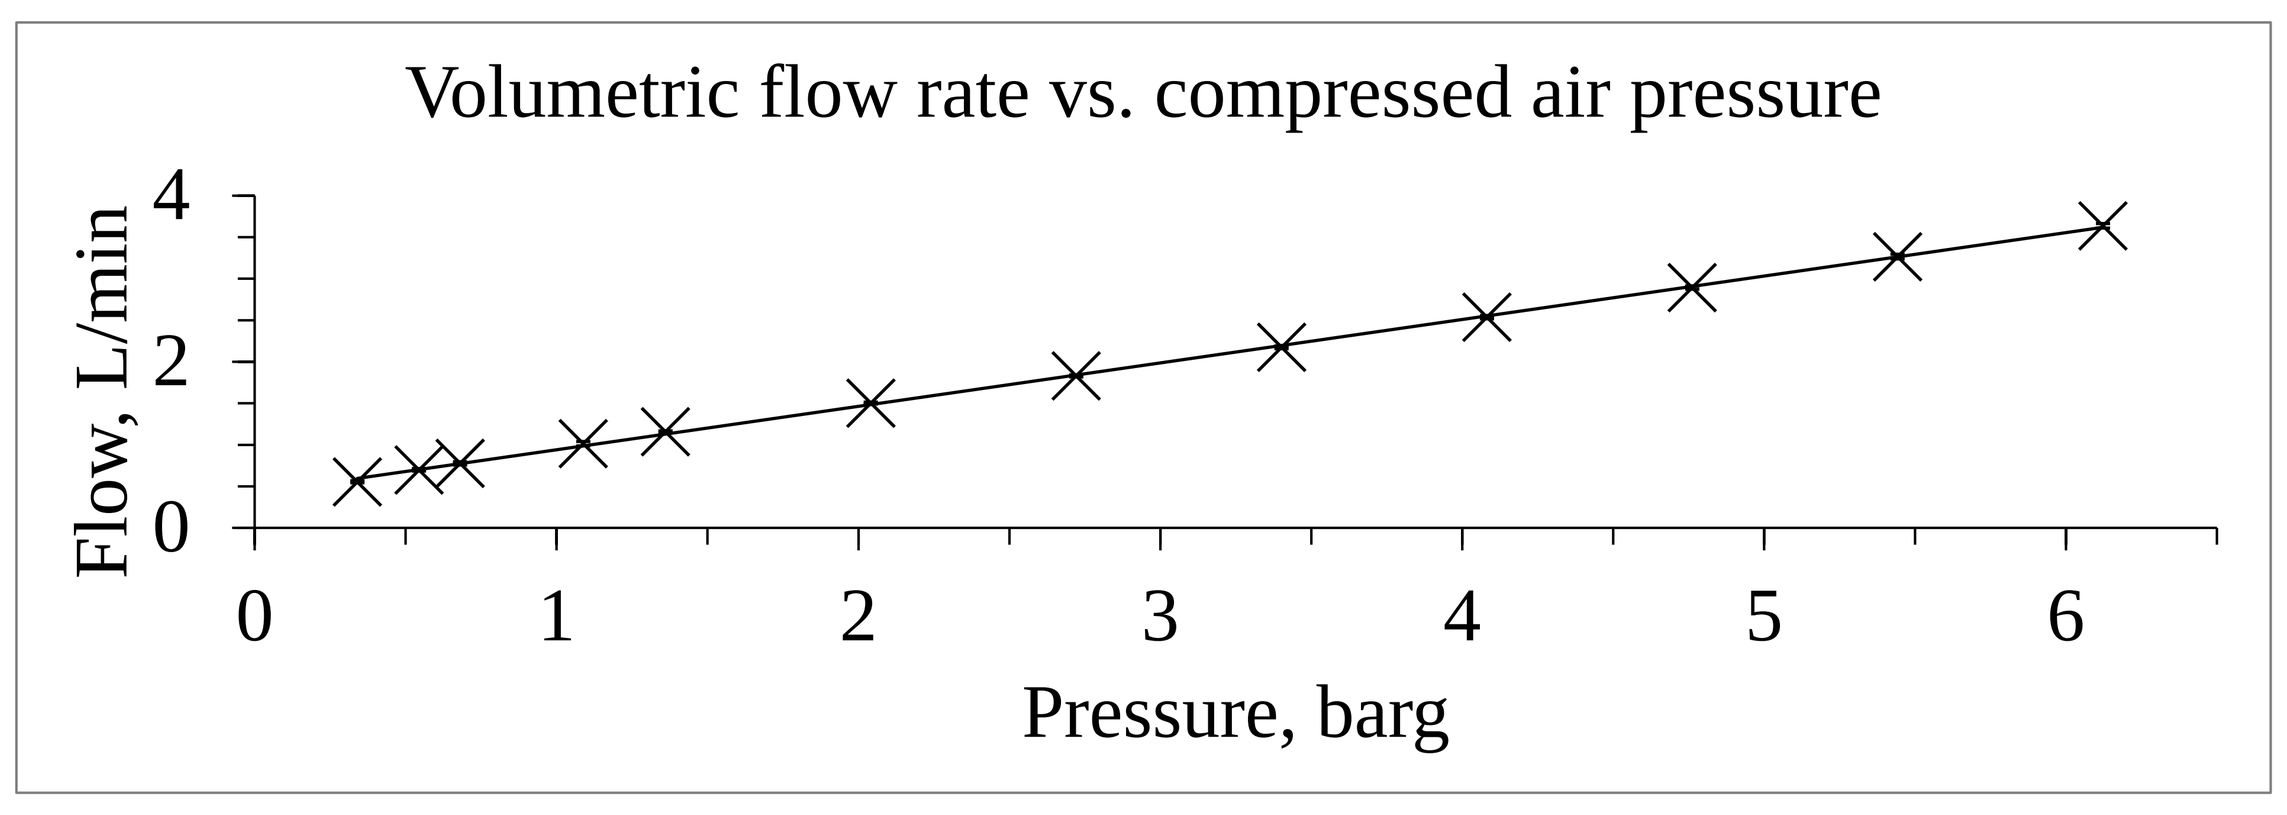
Volumetric flow rate vs. compressed air pressure
Flow, L/min
4
2
0
0
1
2
3
4
5
6
Pressure, barg

Figure 3. Single-orifice BLAM flow rate vs. compressed air pressure.

The pressure of the compressed air supplied to the BLAM is directly proportional to the volumetric flow rate of the aerosols generated. Figure 3 shows the compressed air pressure required to achieve different volumetric flow rates from a single-orifice BLAM.

Stock solution composition

Table 1. Stock solution composition

	Nicotine (Nic)	Propylene glycol (PG)	Glycerol (VG)	Phosphate-buffered saline (PBS)
Composition	5 w/v%	40 w/v%	40 w/v%	Balance
Source	Sigma-Aldrich			
Product code	N3876	P4347	G2289	DB8537

The stock solution was prepared in-house. Hydrochloric acid or sodium hydroxide was used to adjust the pH of the stock solution to neutral (pH 6–8). The composition of the stock solution was verified to be within ±15% of the target value.

