Chronological Changes in BEAS-2B Cells Chronically Exposed to Cigarette Smoke Total Particulate Matter

Marco van der Toorn, Diego Marescotti, Stephanie Johne, Celine Merg, Julia Hoeng, Manuel C. Peitsch, Karsta Luettich

Philip Morris Products S.A. (part of Philip Morris International group of companies), Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland

#GT-10

Abstract

Chronic cigarette smoke exposure is responsible for airway epithelial changes ultimately leading to lung cancer, although our knowledge about the underlying molecular events is still not very detailed. Our aim was to more comprehensively characterize the chronological changes that occur in bronchial epithelial BEAS-2B cells continuously exposed to 3R4F reference cigarette total particulate matter (TPM) for 12 weeks. High-content image analysis was employed to monitor oxidative stress (DHE, GSH), DNA content/cell cycle (Hoechst) and DNA damage (γH2AX), E-cadherin and vimentin expression at regular intervals. MMP-1 and MMP-9 levels were measured in the supernatants using Luminex technology. In addition, cell adhesion, migration and invasion were assessed by real-time impedance, and soft agar assays were performed to examine anchorage independence.

Increased oxidative stress and DNA damage were noticeable within the first two weeks of exposure. At the same time, a significant portion of cells arrested in S-phase, and by week 2, cells were dying or became senescent. However, a small population of surviving treated BEAS-2B cells resumed normal cell cycle shortly thereafter. Following exposure to TPM for 4 weeks, these BEAS-2B cells expressed lower levels of E-cadherin and higher levels of vimentin and displayed fewer cell-to-cell contacts. Increased MMP levels and a decrease in cell adhesion rates were noted, yet the treated cells were unable to invade extracellular matrix or form colonies in soft agar. The latter was only observed when cells were exposed for 12 weeks.

In summary, our experiments indicate that long-term exposure of bronchial epithelial cells to 3R4F TPM induces ongoing phenotypic changes akin to cellular transformation that may eventually lead to carcinogenesis. Further analysis using e.g. 'omics tools may lead to a better mechanistic understanding of the stepwise transformation of normal airway epithelial cells to full malignancy.

Materials & Methods

Generation of TPM - 3R4F reference cigarettes were purchased from the University of Kentucky and stored in a cooling chamber at 5±3°C with uncontrolled humidity in the original packaging (http://www2.ca.uky.edu/refcig/). The cigarettes were conditioned according to ISO standard 3402[1] before total particulate matter (TPM) generation. Cigarette smoke from 3R4F cigarettes was generated on a 20-port Borgwaldt smoking machine (Hamburg, Germany) according to the Intense Health Canada protocol (55 mL puff volume; 2 s puff duration; 2 min^{-1} puff frequency; 100% blocking of filter ventilation holes)[2]. Mainstream smoke from a total of 4 3R4F cigarettes were trapped on one glass fiber filter (44 mm) followed by extraction with an appropriate volume of DMSO to give a final concentration of 25 mg TPM/mL. Five mL DMSO were used to extract TPM from the filters, and aliquots of 25 mg/mL TPM were prepared and stored at -80°C until further use.

Cell Culture and Treatment - The human bronchial epithelial cell line BEAS-2B (LGC Standards GmbH, Wesel, Germany) was grown in complete BEGM™ (Lonza, Basel, Switzerland) on collagen A-coated plates. After two passages of the frozen stock, cells were seeded at an appropriate density and treated with TPM at a final concentration of 7.5 μg/mL. At a confluence of ca. 75%, cells were trypsinized and passaged in fresh treatment medium for a total treatment duration of 12 weeks. Cells cultured without TPM (medium only) and cells cultured in the presence of 0.2% DMSO (vehicle; Sigma Aldrich, Buchs, Switzerland) were included as controls. Every time the cells were passaged, a small aliquot was taken for subsequent gene and miRNA expression analysis. Additionally, treated and control cells were collected once a week for cell counting, high-content screening (HCS) of DNA content (cell cycle), DNA damage and oxidative stress. This experiment was repeated twice, giving a total sample number of 3 per time point/endpoint.

High-Content Screening (HCS) using the Cellomics ArrayScan® VTI HCS reader (Thermo Fisher Scientific Inc., Berkshire, UK) was employed to assess oxidative stress, DNA damage, cell cycle alterations and epithelial-mesenchymal transition (EMT). The abundance of reactive oxygen species (ROS) was determined with a live staining method using Hoechst 33342 (Fisher Scientific, Lucerne, Switzerland) and dihydroethidium (DHE; Sigma Aldrich, Buchs, Switzerland) employing the Compartmental Analysis BioApplication. Alternatively, GSH levels were assessed using monochlorobimane (mBCl; Life Technologies, Zug, Switzerland) together with the Compartmental Analysis BioApplication. The % frequency of cells in each cell cycle phase (G0/G1, S or G2/M) was determined by staining cells in the exponential growth phase with Hoechst 33342 and evaluating DNA content using the Cell Cycle BioApplication. To quantify DNA damage, an anti-phosphorylated H2AX antibody (JBW301, Merck&Cie, Schaffhausen, Switzerland) and secondary fluorescent reporter-tagged anti-mouse IgG antibody (Dylight 650; Fisher Scientific, Reinach, Switzerland) were used. The number of γH2AX-positive foci was determined based on the average fluorescent intensity using the Compartmental Analysis BioApplication. E-cadherin expression was detected using the mouse monoclonal antibody HECD-1 (ab1416, Abcam, Cambridge, UK) together with the Dylight 649 goat anti-mouse secondary antibody (Fisher Scientific). Vimentin expression was detected using a rabbit polyclonal antibody (ab45939, Abcam) together with the Dylight 550 goat anti-rabbit secondary antibody (Life Technologies). Cell nuclei were counterstained with DAPI (Fisher Scientific). Fluorescence intensity of DAPI-stained nuclei, vimentin- and E-cadherin-positive cells was captured using the Compartmental Analysis BioApplication.

Real-time Adhesion and Migration/Invasion Assays - The rate of cell migration was monitored in real-time using collagen A-coated Cell Invasion Migration (CIM) plates and a real-time cell analyzer dual-plate (RTCA-DP) xCELLigence instrument (Bucher Biotec, Basel, Switzerland). Approximately 1 h prior to conducting the experiment, BEAS-2B cells were transferred from BEGM™ to BEBM™ medium. The electrode surface of the CIM plate was coated with collagen A for 1 h. A total of 40,000 BEAS-2B cells were seeded in each well of the upper chamber in BEBM™ medium. The lower chambers were filled with BEBM™ medium containing 10% fetal calf serum (FCS) or no serum as negative control. The CIM plate was left in an incubator for 15 minutes before recording migration. Thereafter, changes in the cell index (impedance), reflecting migration, were recorded in real-time and analyzed using RTCA software v2.0. Cell invasion experiments were carried out similarly, with the addition of Geltrex® Ready-To-Use extracellular matrix (Fisher Scientific).

Soft Agar Assay - Treated and untreated BEAS-2B cells were processed according to the manufacturer's instructions for the CytoSelect™ 96-well cell transformation assay (Cell Biolabs, San Diego, CA, USA). The fluorescence was measured at day 10 with an excitation wavelength of 485 nm through a 520 nm bandpass filter in a 96-well fluorometer (Fluostar Omega Microplate reader, BMG Labtech, Ortenberg, Germany).

Measurement of Total MMP Levels - Total protein levels of MMP-1, -2, -7, -9 and -10 were determined in the cell culture supernatants using a Luminex Performance Human MMP Panel according to the manufacturer's instructions (Merck, Darmstadt, Germany).