Experimental setup

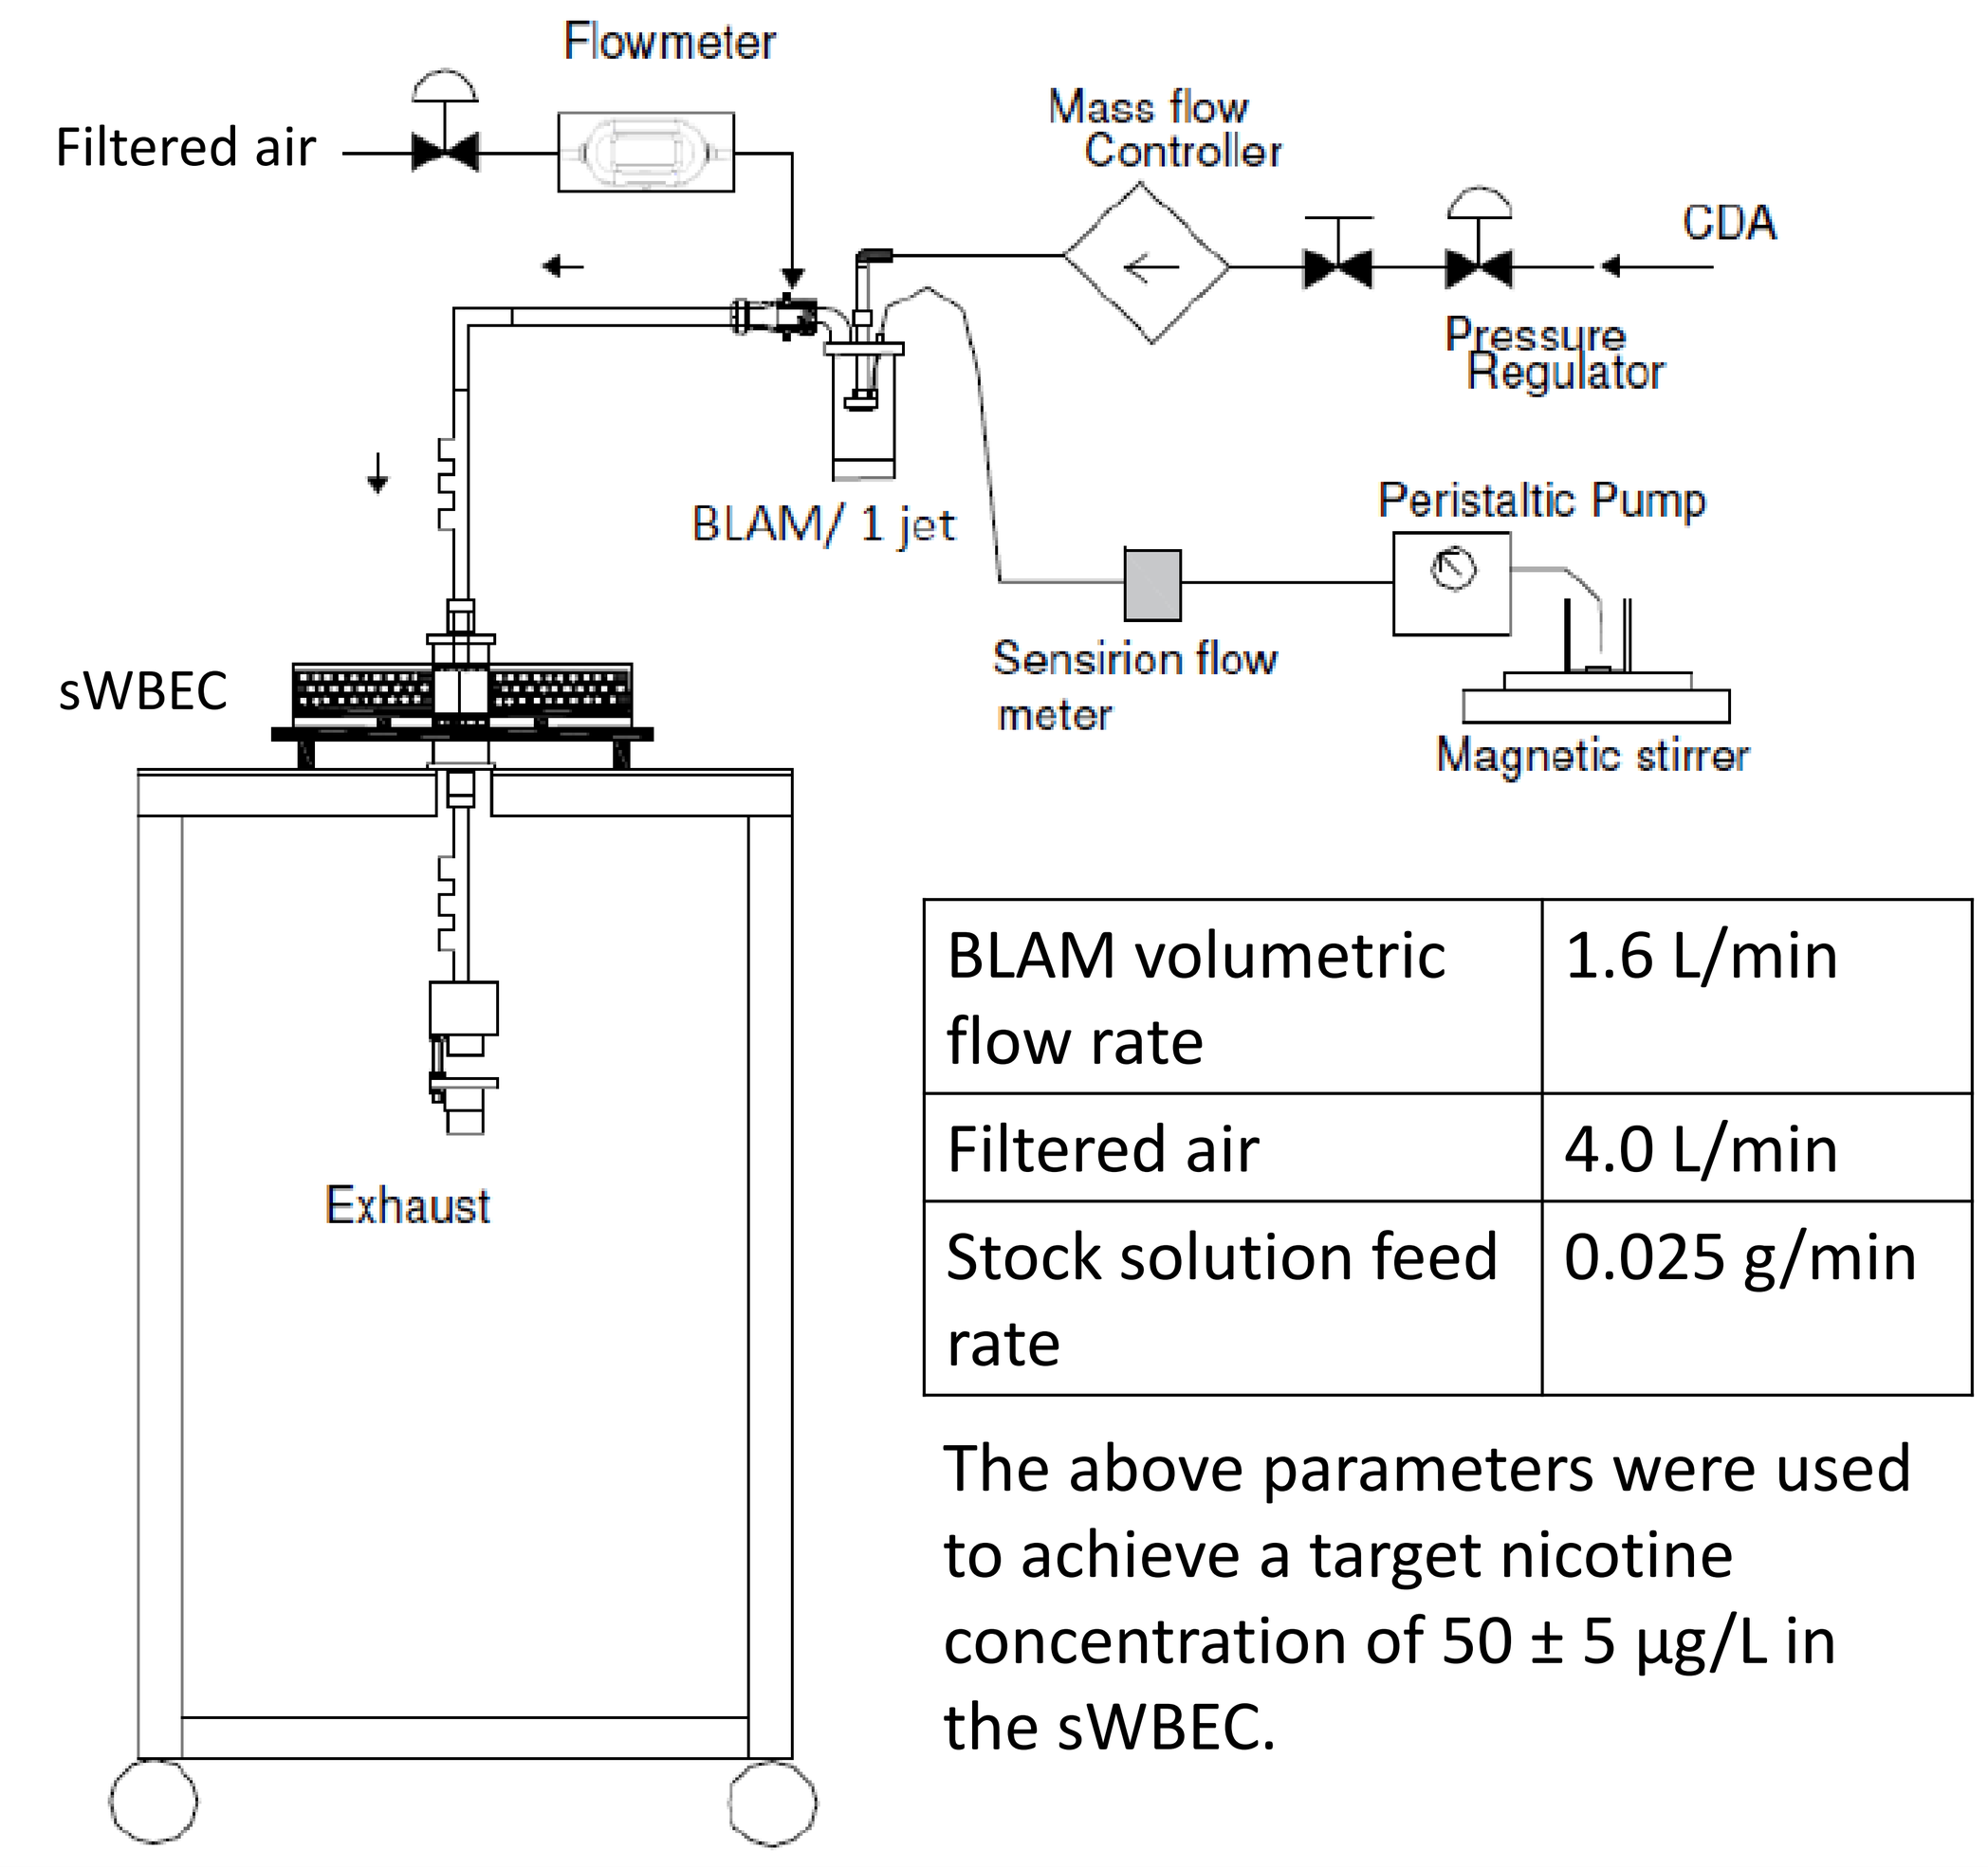
Flowmeter
Filtered air
Mass flow Controller
CDA
Pressure Regulator
BLAM/ 1 jet
Peristaltic Pump
Sensirion flow meter
Magnetic stirrer
sWBEC
Exhaust

BLAM volumetric flow rate	1.6 L/min
Filtered air	4.0 L/min
Stock solution feed rate	0.025 g/min

The above parameters were used to achieve a target nicotine concentration of 50 ± 5 µg/L in the sWBEC.

Spatial homogeneity of aerosol concentration

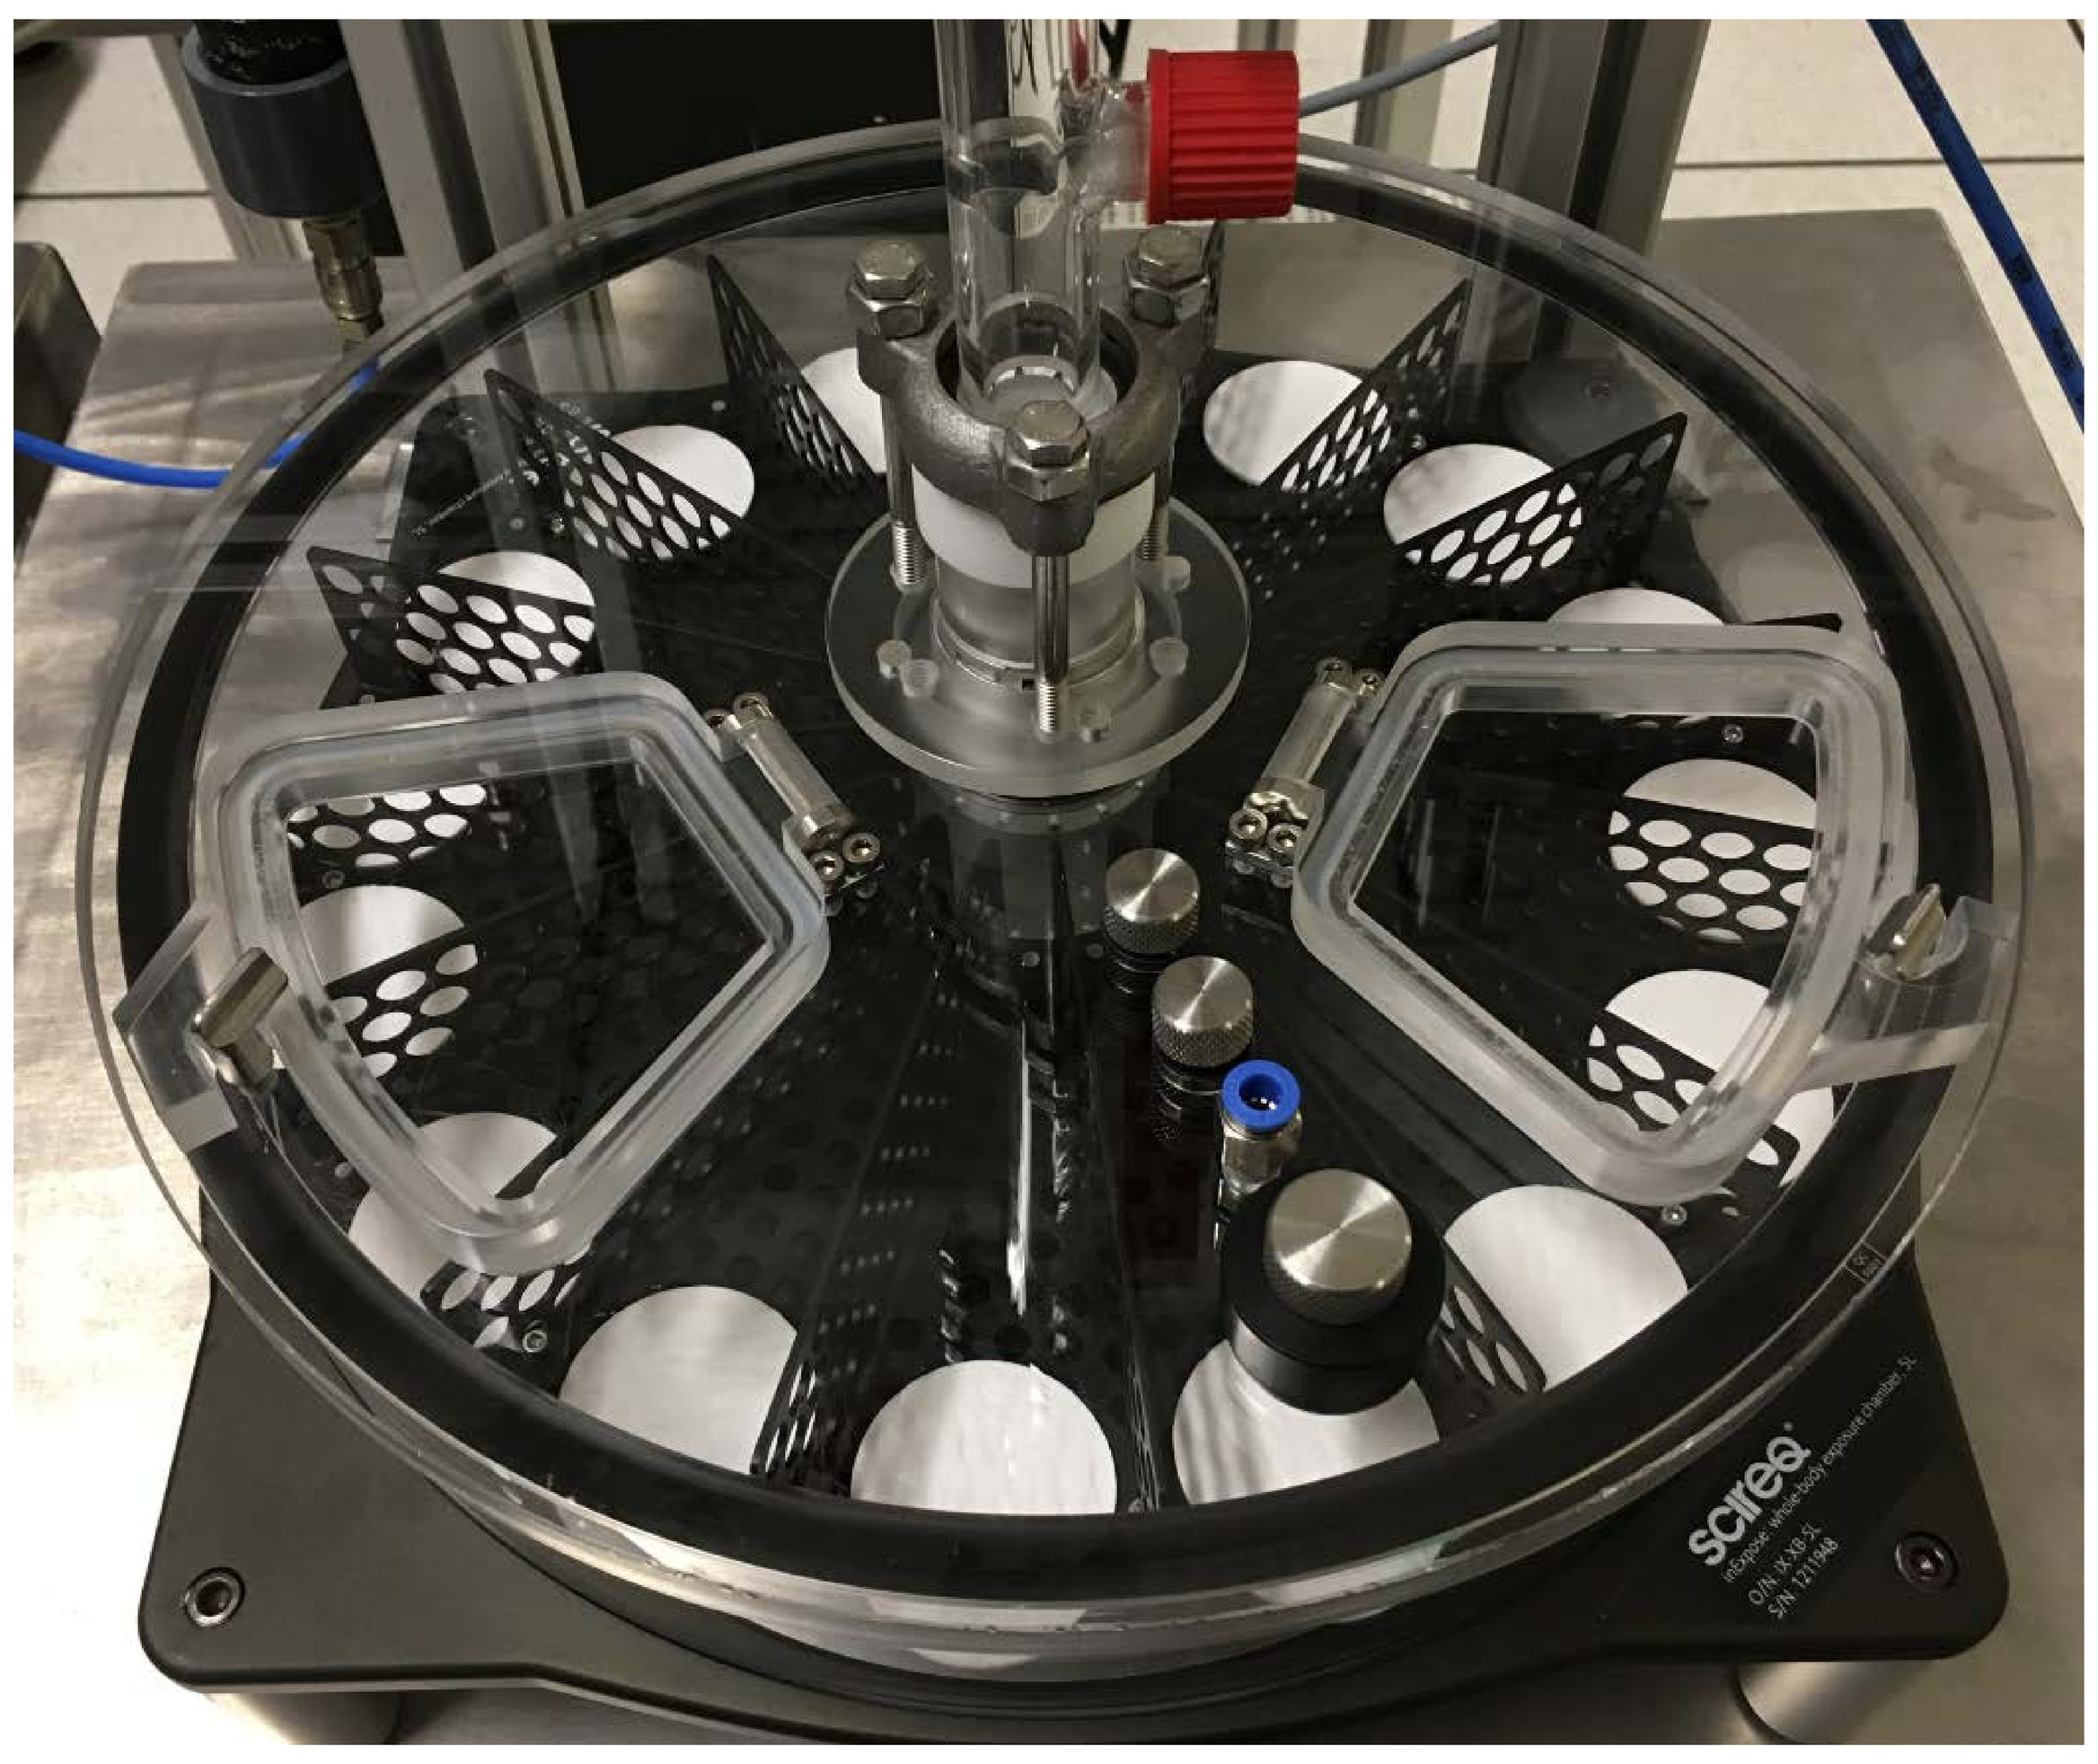

Figure 4. Positioning of filter pads in the sWBEC for determining the spatial homogeneity of aerosol concentration.

To determine the spatial homogeneity of aerosols within the sWBEC, filter pads were placed in the sWBEC, and aerosols were allowed to deposit passively onto the filter pads. This method was used to minimize the influence of sampling on the flow distribution within the sWBEC and will, therefore, provide more representative results on the spatial homogeneity of aerosol concentrations within the sWBEC. To ensure that the mass quantified on the filter pad after sampling was due primarily to the aerosols that were deposited passively, baseline values were determined by supplying compressed air to the sWBEC (Table 2).

Table 2. Baseline values collected on filter pads with compressed air supplied to the sWBEC.

	Position 1	Position 5	Position 9	Position 13
Baseline values with air supplied to the sWBEC	-0.01 mg	-0.05 mg	-0.04 mg	-0.01 mg