Data analysis - All HCS data were exported to GraphPad Prism v6 (GraphPad Software, Inc., La Jolla, CA, USA) and analyzed using Dunnett's multiple comparison test and repeated ANOVA without multiple test correction, respectively. P-values smaller than 0.05 were considered statistically significant and are indicated by an asterisk (*) or hash (#).

Results

Long-term treatment of BEAS-2B cells with 3R4F TPM induces cell cycle changes

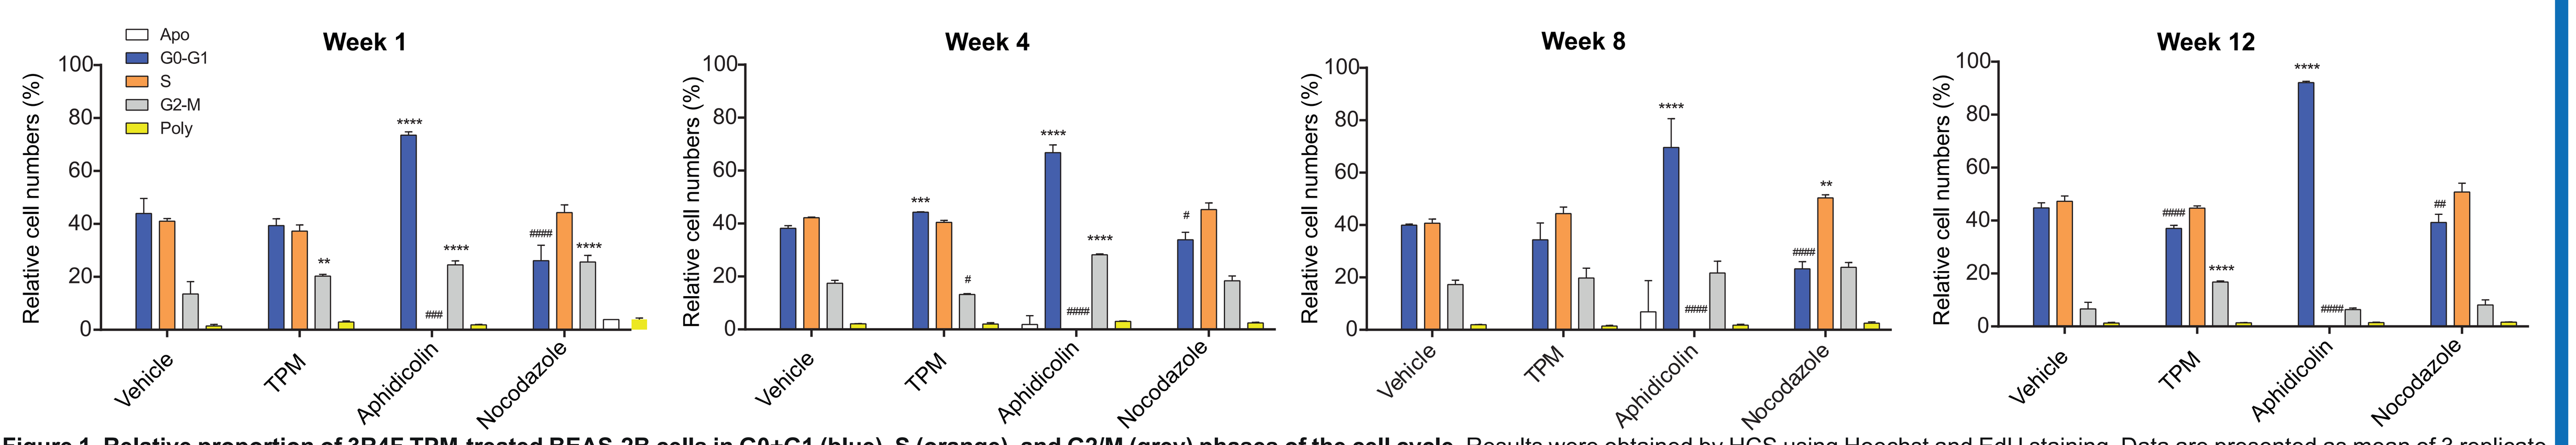
Week 1
Week 4
Week 8
Week 12
Apo
G0-G1
S
G2-M
Poly
Relative cell numbers (%)
Vehicle
TPM
Aphidicolin
Nocodazole

Figure 1. Relative proportion of 3R4F TPM-treated BEAS-2B cells in G0+G1 (blue), S (orange), and G2/M (grey) phases of the cell cycle. Results were obtained by HCS using Hoechst and EdU staining. Data are presented as mean of 3 replicate experiments; error bars indicate standard deviation (SD). * indicates significant increase; # indicates significant decraese. */#P<0.05 *vs* DMSO, **/##P<0.01 *vs* DMSO, ***/###P<0.001 *vs* DMSO and ****/####P<0.0001 vs DMSO by Dunnett's multiple comparison test.

Cell numbers drop significantly after 4 weeks of treatment

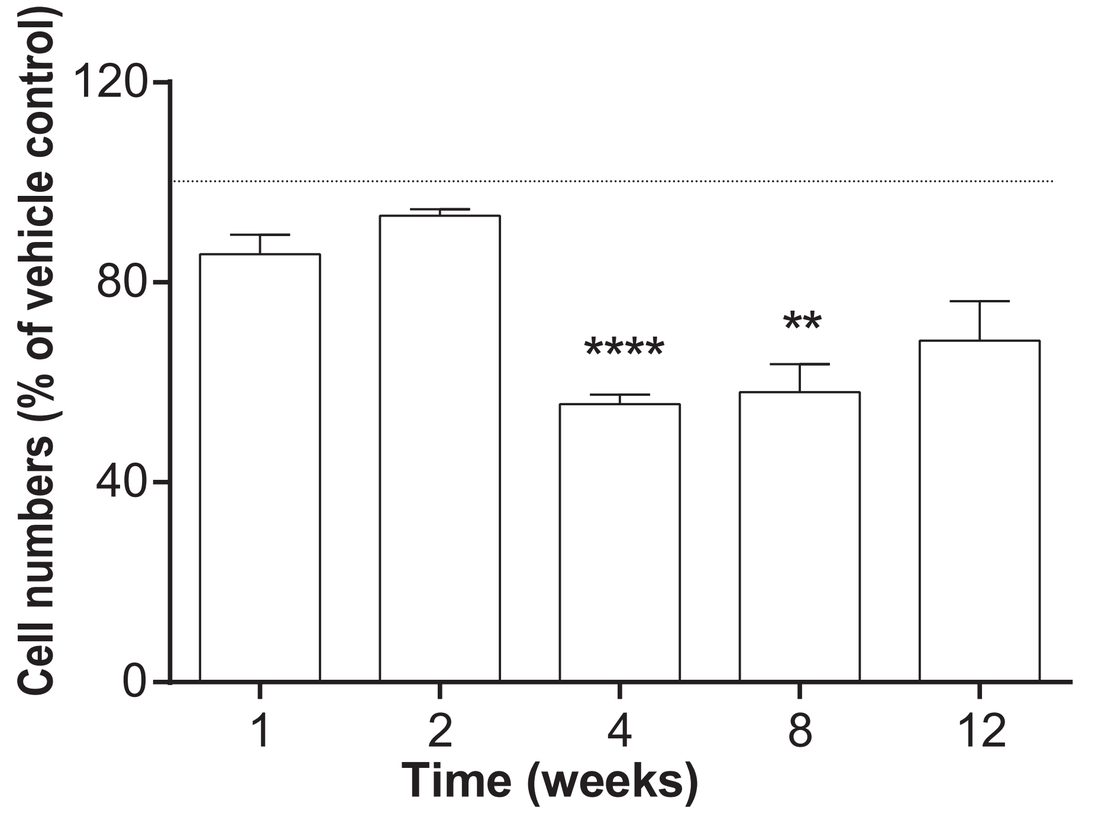
Cell numbers (% of vehicle control)
Time (weeks)

Figure 2. Total cell numbers of TPM-treated BEAS-2B cells over time, expressed as % of control (DMSO). Data are presented as mean of 3 replicates ±SD. * indicates a significant difference relative to vehicle control (Dunnett's test). ** P<0.01, **** P<0.0001

dsDNA damage is only apparent during the first 2 weeks

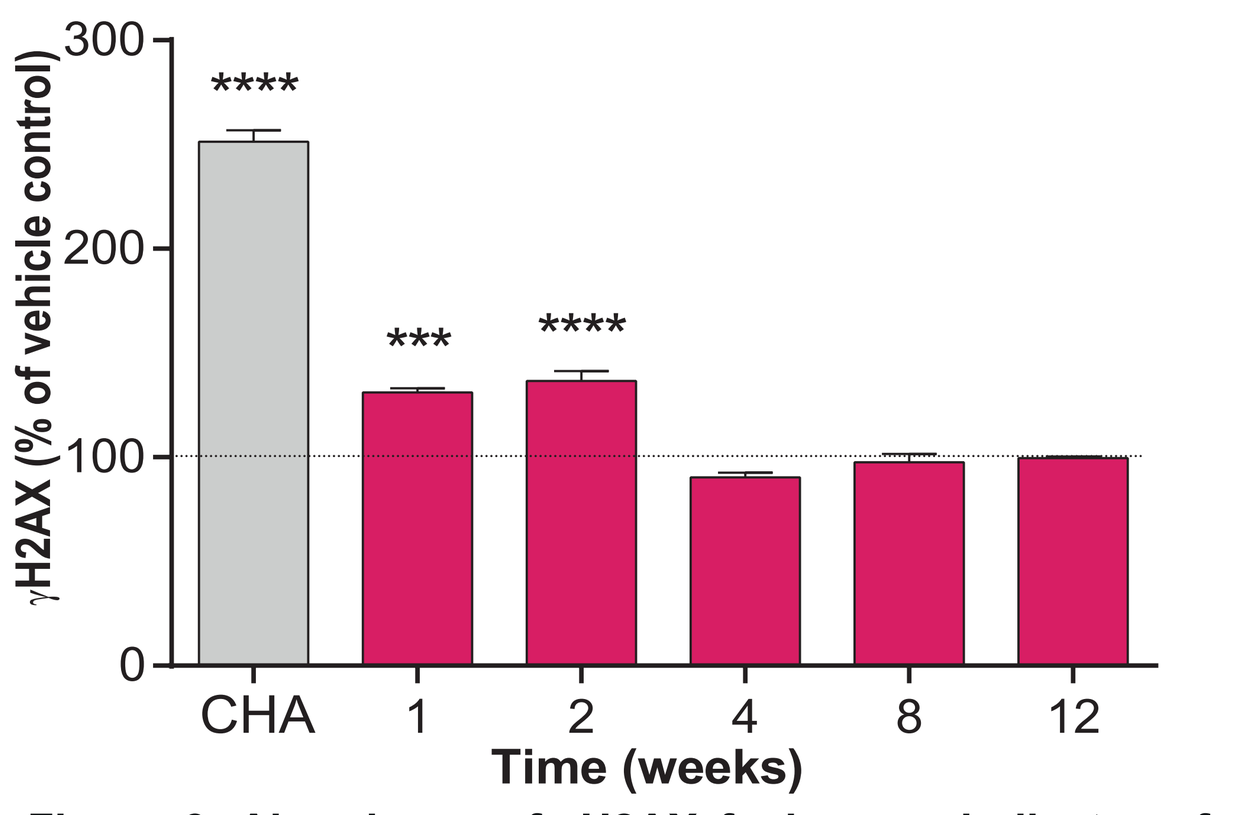
γH2AX (% of vehicle control)
CHA
Time (weeks)

Figure 3. Abundance of γH2AX foci as an indicator of DNA damage in TPM-treated BEAS-2B cells over time, expressed as % of control (DMSO). Data are presented as mean of 3 replicates ±SD. CHA: Chlorambucil (positive control). * indicates a significant difference relative to vehicle control (Dunnett's test). *** P<0.001, **** P<0.0001

Initially, ROS generation is compensated by increased GSH production. However, GSH appears to be depleted after 8 weeks of treatment.

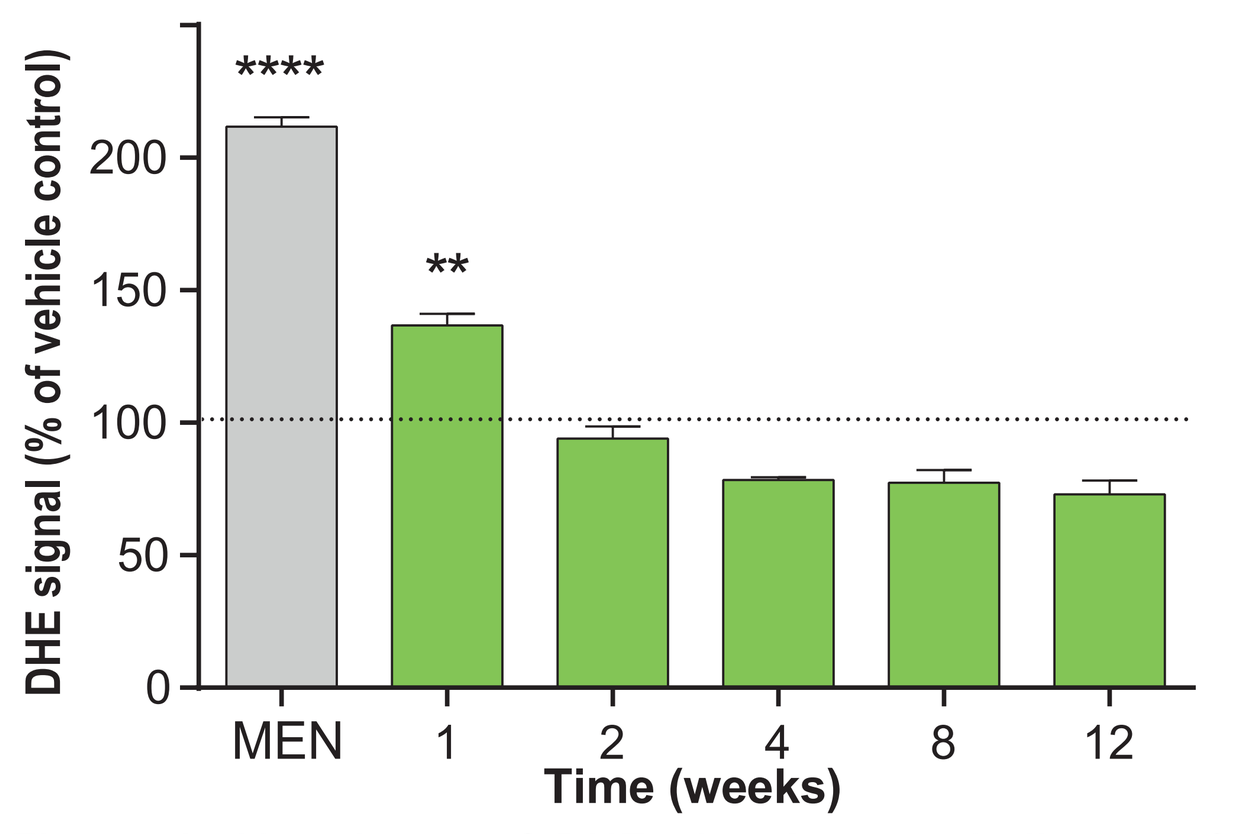
DHE signal (% of vehicle control)
MEN
Time (weeks)