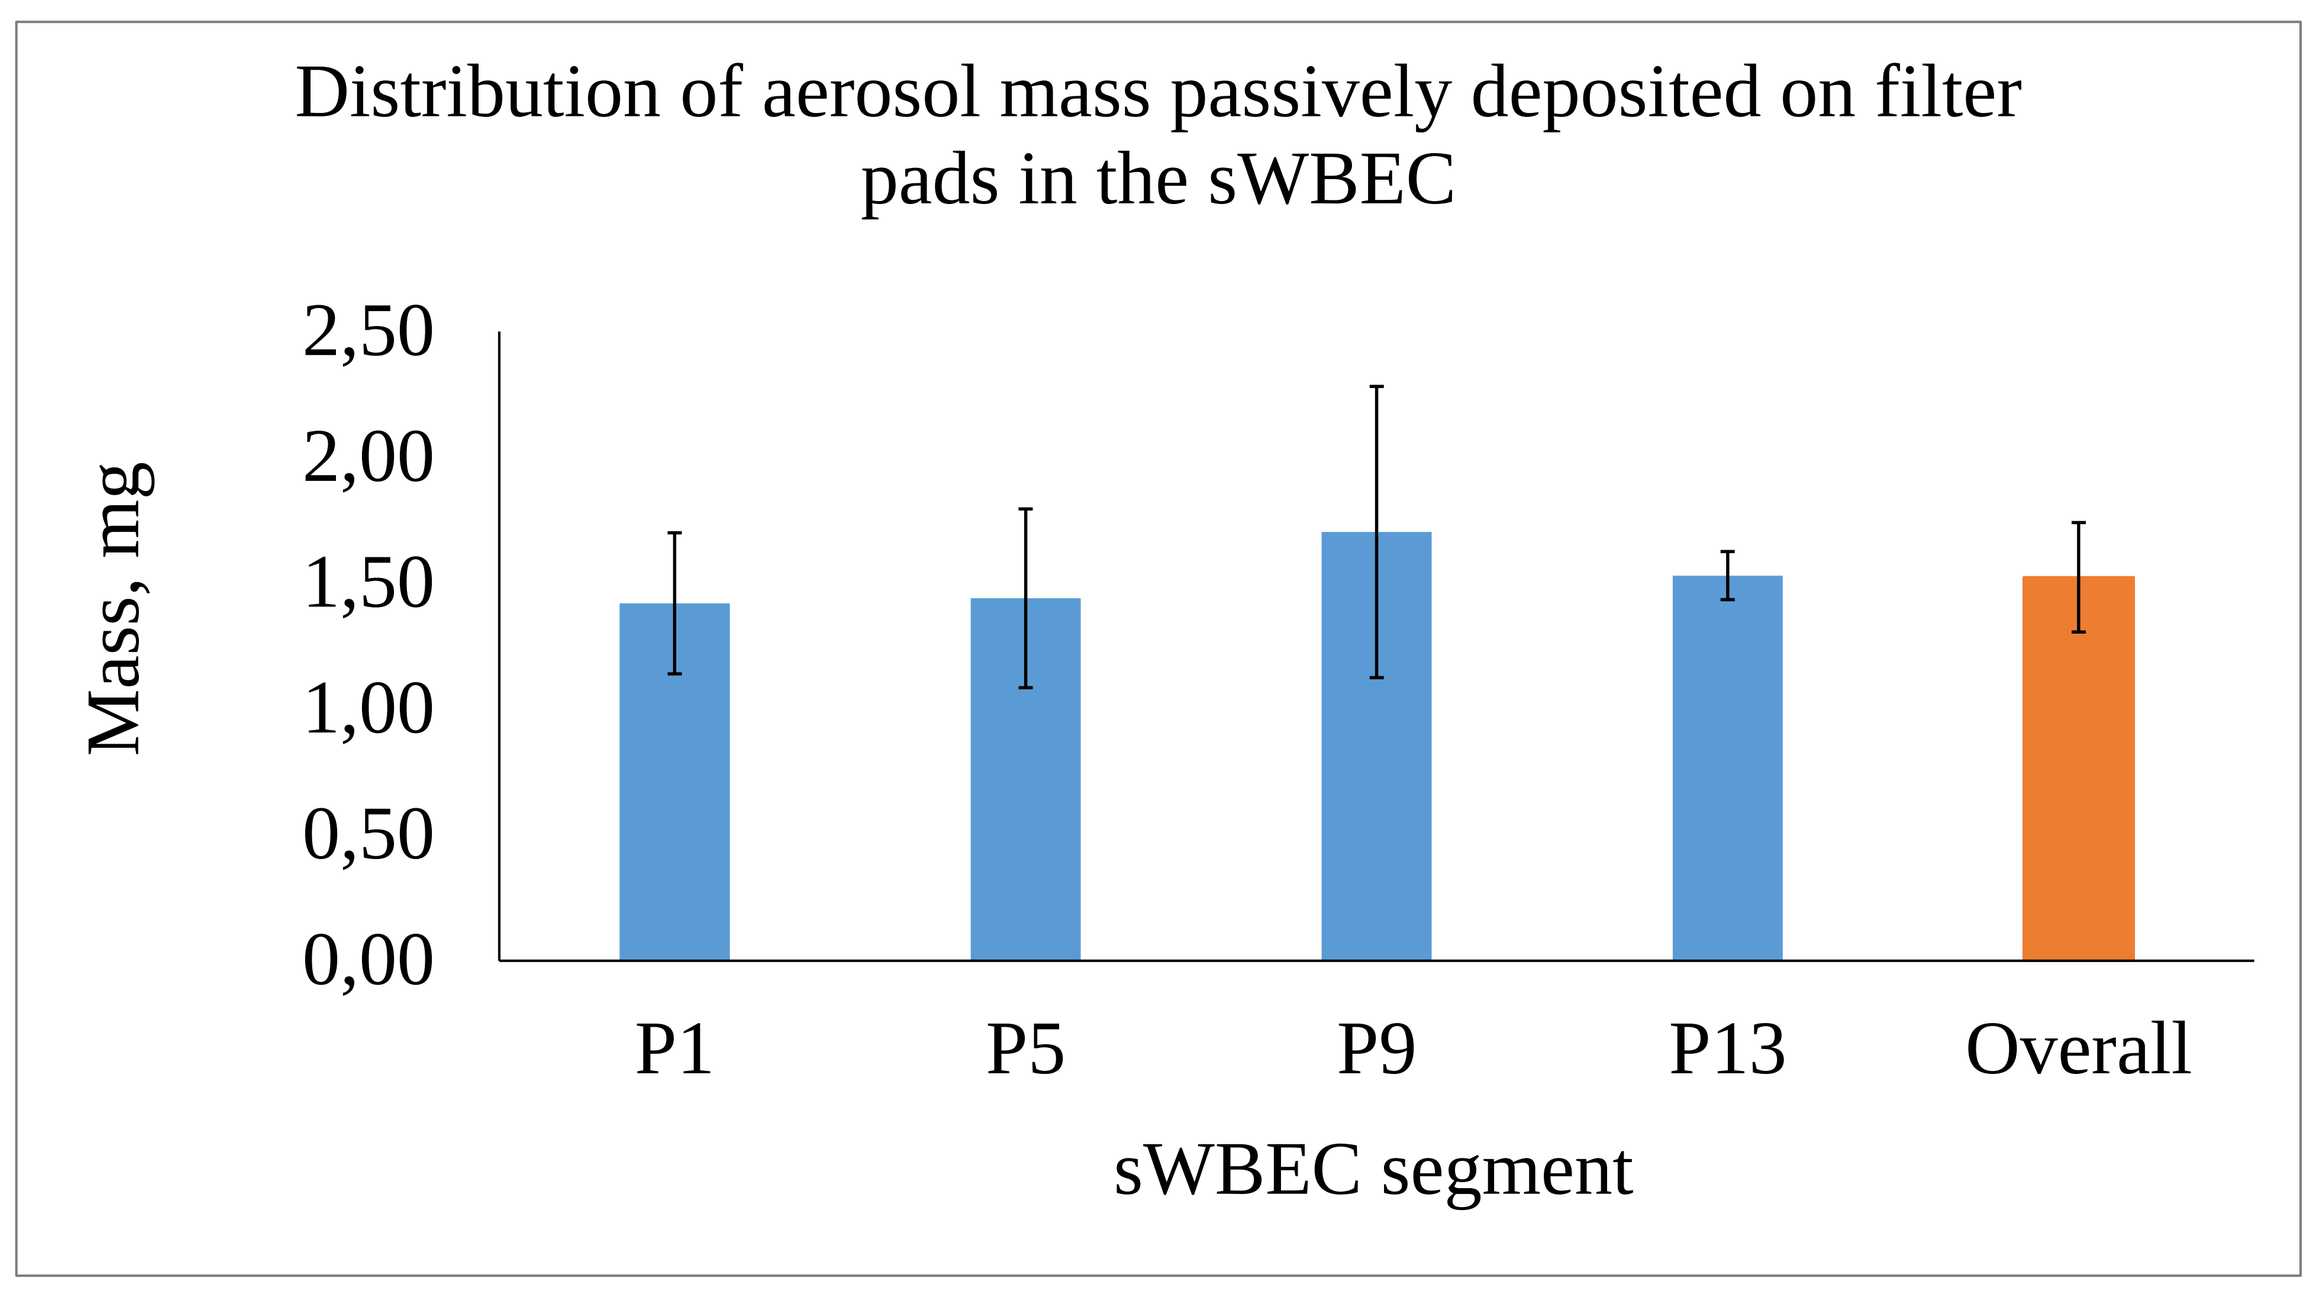
Distribution of aerosol mass passively deposited on filter pads in the sWBEC
Mass, mg
2,50
2,00
1,50
1,00
0,50
0,00
P1
P5
P9
P13
Overall
sWBEC segment

Figure 5. Aerosol mass deposited at different positions in the sWBEC over 60 minutes.

Temporal homogeneity of aerosol concentration

Temporal aerosol homogeneity was quantified by drawing aerosols from the sWBEC to determine the concentration of nicotine at five time points distributed over a 6-hour period. Passive deposition was not applied here, as the concentration of nicotine in the sWBEC cannot be quantified on the basis of the amount deposited passively on the filter pad.

The equation described by Pauluhn et al. (2007) was used as the basis for modelling the concentration profile in the sWBEC. Linearization of the equation and fitting of experimental data to the linearized equation allowed us to determine an empirical equation that accounts for:

- the dynamics of the entire inhalation system and
- the effects on the concentration profile due to factors other than aerosol flow rate and chamber volume (e.g., absorption and/or adsorption of chemicals by/onto surfaces).

Equation for modelling concentration profile:

$$C = C_o\left(1 - e^{-\frac{F}{v}t}\right)$$

Linearized equation for fitting of experimental data:

$$\ln(C_o - C) = \ln C_o - \frac{F}{v}t, \text{ for } C < Co$$

where,
C_o = equilibrium concentration, µg/L
F = aerosol flow rate, L/min
t = time, min
v = chamber volume, L

Substituting C with 0.95C_o gives the following equation for calculating the time to reach 95% of the equilibrium concentration (t_{95}).

$$0.95C_o = C_o\left(1 - e^{-\frac{F}{v}t_{95}}\right)$$

$$t_{95} = \frac{\ln 0.05}{-\frac{F}{v}}$$

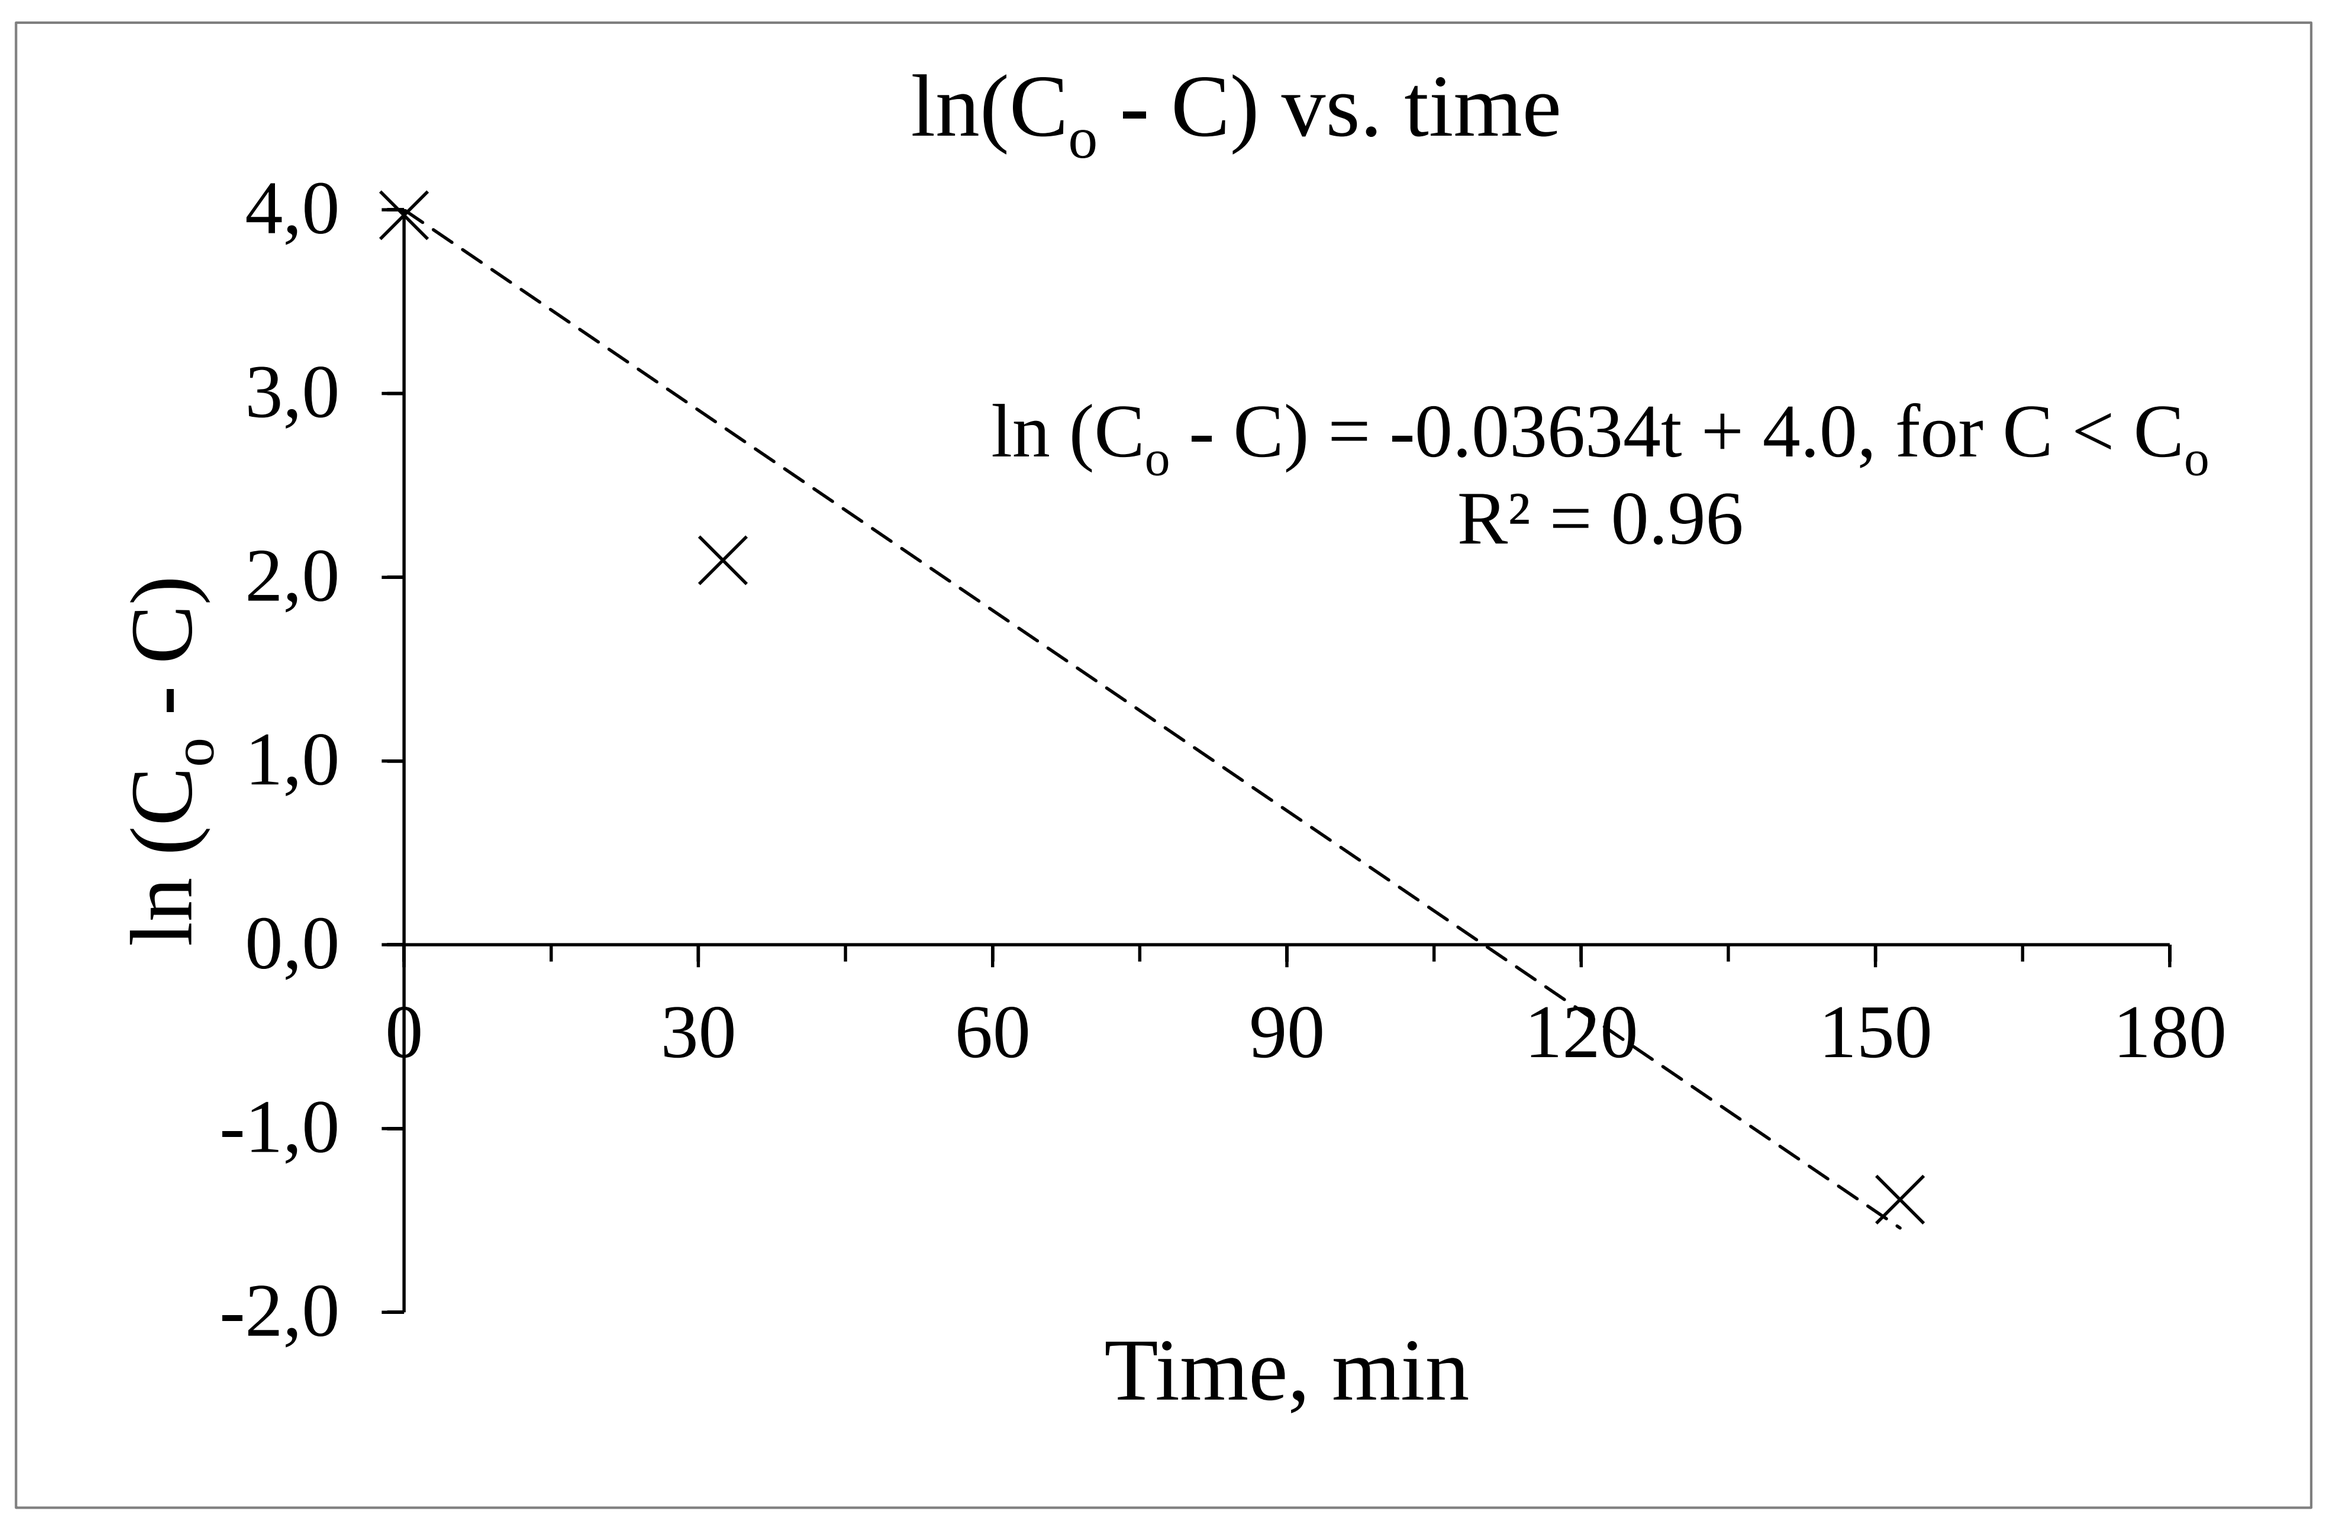
ln(C$_o$ - C) vs. time
ln (C$_o$ - C) = -0.03634t + 4.0, for C < C$_o$
R² = 0.96
ln (C$_o$ - C)
4,0
3,0
2,0
1,0
0,0
-1,0
-2,0
0
30
60
90
120
150
180
Time, min