Figure 4. Abundance of DHE signal as an indicator of oxidative stress in TPM-treated BEAS-2B cells over time, expressed as % of control (DMSO). Data are presented as mean of 3 replicates ±SD. MEN: Menadione (positive control). * indicates a significant difference relative to vehicle control (Dunnett's test). ** P<0.01, **** P<0.0001

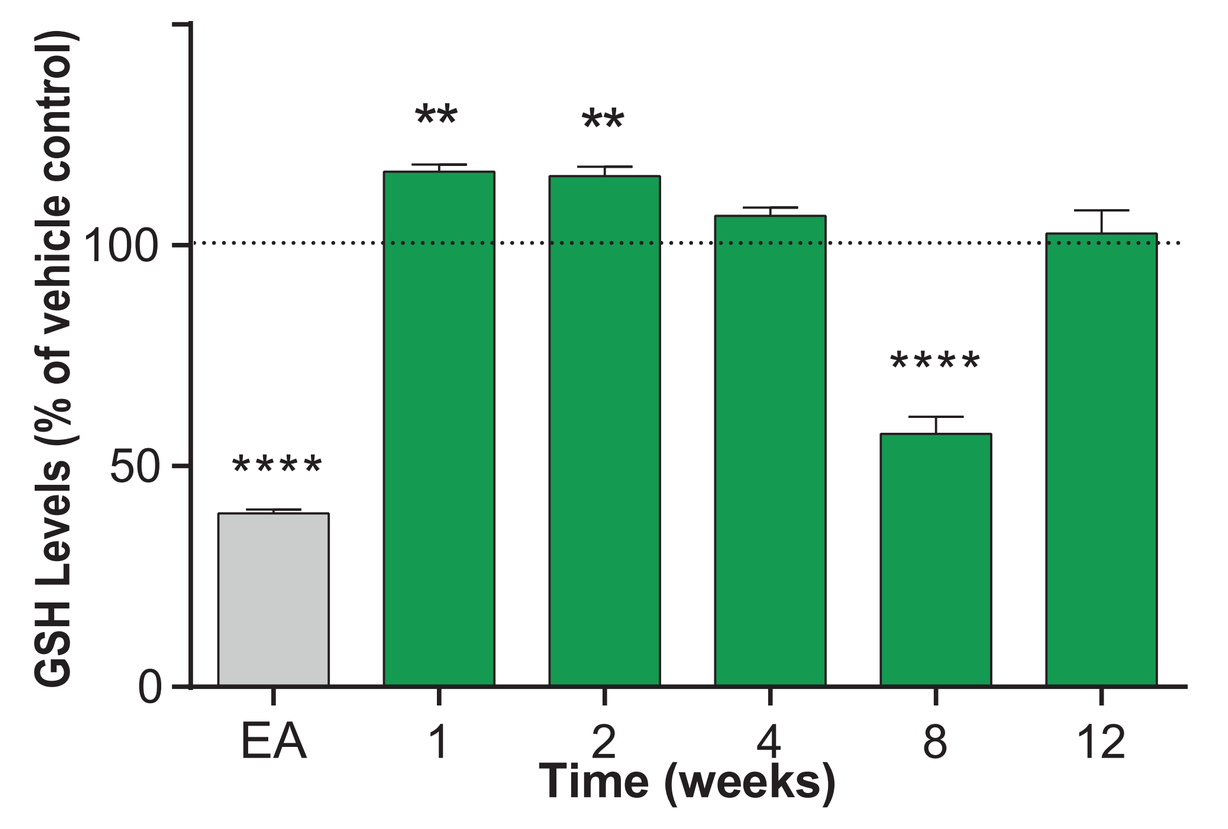
GSH Levels (% of vehicle control)
EA
Time (weeks)

Figure 5. GSH levels as an indicator of oxidative stress in TPM-treated BEAS-2B cells over time, expressed as % of control (DMSO). Data are presented as mean of 3 replicates (RingSpot average intensity) ±SD. EA: Ethancrynic acid (positive control). * indicates a significant difference relative to vehicle control (Dunnett's test). ** P<0.01, **** P<0.0001

Long-term treatment of BEAS-2B cells with 3R4F TPM leads to disruption of adherens junctions, temporary decrease in cell adhesion, and a propensity to form colonies in soft agar

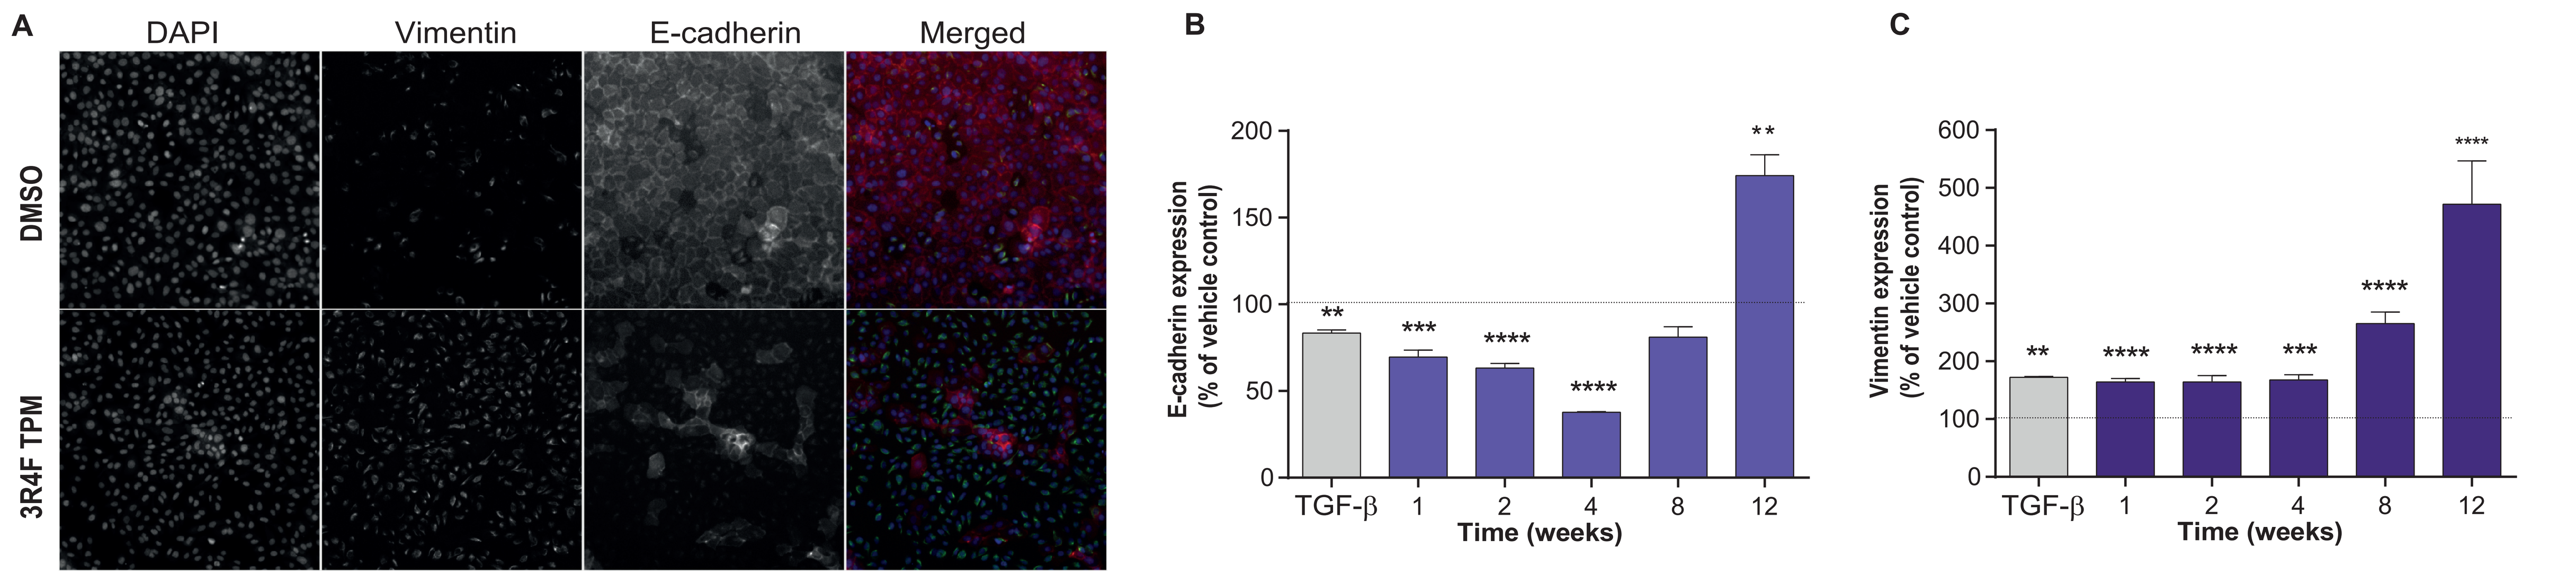
A
DAPI
Vimentin
E-cadherin
Merged
DMSO
3R4F TPM
B
E-cadherin expression (% of vehicle control)
TGF-β
Time (weeks)
C
Vimentin expression (% of vehicle control)
TGF-β
Time (weeks)

Figure 6. Effect of long-term treatment with 3R4F TPM on E-cadherin and vimentin expression in BEAS-2B cells. A) Representative HCS images of DMSO control and 3R4F TPM-treated cells at week 4 stained for E-cadherin and vimentin are shown. Nuclei are counterstained with Hoechst dye. Magnification 40X. **B)** E-cadherin expression, **C)** vimentin expression, **in** TPM-treated BEAS-2B cells over time, expressed as % of vehicle control (DMSO). Data are presented as mean of 3 replicates ±SD. TGFβ: Transforming growth factor beta served as assay positive control. * indicates a significant difference relative to vehicle control (Dunnett's test). * P<0.05, ** P<0.01, *** P<0.001, and **** P<0.0001.

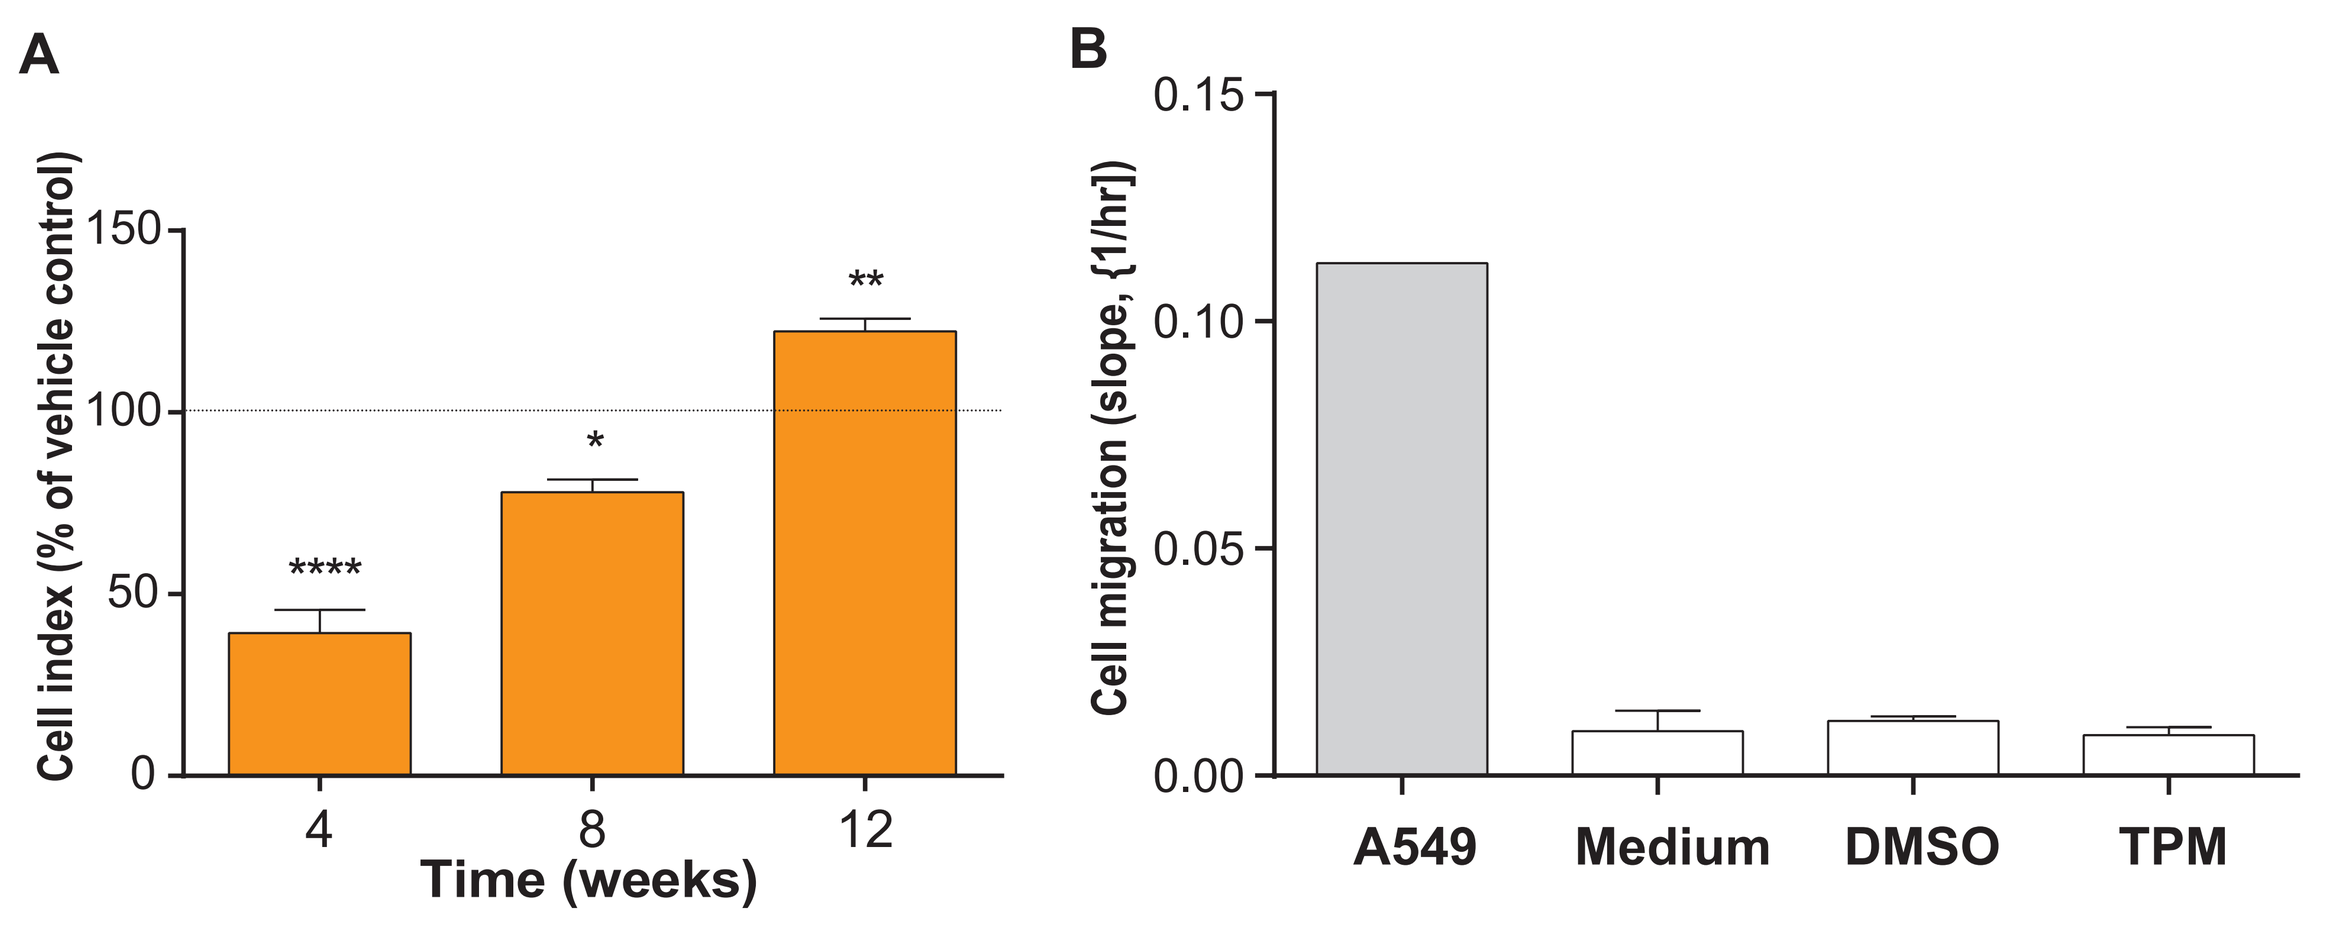
A
Cell index (% of vehicle control)
Time (weeks)
B
Cell migration (slope, {1/hr])
A549
Medium
DMSO
TPM