Figure 6. Plot of ln(C_o – C) vs. time using experimental data using C_o= 53 µg/L

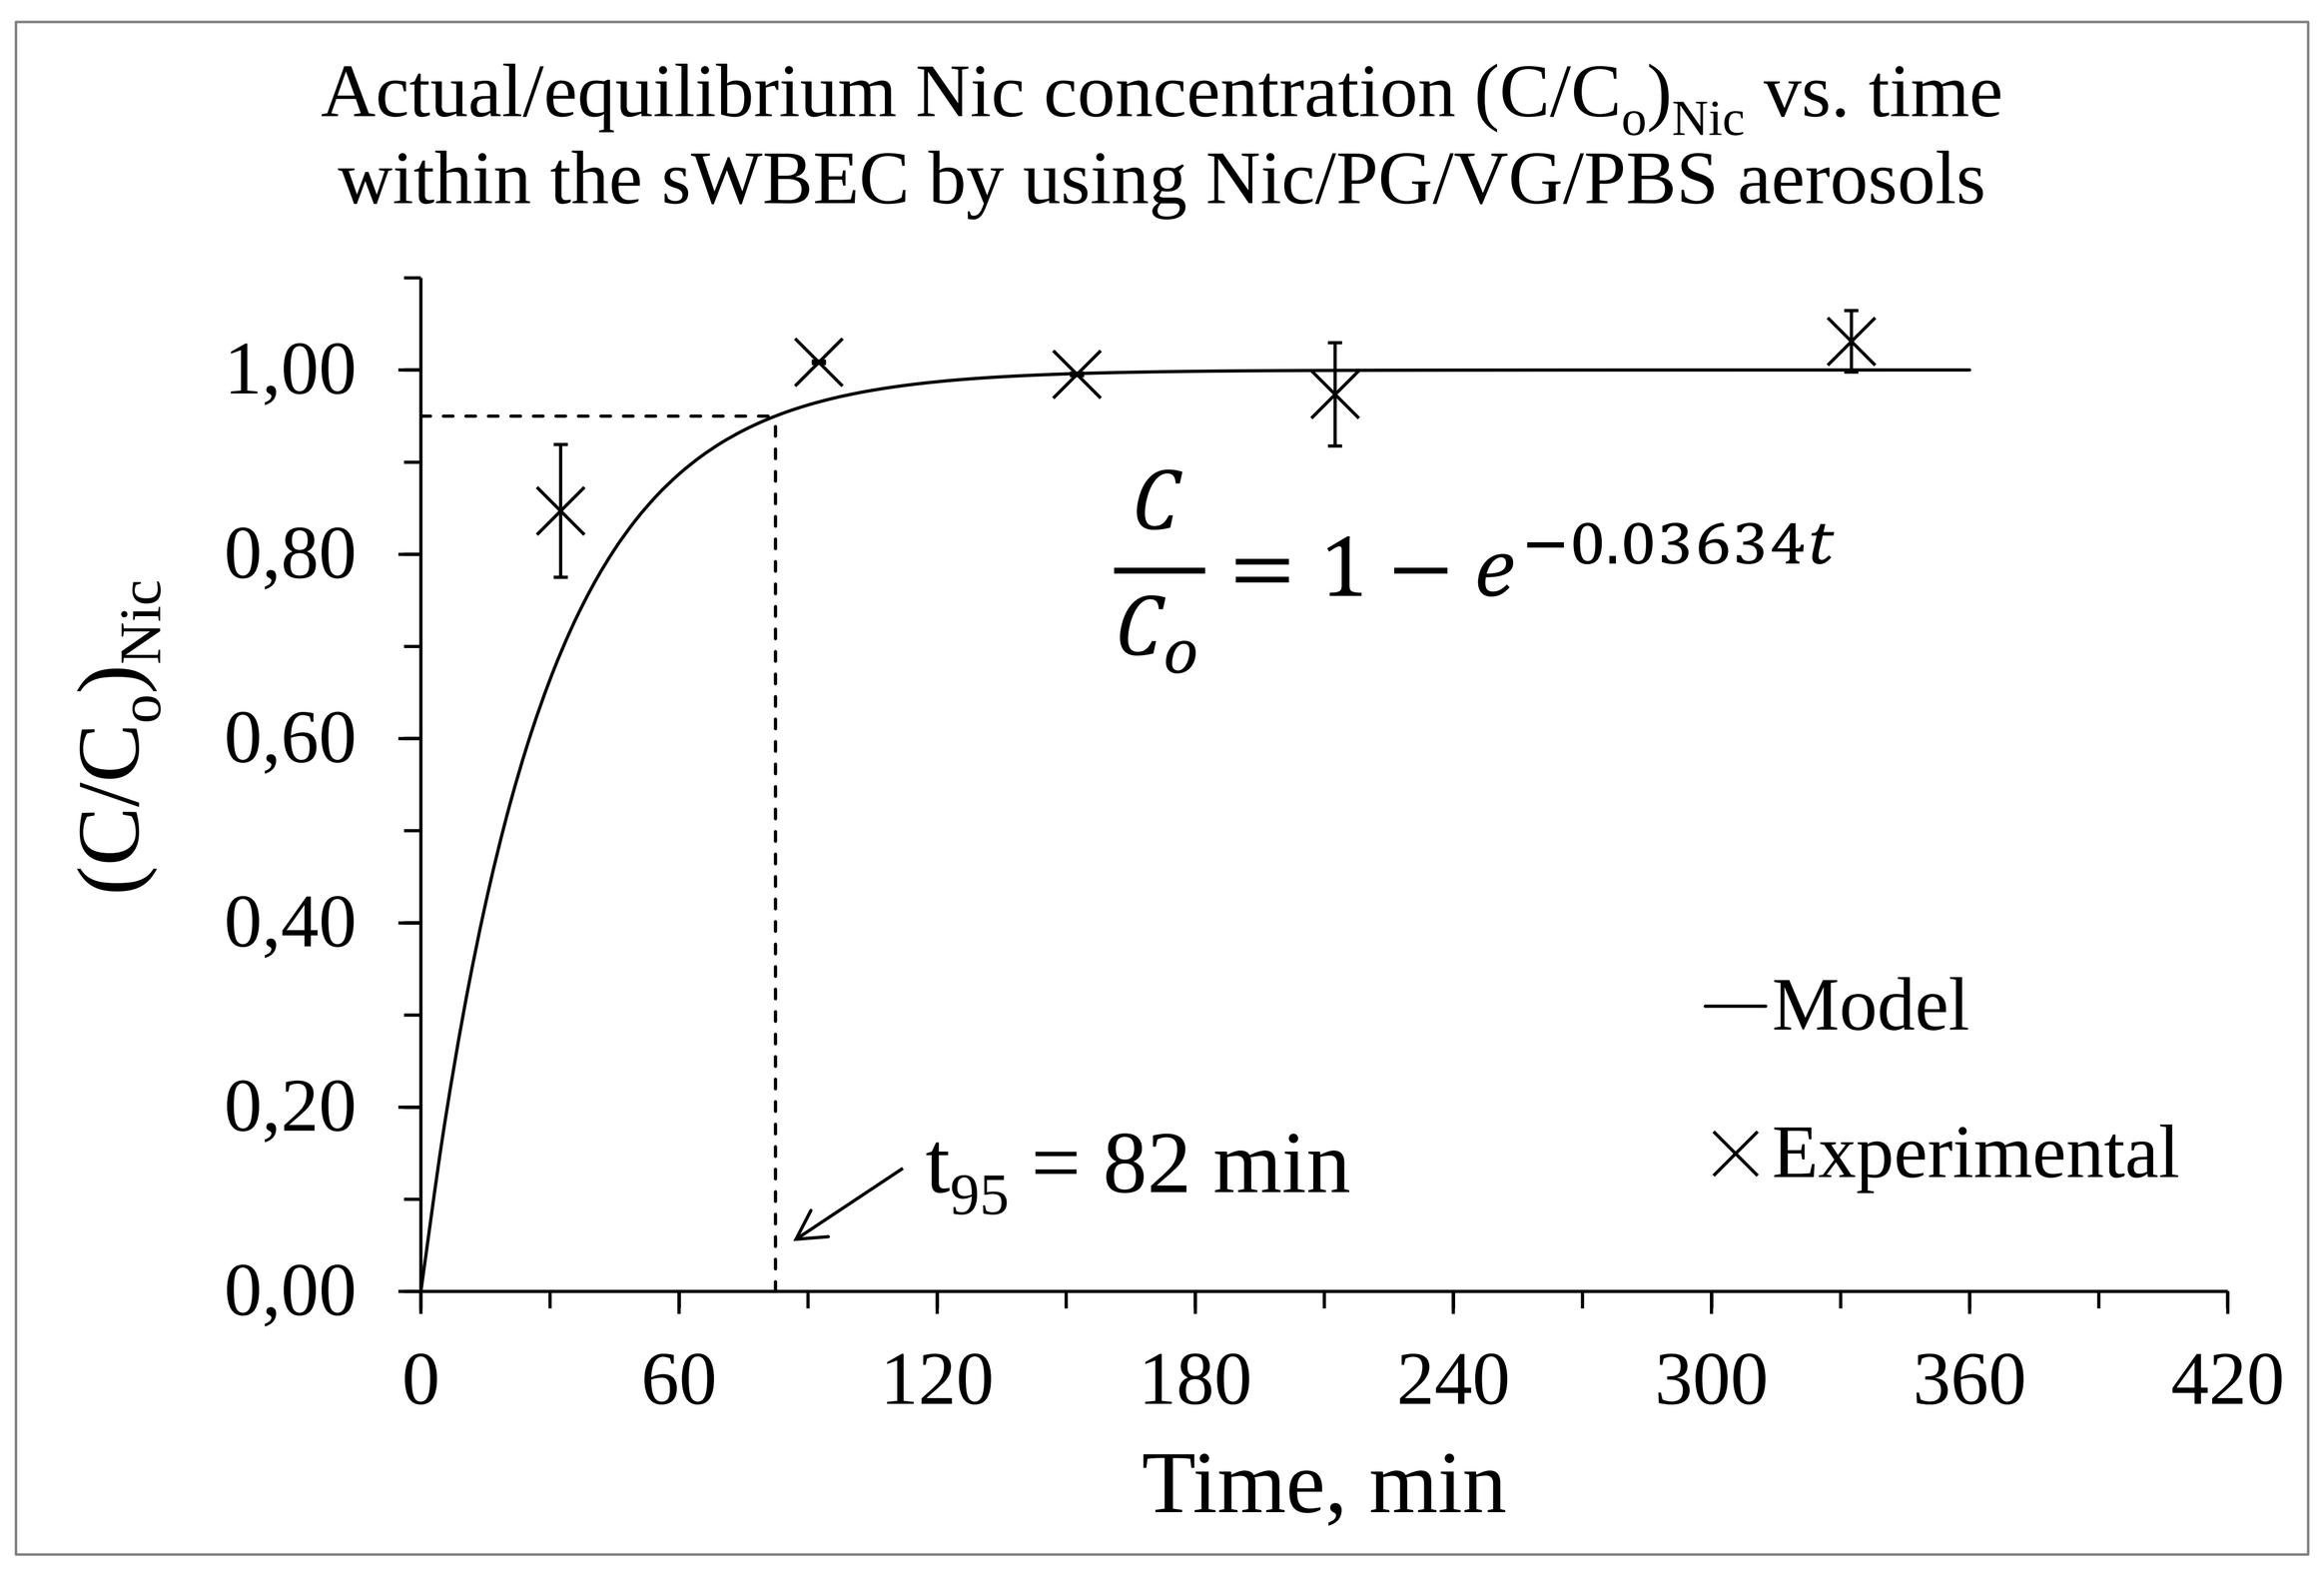
Actual/equilibrium Nic concentration (C/C$_o$)$_{Nic}$ vs. time within the sWBEC by using Nic/PG/VG/PBS aerosols
$\frac{C}{C_o} = 1 - e^{-0.03634t}$
(C/C$_o$)$_{Nic}$
1,00
0,80
0,60
0,40
0,20
0,00
0
60
120
180
240
300
360
420
Time, min
—Model
×Experimental
t$_{95}$ = 82 min

Figure 7. Nicotine concentration profile in the sWBEC, showing t_{95} = 82 min.

Particle size distribution

Particle size distribution of the aerosols was determined by using a nine-stage PIXE cascade impactor (PIXE Inc, FL, USA). Samples were collected from the sWBEC by using a vacuum pump at a flow rate of 1 L/min.

Table 3. Particle size distribution within the sWBEC.

Mass median aerodynamic diameter	Geometric standard deviation
1.54 µm (n = 5, SD = 0.12)	2.02 (n = 5, SD = 0.32)

Discussion and conclusions

Our evaluation of the sWBEC demonstrated that aerosol distribution within the sWBEC is comparable to that within the exposure chambers evaluated by other investigators (O'Shaughnessy 2008, Cheng & Moss 1995). The experimental t95 for nicotine is ~30 times higher than that calculated on the basis of chamber volume and flow rates, which shows the influence of physicochemical effects such as absorption and/or adsorption of chemicals into/onto surfaces.

References:

- Cheng, Y. S. & Moss, O. R. (1995). Inhalation exposure systems. In Concepts in Inhalation Toxicology, 2nd edn, McClellan RO, Henderson RF (eds). Taylor & Francis: Washington DC; 25–66.
- O'Shaughnessy, P. T., Achutan, C., O'Neill, M. E., & Thorne, P. S. (2003). A small whole-body exposure chamber for laboratory use, Inhal. Toxicol., 15, 251–263.
- Pauluhn, J. & Thiel, A. (2007). A simple approach to validation of directed-flow nose-only inhalation chambers, J. Appl. Toxicol., 27, 160–167.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

EAC 2019, Gothenburg

Aug 25–30, 2019

Competing financial interest

Philip Morris International is the sole source of funding and sponsor of this project.