Figure 6. Effect of long-term treatment with 3R4F TPM on BEAS-2B cell adhesion and migration. A) Cell index of 3R4F TPM-treated cells at weeks 4, 8, and 12, relative to vehicle control (DMSO) determined by real-time impedance measurements. **B)** Cell migration rates at week 4, expressed as [1/hr], were derived from the real-time impedance measurements. A549 lung cancer cells were used as positive control. Data are presented as mean of 3 replicates ±SD. * indicates a significant difference relative to vehicle control (Dunnett's test). * P<0.05, ** P<0.01, and **** P<0.0001.

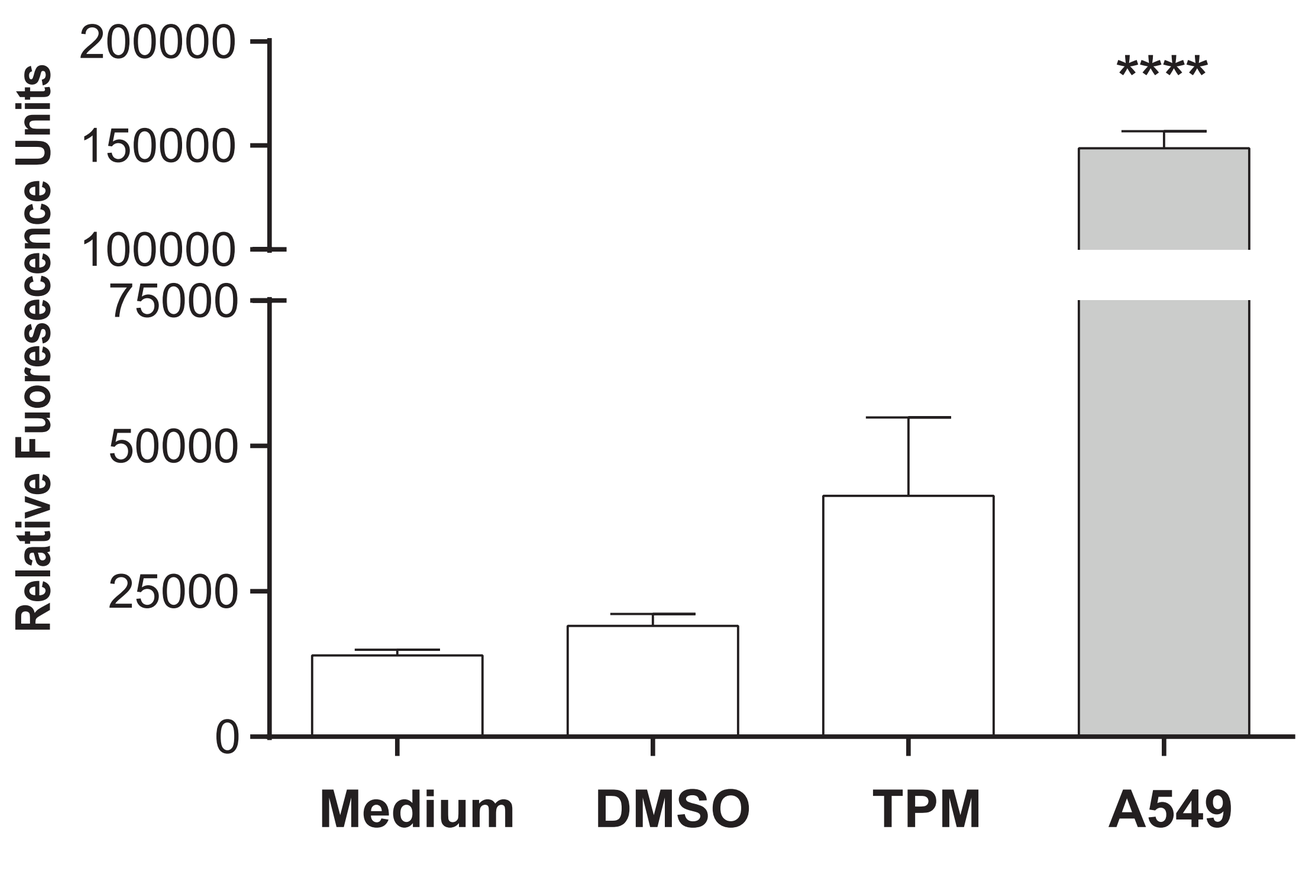
Relative Fuoresecence Units
Medium
DMSO
TPM
A549

Figure 7. Colony formation in soft agar. Relative fluorescence served as a read-out for the number of surviving cells plated in soft agar. A549 lung cancer cells were used as positive control. Data are presented as mean of 3 replicates ±SD. * indicates a significant difference relative to vehicle control (Dunnett's test). **** P<0.0001

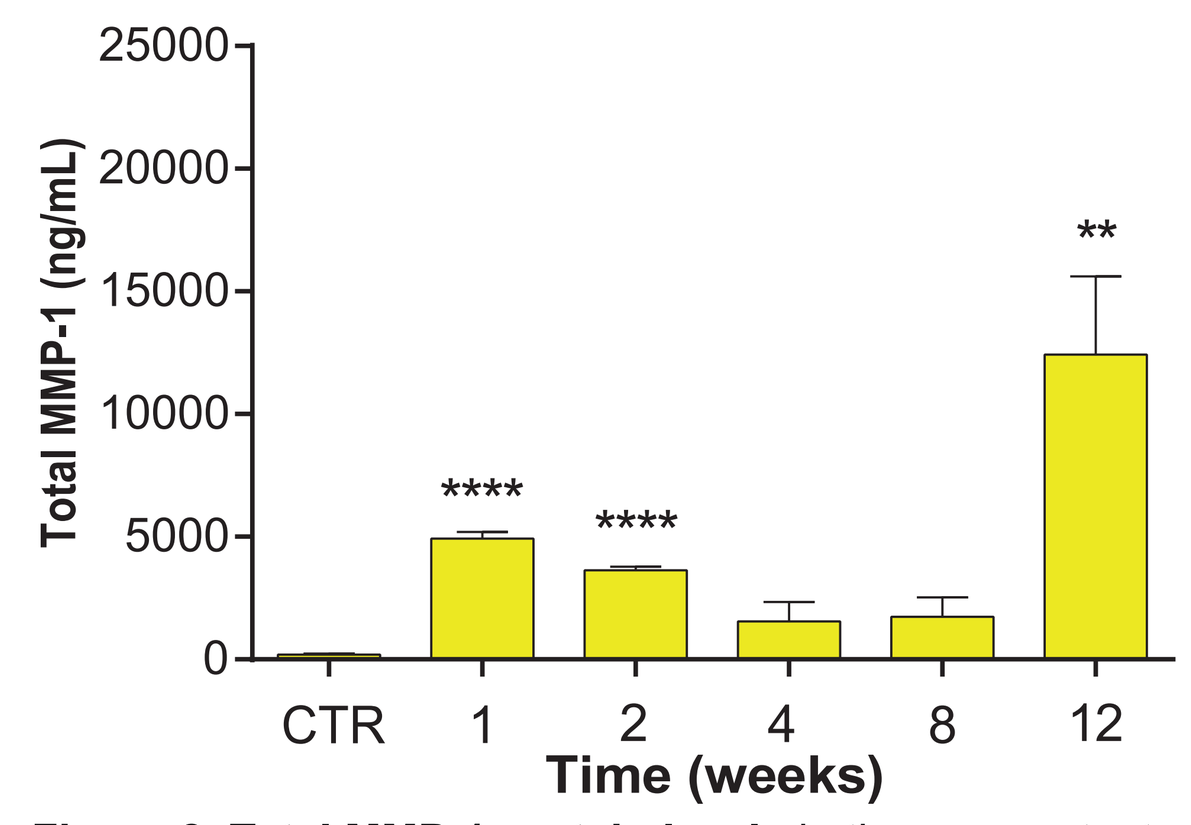
Total MMP-1 (ng/mL)
CTR
Time (weeks)

Figure 8. Total MMP-1 protein levels in the supernatants of BEAS-2B cells subjected to long-term treatment with 3R4F TPM. CTR: DMSO at week 1. Data are presented as mean of 3 replicates ±SD. * indicates a significant difference relative to vehicle control (Dunnett's test). ** P<0.01, and **** P<0.0001

Summary & Conclusions

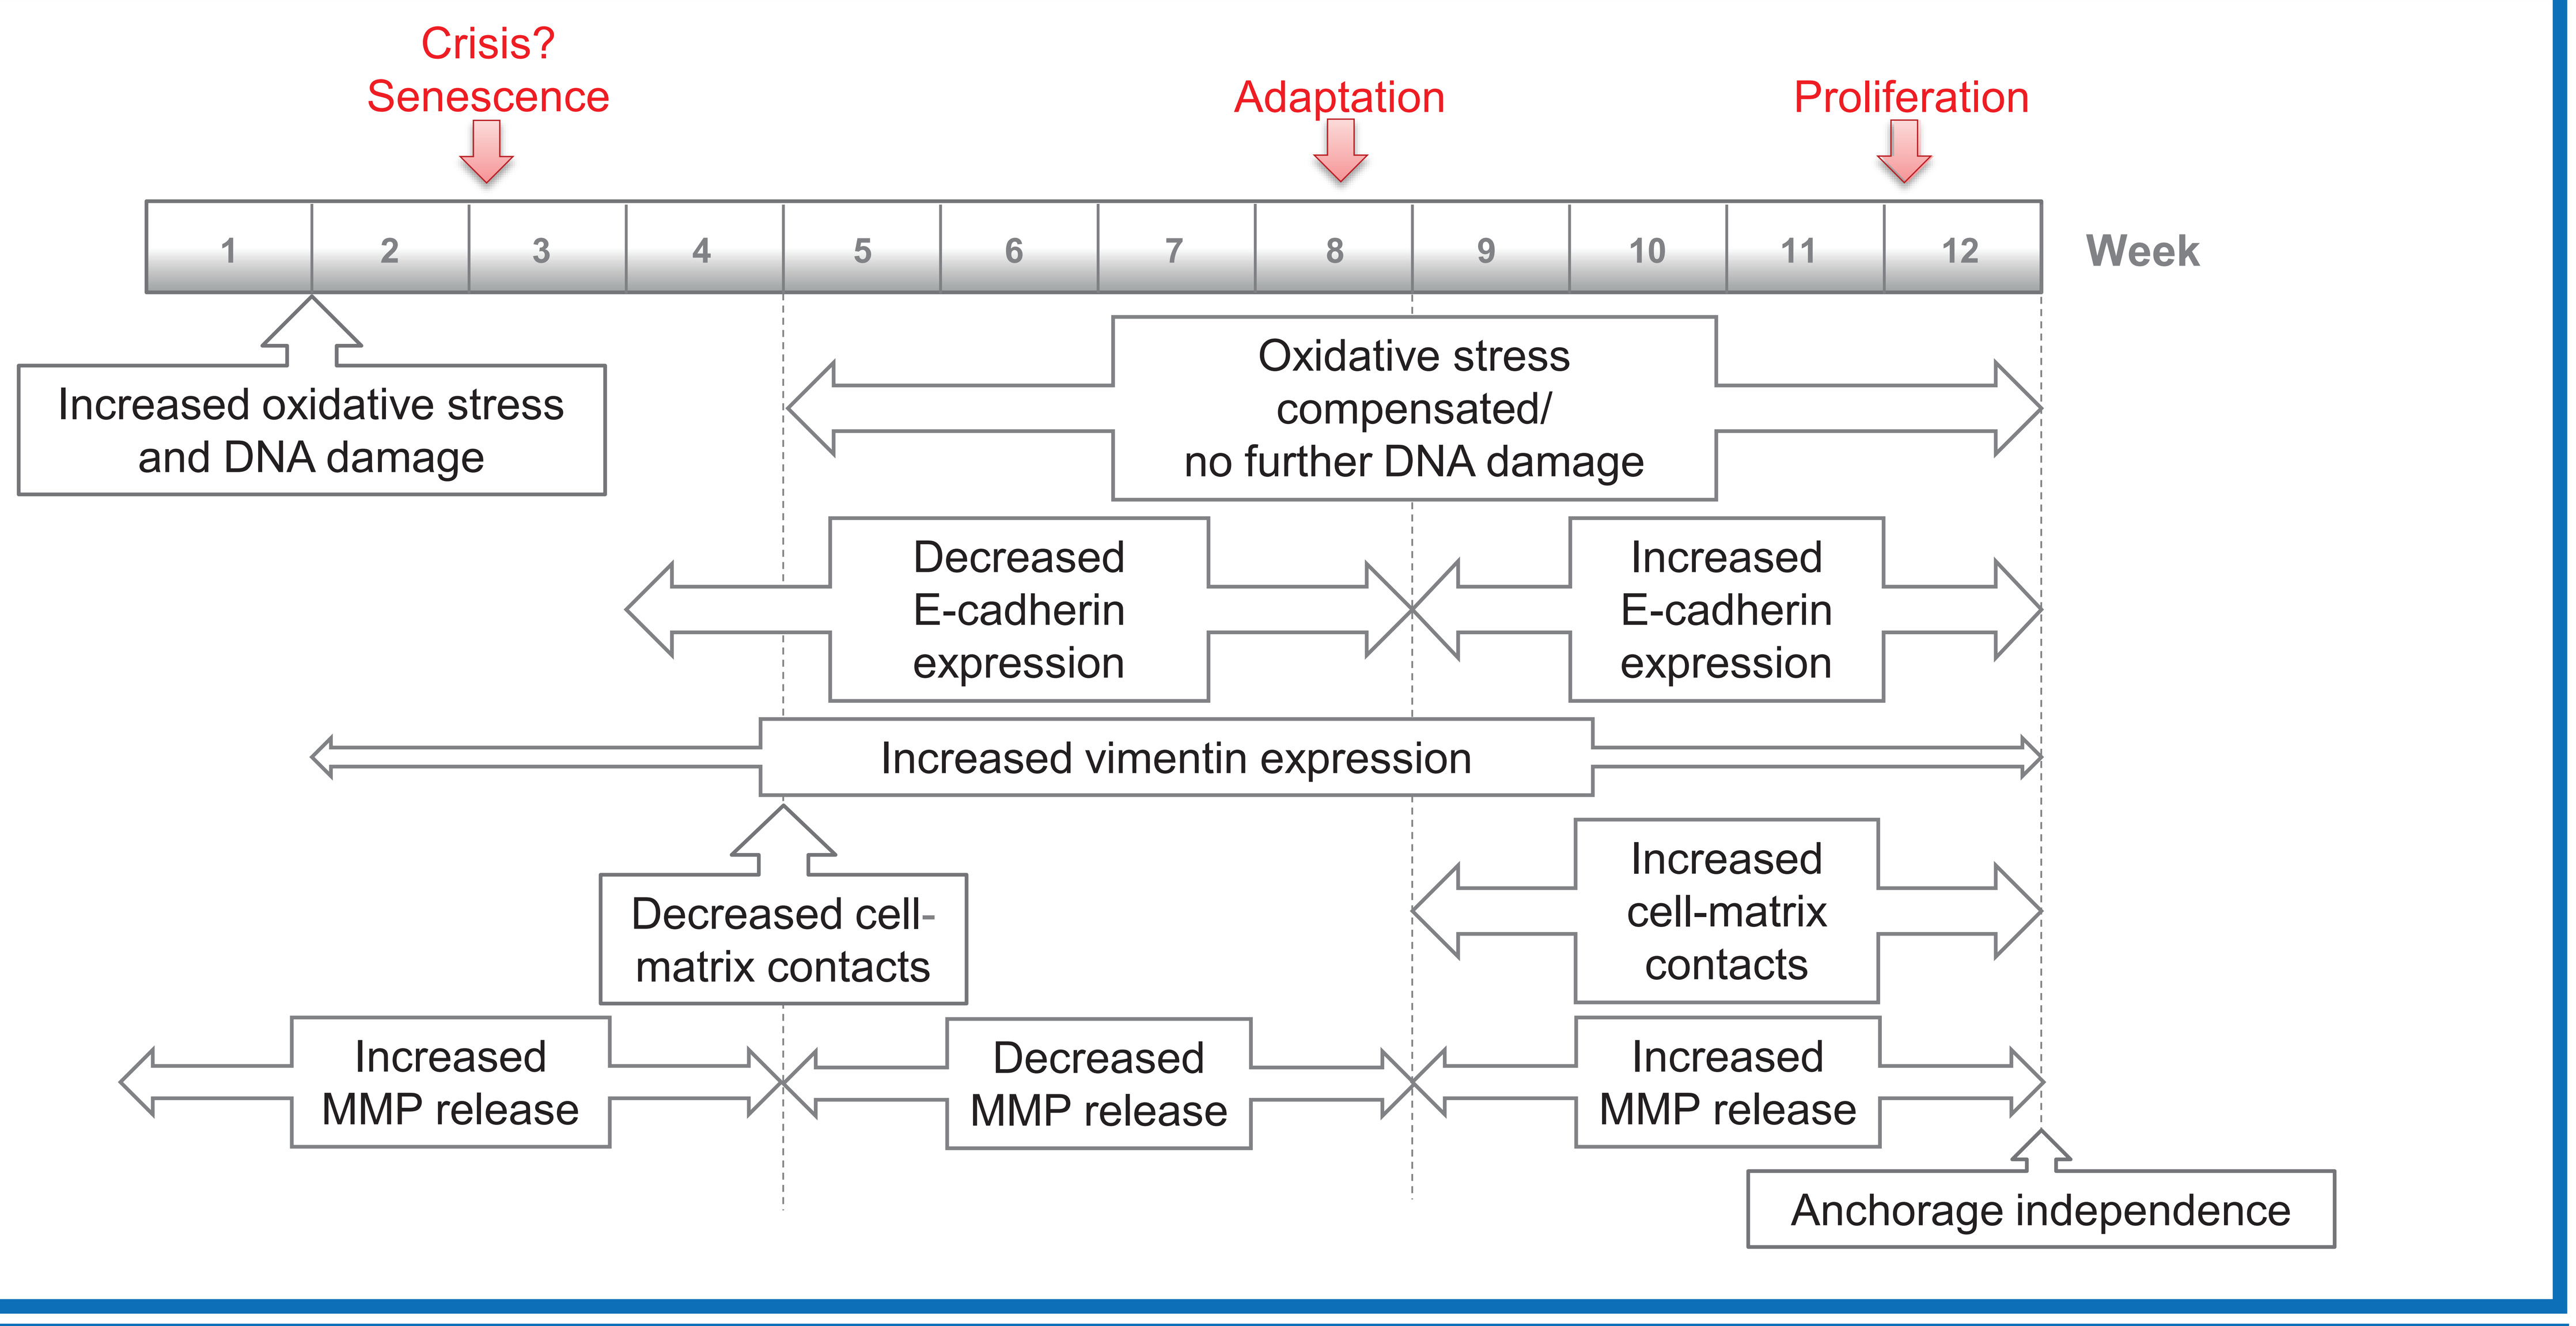
Crisis?
Senescence
Adaptation
Proliferation
1
2
3
4
5
6
7
8
9
10
11
12
Week
Increased oxidative stress and DNA damage
Oxidative stress compensated/ no further DNA damage
Decreased E-cadherin expression
Increased E-cadherin expression
Increased vimentin expression
Decreased cell-matrix contacts
Increased cell-matrix contacts
Increased MMP release
Decreased MMP release
Increased MMP release
Anchorage independence

Conclusions & Perspective

Long-term treatment of BEAS-2B cells with TPM from 3R4F reference cigarettes leads to a sequence of phenotypic changes that suggests that cells successively undergo oxidative stress-induced damage responses including cell death and senescence, transient epithelial-mesenchymal transition leading to loss of cell adhesion (but not migration), and loss of anchorage dependence. Whether the resulting cell population has acquired tumorigenic capacity requires further investigation which is planned as a next step. In addition, cells will also be subjected to comprehensive analysis by transcriptomics using a portfolio of causal networks to gain further insight into the mechanisms underlying the phenotypic changes observed here. Further, mutational profiling of genes known to be involved in lung tumorigenesis may provide additional information about potential drivers of the observed adaptation and/or selection.

References

1. ISO 3402: Tobacco and tobacco products - Atmosphere for conditioning and testing. International Organization for Standardization, Geneva, Switzerland, 1999

2. Health Canada Intense (HCI) Smoking Regimen, Official Method T-115, Determination of "Tar", Nicotine and Carbon Monoxide in Mainstream Tobacco Smoke, December 31, 1999

International Meeting of the German Society for Cell Biology (DGZ)
March 14 - 16, 2016
Munich, Germany

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL
www.pmiscience.com

Philip Morris International is the sole source of funding and sponsor of this project